Reductions in Polypharmacy for Patients With Indolent Systemic Mastocytosis on Avapritinib

Poster Number 696

Cem Akin,[1*] Jason Gotlib,[2] Ivan Alvarez-Twose,[3] Deepti H. Radia,[4] Tsewang Tashi,[5] Cristina Bulai Livideanu,[6] Vito Sabato,[7] Hanneke Oude Elberink,[8] Joseph Jurcic,[9] Paul Van Daele,[10] Andreas Reiter,[11] Prithviraj Bose,[12] Peter Vadas,[13] Patrizia Bonadonna,[14] Daniel J. DeAngelo,[15] Karin Hartmann,[16,17] Stephen Oh,[18] Princess U. Ogbogu,[19] Jocelyn Rice,[1] Mehak Gulati,[1] Ilda Bidollari,[20] Kate J. Newberry,[20] Hui-Min Lin,[20] Mariana Castells[21]

[1]University of Michigan, Ann Arbor, MI, USA; [2]Stanford Cancer Institute/Stanford University School of Medicine, Stanford, CA, USA; [3]Institute of Mastocytosis Studies of Castilla-La Mancha (CLMast), Virgen del Valle Hospital, Toledo, Spain; [4]Guy's & St Thomas' NHS Foundation Trust, London, UK; [5]Huntsman Cancer Institute, University of Utah, Salt Lake City, UT, USA; [6]Département de Dermatologie, Centre de Référence des Mastocytoses, Centre Hospitalier Universitaire de Toulouse, Toulouse, France; [7]Department of Immunology, Allergology and Rheumatology, University of Antwerp, and Antwerp University Hospital, Antwerp, Belgium; [8]Department of Allergology, University Medical Center, Groningen Research Institute Asthma and COPD, University of Groningen, Groningen, Netherlands; [9]Division of Hematology/Oncology, Herbert Irving Comprehensive Cancer Center, Columbia University, New York, NY, USA; [10]Department of Internal Medicine and Immunology, Erasmus Medical Center, Rotterdam, Netherlands; [11]Department of Hematology and Oncology, University Hospital Mannheim, Heidelberg University, Mannheim, Germany; [12]The University of Texas MD Anderson Cancer Center, Houston, TX, USA; [13]Department of Medicine, Division of Clinical Immunology and Allergy, St Michael's Hospital, University of Toronto, Toronto, ON, Canada; [14]Unità Semplice Dipartimentale di Allergologia, Azienda Ospedaliera Universitaria Integrata di Verona, Verona, Italy; [15]Dana–Farber Cancer Institute, Boston, MA, USA; [16]Division of Allergy, Department of Dermatology, University Hospital Basel and University of Basel, Basel, Switzerland; [17]Department of Biomedicine, University Hospital Basel and University of Basel, Basel, Switzerland; [18]Siteman Cancer Center at Barnes-Jewish Hospital and Washington University School of Medicine, St. Louis, MO, USA; [19]Division of Pediatric Allergy, Immunology and Rheumatology, Department of Pediatrics, University Hospitals Rainbow Babies and Children's Hospital, Case Western Reserve University School of Medicine, Cleveland, OH, USA; [20]Blueprint Medicines Corporation, Cambridge, MA, USA; [21]Department of Medicine, Brigham and Women's Hospital, Harvard Medical School, Boston, MA, USA

***Presenting author**

Introduction

- Indolent systemic mastocytosis (ISM) is a clonal mast cell disease driven by the *KIT* D816V mutation in ~95% of patients[1–3]
- Patients with ISM experience life-long debilitating symptoms and poor quality of life (QoL) with significant comorbidity[4–8]
- For the management of these symptoms, most patients rely on polypharmacy with best supportive care (BSC) medications, but no controlled trials of BSC medications exist to provide evidence of clinical benefits[8–10]
 - Furthermore, such polypharmacy can have negative consequences for patients, including increased adverse drug events, harmful drug–drug interactions, and decreased medication adherence[9]
- Avapritinib is a potent, oral, tyrosine kinase inhibitor that selectively targets the *KIT* D816V mutation[11]
- In the PIONEER trial (NCT03731260) studying patients with moderate to severe ISM, avapritinib significantly reduced symptom burden in these patients compared with placebo. Based on this finding, avapritinib is now approved in the USA and Europe for adult patients with ISM[11–13]
- Patients who received avapritinib in PIONEER were more likely to reduce or discontinue BSC medications compared with placebo at Week 24 (24% *vs* 13%, respectively)[13]
- Here, we more deeply examine the changes in polypharmacy that occurred following treatment with avapritinib in patients with ISM

Methods

- PIONEER, a global, randomized, double-blind, placebo-controlled trial, evaluated the safety, efficacy, and QoL in patients with ISM receiving avapritinib plus BSC compared with patients receiving placebo plus BSC
- Eligibility criteria included patients experiencing moderate to severe ISM symptoms despite receiving BSC, including H1 and H2 antihistamines, leukotriene receptor antagonists, cromolyn sodium, proton pump inhibitors, corticosteroids, and omalizumab
- In Part 2, 212 patients were randomly assigned in a 2:1 ratio to receive avapritinib 25 mg orally once daily (QD) plus BSC (avapritinib) or placebo plus BSC (placebo) for 24 weeks. After 24 weeks of treatment was completed, patients were eligible to receive avapritinib 25 mg QD for up to 5 years in Part 3 (**Figure 1**)

Figure 1. PIONEER study design

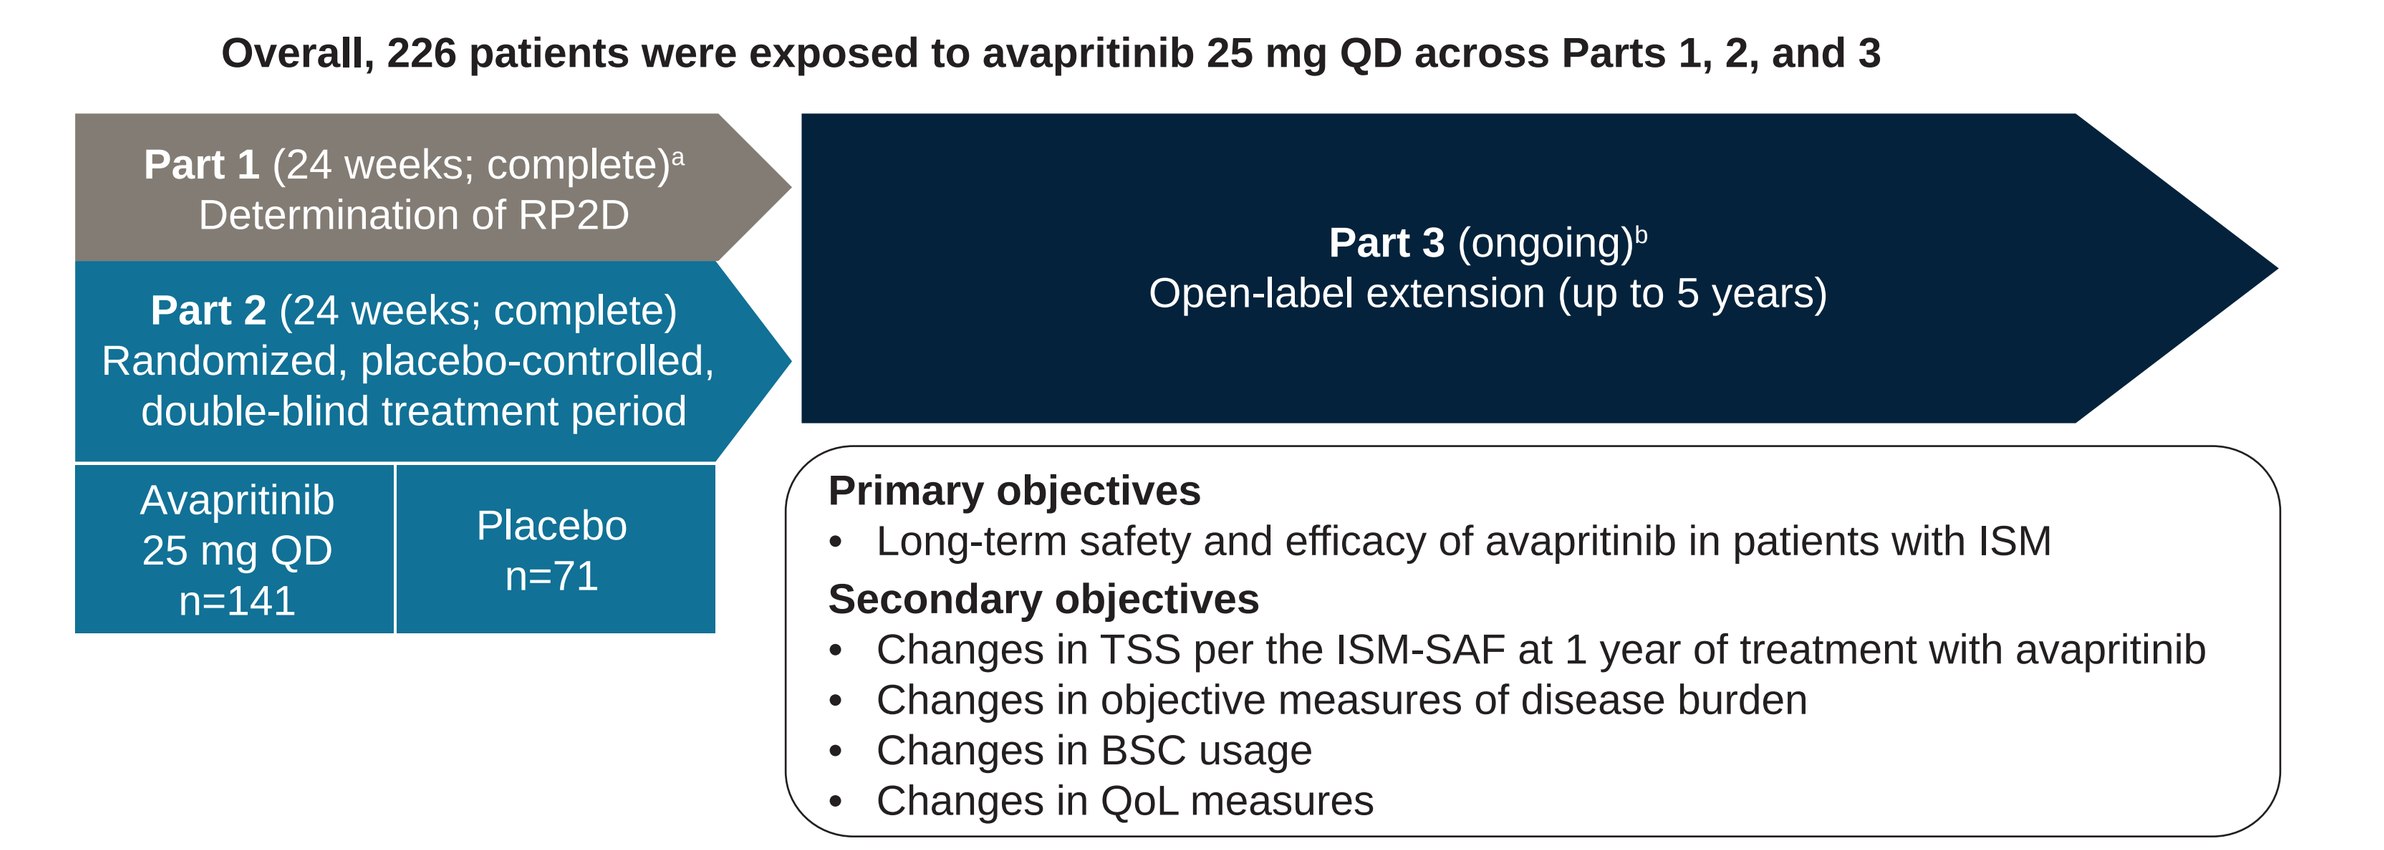
Overall, 226 patients were exposed to avapritinib 25 mg QD across Parts 1, 2, and 3
Part 1 (24 weeks; complete)[a] Determination of RP2D
Part 2 (24 weeks; complete) Randomized, placebo-controlled, double-blind treatment period
Avapritinib 25 mg QD n=141
Placebo n=71
Part 3 (ongoing)[b] Open-label extension (up to 5 years)
Primary objectives
• Long-term safety and efficacy of avapritinib in patients with ISM
Secondary objectives
• Changes in TSS per the ISM-SAF at 1 year of treatment with avapritinib
• Changes in objective measures of disease burden
• Changes in BSC usage
• Changes in QoL measures

