Efficacy and Safety of Avapritinib in Indolent Systemic Mastocytosis: Results From the Double Blinded Placebo-Controlled PIONEER Study

Poster Number S7

Poster available for download at:

Mariana Castells, MD, PhD,[1*] Jason Gotlib, MD, MS,[2*] Matthew Giannetti, MD,[3] Ilda Bidollari MD, MBA,[4] Hui-Min Lin, PhD,[4] Robyn Scherber, MD,[4] Maria Roche, MS,[4] Cem Akin, MD, PhD,[5], and Marcus Maurer, MD,[6,7**]**

[1]Brigham and Women's Hospital, Harvard Medical School, Boston, MA, USA; [2]Stanford Cancer Institute/Stanford University School of Medicine, Stanford, CA, USA; [3]Division of Allergy and Clinical Immunology, Brigham and Women's Hospital, Boston, MA, USA; [4]Blueprint Medicines Corporation, Cambridge, MA, USA; [5]University of Michigan, Ann Arbor, MI, USA; [6]Institute of Allergology, Charité–Universitätsmedizin Berlin, Corporate member of Freie Universität Berlin and Humboldt-Universität zu Berlin, Berlin, Germany; [7]Fraunhofer Institute for Translational Medicine and Pharmacology (ITMP), Allergology and Immunology, Berlin, Germany

**Equally contributing first authors; **Equally contributing last authors*

Background

- Indolent systemic mastocytosis (ISM) is a clonal mast-cell disease driven by the *KIT* D816V mutation in ~95% of adult cases[1–3]
- Patients with ISM often experience life-long debilitating skin, gastrointestinal, neurocognitive, musculoskeletal, and systemic manifestations, including anaphylaxis (**Figure 1**). These symptoms commonly result in impaired daily functioning and poor quality of life (QoL)[4–9]
- Most patients rely on polypharmacy for the management of symptoms with best supportive care (BSC) medications; however, many patients' symptoms are not adequately controlled with these medications and until recently there were no approved therapies that target the KIT D816V-mutated tyrosine kinase in ISM[5,10,11]
- Avapritinib is a potent and highly selective oral therapy targeting KIT D816V, the underlying driver of systemic mastocytosis (**Figure 2**)[12,13]
 - Avapritinib demonstrated reductions in mast-cell burden biomarkers and an improvement in all advanced systemic mastocytosis (AdvSM) symptoms, including skin manifestations and QoL measurements[12–14]
 - In addition, avapritinib demonstrated significantly better survival benefit *versus* best available therapy (including midostaurin and cladribine) in a retrospective real-world analysis[15,16]
- Avapritinib is approved in the USA for adult patients with AdvSM and in Europe for adult patients with AdvSM after ≥1 prior systemic therapy; avapritinib is not recommended for patients with platelet counts <50x10[9]/L[17,18]
- Avapritinib is approved in the USA for adult patients with ISM; avapritinib is not recommended for patients with platelet counts <50x10[9]/L[17]
- PIONEER (NCT03731260), a randomized, double-blind, placebo-controlled trial, evaluated avapritinib 25 mg once daily (QD) plus BSC (avapritinib) compared to placebo plus BSC (placebo) in patients with ISM with uncontrolled moderate to severe symptoms. Primary and secondary endpoints are summarized in **Figure 3**
- Here, we present key efficacy and safety data from Part 2 of the PIONEER trial

Figure 1: ISM symptoms[4–9] assessed by the validated ISM-SAF[a]

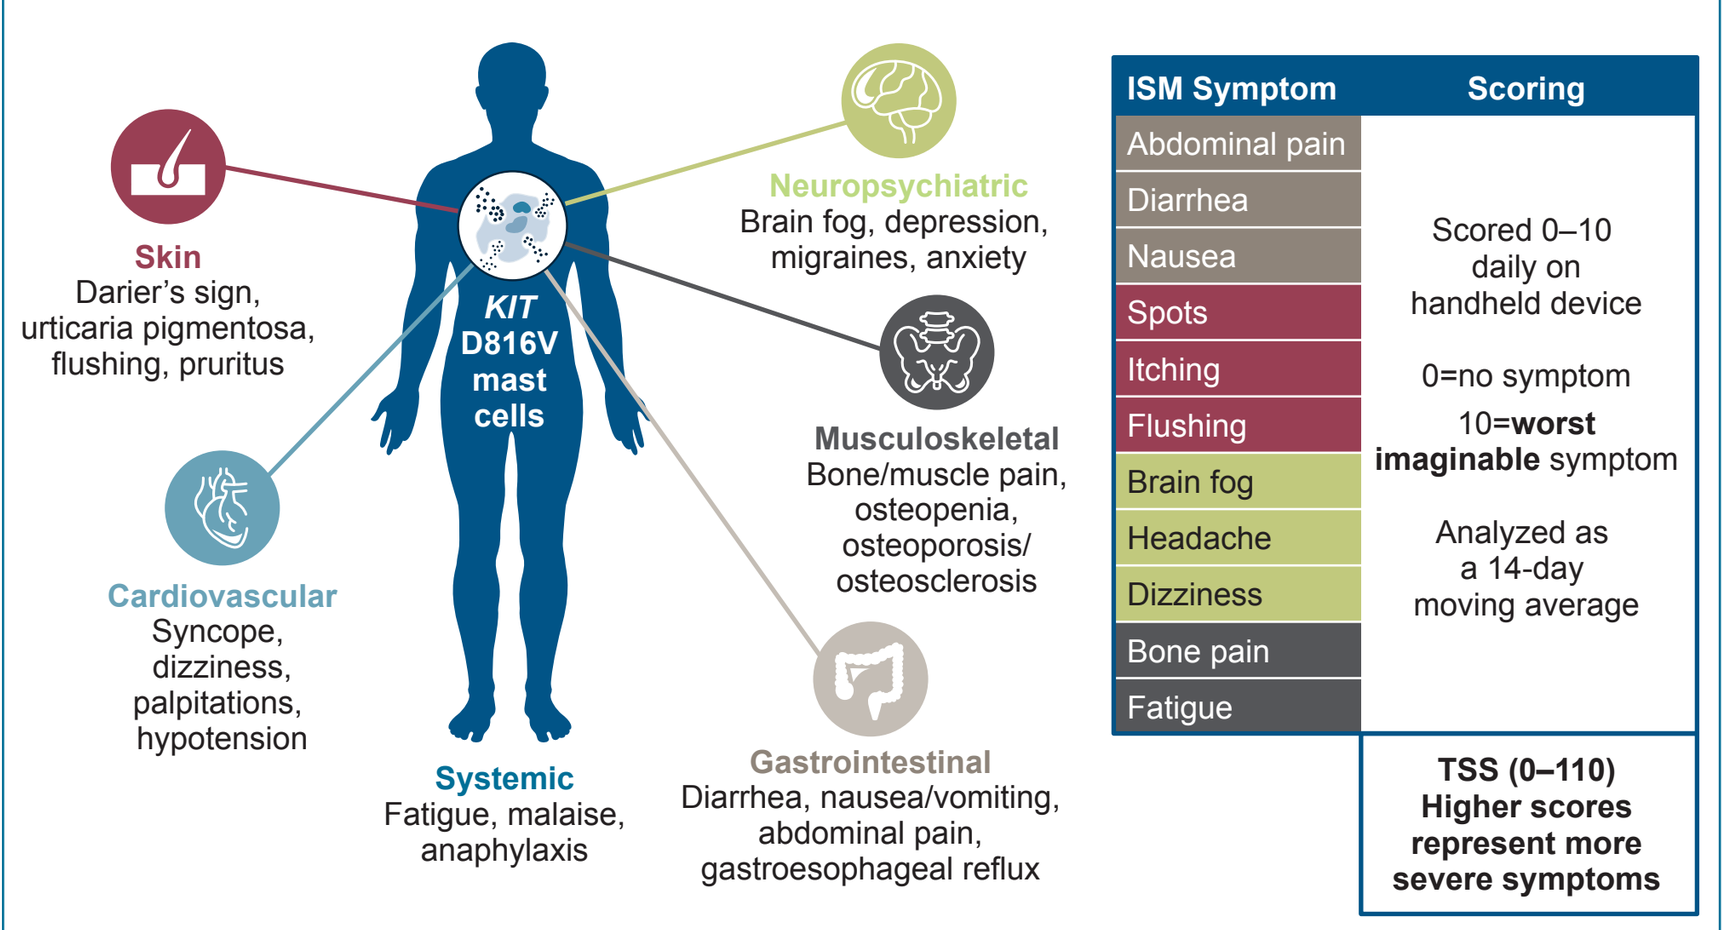

Skin
Darier's sign, urticaria pigmentosa, flushing, pruritus
Neuropsychiatric
Brain fog, depression, migraines, anxiety
KIT D816V mast cells
Musculoskeletal
Bone/muscle pain, osteopenia, osteoporosis/ osteosclerosis
Cardiovascular
Syncope, dizziness, palpitations, hypotension
Systemic
Fatigue, malaise, anaphylaxis
Gastrointestinal
Diarrhea, nausea/vomiting, abdominal pain, gastroesophageal reflux
ISM Symptom
Scoring
Abdominal pain
Diarrhea
Nausea
Spots
Itching
Flushing
Brain fog
Headache
Dizziness
Bone pain
Fatigue
Scored 0–10 daily on handheld device
0=no symptom
10=worst imaginable symptom
Analyzed as a 14-day moving average
TSS (0–110) Higher scores represent more severe symptoms

[a]ISM-SAF is a validated symptom assessment tool specifically developed for evaluation of ISM symptomology.[19–21]
ISM, indolent systemic mastocytosis; ISM-SAF, indolent systemic mastocytosis Symptom Assessment Form; TSS, total symptom score.

