Ultra-sensitive Testing Identifies Previously Undetected *KIT* D816V Mutations in Patients With Indolent Systemic Mastocytosis: Learnings From the PIONEER Study of Avapritinib

Tracy I. George M.D.,[1,2] Cem Akin M.D., Ph.D.,[3] Thanai Pongdee M.D.,[4] Hui-Min Lin Ph.D.,[5] Mark Rosenzweig Ph.D.,[5] Rentian Wu Ph.D.,[5] Guang Yang Ph.D.,[5] Ben Lampson M.D., Ph.D.,[5] Rachel L. Erlich Ph.D.,[5] Mariana Castells M.D., Ph.D.[6]

[1]Huntsman Cancer Institute, University of Utah, Salt Lake City, UT; [2]ARUP Laboratories, Department of Pathology, University of Utah School of Medicine, Salt Lake City, UT; [3]University of Michigan, Ann Arbor, MI; [4]Division of Allergic Diseases, Mayo Clinic, Rochester, MN; [5]Blueprint Medicines Corporation, Cambridge, MA; [6]Brigham and Women's Hospital, Division of Allergy and Clinical Immunology, Boston, MA

Introduction

- Indolent systemic mastocytosis (ISM) is the most common subtype of systemic mastocytosis (SM) and is characterized by long-term, debilitating cutaneous, gastrointestinal, neurological, and musculoskeletal symptoms (**Figure 1**)[1–5]