[a]The recommended Part 2 dose of avapritinib was identified based on efficacy and safety results from Part 1 that included four blinded, randomized cohorts: 25 mg avapritinib (n=10), 50 mg avapritinib (n=10), 100 mg avapritinib (n=10), and placebo (n=9). [b]Part 3 includes 135 patients who received avapritinib in Part 2 and 66 patients who received placebo in Part 2, as well as patients from Part 1.
BSC, best supportive care; ISM, indolent systemic mastocytosis; ISM-SAF, Indolent Systemic Mastocytosis Symptom Assessment Form; QD, once daily; QoL, quality of life; R2PD, recommended Phase 2 dose; TSS, total symptom score.

- The ISM Symptom Assessment Form (ISM-SAF[a]) is a validated symptom assessment tool specifically developed for evaluation of ISM symptomology[14–16]
 - Total symptom score (TSS) is based on the severity of 11 ISM symptoms
 - The ISM-SAF was developed over the past 8 years with input from patients, disease experts, and global regulatory agencies[15]
- Changes in BSC usage (overall and by drug class) for the management of ISM symptoms were assessed
- Part 2 data are presented at a cut-off of June 23, 2022. Part 3 data at Week 48 are presented at a cut-off of April 7, 2023

[a]ISM-SAF © 2018 Blueprint Medicines Corporation.

Results

- In Part 2, baseline characteristics and demographics were balanced between the avapritinib and placebo groups (**Table 1**)
- BSC use at baseline for all patients who received avapritinib 25 mg QD during the study are shown in **Table 2**

Table 1. Baseline characteristics

Patient demographic	Randomized-controlled Part 2: Avapritinib 25 mg QD (n=141)	Placebo (n=71)
Age (years), median (range)	50.0 (18–77)	54.0 (26–79)
Female, n (%)	100 (71)	54 (76)
ISM symptom burden		
TSS, mean (SD)	50.2 (19.1)	52.4 (19.8)
Most severe symptom score, mean (SD)	7.7 (1.7)	7.9 (1.7)
Mast cell burden		
Median serum tryptase (central), ng/mL (range)	38.4 (3.6–256.0)	43.7 (5.7–501.6)
Median bone marrow biopsy mast cells (central), % (range)	7.0 (1.0–50.0)	7.0 (1.0–70.0)
Mast cell aggregates present, n (%)	106 (75)	57 (80)
Median *KIT* D816V VAF in peripheral blood, % (range)[a]	0.4 (0.00–41.3)	0.3 (0.00–36.7)
KIT D816V positivity, n (%)	131 (93)	69 (97)
SM therapy		
Prior cytoreductive therapy, n (%)	19 (13)	7 (10)
Prior TKI therapy	10 (7)	4 (6)

[a]By digital droplet polymerase chain reaction; limit of detection 0.02%.
SD, standard deviation; SM, systemic mastocytosis; TKI, tyrosine kinase inhibitor; VAF, variant allele fraction.

Table 2. BSC usage at baseline

	Randomized-controlled Part 2: Avapritinib 25 mg QD (n=141)	Placebo (n=71)
Number of BSC treatments, median (range)	3 (0–11)	4 (1–8)
BSC use at baseline, n (%)	140 (99)	71 (100)
H1 antihistamines	137 (97)	71 (100)
H2 antihistamines	93 (66)	47 (66)
Leukotriene inhibitors	49 (35)	25 (35)
Cromolyn sodium	43 (30)	25 (35)
Proton pump inhibitors	22 (16)	20 (28)
Corticosteroids	17 (12)	7 (10)
Anti-IgE antibody	14 (10)	7 (10)
Other[a]	33 (23)	19 (27)

[a]Other includes acetaminophen, acetylsalicylic acid, duloxetine, gabapentin, carbasalate, sumatriptan, topical/inhaled steroids, nasal decongestants, bronchodilators, bisphosphonates, antiemetics, antacids, and vitamin/mineral supplements.
IgE, immunoglobulin E.

- At baseline, the majority of patients in the PIONEER trial were using three or more BSC classes (**Figure 2**)
- The number of BSC classes used at baseline was balanced between avapritinib 25 mg QD and placebo treatment groups (**Figure 2**)