Figure 2: Avapritinib kinase inhibitor activity

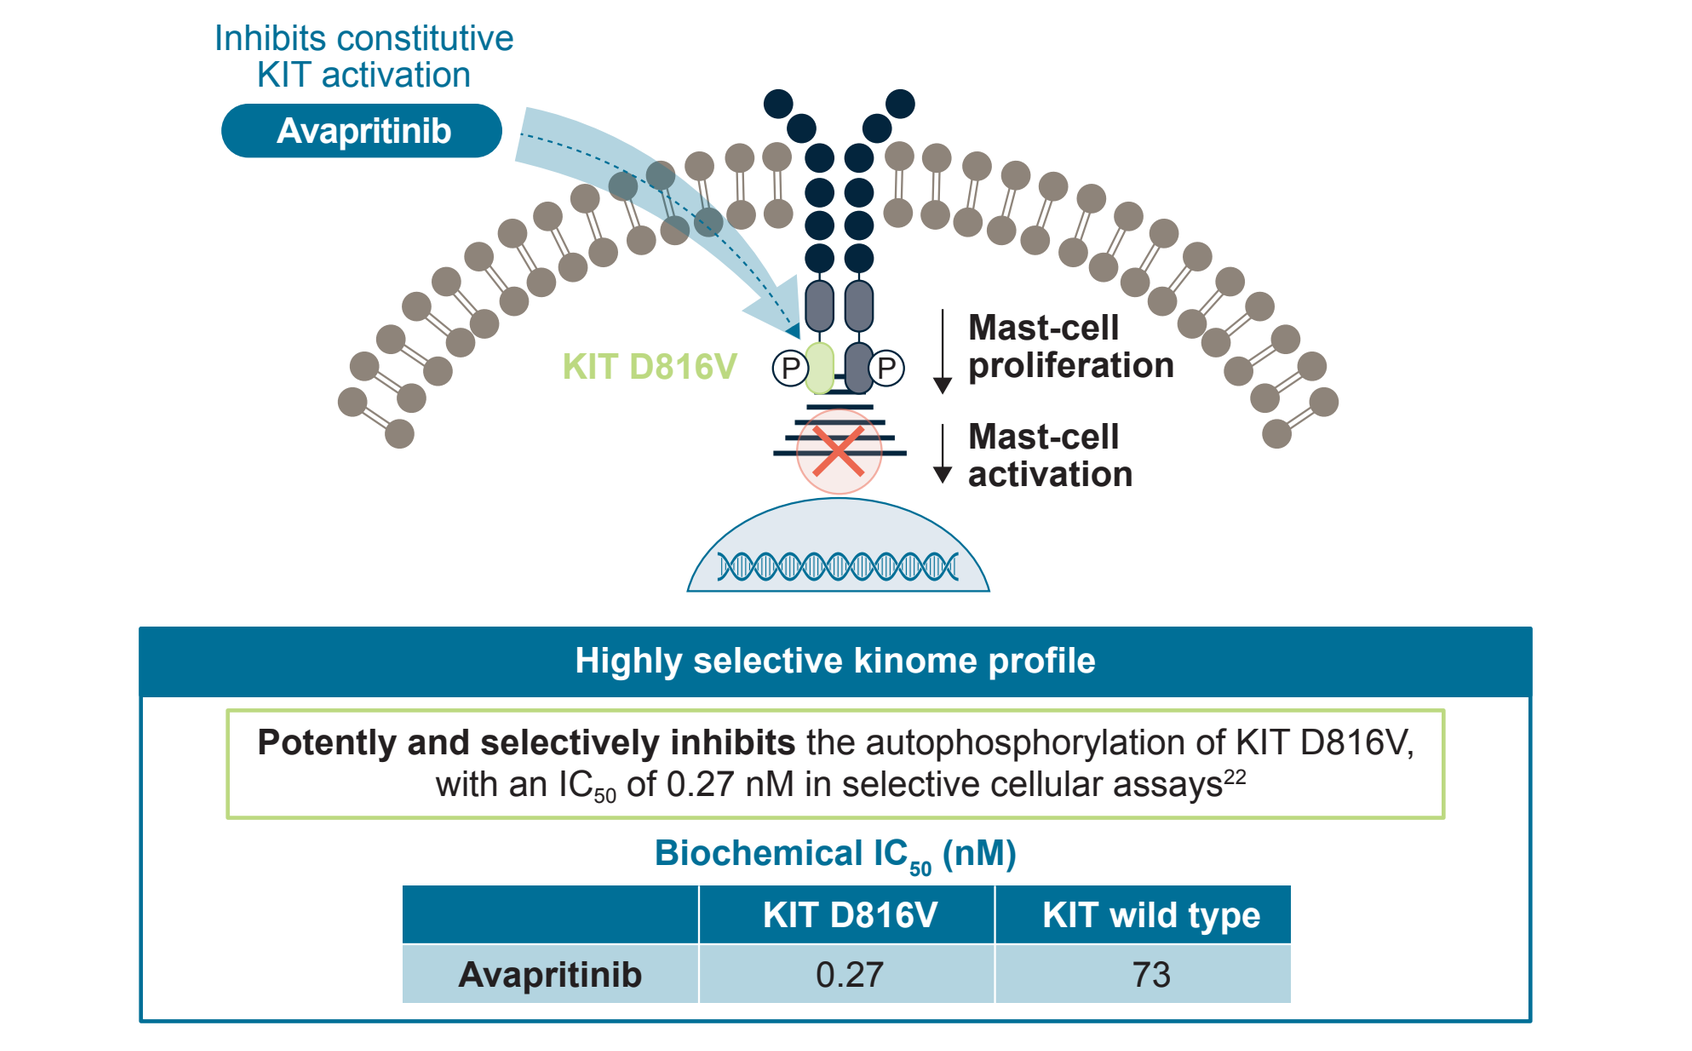

Inhibits constitutive KIT activation
Avapritinib
KIT D816V
Mast-cell proliferation
Mast-cell activation
Highly selective kinome profile
Potently and selectively inhibits the autophosphorylation of KIT D816V, with an IC50 of 0.27 nM in selective cellular assays[22]
Biochemical IC50 (nM)
KIT D816V
KIT wild type
Avapritinib
0.27
73

IC_{50}, half maximal inhibitory concentration; nM, nanomolar.

