Healthcare Resource Utilization and Costs of Medicare Fee for Service Beneficiaries Newly Diagnosed with Moderate to Severe Indolent Systemic Mastocytosis

Erin Sullivan[1], Jenna Cohen[1], Chelsea Norregaard[1], Uyen Nguyen[1], Chris Sloan[1], Allison Petrilla[2], Alison R. Silverstein[2], Anne Murunga[2], Jill Schinkel[2]

[1]*Blueprint Medicines, Cambridge, MA, USA;* [2]*Avalere Health, Washington DC, USA*

Presented at the 63rd American Society of Hematology (ASH) Annual Meeting and Exposition, Atlanta, Georgia, USA, December 11-14, 2021. Publication Number: 4049.

Introduction & Objective

- Systemic mastocytosis (SM) is a rare hematologic disorder driven by the *KIT* D816V mutation in which mast cells accumulate in tissues or organs.[1,2]
- The majority of patients with SM have non-advanced stages of disease, which includes indolent SM (ISM).
- With no approved therapies, treatment is focused on symptom management.[3]
- To date, the economic burden of ISM has not been well-studied among Medicare patients.
- This retrospective study compared direct healthcare resource utilization (HCRU) and healthcare costs in Medicare beneficiaries with ISM and a matched cohort without SM.

Methods

- This study used Centers for Medicare and Medicaid Services-sourced 100% Medicare Fee for Service (FFS) claims (Parts A/B/D) and identified newly diagnosed SM patients who had ≥2 medical claims for SM (ICD-10-CM Dx codes: D47.02, C94.30, C94.31, C94.32, or C96.21) between 1/1/2017 and 12/31/2018. Patients were excluded if they had evidence of advanced SM (i.e., >2 medical claims with diagnosis of aggressive SM, SM with associated hematological neoplasm, or mast cell leukemia). Patients with moderate to severe ISM[4] were identified by use of a tyrosine kinase inhibitor, history of anaphylaxis, splenomegaly, hepatomegaly, osteoporosis or fracture, and/or use of steroids.
- The index date was the date of first observed SM diagnosis. Continuous enrolment in Medicare Parts A/B/D for 12 months pre- and post-index was required.
- ISM patients were direct matched (1:1) on age, sex, race, index year, Medicare-Medicaid dual eligibility, and Charlson Comorbidity Index score to a random sample of Medicare beneficiaries without SM.
- HCRU and costs (based on Medicare payment amounts) were assessed during the 12 months pre- and post-index. Medical costs are reported in 2021 US dollars.

Results

Patient Characteristics

- Post-match, there were 333 ISM and 333 non-SM patients.
- Mean age of the ISM and control cohort was 67 years. Over 25% of patients were <65 years of age at index and originally qualified for Medicare with a disability. Mean CCI score was 2.3 (**Table 1**).
- During the 12-month pre-index period, ISM patients had more specialist physician office visits per patient (mean [SD]: 15 [15]) compared to non-SM patients (10 [13]; p<0.01).
- ISM patients vs. controls had higher prevalence of asthma, malignancy, and osteoporosis (**Table 1**).
- ISM patients were also higher utilizers of corticosteroids (64% vs. 54%, p=0.0094) and epinephrine auto-injectors (31% vs. 1%, p<0.0001) compared to non-SM patients.
- Mean (SD) total medical and pharmacy pre-index costs for the ISM cohort were $1,786 ($2,446) and $1,428 ($2,862) for the non-SM cohort (p=0.084).

Table 1: Beneficiary Baseline Characteristics, 12 Months Pre-Index

Baseline Characteristics	ISM Cohort (N =333) N (%)	Non-SM Cohort (N = 333) N (%)	P-Value
Mean (SD) age at index*	67.3 (11.7)	67.8 (13.3)	0.53
Female*	253 (76%)	253 (76%)	1.00
White*	312 (94%)	312 (94%)	1.00
US Census Region*			0.00
Northeast	73 (22%)	36 (11%)	
Midwest	73 (22%)	90 (27%)	
South	95 (29%)	115 (35%)	
West	91 (27%)	91 (27%)	
Dual eligible for Medicare and Medicaid*	64 (19%)	64 (19%)	1.00
Qualified for Medicare based on Pre-Existing Disability (vs. age)	88 (26%)	87 (26%)	0.93
CCI Score, Mean (SD)*	2.3 (2.1)	2.3 (2.1)	1.00
Comorbidities of interest			
Asthma	95 (29%)	50 (11%)	<0.01
Malignancy (Most prevalent below)	154 (46%)	62 (19%)	<0.01
Mast Cell Neoplasm	139 (42%)	0 (0%)	0.00
Prostate	<11 (<3%)	<11 (<3%)	0.61
Bone Metastasis	<11 (<3%)	0 (0%)	0.03
Female Breast	<11 (<3%)	<11 (<3%)	0.19
Hypertension	193 (58%)	225 (68%)	0.01
Diabetes without complications	62 (19%)	114 (34%)	<0.01
Diabetes with complications	25 (8%)	50 (15%)	<0.01
Osteoporosis	98 (29%)	47 (14%)	<0.01
Renal disease	23 (7%)	38 (11%)	0.04

*Patients were direct matched on age, sex, race, index year, Medicare-Medicaid dual eligibility, and CCI score.