Figure 2. Number of BSC classes used at baseline

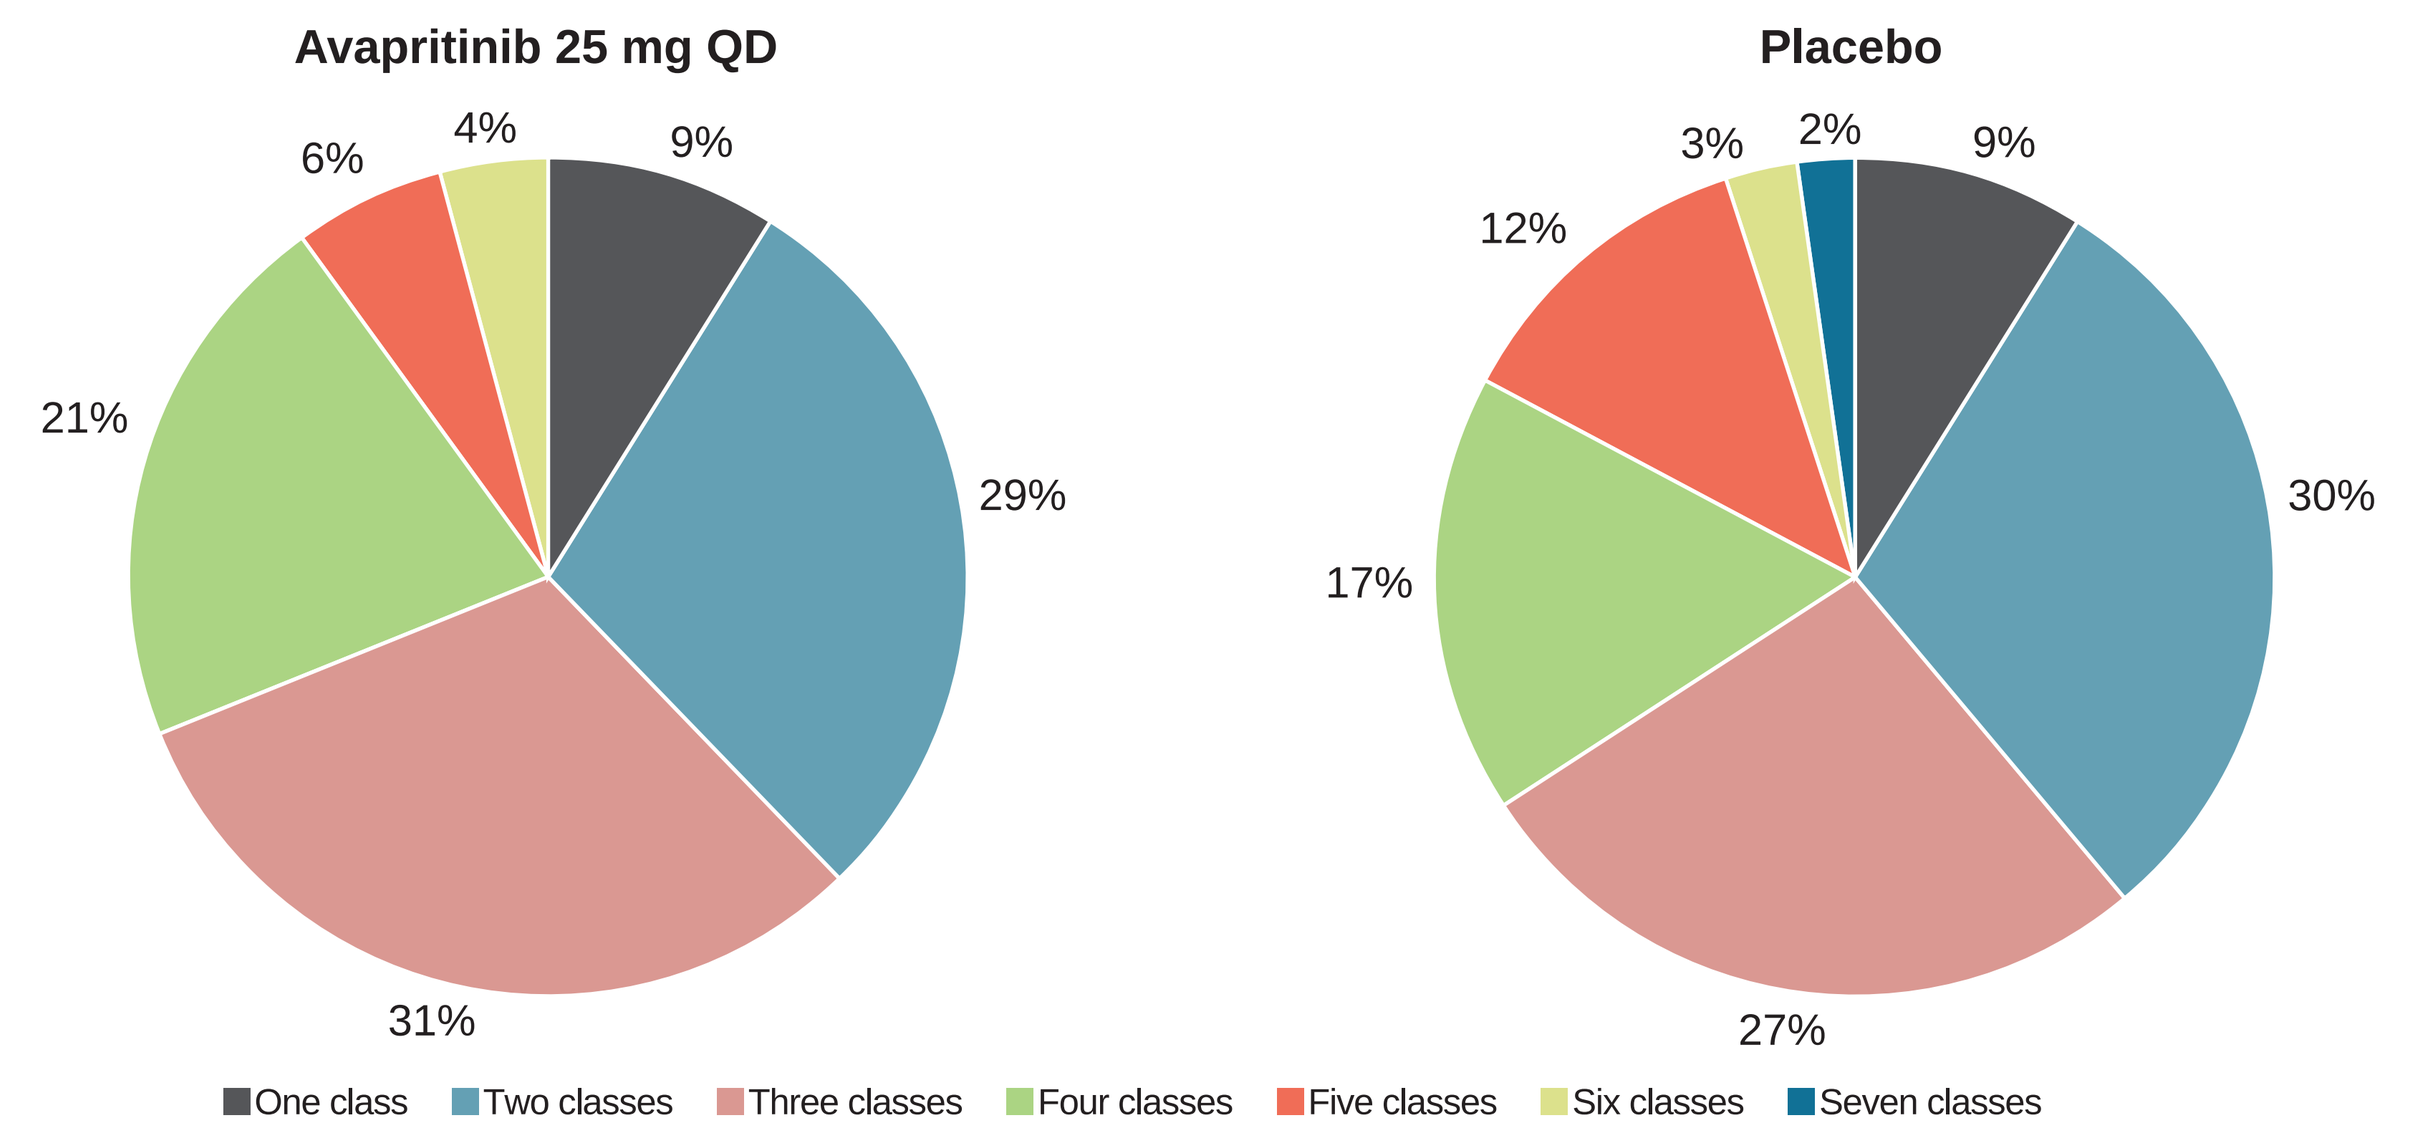
Avapritinib 25 mg QD
9%
29%
31%
21%
6%
4%
Placebo
9%
30%
27%
17%
12%
3%
2%
One class
Two classes
Three classes
Four classes
Five classes
Six classes
Seven classes

