Anaphylaxis in Patients With Indolent Systemic Mastocytosis (ISM): A Severe, Potentially Life-Threatening Complication

M079

Thanai Pongdee,[1] Mariana Castells,[2] Karin Hartmann,[3,4] Cecilia Arana Yi,[5] Cem Akin,[6] Ivan Alvarez-Twose,[7] Sonia Cerquozzi,[8] Jason Gotlib,[9] Jens Panse,[10,11] Peter Vadas,[12] Friederike Wortmann,[13] Ilda Bidollari,[14] Kate Newberry,[14] Ingunn Dybedal,[15] Pankit Vachhani[16]

[1]Division of Allergic Diseases, Mayo Clinic, Rochester, MN, USA; [2]Department of Medicine, Division of Allergy and Clinical Immunology, Brigham and Women's Hospital, Harvard Medical School, Boston, MA, USA; [3]Division of Allergy, Department of Dermatology, University Hospital Basel and University of Basel, Basel, Switzerland; [4]Department of Biomedicine, University Hospital Basel and University of Basel, Basel, Switzerland; [5]Division of Hematology and Medical Oncology, Mayo Clinic, Phoenix, AZ, USA; [6]Division of Allergy and Clinical Immunology, University of Michigan, Ann Arbor, MI, USA; [7]Institute of Mastocytosis Studies of Castilla-La Mancha, Spanish Reference Center of Mastocytosis, Toledo, Spain; [8]Department of Medicine, University of Calgary, Calgary, AB, Canada; [9]Stanford Cancer Institute, Stanford University School of Medicine, Stanford, CA, USA; [10]Department of Oncology, Hematology, Hemostaseology, and Stem Cell Transplantation, University Hospital Aachen, Medical Faculty, Rheinisch-Westfälische Technische Hochschule Aachen University, Aachen, Germany; [11]Center for Integrated Oncology, Aachen, Bonn, Cologne, Düsseldorf (ABCD), Aachen, Germany; [12]Department of Medicine, Division of Clinical Immunology and Allergy, St. Michael's Hospital, University of Toronto, Toronto, ON, Canada; [13]Oberärztin Hämatologie/Onkologie bei Uksh Campus Lübeck, Universitätsklinikum Schleswig-Holstein, Universität zu Lübeck, Lübeck, Schleswig-Holstein, Germany; [14]Blueprint Medicines Corporation, Cambridge, MA, USA; [15]Department of Hematology, Oslo University Hospital, Oslo, Norway; [16]Division of Hematology/Oncology, Department of Medicine, University of Alabama at Birmingham, Birmingham, AL, USA

Background

- ISM is a clonal mast cell (MC) disease, primarily driven by the *KIT* D816V mutation in approximately 95% of cases[1-4]
- Patients often have lifelong debilitating symptoms across multiple organ systems, including uncontrolled, life-threatening anaphylaxis, and reduced quality of life[4-7]
- Frequency of anaphylaxis (20% to 49%) varies due to disease heterogeneity and diverse anaphylaxis classification systems; incidence is higher in patients with systemic mastocytosis (SM) versus the general population[8-10]
- ISM management strategies include trigger avoidance, self-injectable epinephrine, and symptom management with polypharmacy. However, symptoms may not be fully controlled by these measures in some patients, highlighting the need for more effective ISM-targeted therapies[8,11]
- Avapritinib is a potent and highly selective inhibitor of KIT D816V[11]
- Avapritinib is approved in the United States for adult patients with ISM and advanced SM and in Europe for adult patients with advanced SM after ≥1 prior systemic therapy; avapritinib is not recommended for patients with platelet counts $<50\times10^9$/L[12]
- The PIONEER trial (NCT03731260), a global, randomized, placebo-controlled study, assessed efficacy and safety of the oral, highly selective, KIT D816V tyrosine kinase inhibitor avapritinib versus placebo, both with best supportive care, in patients with moderate-to-severe ISM (classified based on the WHO 2016 criteria) over a period of 24 weeks (Part 2 of the study); open-label extension of the trial is in progress[13]
- Here, we provide a descriptive report of anaphylaxis in the PIONEER study, detailing key baseline characteristics and the symptomatic journey of 13 patients, randomized to avapritinib 25 mg or placebo in Part 2 of the PIONEER study, and who had anaphylaxis either during the 12-week screening period (baseline) and/or during 24 weeks of avapritinib treatment (Part 2 of the study)
- We note that the trial was not powered to capture statistically significant changes in anaphylaxis between the avapritinib and placebo arms