Medical and Pharmacy Expenditures

- Total medical + pharmacy costs in the 12-month post-index period were over 25% higher for ISM patients than for non-SM controls but were not statistically different (mean [SD]: $21,096, [$29,586] vs. $16,731 [$35,491]; p=0.0851).
- Pharmacy (Part D only) expenditures were over 2 times higher ($7,085 [$21,007] vs. $3,117 [$10,766], p=0.0023) and accounted for a greater proportion (33.6% vs. 18.6%) of total costs for ISM patients vs. non-SM patients (**Figure 1**).
- Among patients who qualified for Medicare based on non-age-related factors, mean total per patient costs in the 12 months post-index were $31,893 for ISM patients and $19,915 for non-SM patients.

Figure 1: Direct Medical and Pharmacy Costs During 12 Months Post-Index Among ISM and Non-SM Patients

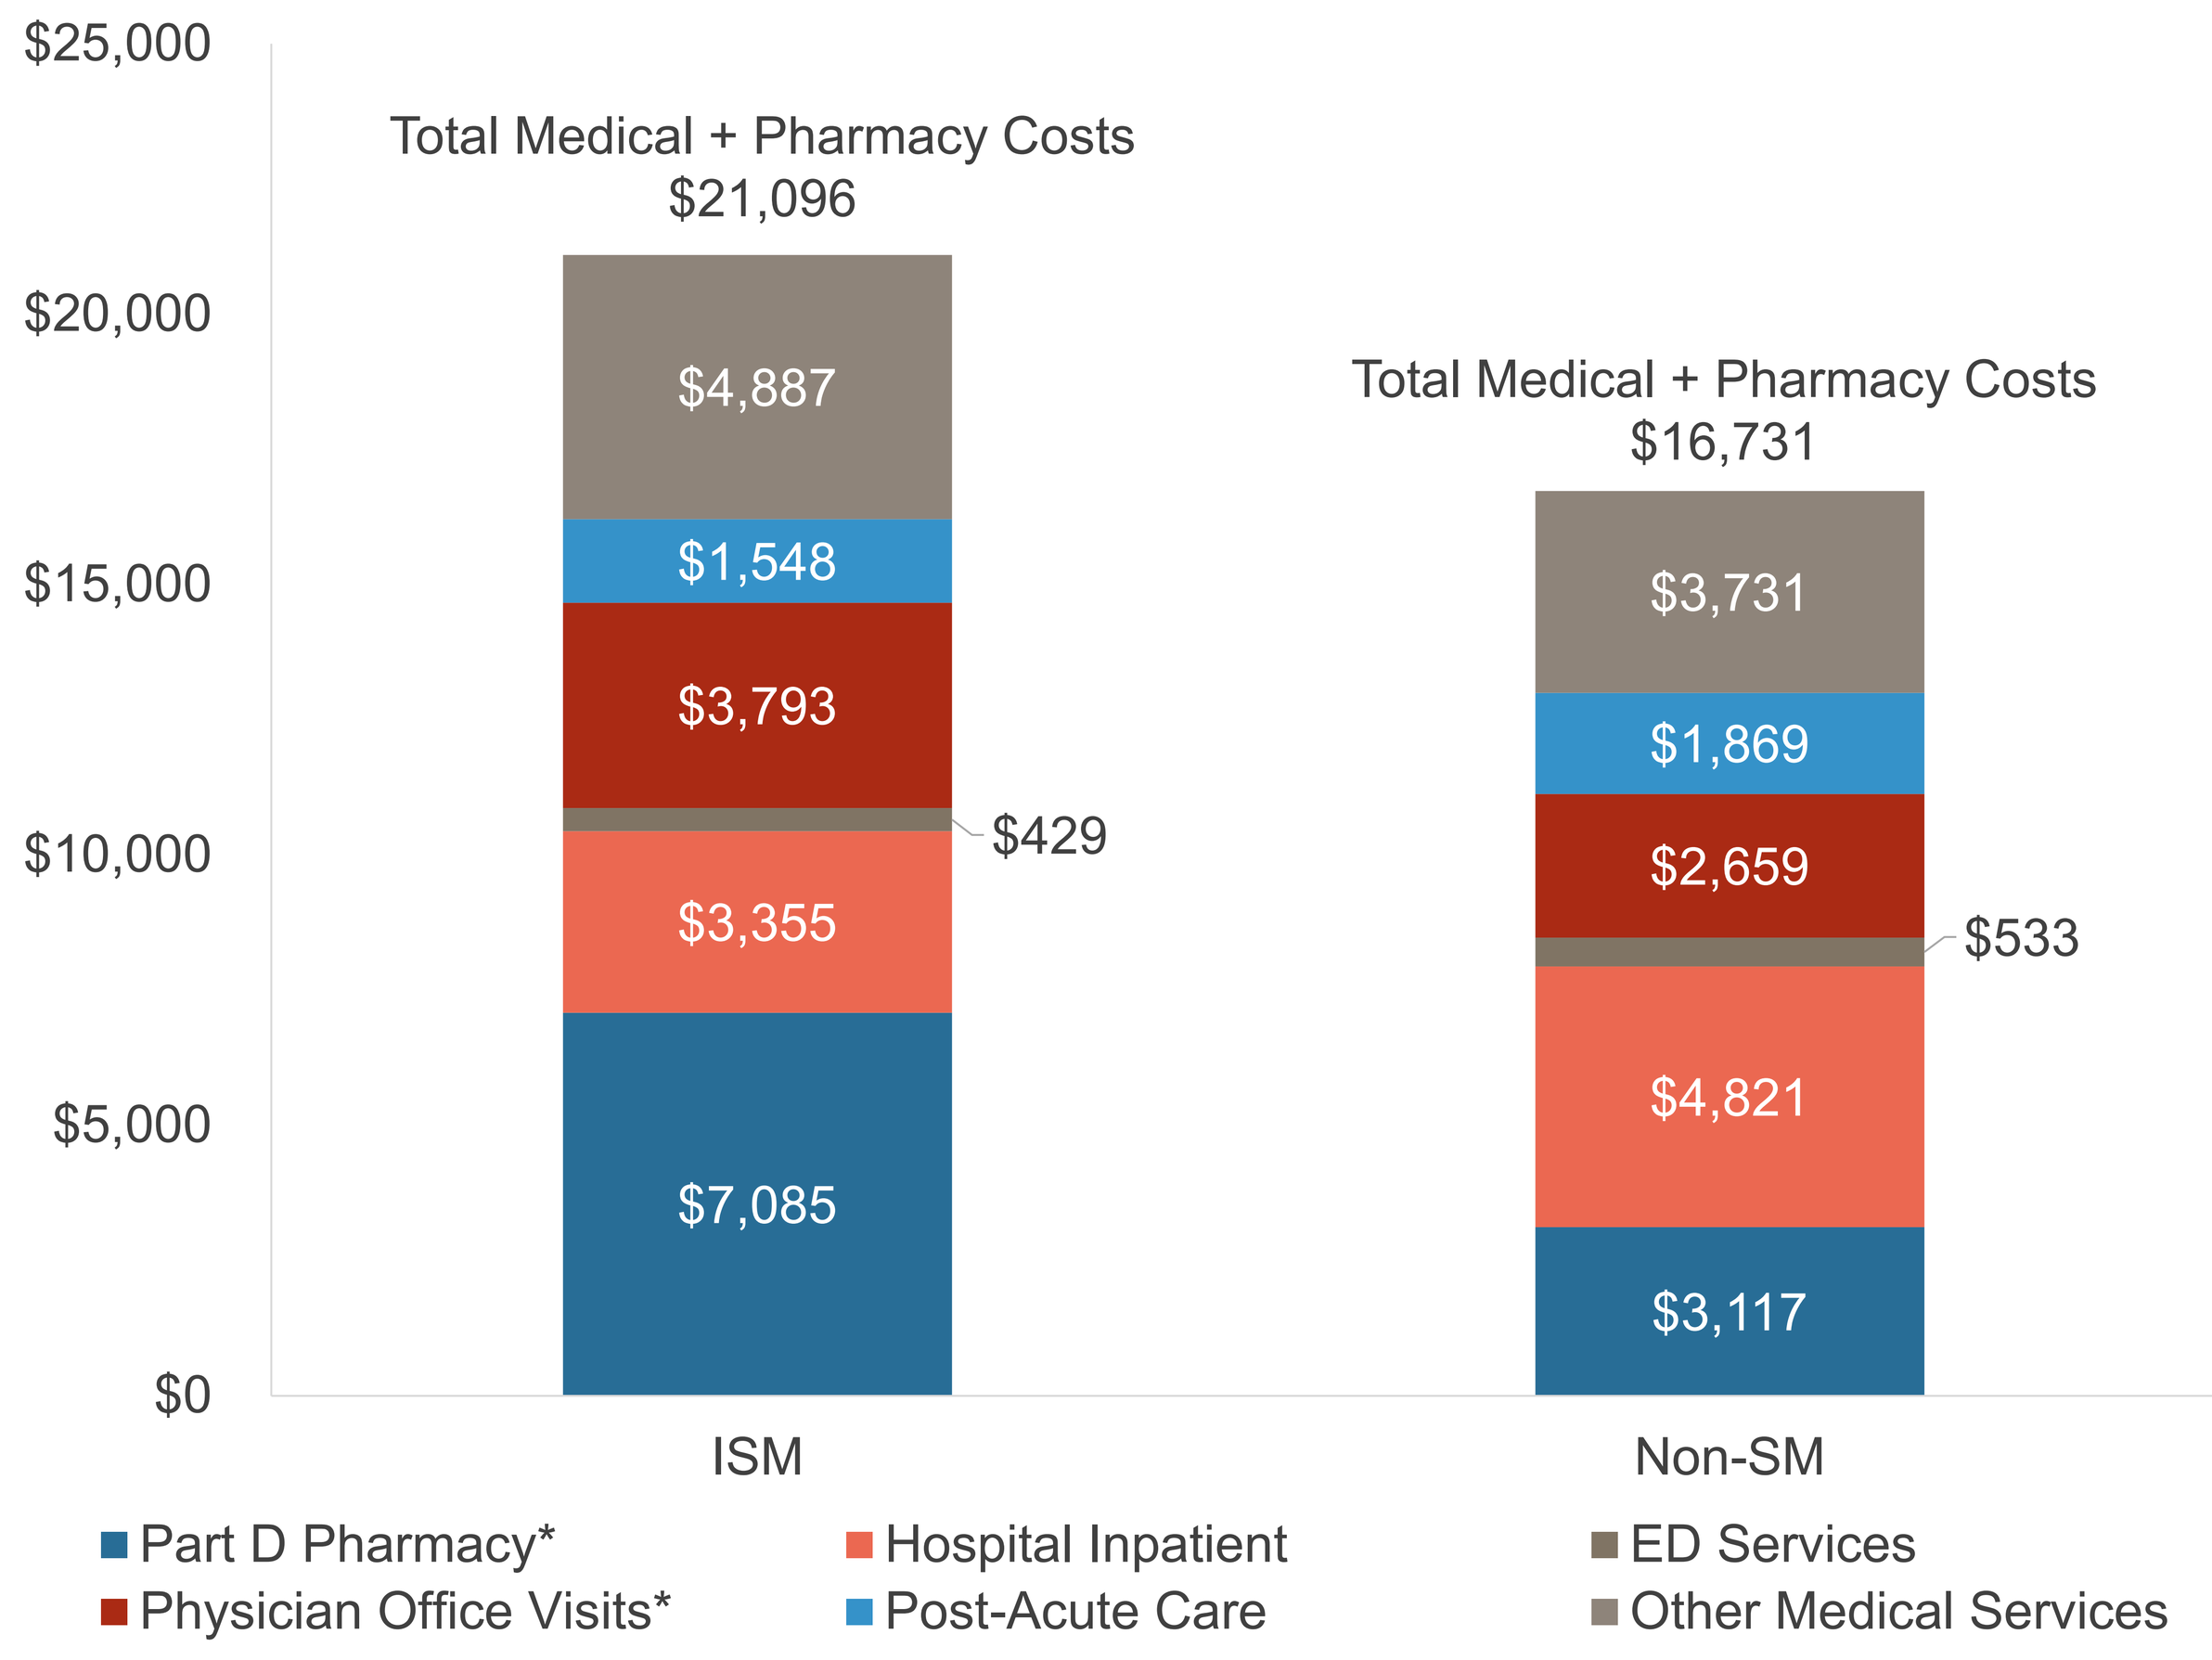
$25,000
$20,000
$15,000
$10,000
$5,000
$0
Total Medical + Pharmacy Costs $21,096
$4,887
$1,548
$3,793
$429
$3,355
$7,085
Total Medical + Pharmacy Costs $16,731
$3,731
$1,869
$2,659
$533
$4,821
$3,117
ISM
Non-SM
Part D Pharmacy*
Physician Office Visits*
Hospital Inpatient
Post-Acute Care
ED Services
Other Medical Services