Figure 3. Decrease in BSC use at Week 48

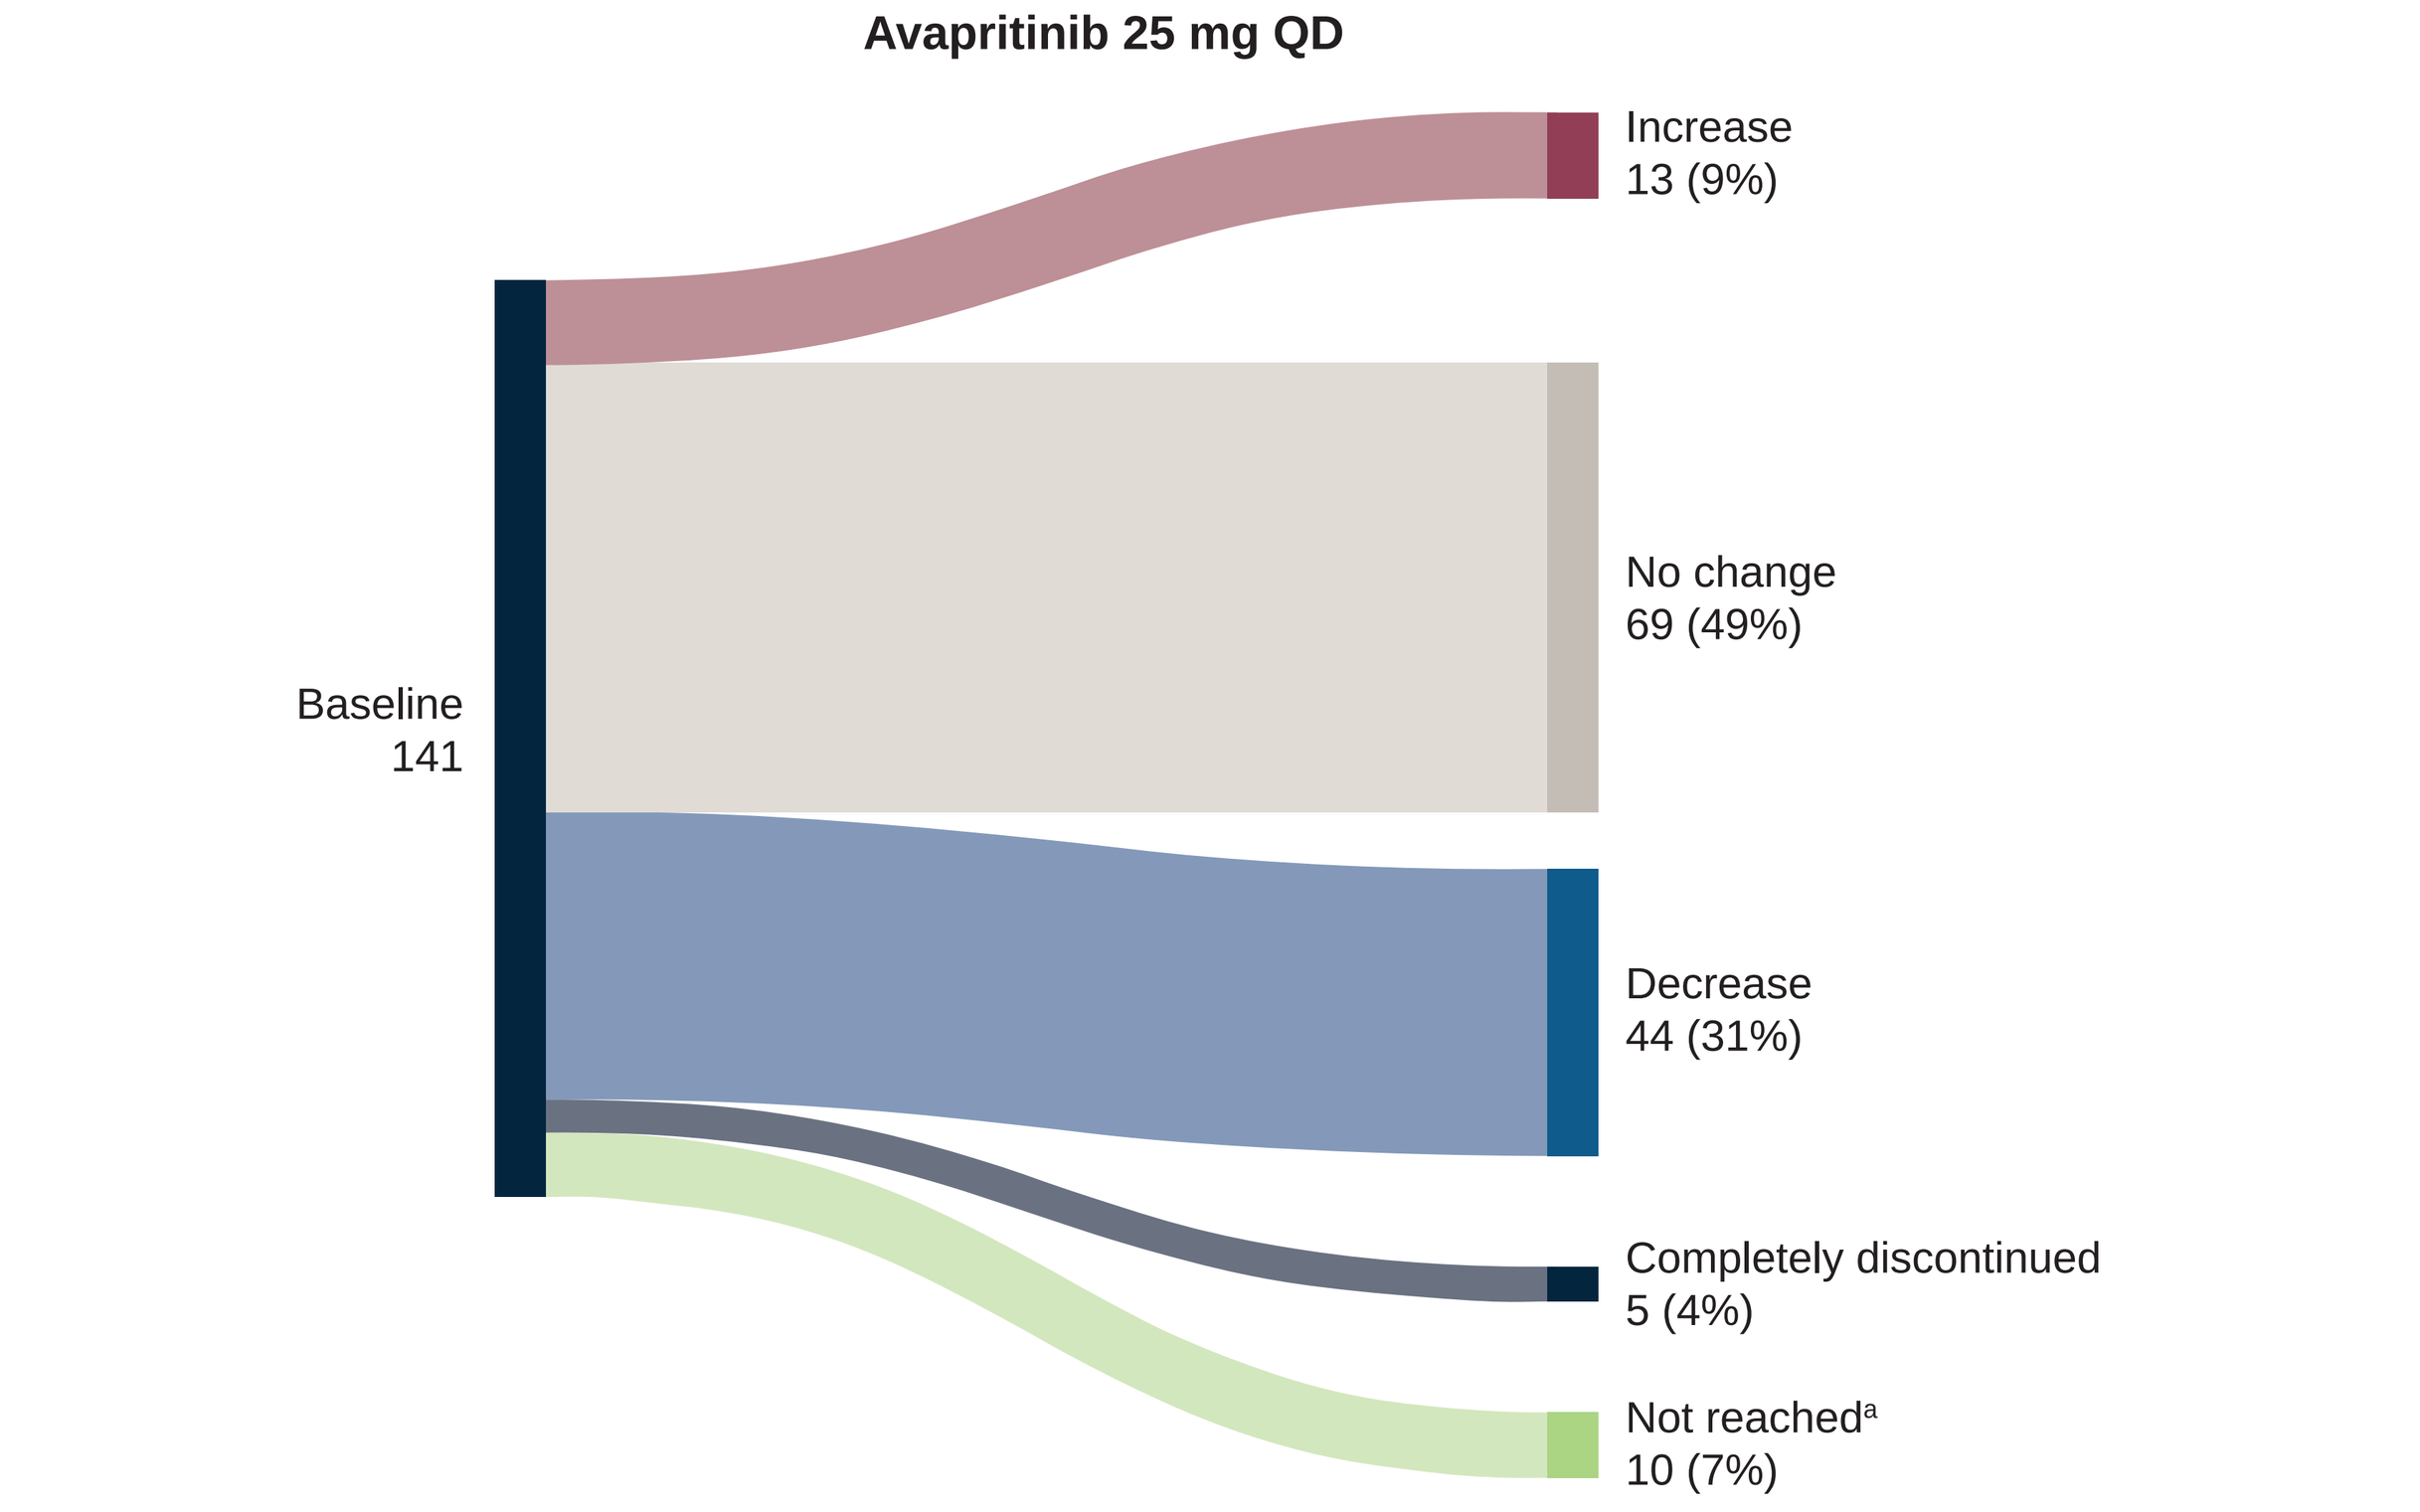
Avapritinib 25 mg QD
Baseline
141
Increase
13 (9%)
No change
69 (49%)
Decrease
44 (31%)
Completely discontinued
5 (4%)
Not reached[a]
10 (7%)

[a]Includes patients who had not yet reached 48 weeks of treatment or had discontinued treatment.

- By Week 24, in patients treated with avapritinib 25 mg QD, 21% (30/141) had decreased BSC *versus* 13% (9/71) of patients treated with placebo
 - With avapritinib treatment, 3% (4/141) were able to completely discontinue BSC *versus* 0% treated with placebo
- After 48 weeks of avapritinib 25 mg QD treatment, 31% (44/141) of patients had decreased BSC and 4% (5/141) were able to completely discontinue BSC (**Figure 3**)