Methods

Figure 3: Study design

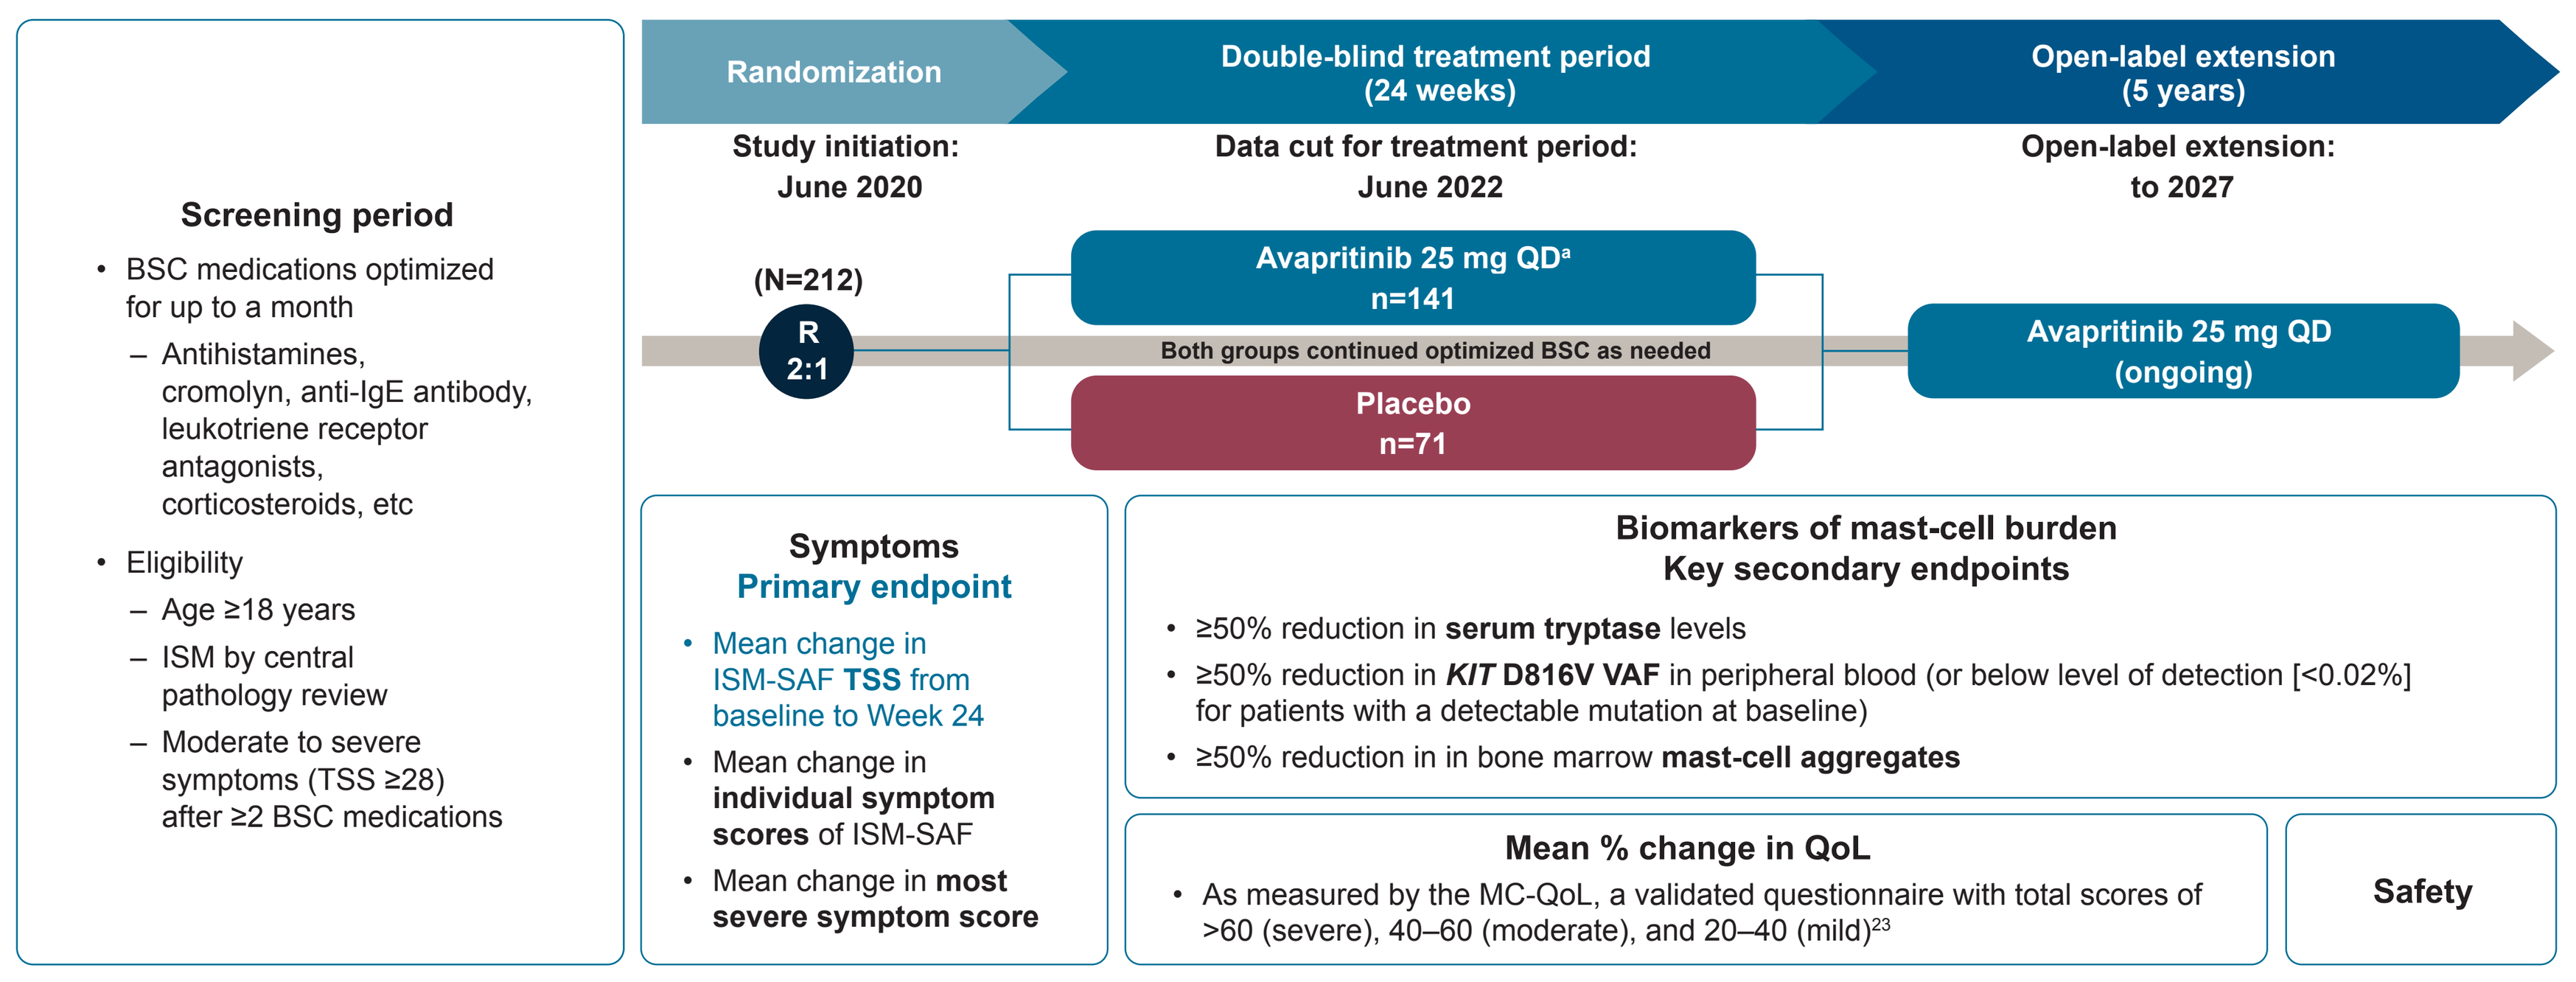

Screening period
BSC medications optimized for up to a month
– Antihistamines, cromolyn, anti-IgE antibody, leukotriene receptor antagonists, corticosteroids, etc
Eligibility
– Age ≥18 years
– ISM by central pathology review
– Moderate to severe symptoms (TSS ≥28) after ≥2 BSC medications
Randomization
Double-blind treatment period (24 weeks)
Open-label extension (5 years)
Study initiation: June 2020
Data cut for treatment period: June 2022
Open-label extension: to 2027
(N=212)
R 2:1
Avapritinib 25 mg QD[a] n=141
Both groups continued optimized BSC as needed
Placebo n=71
Avapritinib 25 mg QD (ongoing)
Symptoms
Primary endpoint
Mean change in ISM-SAF TSS from baseline to Week 24
Mean change in individual symptom scores of ISM-SAF
Mean change in most severe symptom score
Biomarkers of mast-cell burden
Key secondary endpoints
≥50% reduction in serum tryptase levels
≥50% reduction in KIT D816V VAF in peripheral blood (or below level of detection [<0.02%] for patients with a detectable mutation at baseline)
≥50% reduction in in bone marrow mast-cell aggregates
Mean % change in QoL
As measured by the MC-QoL, a validated questionnaire with total scores of >60 (severe), 40–60 (moderate), and 20–40 (mild)[23]
Safety

[a]The recommended dose of avapritinib for the double-blind period and open-label extension was identified based on efficacy and safety results from Part 1 that included four cohorts: 25 mg avapritinib (n=10), 50 mg avapritinib (n=10), 100 mg avapritinib (n=10) and placebo (n=9). Patients treated with high dose steroids within 7 days of primary endpoint (n=4) were excluded from the Week 24 analysis but included at other timepoints of the study. Percentages were calculated based on available data at the timepoint. One-sided P-values are reported for primary and key secondary endpoints. BSC, best supportive care; IgE, immunoglobulin E; MC-QoL, Mastocytosis Quality of Life Questionnaire; QD, once daily; QoL, quality of life; R, randomized; VAF, variant allele fraction.

Results

- Baseline characteristics are described in **Table 1**
- More than 70% (n=167) of PIONEER patients had skin involvement at baseline; the patient population with skin biopsies at baseline had similar baseline characteristics to the intent-to-treat population (**data not shown**)

Improvements in TSS

- Avapritinib demonstrated significantly greater improvement in mean TSS (95% confidence interval [CI]) at 24 weeks *versus* placebo (–15.6 [–18.6 to –12.6] *vs* –9.2 [–13.1 to –5.2]; *P*=0.003) (**Figure 4**)
- A total of 66 patients crossed over from placebo to avapritinib in Part 3 and improvements in TSS were observed; in the 26 patients who completed an additional 24 weeks of treatment, the mean (95% CI) change in TSS was –21.4 (–28.0 to –14.9) at 48 weeks

Improvements in symptoms

- At 24 weeks, avapritinib demonstrated improvement in all individual ISM symptoms *versus* placebo, including the most severe symptom at baseline (**Figure 5**)
 - Regardless of which symptom was rated most severe at baseline, avapritinib patients had a notable reduction in this *versus* placebo
- In the majority of patients, the most severe symptom domain at baseline was the skin domain
 - At 24 weeks, in patients with skin disease, mean standard deviation (SD) change in skin domain was greater with avapritinib (n=107; −7.2 [6.4]) *vs* placebo (n=60; −2.8 [4.2]), *P*<0.0001; avapritinib *versus* placebo also improved spots (–2.3 [2.1] *vs* –0.6 [1.5]; *P*<0.0001), itching (–2.6 [2.8] *vs* –1.2 [2.0]; *P*<0.001), and flushing (–2.2 [2.6] *vs* –1.0 [1.9]; *P*<0.001)

Figure 4: Avapritinib demonstrated significant and durable improvement in symptoms *versus* placebo