Methods

PIONEER Study Design

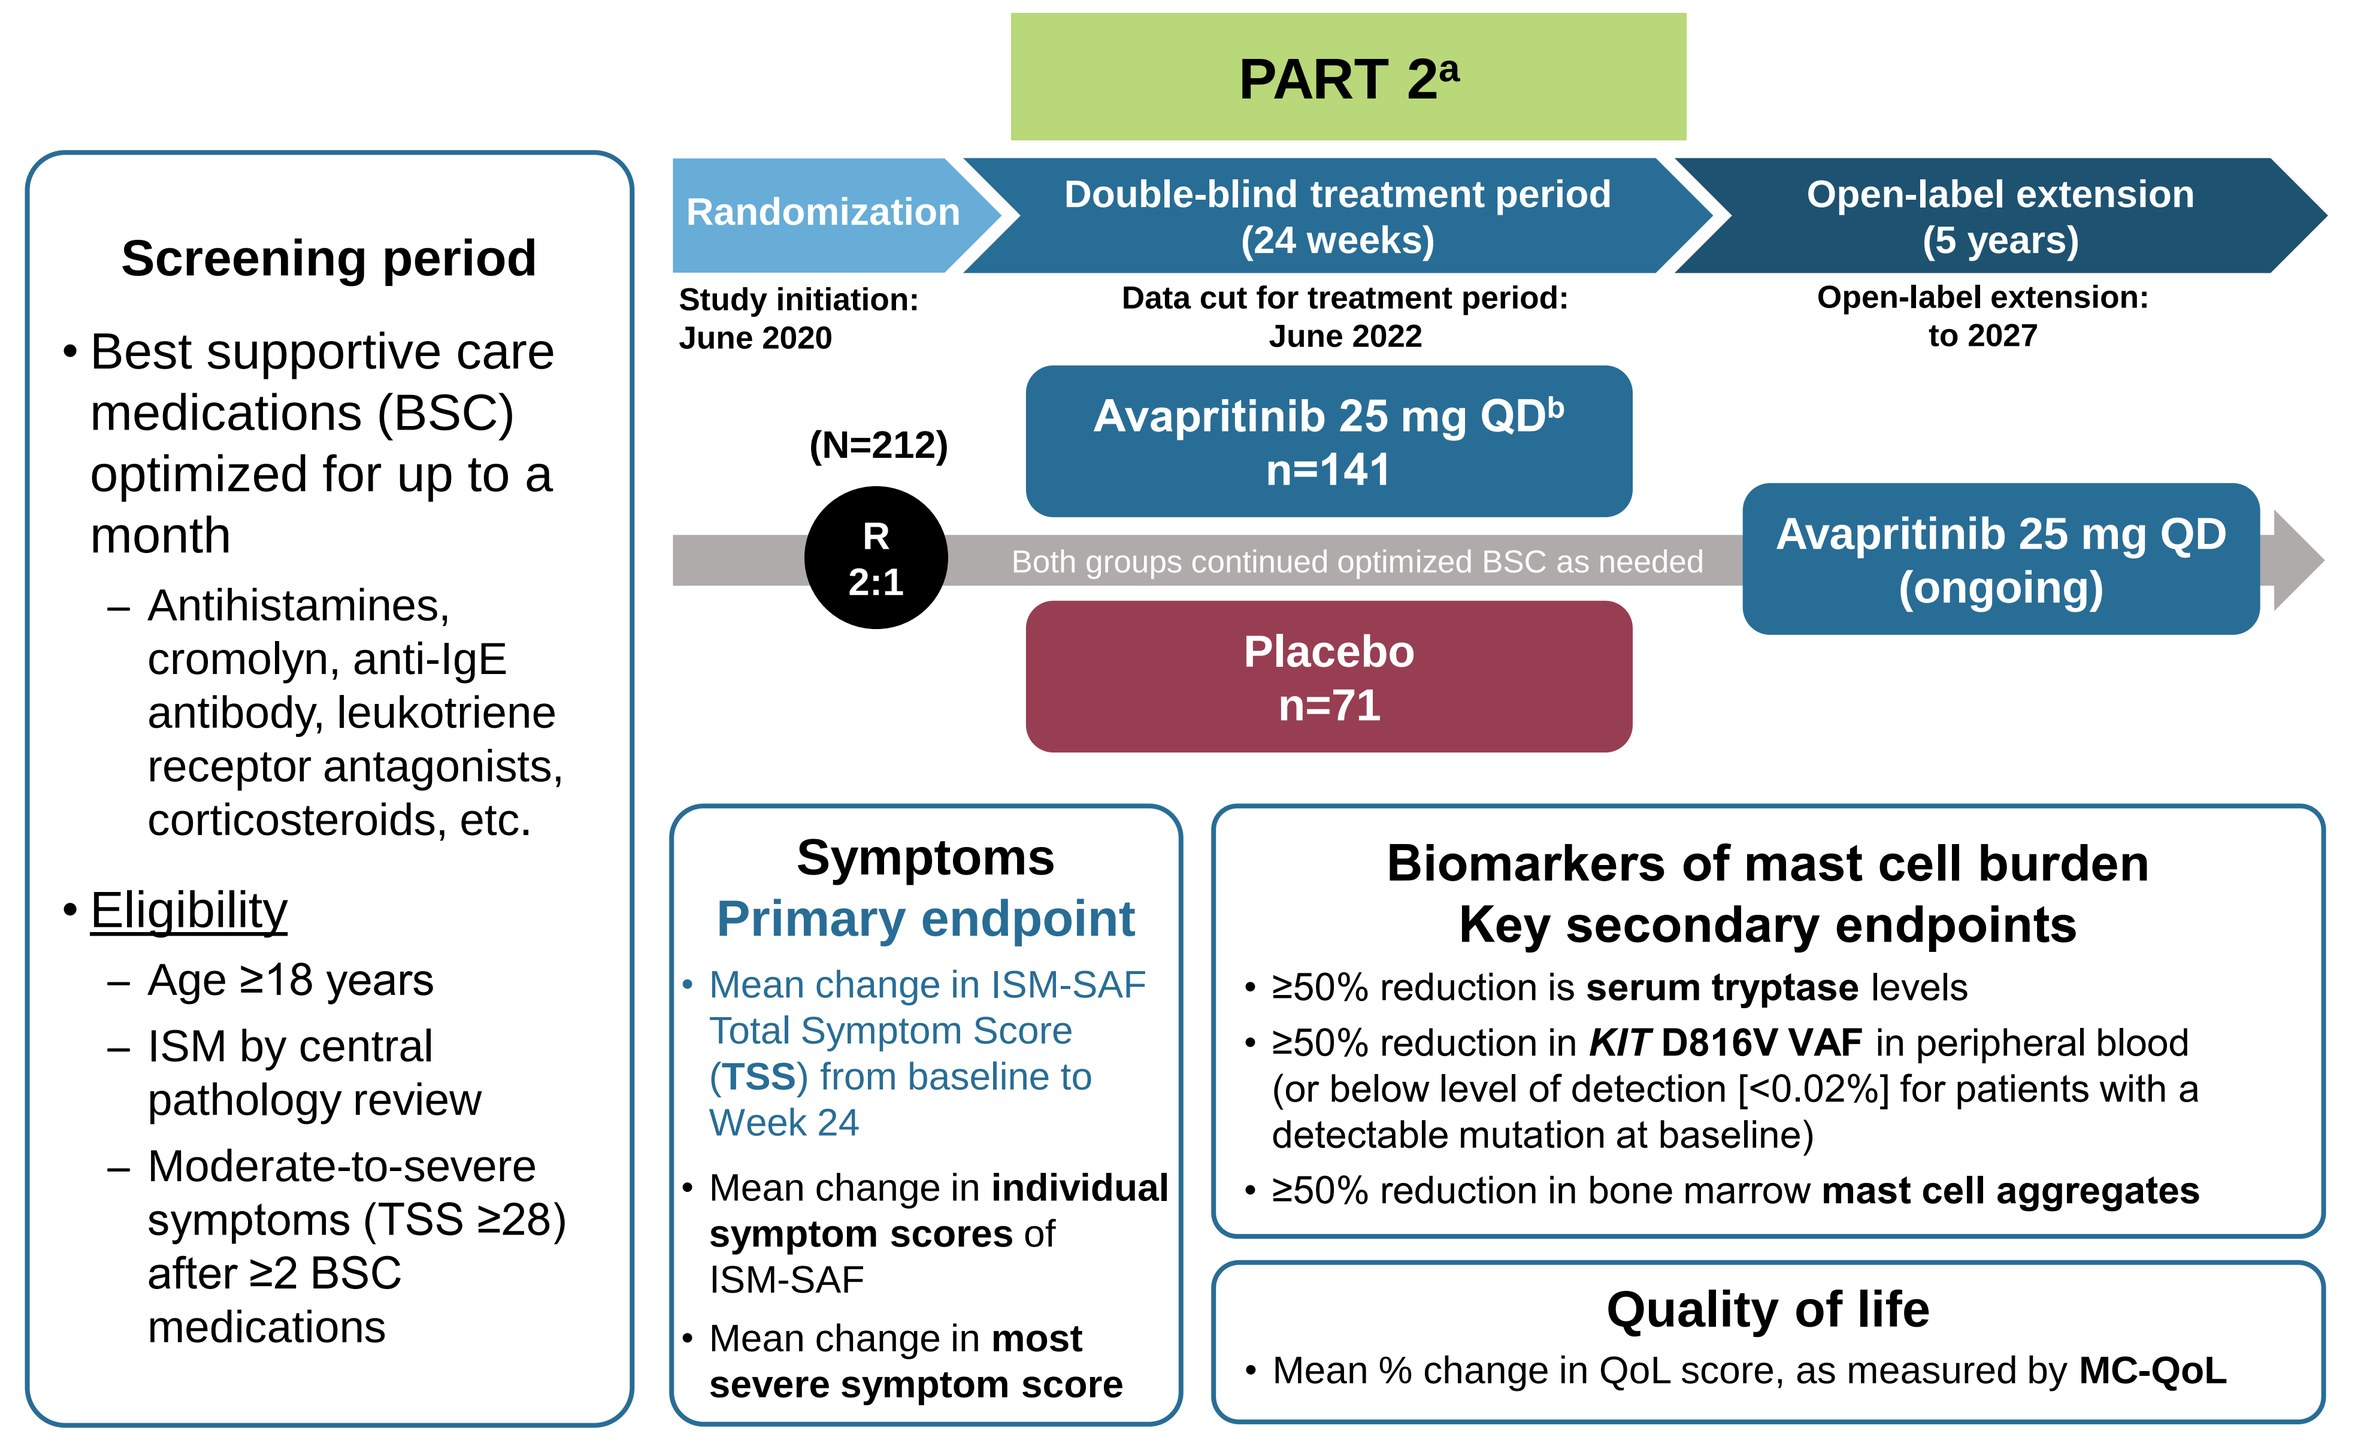
PART 2[a]
Screening period
• Best supportive care medications (BSC) optimized for up to a month
– Antihistamines, cromolyn, anti-IgE antibody, leukotriene receptor antagonists, corticosteroids, etc.
• Eligibility
– Age ≥18 years
– ISM by central pathology review
– Moderate-to-severe symptoms (TSS ≥28) after ≥2 BSC medications
Randomization
Double-blind treatment period (24 weeks)
Open-label extension (5 years)
Study initiation: June 2020
Data cut for treatment period: June 2022
Open-label extension: to 2027
(N=212)
R 2:1
Avapritinib 25 mg QD[b] n=141
Both groups continued optimized BSC as needed
Placebo n=71
Avapritinib 25 mg QD (ongoing)
Symptoms
Primary endpoint
• Mean change in ISM-SAF Total Symptom Score (TSS) from baseline to Week 24
• Mean change in individual symptom scores of ISM-SAF
• Mean change in most severe symptom score
Biomarkers of mast cell burden
Key secondary endpoints
• ≥50% reduction is serum tryptase levels
• ≥50% reduction in KIT D816V VAF in peripheral blood (or below level of detection [<0.02%] for patients with a detectable mutation at baseline)
• ≥50% reduction in bone marrow mast cell aggregates
Quality of life
• Mean % change in QoL score, as measured by MC-QoL