Figure 4. Decreases in individual BSC classes at Week 48

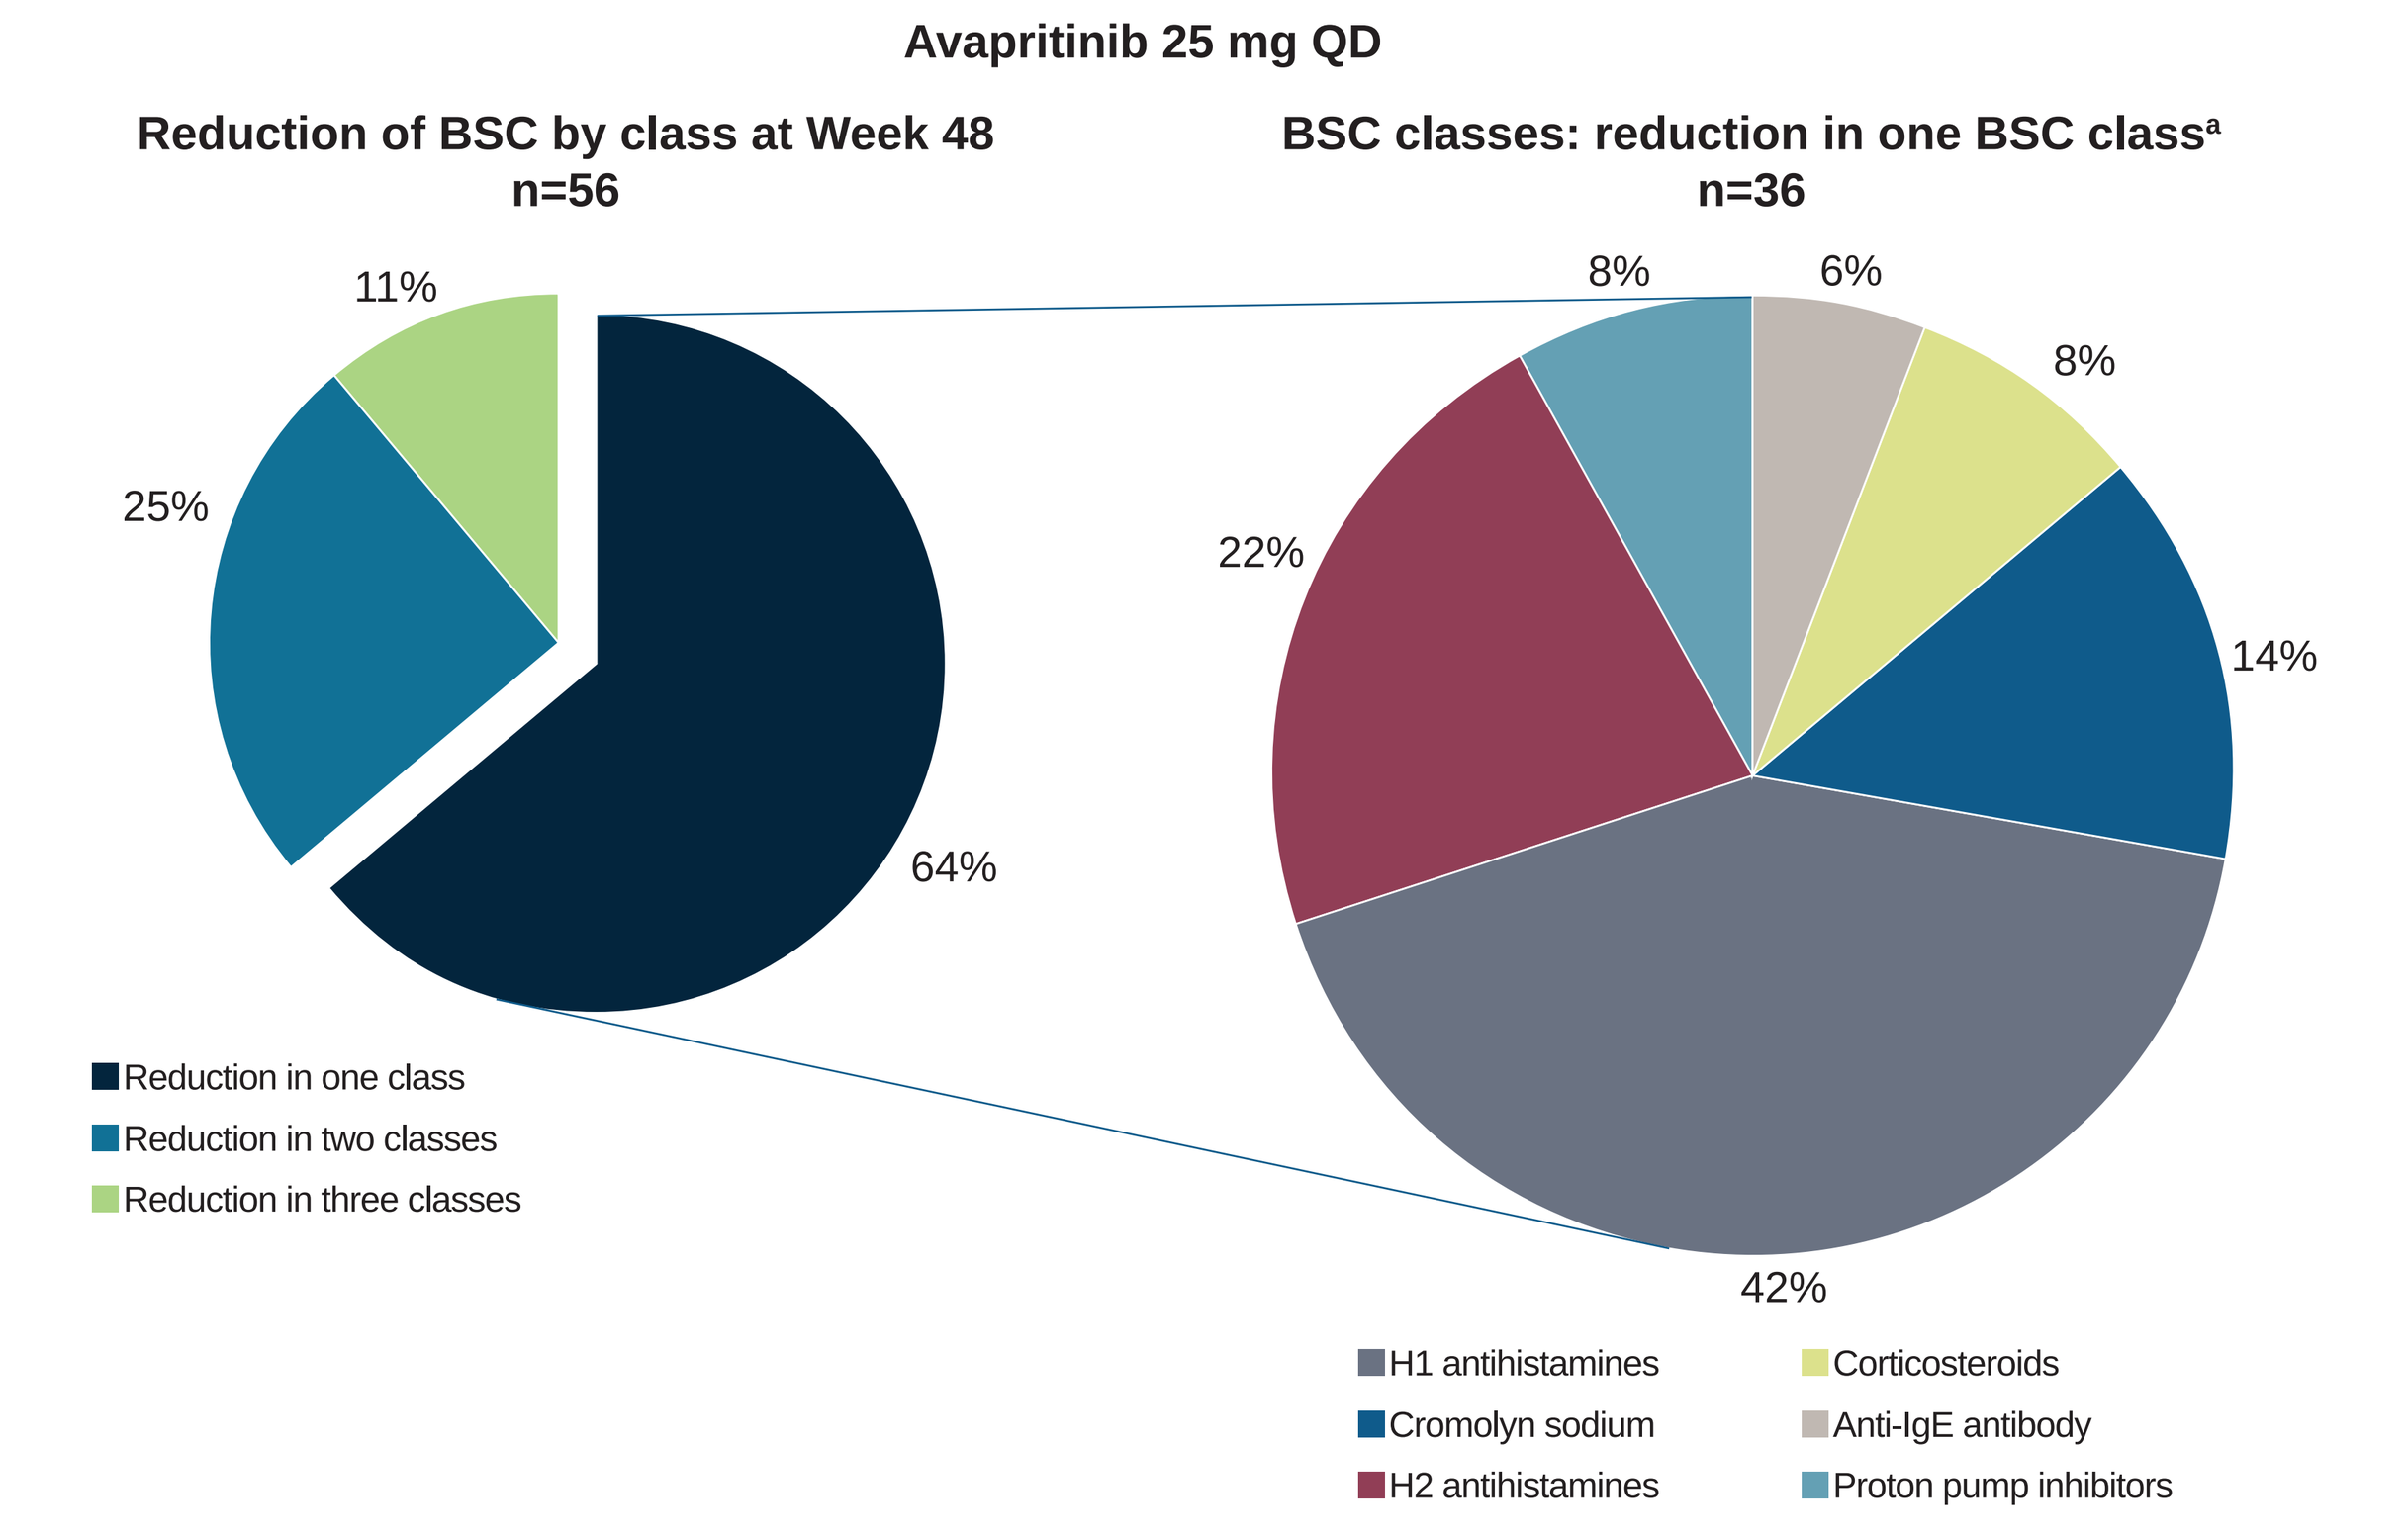
Avapritinib 25 mg QD
Reduction of BSC by class at Week 48
n=56
64%
25%
11%
Reduction in one class
Reduction in two classes
Reduction in three classes
BSC classes: reduction in one BSC class[a]
n=36
42%
22%
14%
8%
8%
6%
H1 antihistamines
Cromolyn sodium
H2 antihistamines
Corticosteroids
Anti-IgE antibody
Proton pump inhibitors

[a]At Week 48, there were no patients reporting one class improvement of leukotriene inhibitors.