ISM: Indolent Systemic Mastocytosis; ED: emergency department; SM: Systemic Mastocytosis
*Differences between ISM and Non-SM cohort were statistically significant at p=0.05
Other Medical Costs include: outpatient visits other than ED, laboratory tests, radiology encounters, and other physician services

Healthcare Resource Utilization

- ISM patients were high utilizers of specialty physician office visits post-index compared to non-SM controls; more ISM patients had ≥1 oncology/hematology visit (32.7% vs. 10.2%; p<0.0001), or allergy/immunology visit (43.2% vs. 3.3%; p<0.0001) (**Figure 2**).

Figure 2: Healthcare Resource Utilization During The 12-Month Post-Index Period Among ISM and Non-SM Patients

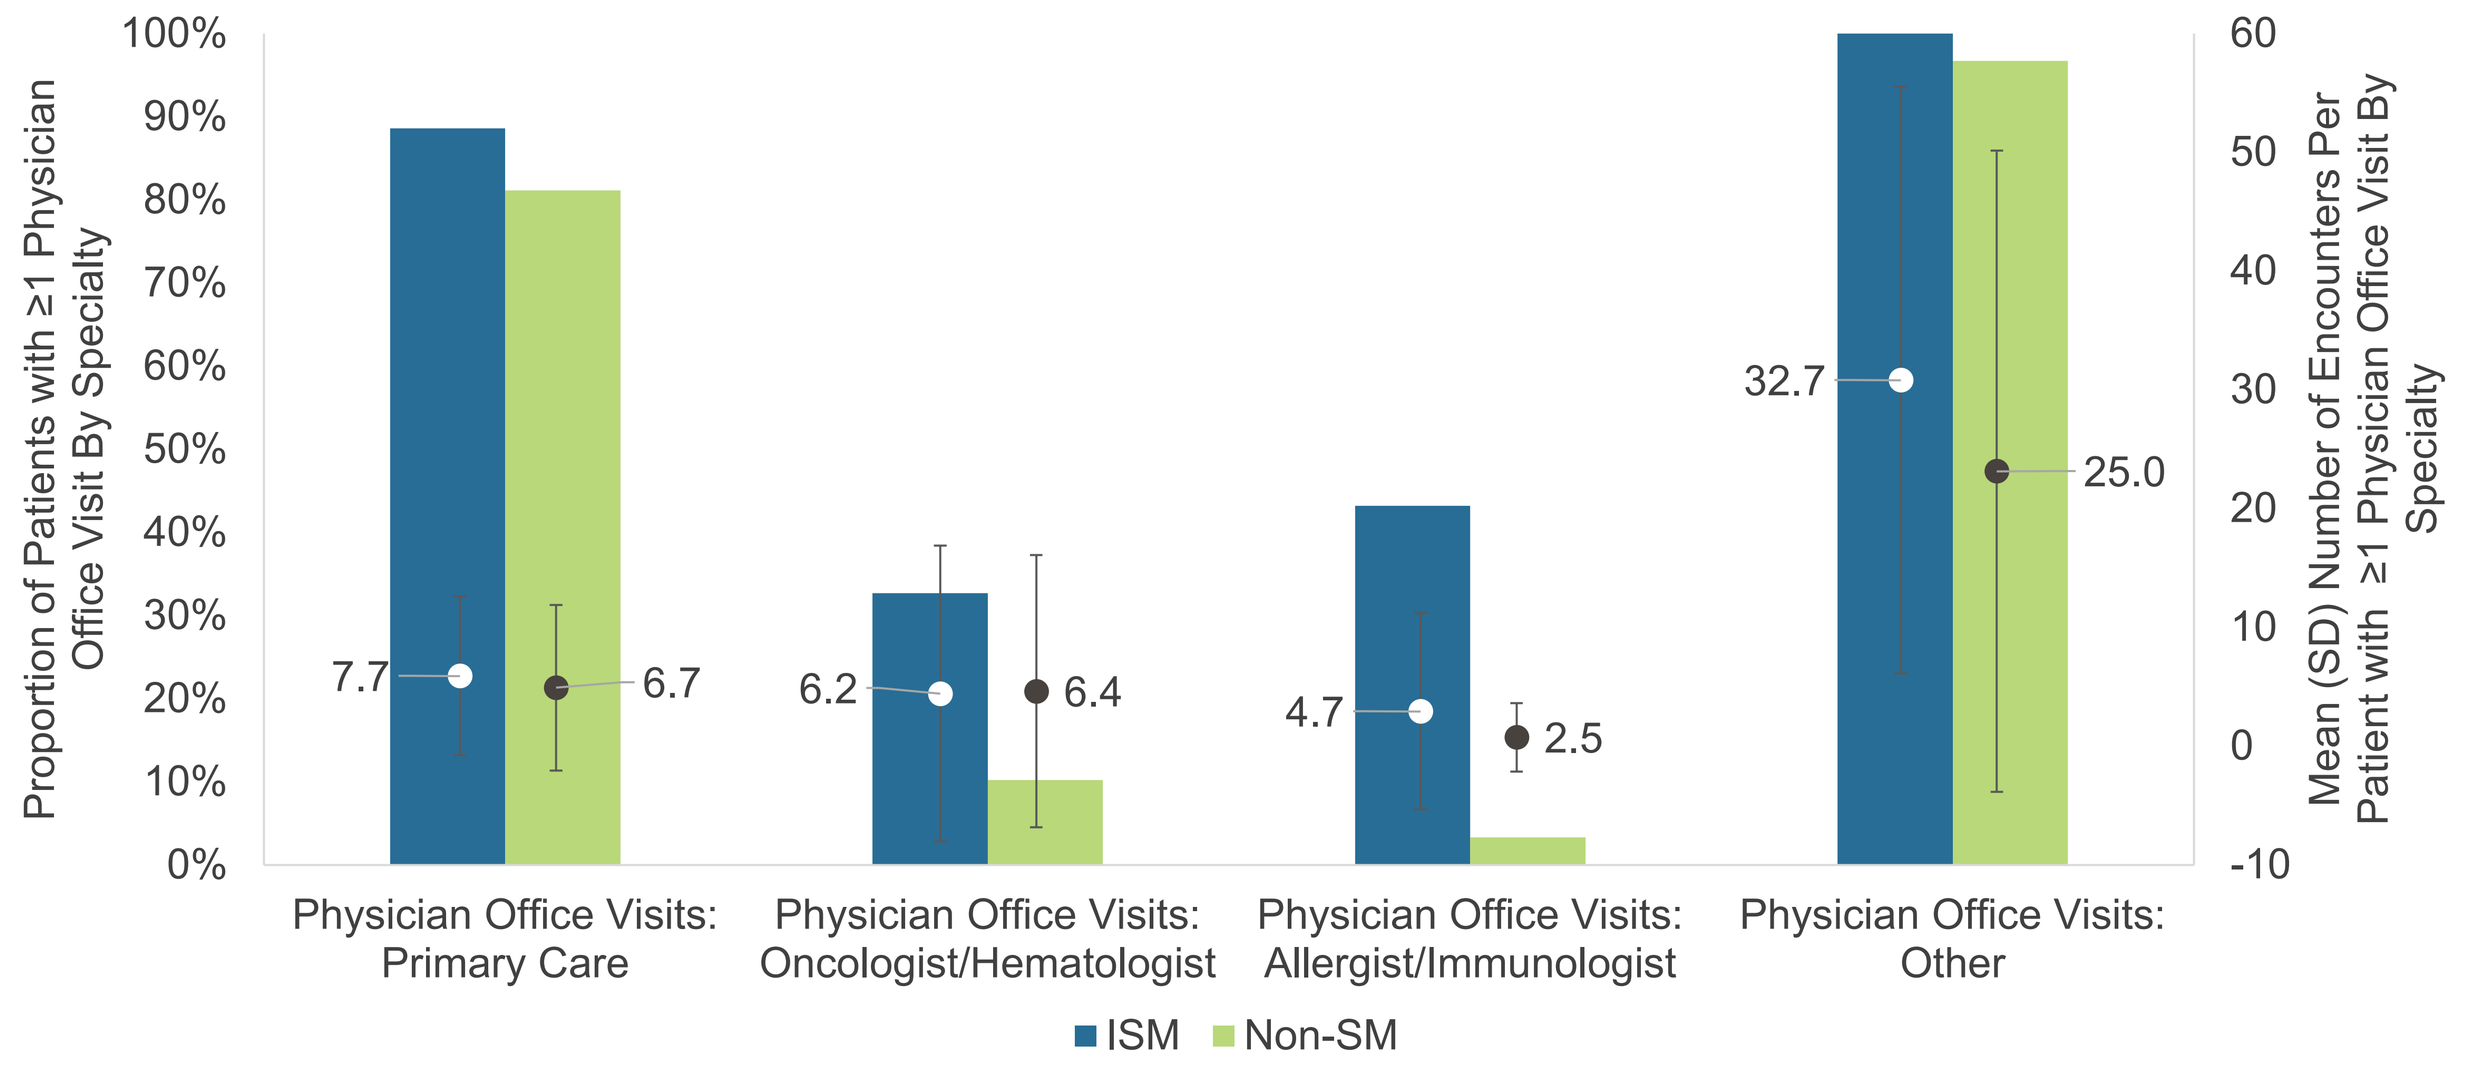
Proportion of Patients with ≥1 Physician Office Visit By Specialty
100%
90%
80%
70%
60%
50%
40%
30%
20%
10%
0%
7.7
6.7
6.2
6.4
4.7
2.5
32.7
25.0
Mean (SD) Number of Encounters Per Patient with ≥1 Physician Office Visit By Specialty
60
50
40
30
20
10
0
-10
Physician Office Visits: Primary Care
Physician Office Visits: Oncologist/Hematologist
Physician Office Visits: Allergist/Immunologist
Physician Office Visits: Other
ISM
Non-SM