IgE, immunoglobulin E; ISM-SAF, Indolent Systemic Mastocytosis-Symptom Assessment Form; MC-QoL, Mastocytosis Quality of Life Questionnaire; QD, once daily; QoL, quality of life; R, randomization; VAF, variant allele fraction.
[a]Anaphylactic events leading to treatment with epinephrine were considered for this subgroup analysis. [b]The recommended dose of avapritinib for the double-blind period and open-label extension was identified based on efficacy and safety results from Part 1 that included 4 cohorts: 25 mg avapritinib (n=10), 50 mg avapritinib (n=10), 100 mg avapritinib (n=10), and placebo (n=9). Patients treated with high-dose steroids within 7 days of primary endpoint (n=4) were excluded from the Week 24 analysis, but included at other timepoints of the study. Percentages were calculated based on available data at the timepoint. One-sided *P* values are reported for primary and key secondary endpoints.

- Information on anaphylactic events requiring the use of epinephrine was collected during the study, including precipitating factors, associated symptoms, dose and frequency of epinephrine, steroids, and other medications administered for the event
- Baseline characteristics, known anaphylactic triggers, frequency of events on study, and safety are summarized

Results

Demographics, Clinical Characteristics, and Treatment History

- Baseline demographics and clinical characteristics varied between the patients (**Table 1**)
 - Numbers of known allergens/triggers for anaphylactic events were widely variable, with 13 being the highest
 - Patients received multiple prior treatments for ISM including omalizumab, tyrosine kinase inhibitors (TKIs), and cytoreductive therapy

Table 1. Demographics, Clinical Characteristics, and Treatment History

	Patients with anaphylactic events[a]	Age, years	Sex	Time from diagnosis to randomization, years	Known allergens/triggers	B findings	Omalizumab	Prior cytoreductive therapy/ TKI	# of best supportive care (BSC) medications
Avapritinib n=10/141	1	39	F	11	Not available	No	Yes	Hydroxyurea	6
	2	35	F	12	Vaccine	Hypercelluar BM[b]	No	No	4
	3	61	F	4	Unknown	No	No	No	4
	4	50	F	6	Bee venom	No	No	No	5
	5	48	M	10	Possible stress, penicillin, contrast dye	No	No	No	5
	6	54	F	4	Fish, pork	No	No	No	2
	7	39	F	2	Adhesives, Augmentin, contrast dye, insect bite, strawberries, tramadol hydrochloride, wine, acetaminophen, aspirin, nizatidine, pineapple cake, mint, dust mites	No	Yes	Interferon alpha, midostaurin	9
	8	43	F	11	Mosquito bite, honey, tomato-based products, yeast-fermented sourdough bread, spicy food, morphine, ethylmorphine, tramadol, penicillin	No	No	No	4
	9	39	F	10	Bee and wasp stings, contrast agents, physical stress, friction, cold/heat, alcohol	No	No	Interferon alpha, dasatinib, cladribine (2CdA), midostaurin	6
	10	56	M	1	Unknown	No	No	No	3
Placebo n=3/71	11	34	F	1	Scented candle	No	Yes	No	6
	12	66	F	15	Not available	MC in BM, tryptase >200 ng/mL[c]	Yes	No	7
	13	30	F	3	Not available	No	Yes	No	6

BM, bone marrow; MC, mast cell; MDS, myelodysplastic syndrome; MPN, myeloproliferative neoplasm; TKI, tyrosine kinase inhibitor; WHO, World Health Organization.
[a]Refers to overall number of patients who experienced anaphylactic events either during screening or during treatment with avapritinib. [b]Patient had hypercellular BM with loss of fat cells, discrete signs of dysmyelopoiesis without substantial cytopenias or WHO criteria for an MDS or MPN. [c]B findings represent MC infiltration in BM >30% by histology and baseline serum tryptase >200 ng/mL.

Baseline Diagnostics and Symptomatology

- Serum tryptase levels ranged from 3.6 ng/mL to 235.2 ng/mL; bone marrow mast cell burden varied from 1% to 25% (**Table 2**)
- 11 of the 13 patients had a detectable *KIT* D816V mutation with an allele burden that ranged from 0.07% to 6.75%
 - *KIT* D816V mutations were not detected in 2 of the 13 patients
- Baseline TSS (Total Symptom Score) ranged between 31.0 to 85.6

Table 2. Baseline Diagnostics and Symptomatology

	Patients with anaphylactic events	Serum tryptase (ng/mL)	Bone marrow mast cells, %	*KIT* D816V mutation allele burden as measured by VAF using ddPCR, %	Mast cells (counts/mm²) in skin, lesional/ non-lesional	TSS (range: 0-110)[a]	GI domain score[b]	Skin domain score[c]	Neurocognitive domain score[d]	Skin involvement
Avapritinib n=10/141	1	3.6	3	Mutation not detected	150/148	33.0	3.4	11.1	9.7	Yes
	2	104.0	20	4.54	587/307	34.8	5.5	12.4	11.5	Yes
	3	38.6	15	0.96	NA	57.4	16.9	23.5	18.3	No
	4	63.8	7	0.08	237/152	60.6	11.9	18.1	16.1	Yes
	5	62.2	20	1.14	967/102	31.3	9.1	10.2	6.2	Yes
	6	75.4	25	0.91	376/250	36.5	3.4	5.1	15.4	Yes
	7	10.6	1	0.07	NA	74.2	18.9	16.2	22.5	No
	8	27.5	5	0.26	385/104	82.5	15.6	21.1	26.5	Yes
	9	25.1	7	Mutation not detected	109/117	85.0	18.6	22.5	26.0	Yes
	10	4.2	3	0.77	NA	34.6	10.2	1.9	7.3	No
Placebo n=3/71	11	25.5	10	0.07	370/148	85.6	21.5	24.5	23.4	Yes
	12	235.2	15	6.75	739/311	31.0	0.9	13.1	8.7	Yes
	13	26.3	7	0.68	535/72	38.2	4.9	12.5	10.3	Yes

AE, adverse event; ddPCR, digital droplet polymerase chain reaction; GI, gastrointestinal; NA, not available; TSS, Total Symptom Score; VAF, variant allele fraction.
[a]TSS is based on severity of 11 ISM symptoms, each assessed from 0 (no symptom) to 10 (worst imaginable symptom). [b]GI domain score includes abdominal pain, diarrhea, and nausea. [c]Skin domain score includes spots, itching, and flushing. [d]Neurocognitive domain score includes brain fog, headache, and dizziness.