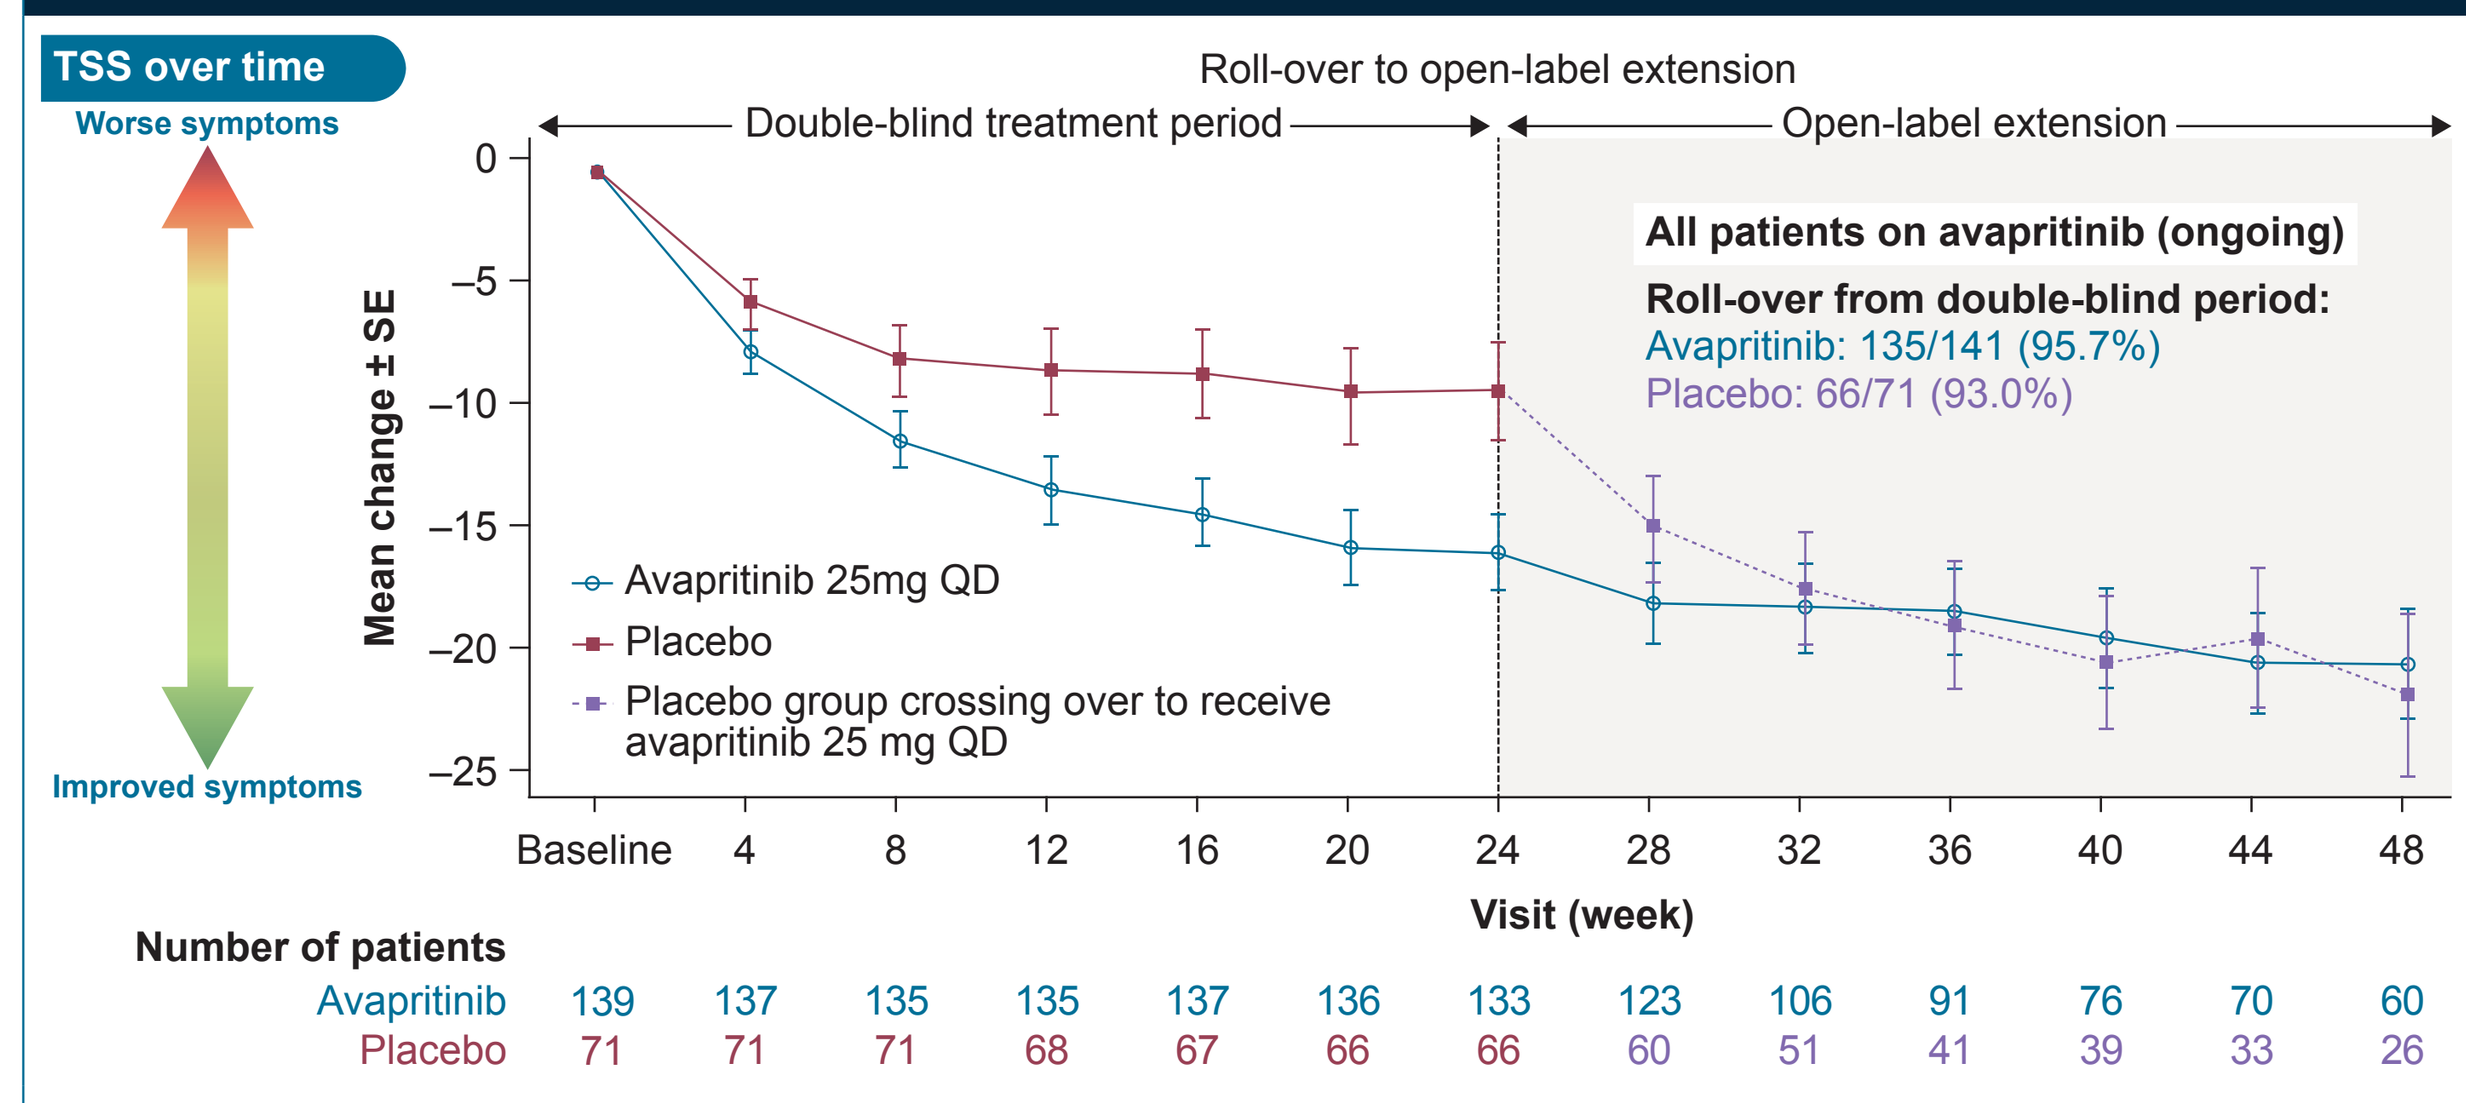

TSS over time
Worse symptoms
Improved symptoms
Roll-over to open-label extension
Double-blind treatment period
Open-label extension
All patients on avapritinib (ongoing)
Roll-over from double-blind period:
Avapritinib: 135/141 (95.7%)
Placebo: 66/71 (93.0%)
Mean change ± SE
Avapritinib 25mg QD
Placebo
Placebo group crossing over to receive avapritinib 25 mg QD
Baseline 4 8 12 16 20 24 28 32 36 40 44 48
Visit (week)
Number of patients
Avapritinib 139 137 135 135 137 136 133 123 106 91 76 70 60
Placebo 71 71 71 68 67 66 66 60 51 41 39 33 26

Primary endpoint

A one-sided *P*-value of <0.025 was needed to declare avapritinib as superior in reducing TSS *versus* placebo.

At Week 24	Avapritinib 25 mg QD (n=141)	Placebo (n=71)	*P*-value
Mean change in TSS (95% CI)	–15.58 (–18.61 to –12.55)	–9.15 (–13.12 to –5.18)	0.003

CI, confidence interval; SE, standard error of the mean.