ISM: Indolent Systemic Mastocytosis; SM: Systemic Mastocytosis
*Differences between ISM and Non-SM cohorts were not statistically significant at p=0.05 for mean number of primary care, oncology/hematology, or allergy/immunology visits

- Approximately 30% of ISM patients filled ≥1 prescription for an epinephrine auto-injector compared with <3% in non-SM patients (p<0.0001). More ISM patients had prescriptions for H1 antihistamines (11.7% vs. 3.6%, p<0.0001), oral and systemic corticosteroids (41.4% vs. 22.8%, p<0.0001; 38.4% vs. 29.4%, p=0.0141, respectively), leukotriene antagonists (34.8% vs. 7.2%; p<0.0001), and omalizumab (6.0% vs. 0.0%, p<0.0001) (**Figure 3**).

Figure 3: Select Medication Utilization During the 12-Month Post-Index Period Among ISM and Non-SM Patients

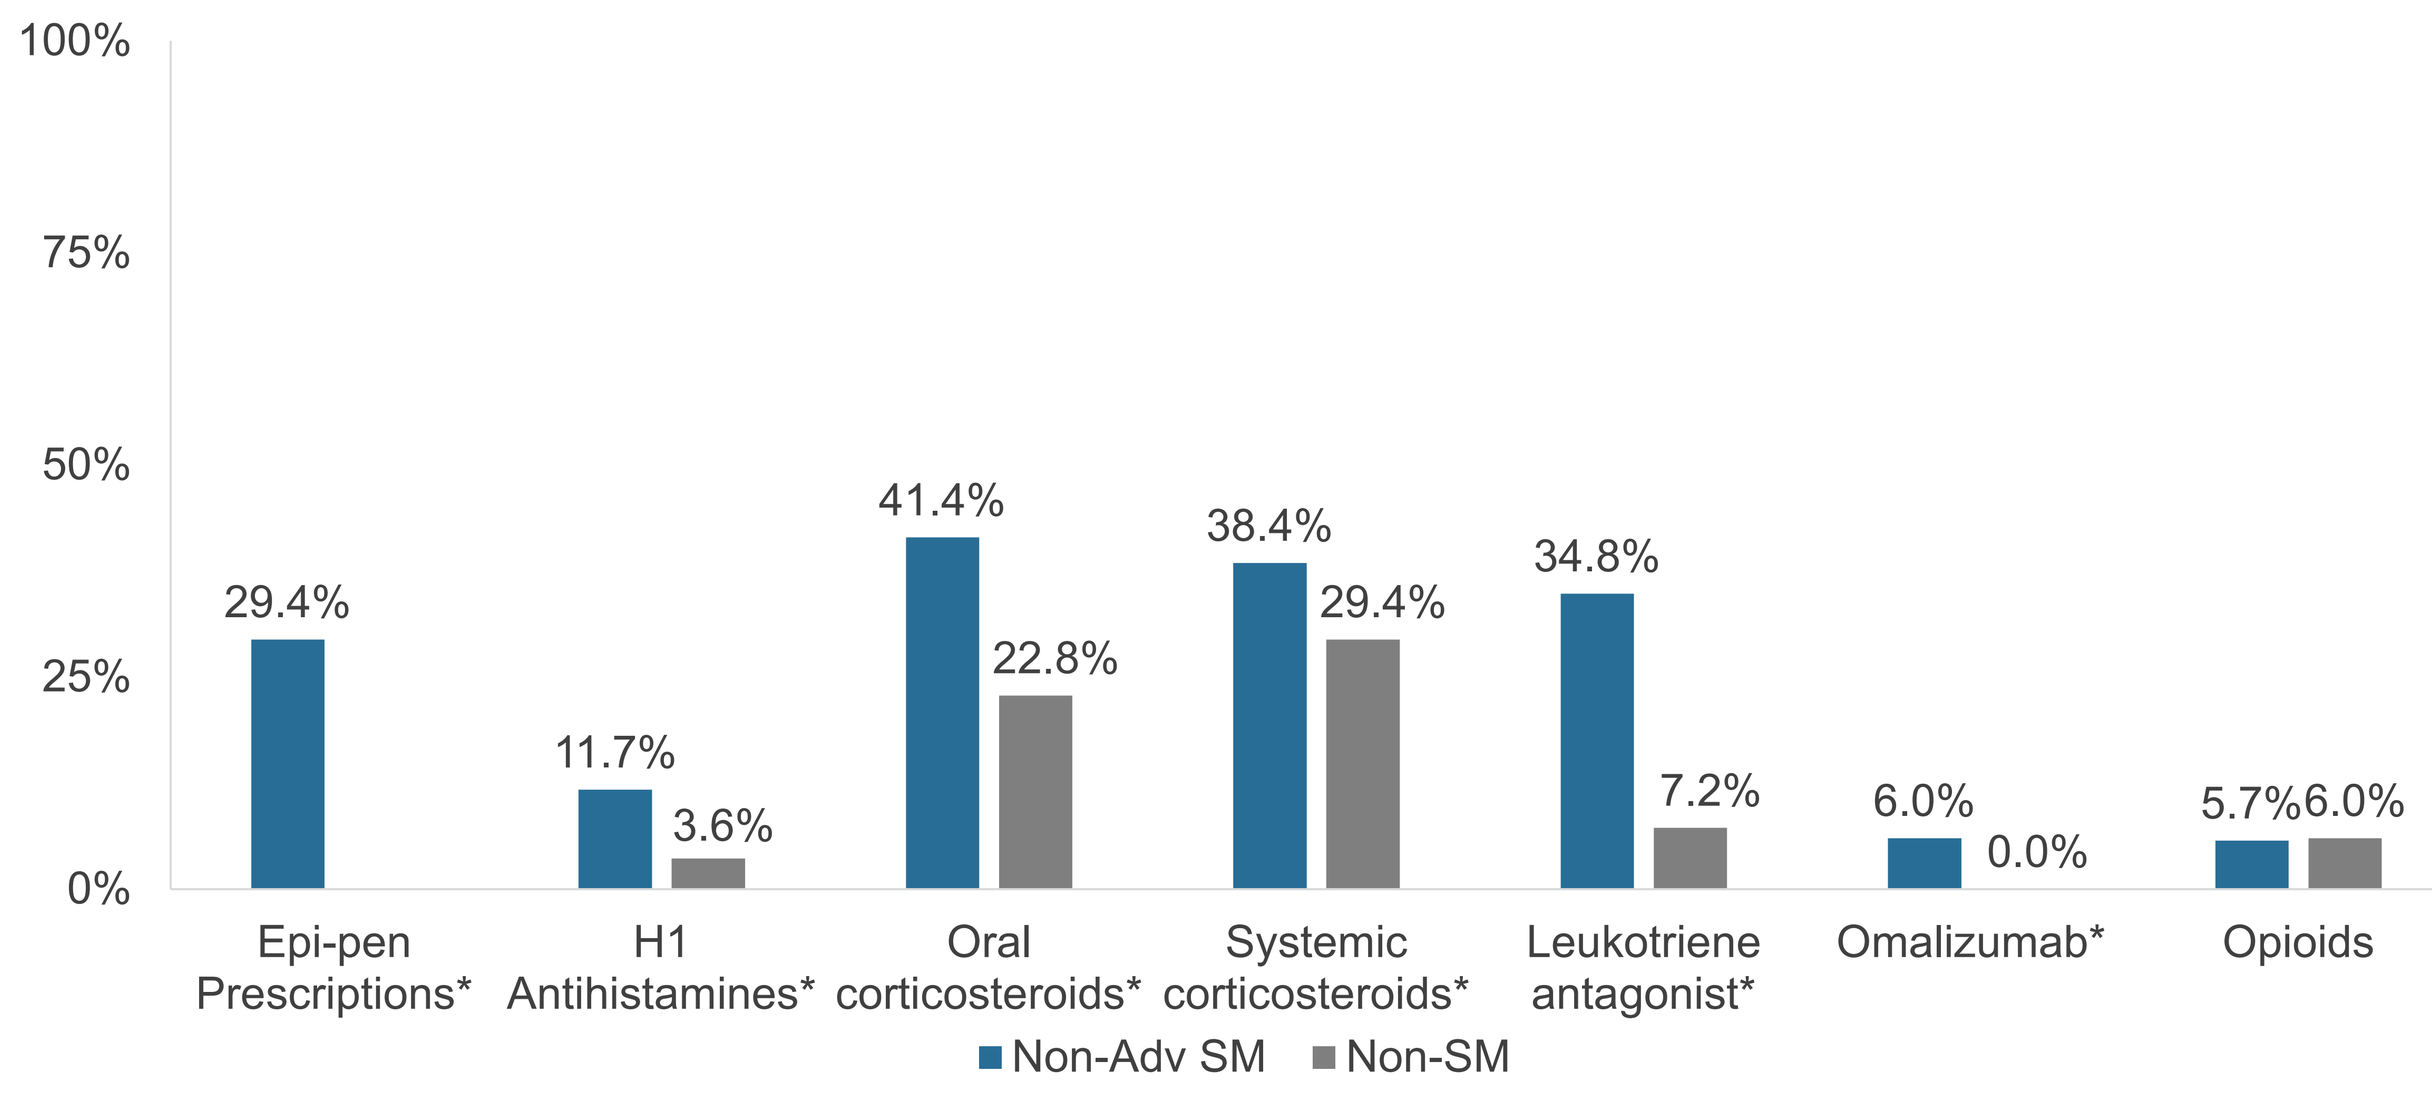
100%
75%
50%
25%
0%
29.4%
11.7%
3.6%
41.4%
22.8%
38.4%
29.4%
34.8%
7.2%
6.0%
0.0%
5.7%
6.0%
Epi-pen Prescriptions*
H1 Antihistamines*
Oral corticosteroids*
Systemic corticosteroids*
Leukotriene antagonist*
Omalizumab*
Opioids
Non-Adv SM
Non-SM