Anaphylaxis Events

- Overall, in the avapritinib group, 10 patients experienced anaphylactic events either during the screening period or during treatment in Part 2 of the study (**Figure 1**)
 - 8 of the 10 patients in the avapritinib group experienced anaphylaxis during the screening period, prior to treatment with avapritinib
 - 6 of these 8 patients did not experience anaphylaxis during treatment with avapritinib
 - 2 of the 10 patients in the avapritinib group did not experience anaphylaxis during screening but reported it during the treatment period
- Within the placebo group, none of the 3 patients experienced anaphylaxis during screening (**Figure 1**)
 - All 3 patients experienced anaphylaxis during the treatment period (Part 2 of the study)
- Symptoms during the anaphylactic events varied, with skin reactions; swelling of the eyes, face or throat; dizziness; fainting and unconsciousness being most common
- The majority of events (83%) required the use of at least 1 dose of epinephrine (**Table 3**)

Figure 1. Anaphylaxis Events Relative to Randomization

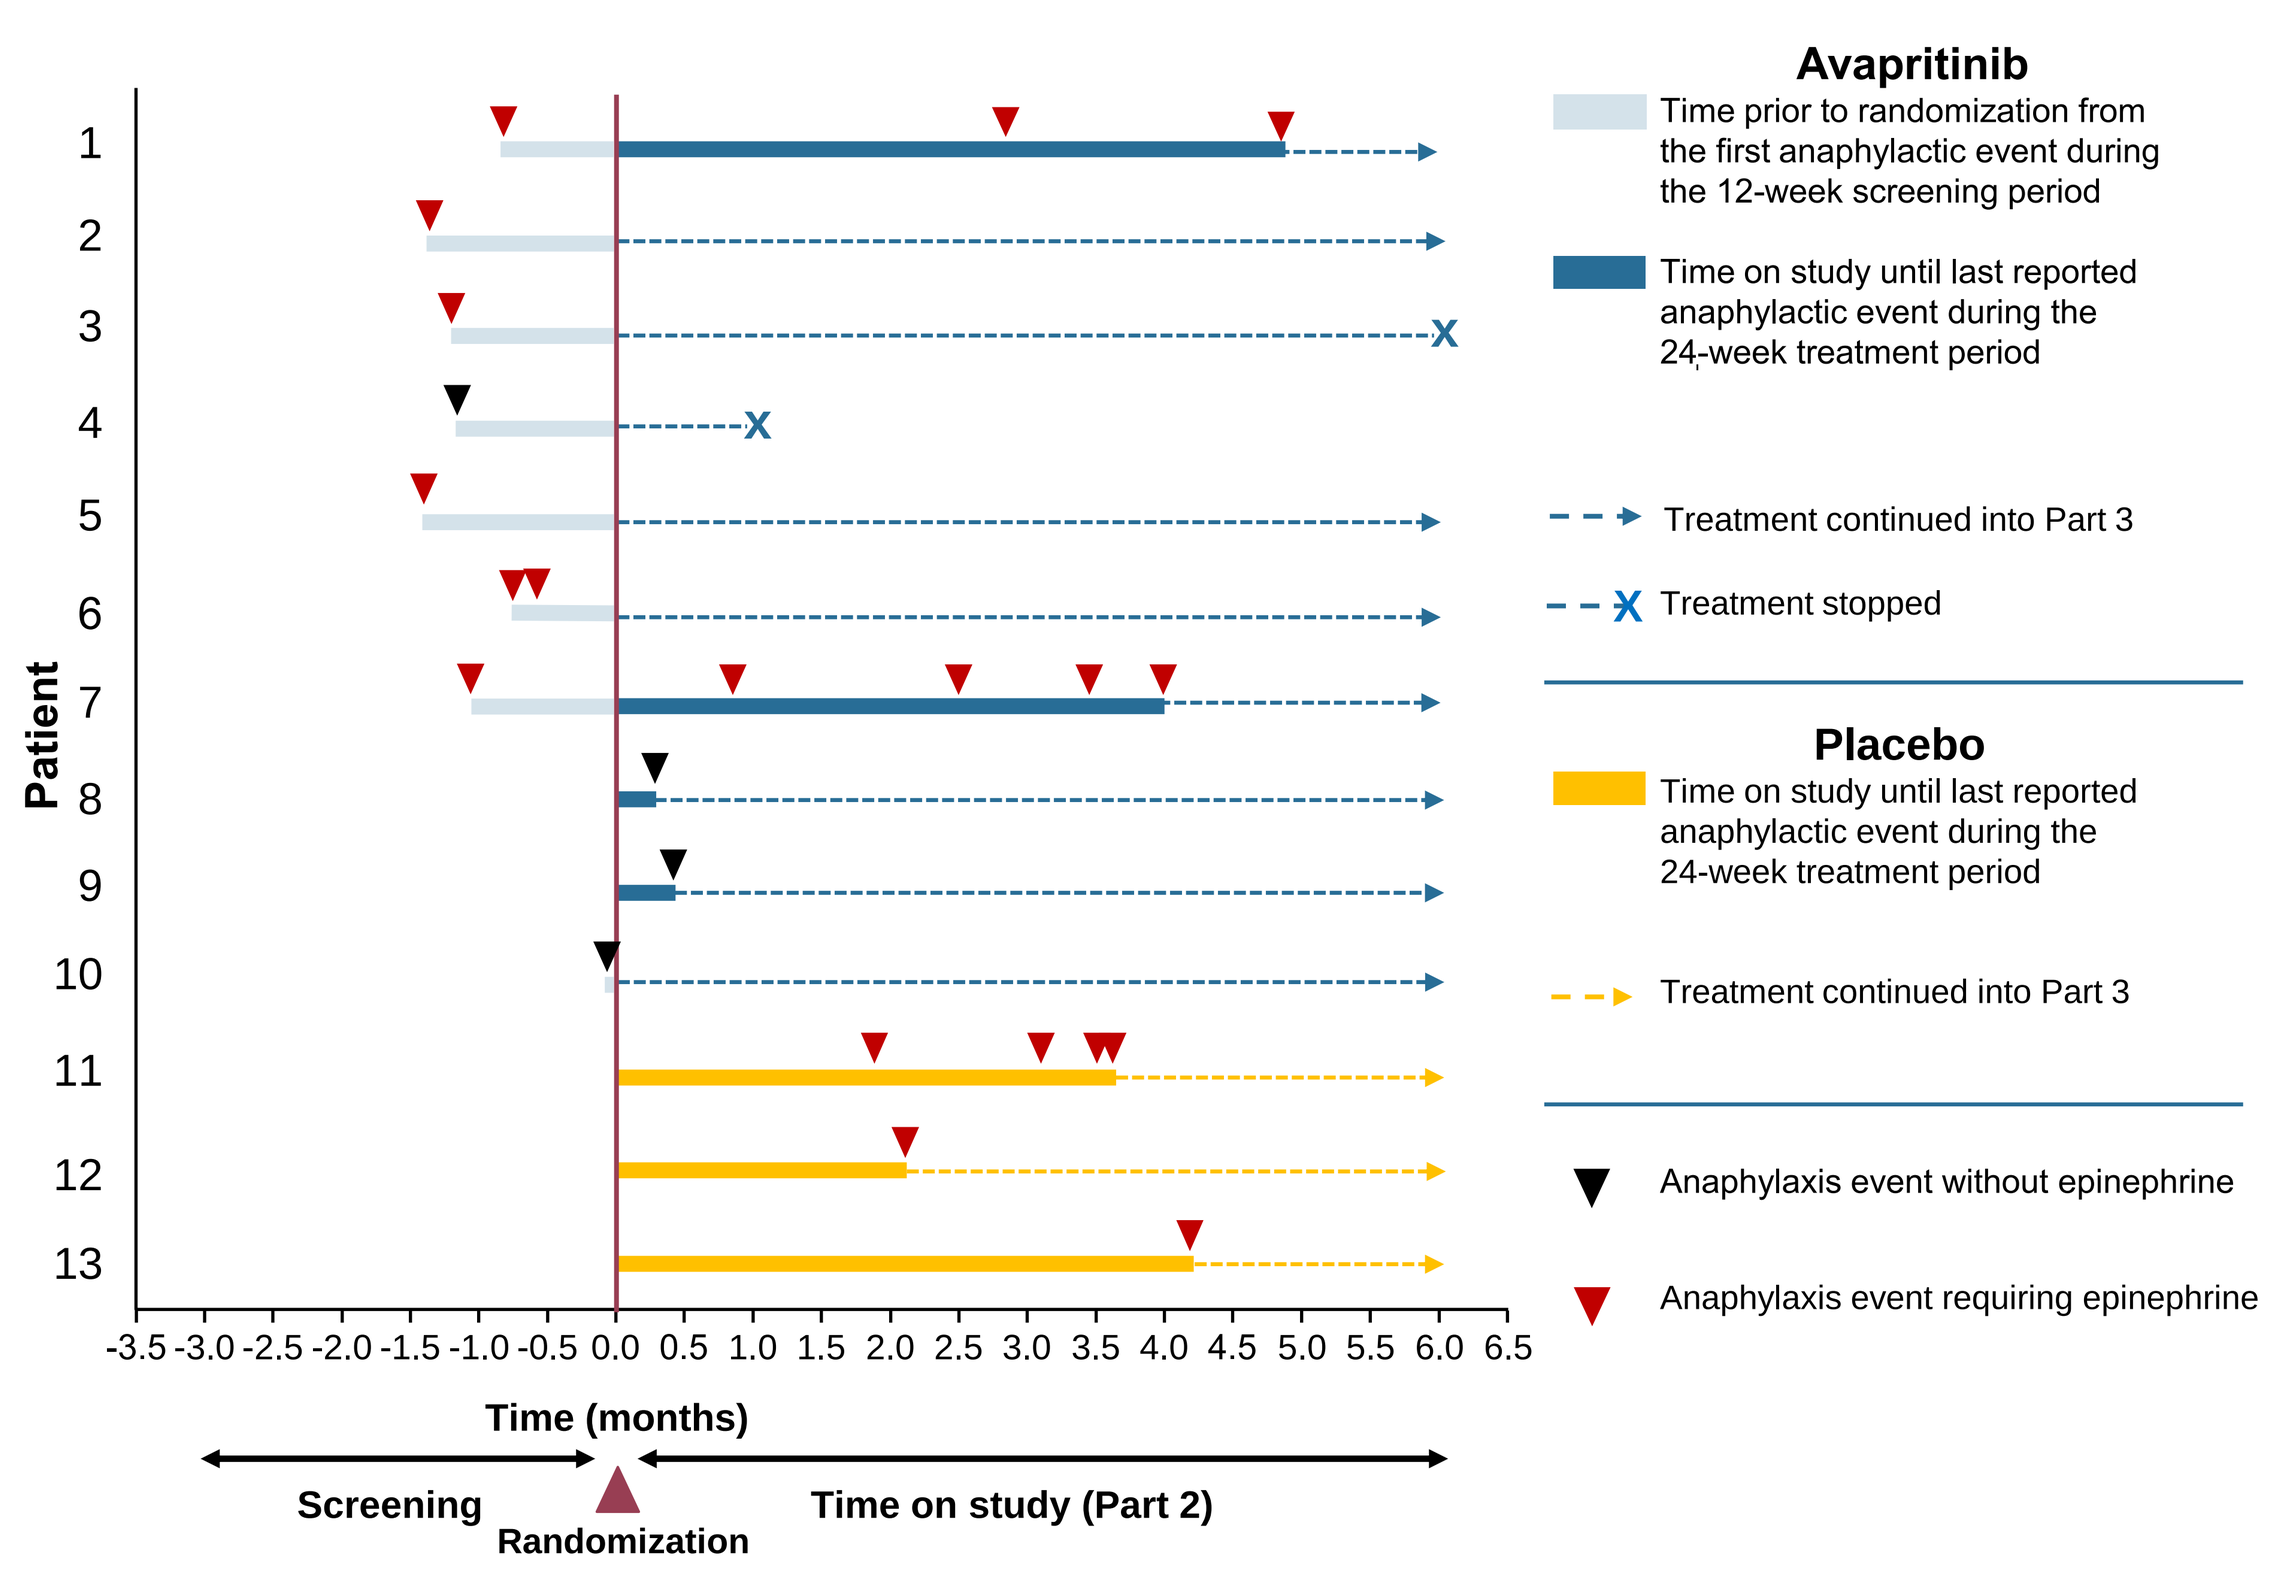
Patient
1
2
3
4
5
6
7
8
9
10
11
12
13
-3.5 -3.0 -2.5 -2.0 -1.5 -1.0 -0.5 0.0 0.5 1.0 1.5 2.0 2.5 3.0 3.5 4.0 4.5 5.0 5.5 6.0 6.5
Time (months)
Screening
Randomization
Time on study (Part 2)
Avapritinib
Time prior to randomization from the first anaphylactic event during the 12-week screening period
Time on study until last reported anaphylactic event during the 24-week treatment period
Treatment continued into Part 3
Treatment stopped
Placebo
Time on study until last reported anaphylactic event during the 24-week treatment period
Treatment continued into Part 3
Anaphylaxis event without epinephrine
Anaphylaxis event requiring epinephrine