Figure 5: Avapritinib improved all individual ISM symptoms *versus* placebo at 24 weeks and demonstrated continued improvement up to 48 weeks

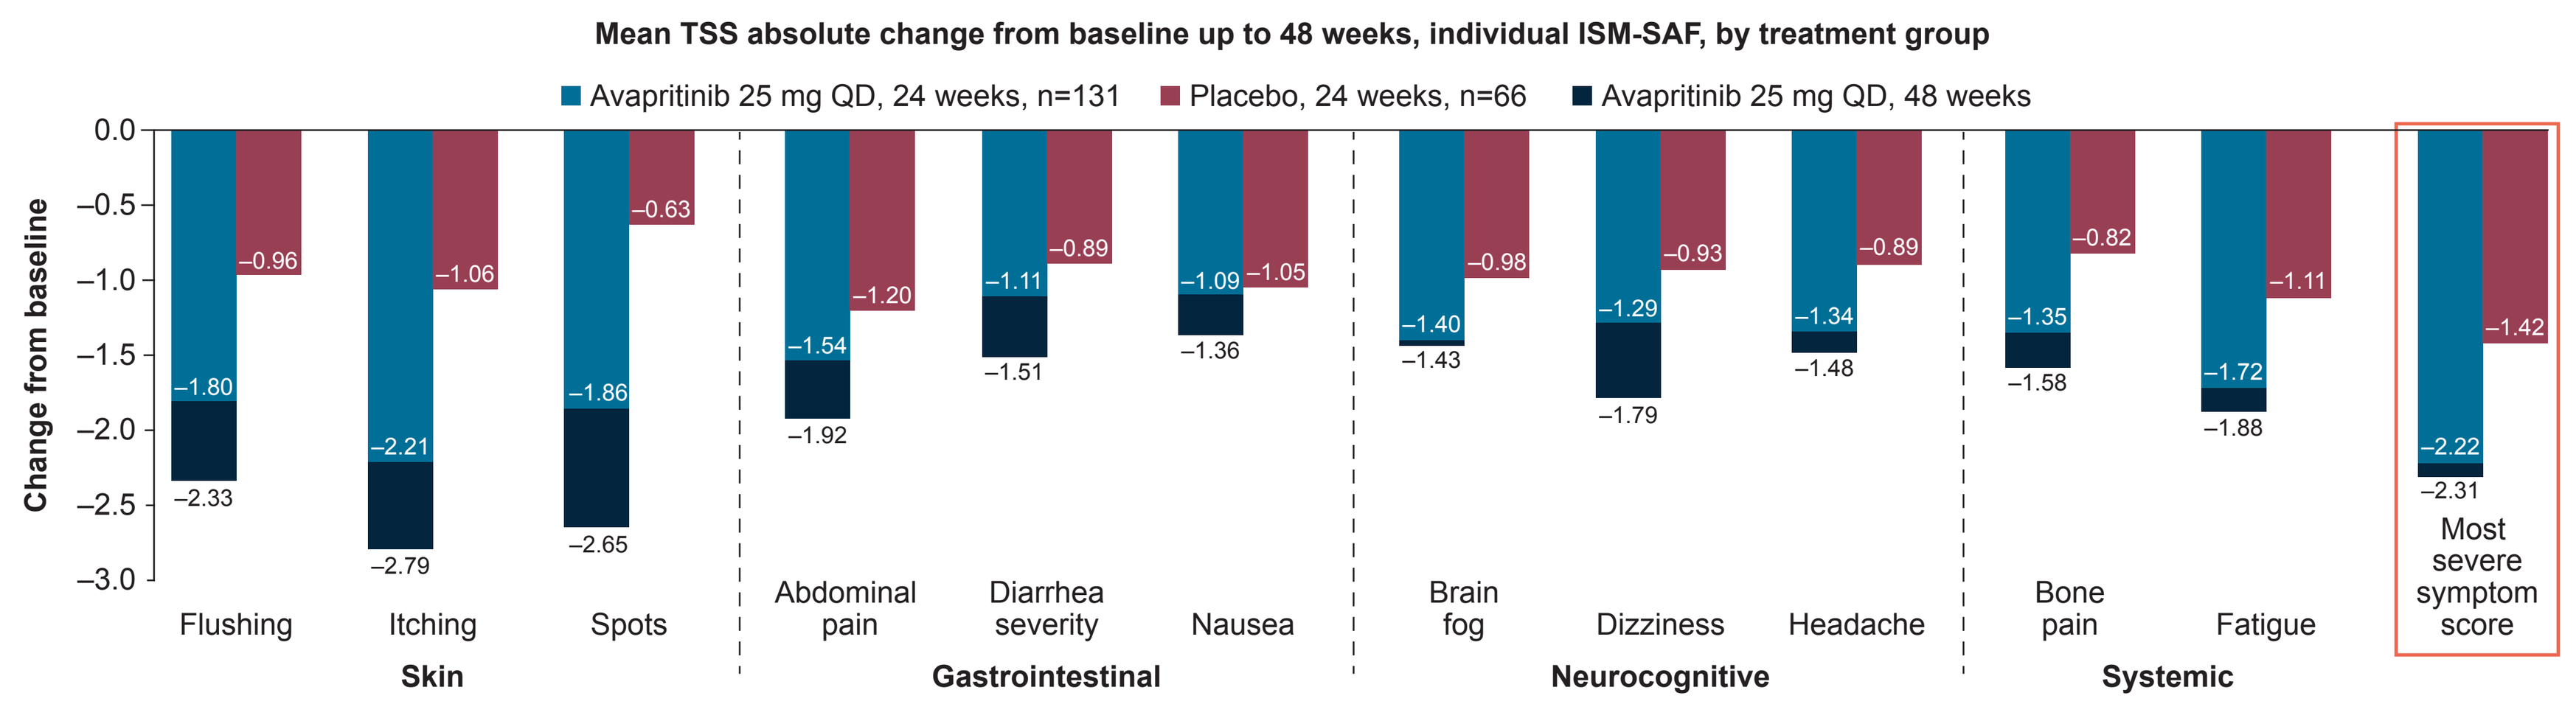

Mean TSS absolute change from baseline up to 48 weeks, individual ISM-SAF, by treatment group
Avapritinib 25 mg QD, 24 weeks, n=131
Placebo, 24 weeks, n=66
Avapritinib 25 mg QD, 48 weeks
Change from baseline
Flushing –1.80 –0.96 –2.33
Itching –2.21 –1.06 –2.79
Spots –1.86 –0.63 –2.65
Skin
Abdominal pain –1.54 –1.20 –1.92
Diarrhea severity –1.11 –0.89 –1.51
Nausea –1.09 –1.05 –1.36
Gastrointestinal
Brain fog –1.40 –0.98 –1.43
Dizziness –1.29 –0.93 –1.79
Headache –1.34 –0.89 –1.48
Neurocognitive
Bone pain –1.35 –0.82 –1.58
Fatigue –1.72 –1.11 –1.88
Systemic
Most severe symptom score –2.22 –1.42 –2.31

At Week 24	Avapritinib 25 mg QD (n=131)	Placebo (n=66)	*P*-value
Mean change in most severe symptom score (SD)	–2.22 (2.30)	–1.42 (1.88)	0.015

SD, standard deviation.

- At 48 weeks, continued improvement was observed in all individual symptoms among avapritinib-treated patients (**Figure 5**)

Reductions in measures of mast-cell burden

- At Week 24, a significantly greater proportion of avapritinib-treated patients achieved ≥50% reductions in measures of mast-cell burden, including:
 - Serum tryptase levels (54% [95% CI, 45 to 62] *vs* 0% [0 to 5], *P*<0.0001; **Figure 6A**)
 - Bone marrow mast-cell burden (53% [95% CI, 43 to 63], *vs* 23% [13 to 36]; *P*<0.0001)
 - *KIT* D816V VAF (68% [95% CI, 59 to 76], *vs* 6% [2 to 16]; *P*<0.0001; **Figure 6B**)

Figure 6: Rapid and sustained reductions in biomarkers of mast-cell burden were observed in avapritinib-treated patients *versus* placebo