ISM: Indolent Systemic Mastocytosis; SM: Systemic Mastocytosis
*Differences between ISM and Non-SM cohorts were not statistically significant at p=0.05
<3% Non-SM patients had an epi-pen prescription; 0% Non-SM patients had a prescription for omalizumab

Limitations

- Medicare beneficiaries with SM were identified beginning January 1, 2017, but the ICD-10 diagnosis code for SM (D47.02) did not go into effect until October 1, 2017.
- Although we limited the ISM cohort to those with no evidence of SM in the 12-month pre-index period, patients may have been originally diagnosed outside of the observation period. Complete medical history was not available. Misidentification of patients may contribute to an underestimation of SM patients in the study sample. Future research may consider using a refined patient identification algorithm to identify patients who likely have ISM but are not yet diagnosed and to delineate between patients with ISM and smoldering SM.
- Use of age and comorbidity score in the cohort matching process resulted in a larger proportion of ISM and non-SM patients who qualified for Medicare due to pre-existing disability rather than age (as compared to the full Medicare population). The matching process did not fully account for the reason for disability and other comorbid factors which may undercount the true difference in HCRU and costs between ISM and non-SM populations.
- Results from this study may not be generalizable to populations who have less access to the US health care system and/or who are uninsured.
- This study cannot be used to determine cause and effect; claims data only capture those disease entities and variables that have their own specific billing codes. As such, temporality cannot be truly established with the use of claims data.

Conclusions

- Compared to a matched cohort of non-SM Medicare FFS patients, ISM Medicare patients had 26% higher mean per patient total healthcare expenditures ($21,096 vs. $16,731), driven by higher utilization of outpatient resources, specifically visits to oncologists/hematologists and allergists/immunologists, and prescription medications.
- Further research to understand the basis of the higher proportion of ISM patient in this analysis who were <65 years and qualified for Medicare with a disability (vs. 14% in all of Medicare), and the corresponding long-term medical costs among these patients is warranted.

References

1. Valent, P., Akin, C., Gleixner, K. V., Sperr, W. R., Reiter, A., Arock, M., & Triggiani, M. (2019). Multidisciplinary challenges in mastocytosis and how to address with personalized medicine approaches. *International journal of molecular sciences*, *20*(12), 2976.
2. Shomali, W., & Gotlib, J. (2018). The new tool "KIT" in advanced systemic mastocytosis. *Hematology 2014, the American Society of Hematology Education Program Book*, *2018*(1), 127-136.
3. Pardanani, A. (2019). Systemic mastocytosis in adults: 2019 update on diagnosis, risk stratification and management. *American journal of hematology*, *94*(3), 363-377.
4. Padilla, B., Shields, A. L., Taylor, F., Li, X., Mcdonald, J., Green, T., ... & Mar, B. (2021). Psychometric evaluation of the Indolent Systemic Mastocytosis Symptom Assessment Form (ISM-SAF) in a phase 2 clinical study. *Orphanet journal of rare diseases*, *16*(1), 1-10.

This study was sponsored by Blueprint Medicines. Blueprint Medicines and associated logo are trademarks of Blueprint Medicines Corporation. © 2021 Blueprint Medicines Corporation.