Table 3. Anaphylaxis Events

	Patients with anaphylactic events	Number of anaphylactic events during screening	Number of anaphylactic events during Part 2	Number of anaphylactic events requiring Epi-pen use	Event rate during screening (events/mo)	Event rate during Part 2 (events/mo)
Avapritinib n=10/141	1	1	2	3	0.33	0.33
	2	1	0	1	0.33	0.00
	3	1	0	1	0.33	0.00
	4	1	0	0	0.33	0.00
	5	1	0	1	0.33	0.00
	6	2	0	2	0.67	0.00
	7	1	4	5	0.33	0.67
	8	0	1	0	0.00	0.17
	9	0	1	0	0.00	0.17
	10	1	0	0	0.33	0.00
Placebo n=3/71	11	0	4	4	0.00	0.67
	12	0	1	1	0.00	0.17
	13	0	1	1	0.00	0.17

Figure 2. Case Study: In addition to an improvement in the area and color of skin lesions, patient had an improvement in incidence of anaphylaxis at Week 24 with avapritinib treatment

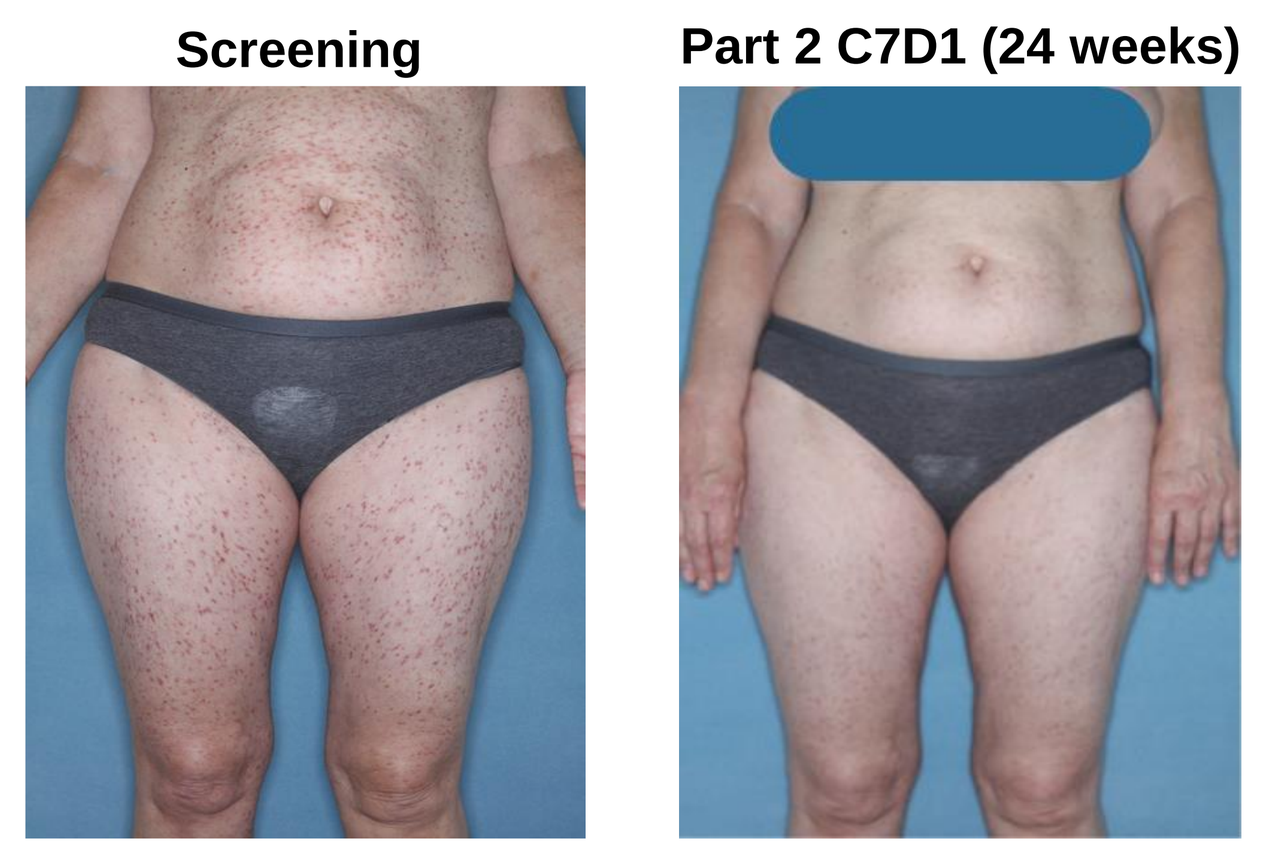
Screening
Part 2 C7D1 (24 weeks)

Patient Case Study

- The patient (#6), a 54-year-old woman, was diagnosed with ISM in 2017
- Prior to study entry, patient experienced severe headaches, nausea, joint and muscle pain, severe fatigue, and frequent anaphylactic reactions (3 anaphylactic events reported in the year before study entry; 2 anaphylactic events during the 12-week screening period, **Figure 1**)
- During the 24 weeks of avapritinib treatment, reduction in skin lesions (**Figure 2**), improvement in pain and increased energy were observed without the occurrence of anaphylactic events (**Figure 1**)
- From baseline to Week 24, the patient had an ~34% decrease in TSS