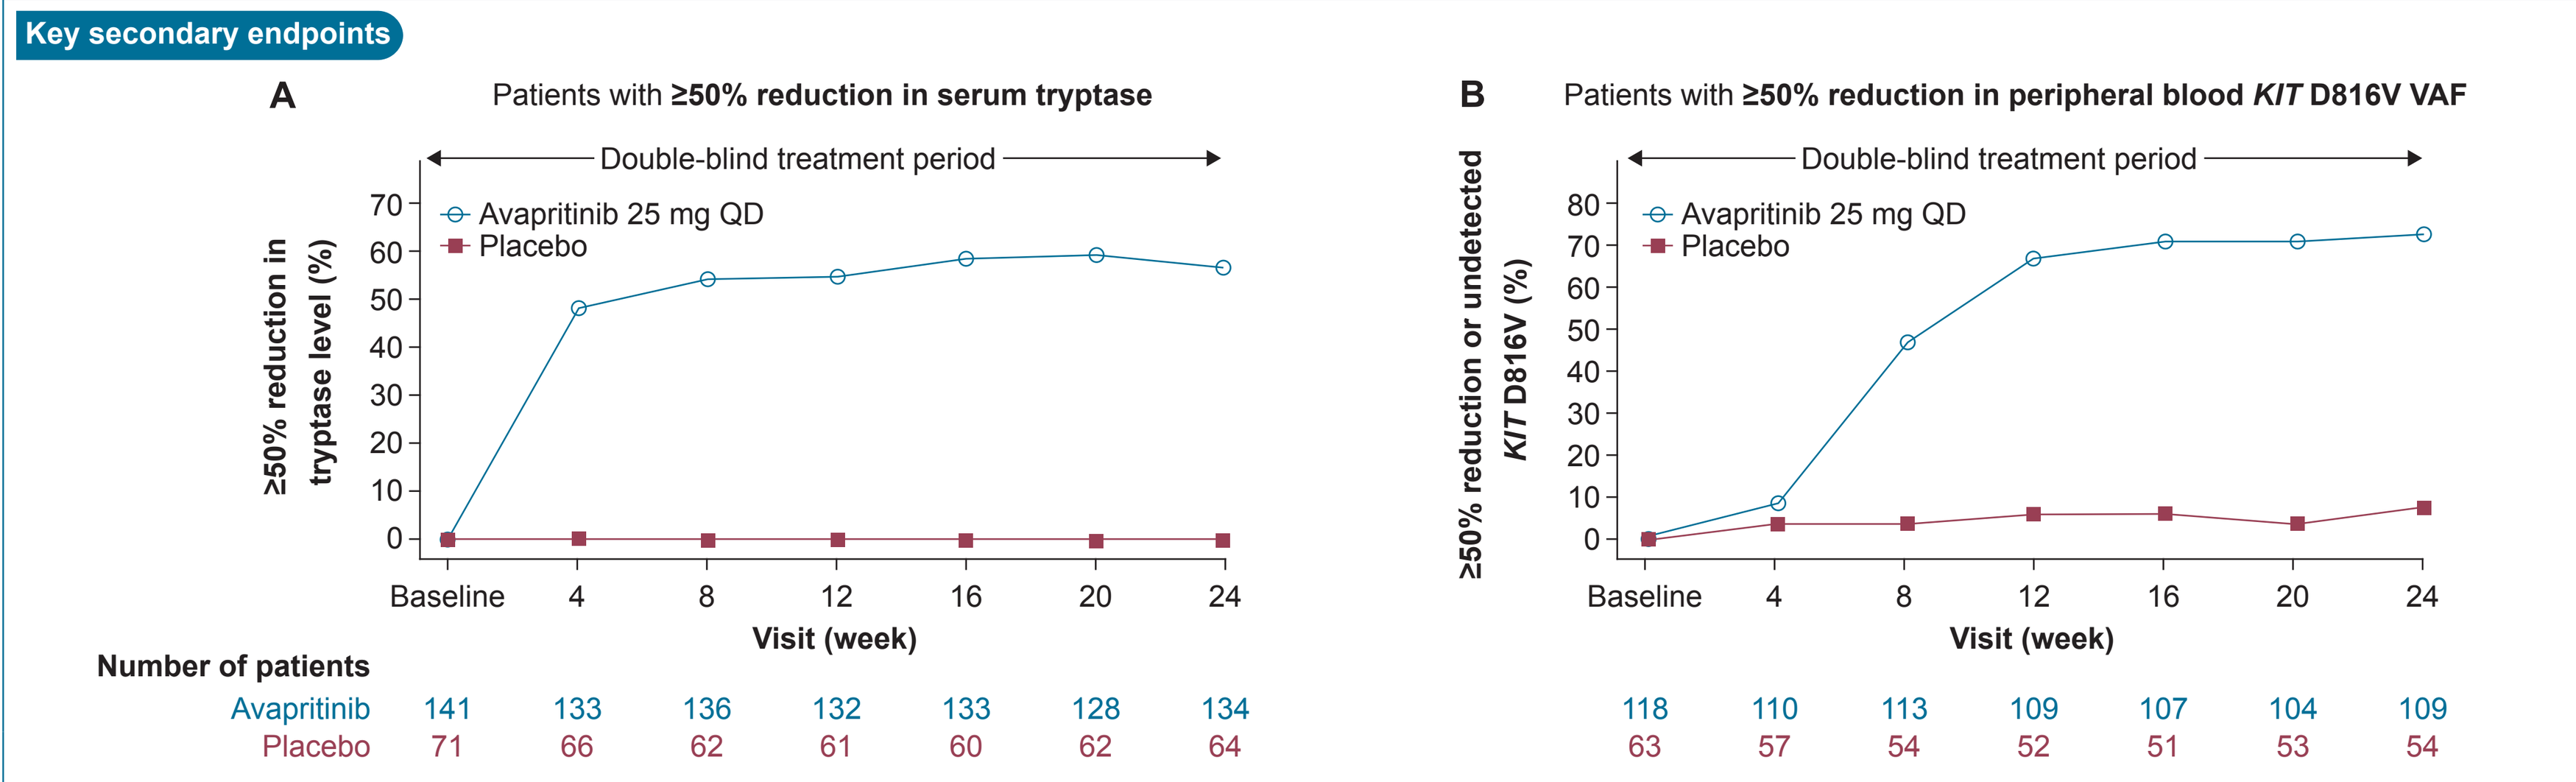

Key secondary endpoints
A
Patients with ≥50% reduction in serum tryptase
Double-blind treatment period
≥50% reduction in tryptase level (%)
Avapritinib 25 mg QD
Placebo
Baseline 4 8 12 16 20 24
Visit (week)
Number of patients
Avapritinib 141 133 136 132 133 128 134
Placebo 71 66 62 61 60 62 64
B
Patients with ≥50% reduction in peripheral blood KIT D816V VAF
≥50% reduction or undetected KIT D816V (%)
118 110 113 109 107 104 109
63 57 54 52 51 53 54

Table 1: Baseline patient demographics and characteristics

Patient demographics	ITT population (N=212) Avapritinib 25 mg QD (n=141)	Placebo (n=71)
Age (years), median (range)	**50.0** (18–77)	**54.0** (26–79)
Female, n (%)	**100** (70.9)	**54** (76.1)
ISM symptom burden		
TSS score, mean (SD)[a,b]	**50.2** (19.1)	**52.4** (19.8)
Most severe symptom score, mean (SD)	**7.7** (1.7)	**7.9** (1.7)
Mast-cell burden		
Median serum tryptase (central), ng/mL (range)	**38.4** (3.6–256.0)	**43.7** (5.7–501.6)
Median bone marrow biopsy mast cells (central), % (range)	**7.0** (1.0–50.0)	**7.0** (1.0–70.0)
Mast-cell aggregates present, n (%)	**106** (75.2)	**57** (80.3)
Median *KIT* D816V VAF in peripheral blood, % (range)[c]	**0.4** (0.02–41.3)	**0.3** (0.02–36.7)
SM Therapy		
Prior cytoreductive therapy, n (%)[d]	**19** (13.5)	**7** (9.9)
Prior TKI therapy, n (%)	**10** (7.1)	**4** (5.6)
BSC use		
Number of BSC treatments, median (range)[e]	**3** (0–11)	**4** (1–8)

[a]Eligibility for enrollment was based on TSS ≥28 at screening; patients may have a score <28 at baseline. [b]Two patients in the avapritinib group had missing baseline TSS values; therefore, the denominator was based on patients with available data at baseline (n=139). [c]The limit of detection was 0.02%. [d]Cytoreductive therapies included dasatinib, imatinib, masitinib, nilotinib, midostaurin, brentuximab vedotin, cladribine, hydroxyurea, rapamycin, and interferon alpha. Includes treatments received by patients at baseline; patients may have received BSC treatments previously that had been discontinued at the time of enrollment/baseline. [e]All patients had at least two BSC prior to or at screening. A total of 10 (7.1%) patients treated with avapritinib and five (7.0%) patients treated with placebo had <2 BSC at the start of the study.
Note: A total of 72.3% of patients on avapritinib and 74.6% of patients receiving the placebo had PI-assessed skin involvement.
ITT; intent-to-treat; PI, principal investigator; TKI, tyrosine kinase inhibitor.

Figure 7: Avapritinib demonstrated sustained improvement in MC-QoL *versus* placebo