- At Week 48, 64% (36/56) of patients experienced a reduction in one class of BSC (**Figure 4, left**)
- Of the patients who experienced a reduction in one class of BSC, the most reductions were seen in H1 antihistamines (42%), H2 antihistamines (22%), and cromolyn sodium (14%) (**Figure 4, right**)
- After 48 weeks of treatment with avapritinib 25 mg QD, the BSC classes with the largest proportion of patients who decreased or discontinued use were cromolyn sodium (17/67; 25%), H1 antihistamines (42/193; 22%), H2 antihistamines (30/139; 22%), and proton pump inhibitors (6/41; 15%)
- Patients who reduced or discontinued cromolyn sodium (n=14) after 48 weeks on avapritinib had significant improvements in their gastrointestinal (GI) symptoms (**Figure 5**)

Figure 5. Reduction in cromolyn sodium use at Week 48 aligns with improvements in GI symptoms

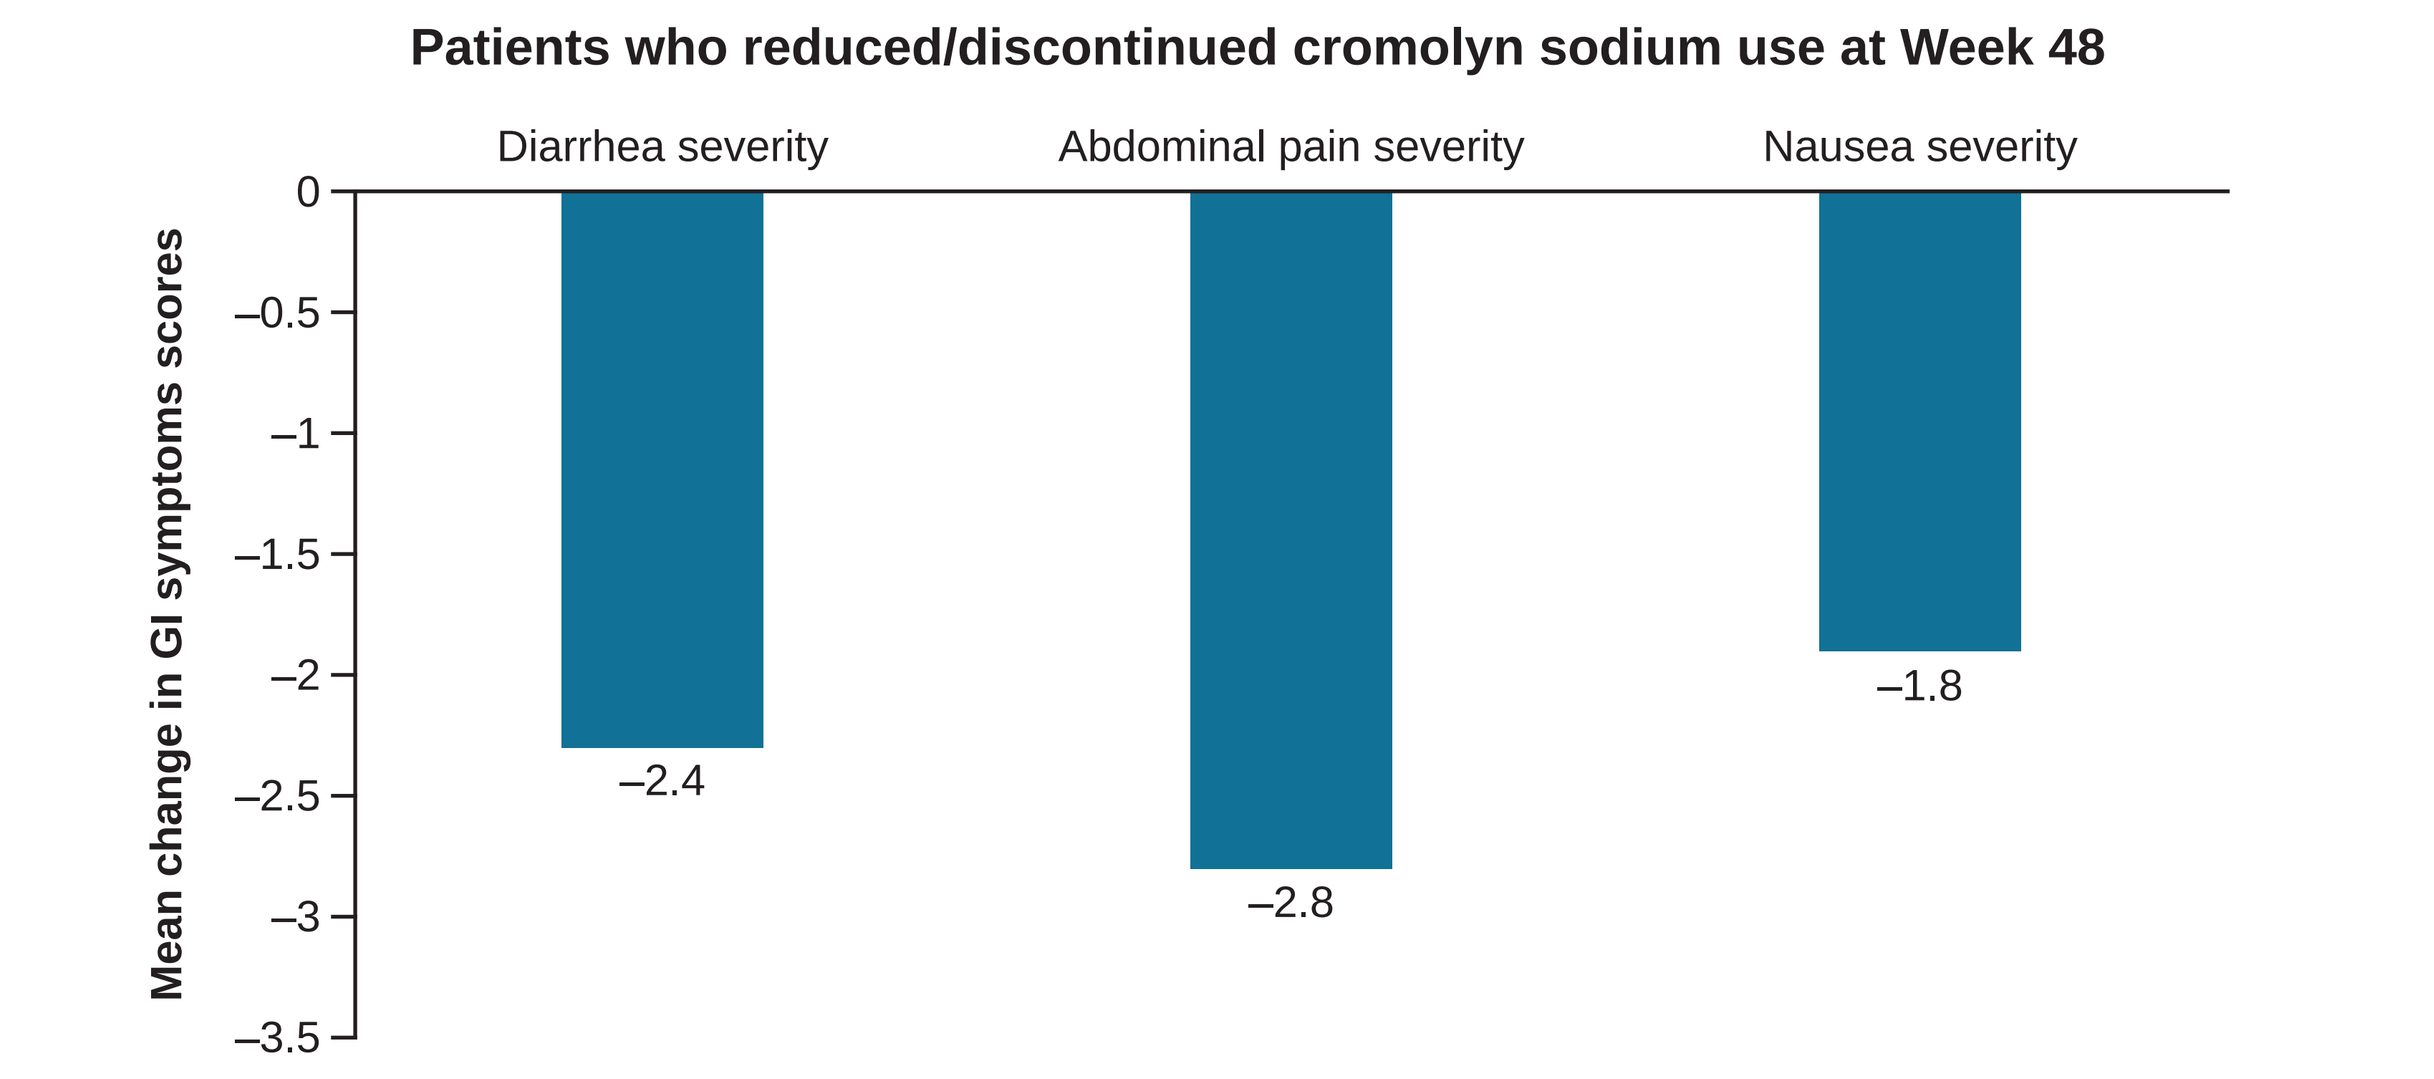
Patients who reduced/discontinued cromolyn sodium use at Week 48
Diarrhea severity
Abdominal pain severity
Nausea severity
Mean change in GI symptoms scores
0
–0.5
–1
–1.5
–2
–2.5
–3
–3.5
–2.4
–2.8
–1.8

GI, gastrointestinal.