Quality of Life

- In PIONEER, avapritinib significantly improved symptoms and quality of life; patients reported improvement by Week 4 of treatment that was sustained through Week 24 of Part 2[14]
- After treatment with avapritinib in Part 2, patients with anaphylaxis showed a reduction in MC-QoL scores (58.4 at baseline to 46.8 at Week 24), indicative of improvement from near-severe to mild disease
- Short Form 12 (SF-12) physical scores (PCS) improved rapidly after Week 16 (30.2 at baseline to 34.9 at Week 24); SF-12 mental scores (MCS) also improved (42.2 at baseline to 47.0 at Week 24)
- Patient Global Impression Severity (PGI-S) scores, indicative of global symptom severity, decreased throughout Part 2 (3.1 at baseline to 2.4 at Week 24)

Safety Outcomes

- Avapritinib was well tolerated with a safety profile similar to that of placebo
- Anaphylaxis was the only Grade 3 adverse event (AE) experienced in most patients, demonstrating the impact on the quality of life of these patients
- Except for anaphylaxis, most of the patients experienced AEs that were Grade 1 or 2
 - 1 out of 10 patients in the avapritinib group experienced Grade 3 AEs (dizziness, rash maculopapular, pruritus, chills, and feeling abnormal), assessed as not related to study drug
 - 2 out of 3 patients in the placebo group experienced Grade 3 AEs (adenovirus infection in the first patient, events of gastroenteritis and mastocytosis in the second patient), assessed as not related to study drug
- There were no Grade ≥3 treatment-related AEs
- None of the AEs resulted in treatment discontinuation

Conclusions

- The overall results of the PIONEER study demonstrated that avapritinib significantly improved symptoms, reduced biomarkers of MC burden, and improved quality of life, with a well-tolerated safety profile similar to placebo
 - Avapritinib represents a promising new treatment for patients with this rare, debilitating disease
- In Part 2 of the PIONEER study, patients who received avapritinib 25 mg had fewer anaphylaxis episodes over time compared to patients who received placebo
 - 6 of the 8 patients in the avapritinib group who had baseline anaphylaxis had no events during Part 2 of the study
- We note that the true burden of anaphylaxis is unlikely captured by the short study duration and limited number of cases

References

1. Kristensen T, et al. *J Mol Diagn*. 2011;13(2):180-188.
2. Cohen SS, et al. *Br J Haematol*. 2014;166(4):521-528.
3. Garcia-Montero A, et al. *Blood.* 2006;108(7):2366-2372.
4. Mesa RA, et al. *Cancer*. 2022;128(20):3691-3699.
5. Hermine O, et al. *PLoS One*. 2008;3(5):e2266.
6. van Anrooij B, et al. *Allergy*. 2016;71(11):1585-1593.
7. Akin C, et al. *J Allergy Clin Immunol.* 2022;149(6):1912-1918.
8. Pardanani A. *Blood*. 2013;121(16):3085-3094.
9. Gulen T, et al. *Immunol Allergy Clin North Am.* 2022;42(1):45-63.
10. Buonomo A, et al. *Mediterr J Hematol Infect Dis.* 2022;14(1):e2022040.
11. Pardanani A. *Am J Hematol.* 2021;96(4):508-525.
12. AYVAKIT (avapritinib) [package insert]. Cambridge, MA: Blueprint Medicines Corporation; 2023.
13. Gotlib J, et al. *NEJM Evid*. 2023;2(6). doi: 10.1056/EVIDoa2200339.
14. Akin C, et al. AAAAI 2023 Annual Meeting. Abstract L151 [poster].

Acknowledgments

- We thank the patients and their families for making this trial possible
- We thank the investigators and clinical trial teams who participated in the trial
- This study was supported by Blueprint Medicines Corporation, Cambridge, MA
- Medical writing and editorial support was provided by Srividya Venkitachalam, PhD, of ProEd Communications, supported by Blueprint Medicines Corporation, Cambridge, MA, according to Good Publication Practice guidelines

Disclosures

Dr Pongdee has acted as a consultant for Blueprint Medicines Corporation. **Dr Castells** has no conflict of interests to disclose. **Dr Hartmann** is a consultant for ALK-Abelló, Allergopharma, Blueprint Medicines Corporation, Cogent, KalVista, Leo, Menarini, Novartis, Pfizer, Sanofi, Takeda, and Thermo Fisher, and has received a grant/contract from Thermo Fisher Scientific. **Dr Yi** has received advisory board fees from Blueprint Medicines Corporation and Cogent Therapeutics LLC. **Dr Akin** has received research funding from Blueprint Medicines Corporation and Cogent and is a consultant for Blueprint Medicines Corporation, Cogent, and Novartis. **Dr Alvarez-Twose** is a consultant/speaker for and has received honoraria from Blueprint Medicines Corporation and Novartis. **Dr Cerquozzi** is a consultant/speaker for and/or has worked on advisory boards for AbbVie, Bristol Myers Squibb, GlaxoSmithKline, Jazz Pharmaceuticals, Novartis, Paladin Laboratories, and Pfizer. **Dr Gotlib** is a consultant for Blueprint Medicines Corporation, Cogent, Imago, Incyte, Kartos, Novartis, PharmaEssentia, and Protagonist Therapeutics, and has received grants from Novartis and Protagonist Therapeutics. **Dr Panse** is a consultant, speaker, and/or has worked on scientific advisory boards for Alexion, Apellis, AstraZeneca, Blueprint Medicines Corporation, Boehringer Ingelheim, Bristol Myers Squibb, Chugai Pharmaceutical, F. Hoffmann-La Roche, Novartis, Pfizer, and Sanofi-Pasteur. **Dr Vadas** has no conflict of interests to disclose. **Dr Wortmann** has been a speaker for Blueprint Medicines Corporation. **Ilda Bidollari** is a current employee of Blueprint Medicines Corporation. **Kate Newberry** is a current employee and shareholder of Blueprint Medicines Corporation. **Dr Dybedal** has no conflict of interests to disclose. **Dr Vachhani** has received advisory board fees from AbbVie, Amgen, Blueprint Medicines Corporation, Cogent Biosciences, CTI BioPharma Corp, Daiichi Sankyo, Genentech, GlaxoSmithKline, Incyte, MorphoSys, Novartis, Pfizer, Servier, and Stemline.

Presented at the **American College of Allergy, Asthma & Immunology (ACAAI) Annual Meeting**, November 9-13, 2023, Anaheim, CA, USA