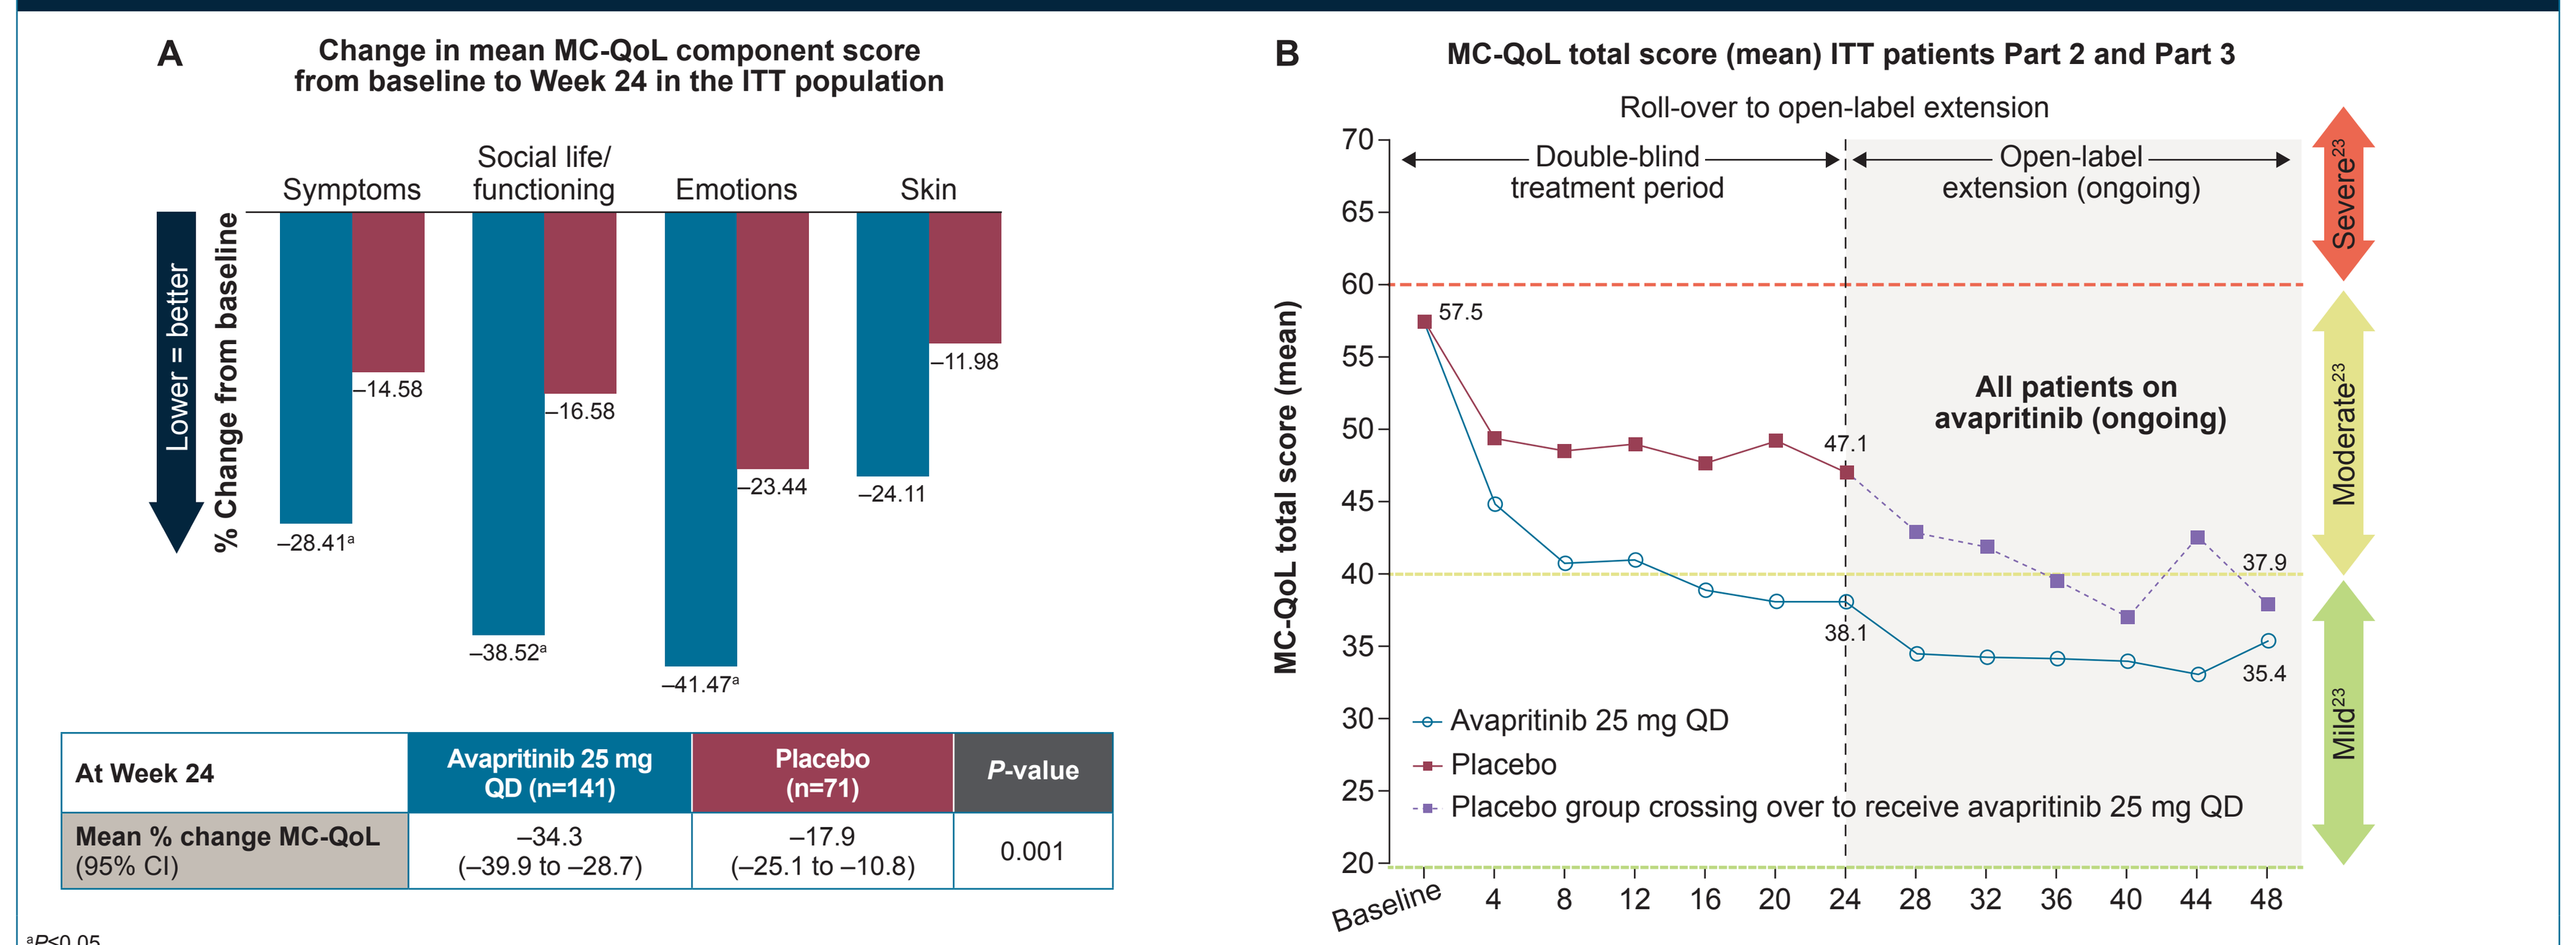

A
Change in mean MC-QoL component score from baseline to Week 24 in the ITT population
Lower = better
% Change from baseline
Symptoms –28.41[a] –14.58
Social life/ functioning –38.52[a] –16.58
Emotions –41.47[a] –23.44
Skin –24.11 –11.98
B
MC-QoL total score (mean) ITT patients Part 2 and Part 3
Roll-over to open-label extension
Double-blind treatment period
Open-label extension (ongoing)
All patients on avapritinib (ongoing)
MC-QoL total score (mean)
57.5
47.1
38.1
37.9
35.4
Severe[23]
Moderate[23]
Mild[23]
Avapritinib 25 mg QD
Placebo
Placebo group crossing over to receive avapritinib 25 mg QD
Baseline 4 8 12 16 20 24 28 32 36 40 44 48

At Week 24	Avapritinib 25 mg QD (n=141)	Placebo (n=71)	*P*-value
Mean % change MC-QoL (95% CI)	–34.3 (–39.9 to –28.7)	–17.9 (–25.1 to –10.8)	0.001

[a]*P*≤0.05.

Improvement in QoL

- MC-QoL scores showed greater improvement with avapritinib *versus* placebo across domains (**Figure 7A**)
- Improvement in MC-QoL mean total score (SD) was greater with avapritinib (57.5 [16.0] to 38.1 [21.6]) *vs* placebo (57.5 [17.2] to 47.1 [20.9]), respectively, at baseline to Week 24 and overtime through to Week 48 (**Figure 7B**)

Well tolerated safety profile

- Avapritinib was well tolerated, with a safety profile similar to placebo (**Table 2**):
 - Majority of adverse events (AEs) were Grade 1 or 2 with a low rate of discontinuation (~1% in both arms)
 - Serious adverse events (SAEs) were reported more frequently in the placebo group (no treatment-related SAEs in either group)
 - Edema AEs were higher in the avapritinib group (majority Grade 1, and did not result in discontinuation)
- In case studies, where photos of patient's skin lesions were assessed, area and color of skin lesions were shown to improve at Week 24 with avapritinib treatment (**Figure 8**)

Table 2: Summary of adverse events

	Avapritinib 25 mg QD (n=141)	Placebo (n=71)
Any AEs[a,b], n (%)	128 (90.8)	66 (93.0)
Grade 1–2 AEs	98 (69.5)	51 (71.8)
Grade 1–2 TRAEs[c]	74 (52.5)	30 (42.3)
Grade ≥3 AEs	30 (21.3)	15 (21.1)
Grade ≥3 TRAEs	3 (2.1)	2 (2.8)
SAEs, n (%)	7 (5.0)	8 (11.3)
Any grade TRAEs	77 (54.6)	32 (45.1)
Most frequently reported TRAEs (≥5% of patients at any group)		
Headache	11 (7.8)	7 (9.9)
Nausea	9 (6.4)	6 (8.5)
Peripheral edema	9 (6.4)	1 (1.4)
Periorbital edema	9 (6.4)	2 (2.8)
Dizziness	4 (2.8)	5 (7.0)
TRAEs leading to discontinuation	2 (1.4)	1 (1.4)

[a]AEs refer to TEAEs, defined as any AE that occurred between Day 1 of Part 2 through to a day prior to Day 1 of Part 3 if the patient crossed over to Part 3; if the patient did not cross over, then through 30 days after the last dose of study drug. [b]There were too few events (2 events in the avapritinib group and 3 events in the placebo group) to assess the impact of avapritinib on anaphylaxis [c]TRAEs refer to treatment-related AEs as assessed by investigators.
AEs, adverse events; SAEs, serious adverse events; TEAEs, treatment-emergent adverse events; TRAEs, treatment-related adverse events.