Table 3. Summary of AEs

	Randomized-controlled Part 2: Avapritinib 25 mg QD (n=141)	Placebo (n=71)	All patients who received avapritinib 25 mg QD during Parts 1, 2, or 3 (n=226)[a]
Any AEs, n (%)	128 (91)[b]	66 (93)[b]	223 (99)
Grade ≥3 AEs	30 (21)[b]	15 (21)[b]	86 (38)
Any grade TRAEs, n (%)	77 (55)	32 (45)	151 (67)
Grade ≥3 TRAEs	3 (2)	2 (3)	11 (5)
SAEs, n (%)	7 (5)	8 (11)	31 (14)
Most frequently reported TRAEs (≥5% of patients)			
Headache	11 (8)	7 (10)	20 (9)
Nausea	9 (6)	6 (8)	17 (8)
Peripheral edema	9 (6)	1 (1)	23 (10)
Periorbital edema	9 (6)	2 (3)	18 (8)
Dizziness	4 (3)	5 (7)	9 (4)
TRAEs leading to discontinuation	2 (1)	1 (1)	6 (3)

[a]This includes patients from Part 1 who continued avapritinib 25 mg QD or crossed over from placebo to avapritinib 25 mg QD. This also includes patients from Part 2 who received avapritinib 25 mg QD or who crossed over from placebo to avapritinib 25 mg QD. [b]AEs refer to treatment-emergent AEs, defined as any AE that occurred between day 1 of Part 2 through to a day prior to day 1 of Part 3 if the patient crossed over to Part 3; if the patient did not cross over, then through 30 days after the last dose of study drug.
AEs, adverse events; SAEs, serious adverse events; TRAEs, treatment-related adverse events

Placebo-controlled evaluation of safety

- Avapritinib 25 mg QD was generally well tolerated, with a similar safety profile to placebo during the blinded, randomized Part 2 (median follow-up of 5.5 months; **Table 3**)
- The majority of adverse events (AEs) were Grade 1 or 2 with a low rate of discontinuation
- Serious AEs (SAEs) were reported more frequently in the placebo group (no treatment-related SAEs in either group)
- Edema AEs were higher in the avapritinib group (majority Grade 1), and did not result in discontinuation
- AEs of special interest included intracranial hemorrhage (ICH) and cognitive effects. No ICHs were observed. The rate of cognitive effects in patients treated with avapritinib (3%) and placebo (4%) were similar

Longer-term open-label evaluation of safety

- The Part 3 open-label extension of PIONEER allowed for the assessment of longer-term safety of avapritinib at 25 mg QD in 226 patients (median follow-up of 18 months)
- No new safety concerns were observed with longer follow-up; the most common treatment-related AEs (TRAEs) (≥5% of patients) remained consistent to those reported during Part 2 (**Table 3**)
- No ICHs were observed. The rate of cognitive effects remained low
- The number of TRAEs leading to discontinuation remained low
- Drug interruptions were predominantly for non-TRAEs and other reasons
 - For example, a 33-year-old male interrupted treatment to successfully father a pregnancy and then resumed avapritinib 190 days later