Figure 8: Case study

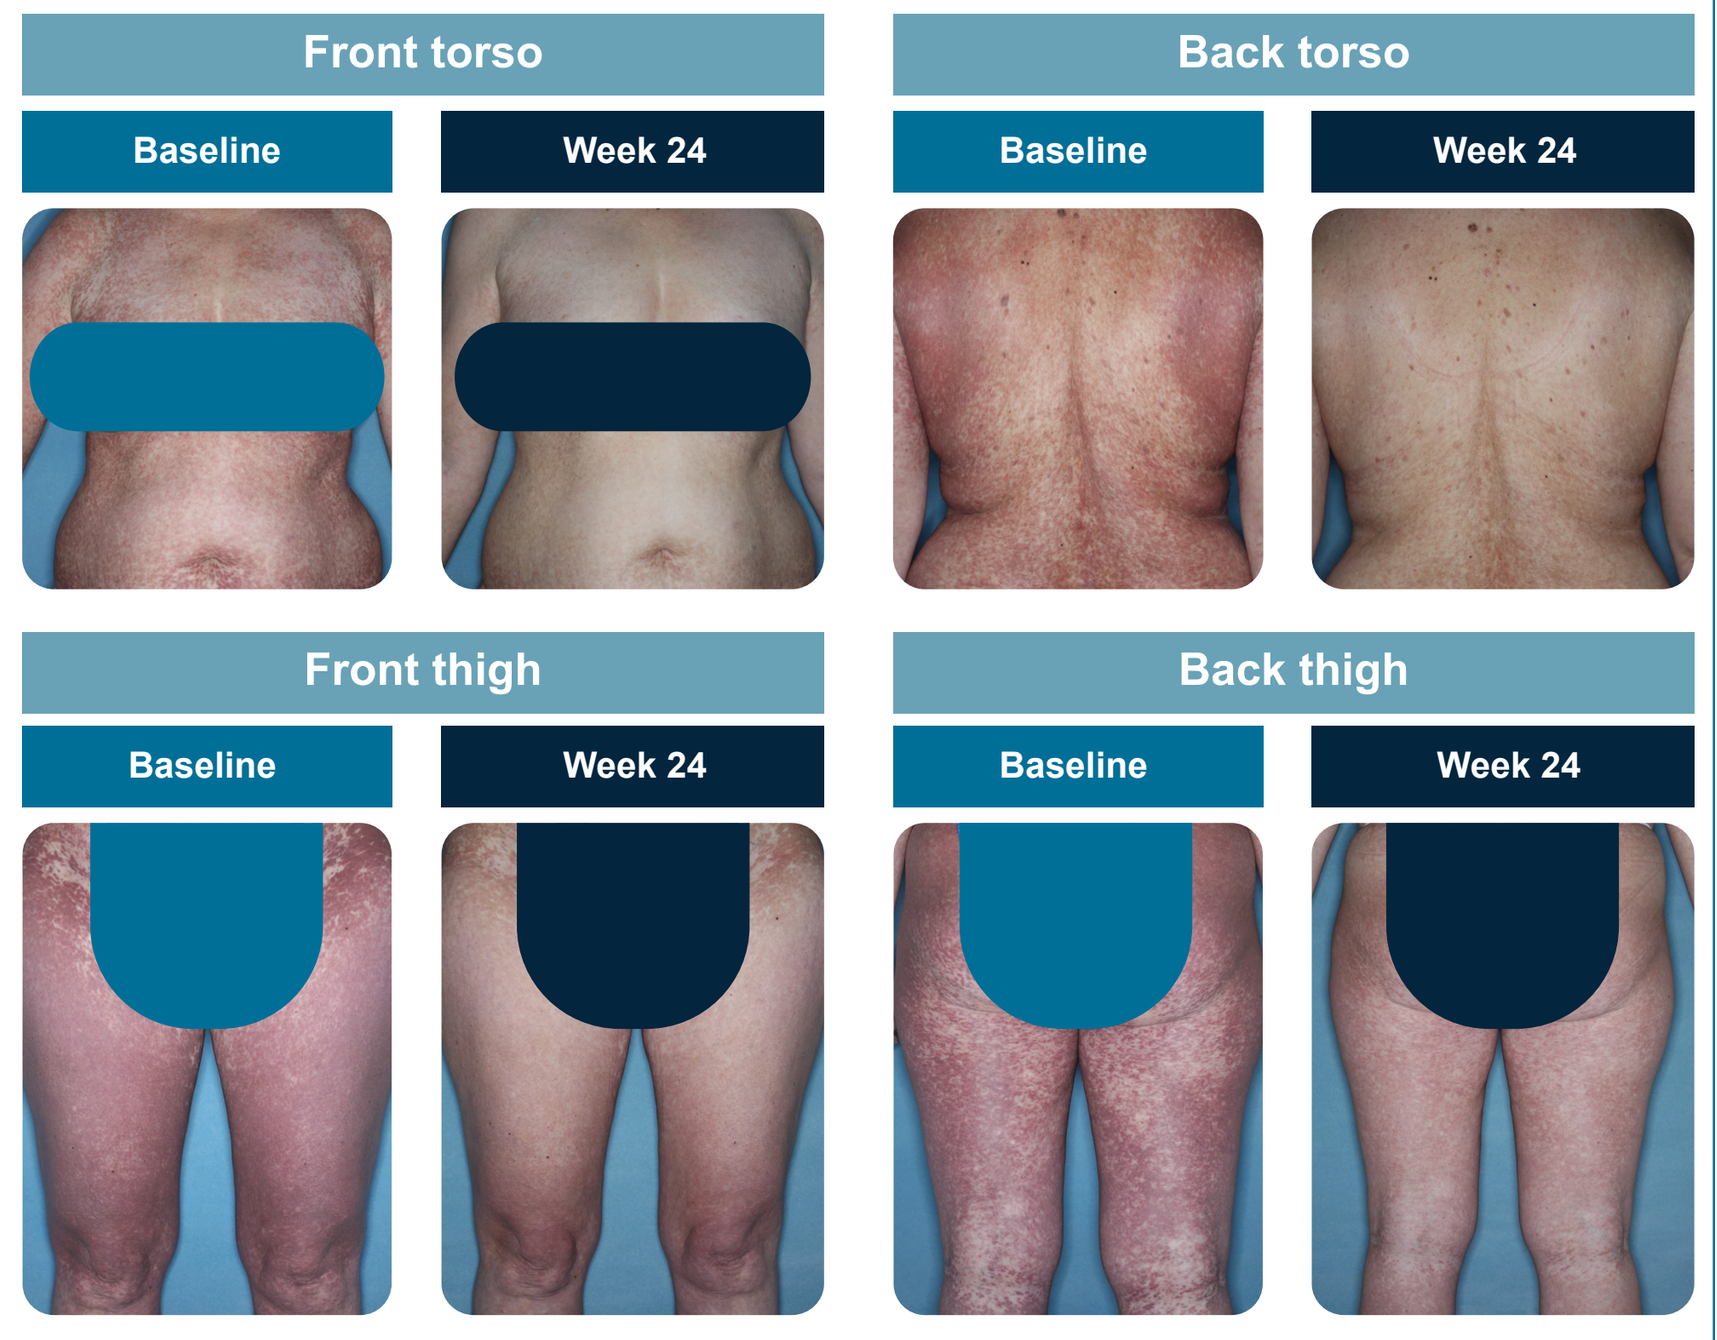

Front torso
Back torso
Baseline
Week 24
Front thigh
Back thigh

Patient permission granted for use of photos.

Conclusions

- By selectively targeting KIT D816V, the underlying driver of ISM, avapritinib treatment provided significant and clinically meaningful improvements in mast-cell burden, symptoms, and QoL compared with placebo
- Of the patients with skin involvement, those treated with avapritinib experienced marked reductions in skin symptoms, skin color, and surface area of skin lesions
- Avapritinib treatment was well tolerated, with a similar safety profile to placebo
- Avapritinib treatment offers a promising new treatment option for patients with ISM

References

1. Kristensen T et al. *J Mol Diagn*. 2011;13:180–188; 2. Cohen SS et al. *Br J Haematol*. 2014;166:521–528; 3. Arber DA et al. *Blood*. 2022;140:1200–1228; 4. Hermine O et al. *PLoS One*. 2008;3:e2266; 5. Akin C et al. *J Allergy Clin Immunol*. 2022;149:1912–1918; 6. Mesa RA et al. *Cancer*. 2022;128:3691–3699; 7. Van Anrooij B et al. *Allergy*. 2016;71:1585–1593; 8. Hartmann K et al. *J Allergy Clin Immunol*. 2016;137:35–45; 9. Pieri L et al. *Am J Hematol*. 2016;91:692–699; 10. Pardanani A. *Blood*. 2013;121:3085–3094; 11. Pardanani A. *Am J Hematol*. 2021;96:508–525; 12. DeAngelo DJ et al. *Nat Med*. 2021; 27:2183–2191; 13. Gotlib J et al. *Nat Med*. 2021;12:2192–2199; 14. Hartmann K et al. Presented at EAACI 2022. Poster #000623; 15. Reiter A et al. *Leukemia*. 2022;36:2108–2120; 16. Radia DH et al. Oral presentation at ASH 2022. *Blood*. 2021;140:Abstract 625; 17. Ayvakit® (avapritinib) [package insert]. 2023. Blueprint Medicines Corporation; 18. Ayvakyt® (avapritinib) Summary of Product Characteristics. 2023. Blueprint Medicines Corporation; 19. Shields AL et al. *Orphanet J Rare Dis*. 2023;18:69; 20. Padilla B et al. *Orphanet J Rare Dis*. 2021;16:434; 21. Taylor F et al. *Orphanet J Rare Dis*. 2021;16:414; 22. Evans EK et al. *Sci Transl Med*. 2017;9:eaao1690; 23. Pulfer S et al. *Allergy Clin Immunol Pract*. 2021;9:3166–3175.

Acknowledgements

Medical writing support was provided by Mathilde Sanson, PhD, and editorial support was provided by Travis Taylor, BA, all of Paragon, Knutsford, UK, supported by Blueprint Medicines Corporation, Cambridge, MA, according to Good Publication Practice guidelines.

Figures 3, 4, 6, 7 and 8 were adapted from *NEJM Evidence*, Gotlib J et al, Avapritinib versus Placebo in Indolent Systemic Mastocytosis, Volume 2 No. 6. Copyright © 2023 Massachusetts Medical Society. Reprinted with permission from Massachusetts Medical Society.

Disclosures

Blueprint Medicines Corporation funded this research. Blueprint Medicines Corporation reviewed and provided feedback on the poster. The authors had full editorial control of the poster and provided their final approval of all content. Dr Castells has served as a consultant for Blueprint Medicines Corporation and is a PI on several clinical trials for Blueprint Medicines Corporation. Dr Castells has received author fees from UpToDate and the Editorial Board for *Annals of Allergy, Asthma & Immunology*.

Presented at the **Eastern Allergy Conference (EAC), June 1–4, 2023**. Please contact medinfo@blueprintmedicines.com for permission to reprint and/or distribute.