Figure 6. Clinical vignette

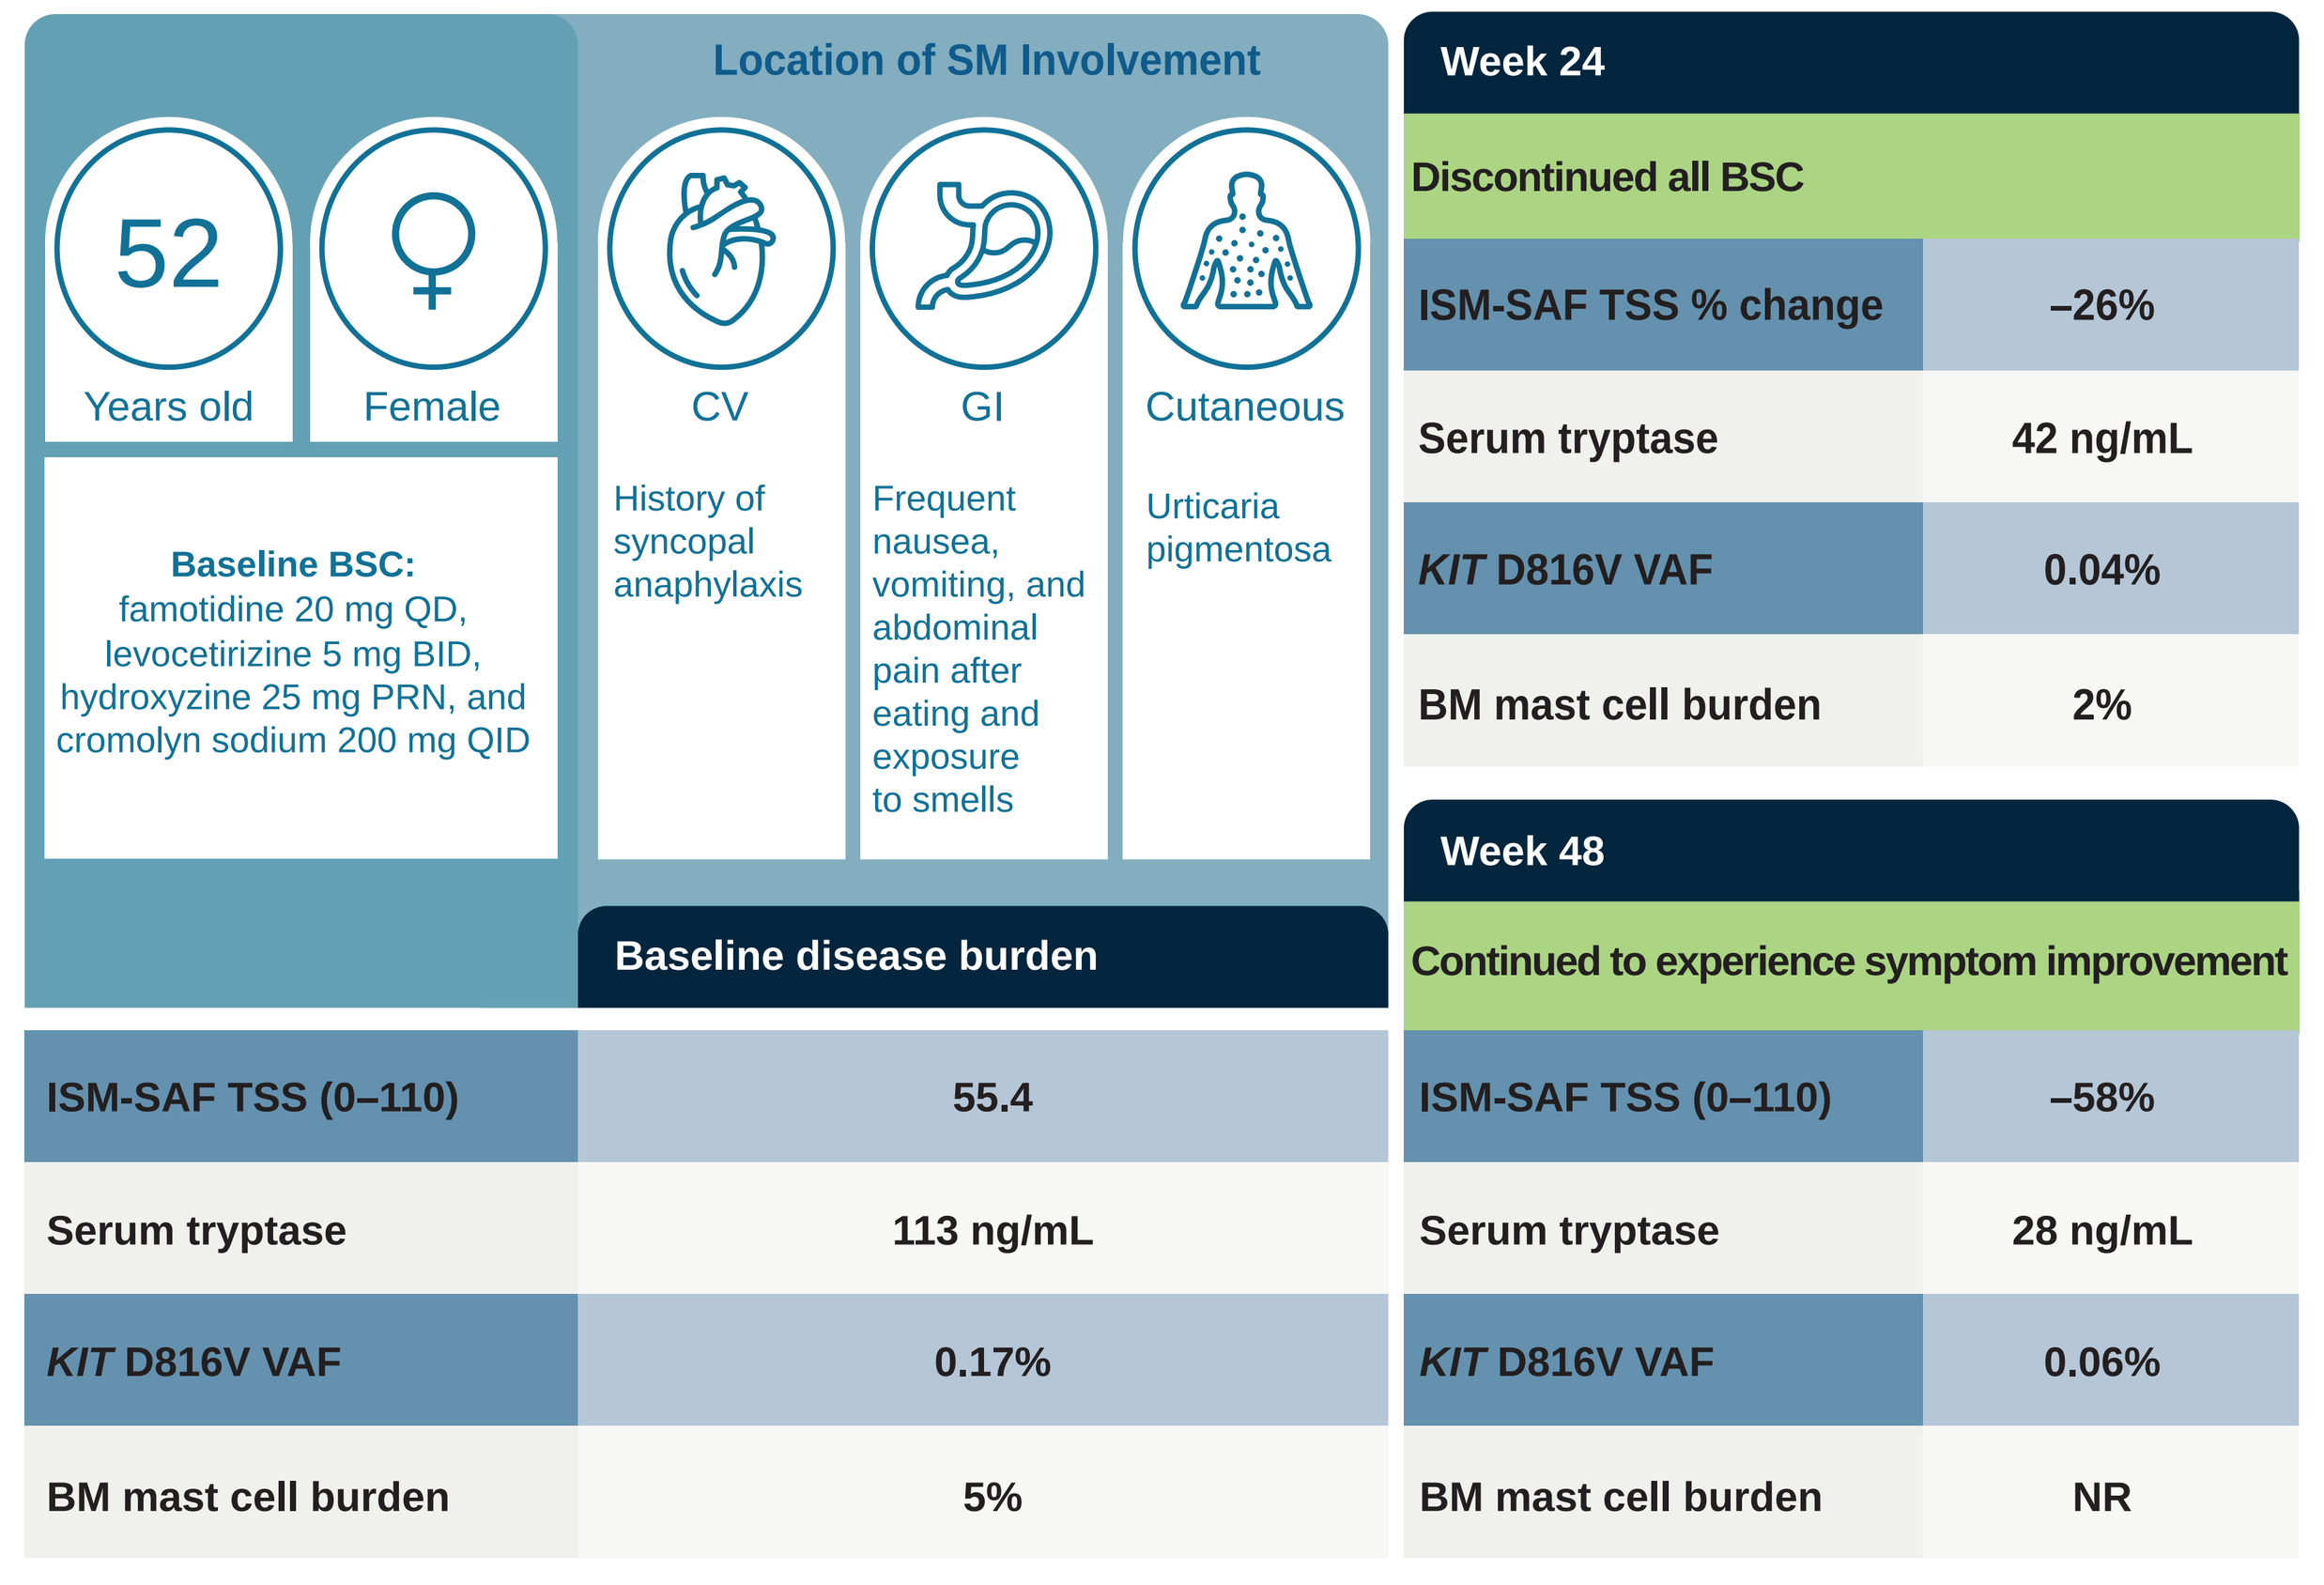
52
Years old
Female
Baseline BSC: famotidine 20 mg QD, levocetirizine 5 mg BID, hydroxyzine 25 mg PRN, and cromolyn sodium 200 mg QID
Location of SM Involvement
CV
History of syncopal anaphylaxis
GI
Frequent nausea, vomiting, and abdominal pain after eating and exposure to smells
Cutaneous
Urticaria pigmentosa
Baseline disease burden
ISM-SAF TSS (0–110) 55.4
Serum tryptase 113 ng/mL
KIT D816V VAF 0.17%
BM mast cell burden 5%
Week 24
Discontinued all BSC
ISM-SAF TSS % change –26%
Serum tryptase 42 ng/mL
KIT D816V VAF 0.04%
BM mast cell burden 2%
Week 48
Continued to experience symptom improvement
ISM-SAF TSS (0–110) –58%
Serum tryptase 28 ng/mL
KIT D816V VAF 0.06%
BM mast cell burden NR

BID, twice daily; BM, bone marrow; CV, cardiovascular; NR, not reported; PRN, as needed; QID, four times daily.

- A 52-year-old female diagnosed with ISM in 2015 had symptoms that severely restricted her QoL (**Figure 6**)
- She was randomized to avapritinib 25 mg QD in Part 2 of the PIONEER study
- After 24 weeks on avapritinib, she experienced improvements in symptoms and measures of disease burden and was able to stop all BSC medications
- After 48 weeks on avapritinib, she continued to experience improvement in symptom and disease burden

Conclusions

- Patients enrolled in PIONEER had moderate to severe disease symptoms as assessed by TSS, despite the majority of patients taking three or more classes of BSC
- Treatment with avapritinib reduced the use of BSC medications in patients with ISM, with further reductions seen with longer-term use at Week 48
- Classes of BSC medications with the greatest reductions in use were cromolyn sodium, proton pump inhibitors, H1 antihistamines, and H2 antihistamines
- Patients who reduced cromolyn sodium use continued to have markedly improved GI symptoms
- Avapritinib was generally well tolerated, with a similar safety profile to placebo and no new safety concerns observed with a median treatment duration of 18 months

References

1. Kristensen T et al. *J Mol Diagn*. 2011;13:180–188; 2. Cohen SS et al. *Br J Haematol*. 2014;166:521–528; 3. Arber DA et al. *Blood*. 2022;140:1200–1228; 4. Mesa RA et al. *Cancer*. 2022;128:3691–3699; 5. Hermine O et al. *PLoS One*. 2008;3:e2266; 6. van Anrooij B et al. *Allergy*. 2016;71:1585–1593; 7. Hartmann K et al. *J Allergy Clin Immunol*. 2016;137:35–45; 8. Akin C et al. *J Allergy Clin Immunol*. 2022;149:1912–1918; 9. Pardanani A. *Blood*. 2013;121:3085–3094; 10. Pardanani A. *Am J Hematol*. 2021;96:508–525; 11. Blueprint Medicines Corporation. AYVAKIT® (avapritinib). Prescribing Information. 2023. https://www.accessdata.fda.gov/drugsatfda_docs/label/2021/212608s007lbl.pdf. Accessed August 6, 2023; 12. Blueprint Medicines Corporation. AYVAKYT® (avapritinib). Summary of Product Characteristics. 2023. https://www.ema.europa.eu/en/documents/product-information/ayvakyt-epar-product-information_en.pdf. Accessed August 6, 2023; 13. Gotlib J et al. *NEJM Evid*. 2023;2:EVIDoa2200339; 14. Shields AL et al. *Orphanet J Rare Dis*. 2023;18:69; 15. Taylor F et al. *Orphanet J Rare Dis*. 2021;16:414; 16. Padilla B et al. *Orphanet J Rare Dis*. 2021;16:434.

Acknowledgements

Medical writing support was provided by Isabella Muñoz, BS, and Travis Taylor, BA, both of Paragon (a division of Prime, Knutsford, UK). Funded by Blueprint Medicines Corporation. The sponsor reviewed and provided feedback on the poster. However, the authors had full editorial control and provided final approval of all content.

Disclosures

Dr Akin has received consulting fees and research support from Blueprint Medicines Corporation and Cogent Biosciences, and consulting fees from Novartis. For all author disclosures, please contact medinfo@blueprintmedicines.com.

Presented at the American Academy of Allergy Asthma & Immunology (AAAAI) February 23–26, 2024, in Washington, DC, USA. Please contact medinfo@blueprintmedicines.com for permission to reprint and/or distribute.