Figure 1. Clinical manifestations of ISM

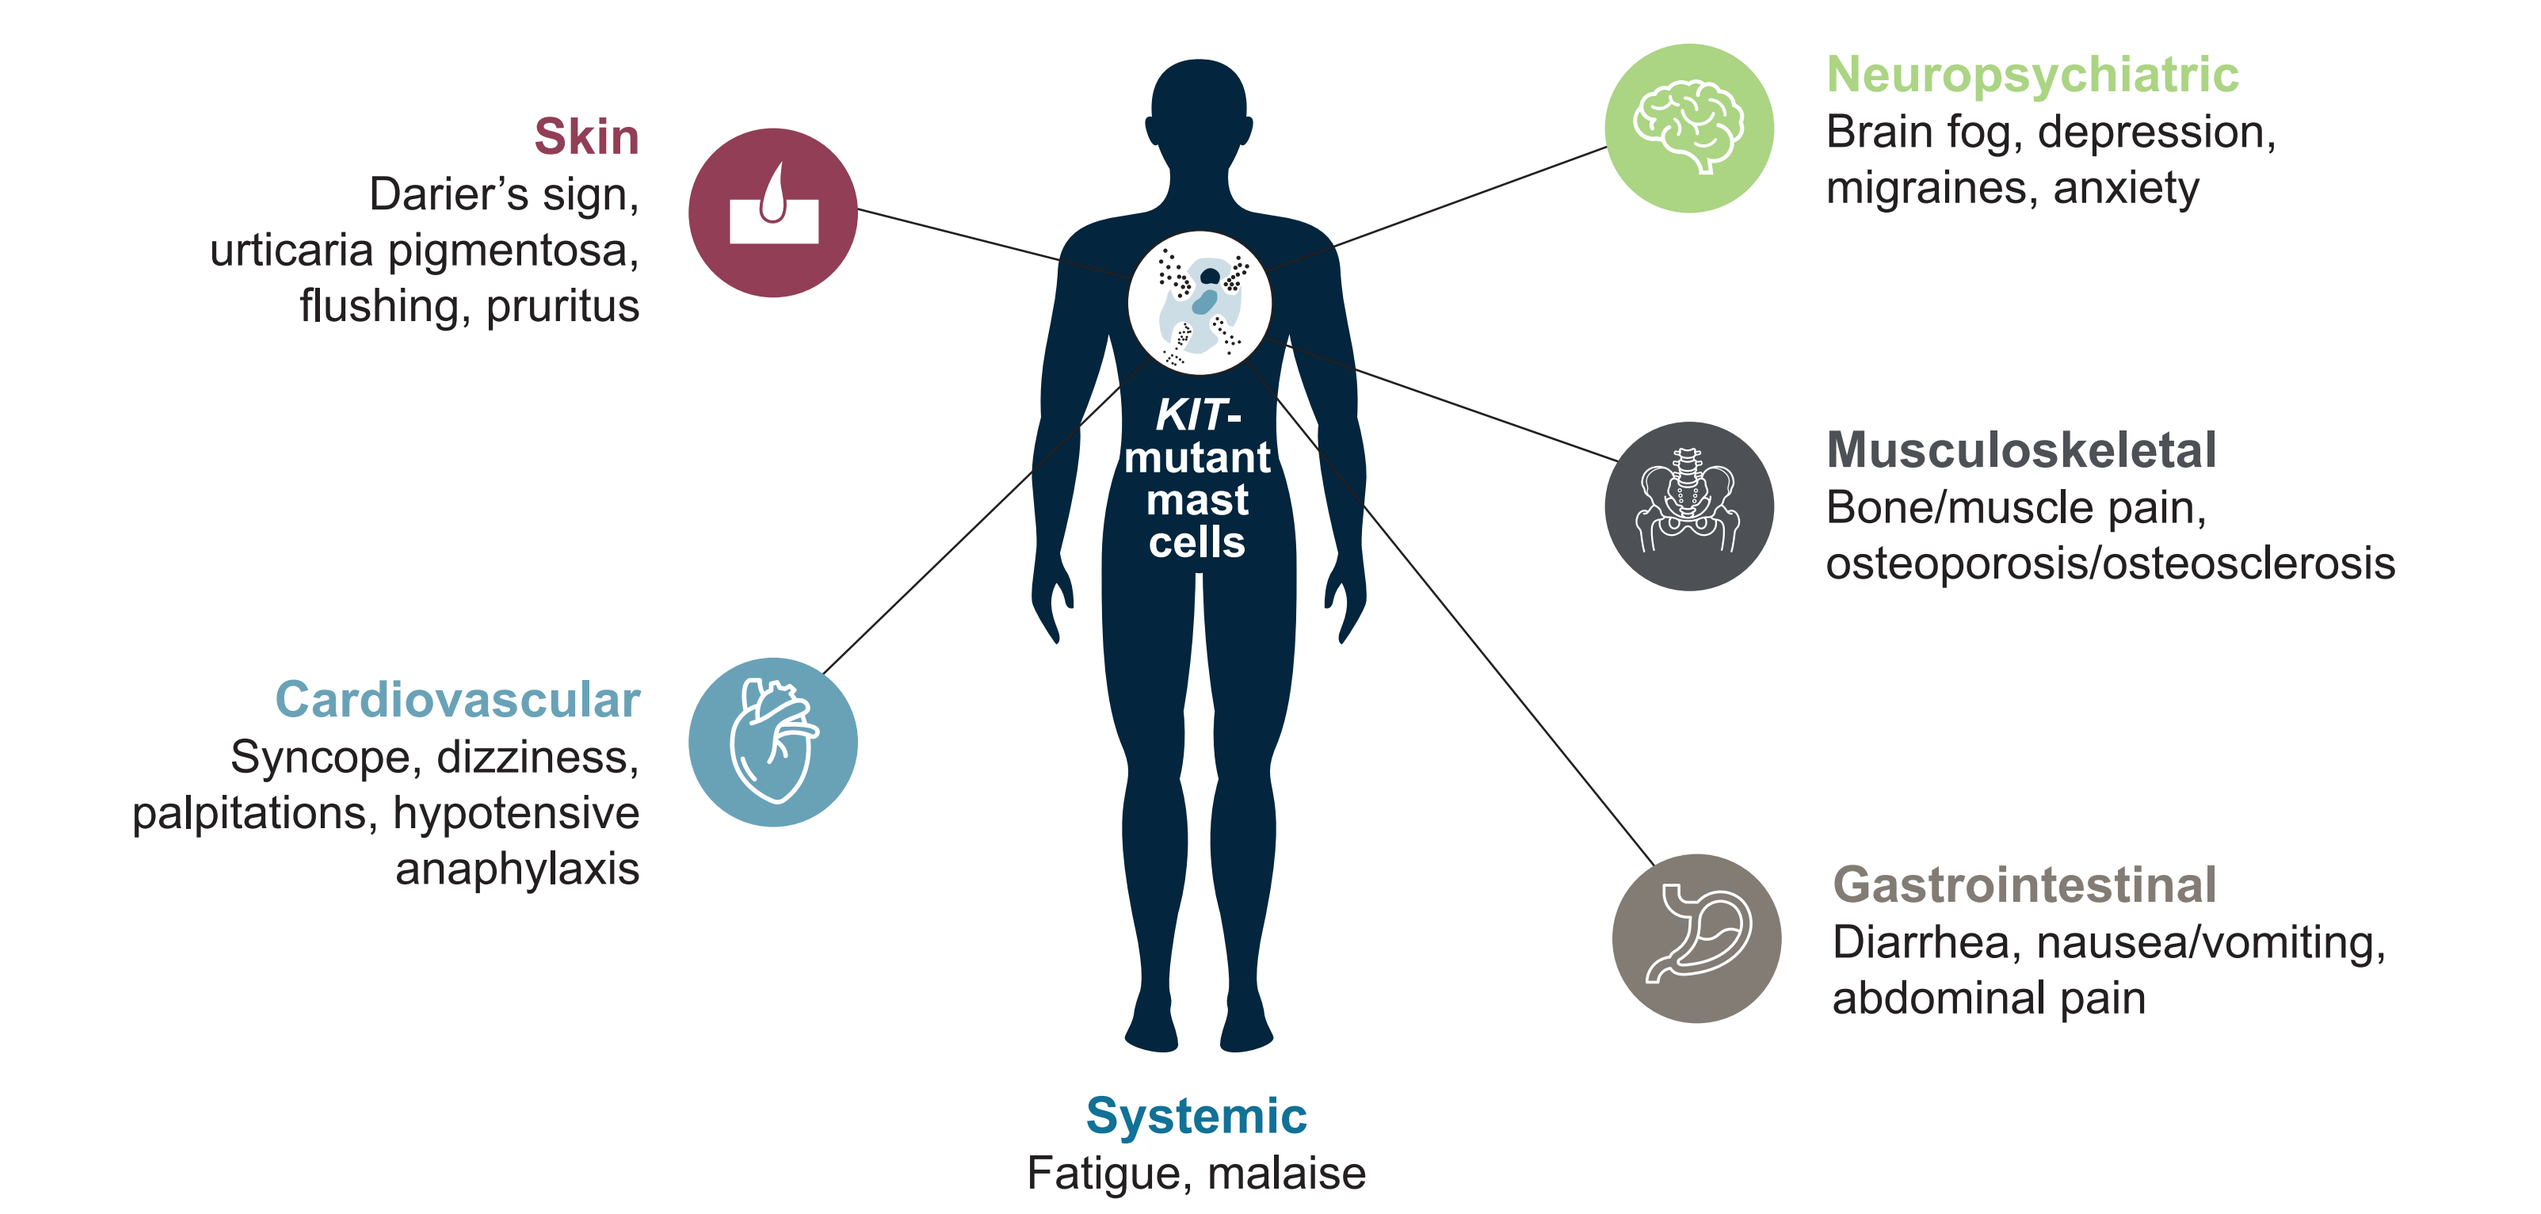

Skin
Darier's sign, urticaria pigmentosa, flushing, pruritus
Neuropsychiatric
Brain fog, depression, migraines, anxiety
KIT-mutant mast cells
Musculoskeletal
Bone/muscle pain, osteoporosis/osteosclerosis
Cardiovascular
Syncope, dizziness, palpitations, hypotensive anaphylaxis
Gastrointestinal
Diarrhea, nausea/vomiting, abdominal pain
Systemic
Fatigue, malaise

ISM, indolent systemic mastocytosis.

- The prevalence of SM has been estimated at up to 1 in 5000 people[6–9]
- ISM is driven by the *KIT* D816V mutation in ~95% of patients and the presence of this mutation is one of the four minor diagnostic criteria for SM[10–16]
- Droplet digital polymerase chain reaction (ddPCR) is the current gold standard test for the detection of the *KIT* D816V mutation (**Table 1**)

Table 1. Testing modalities for *KIT* D816V mutations

Technology	Assay status	Relative sensitivity	*KIT* mutations that can be detected	Sample input	Useful for ISM diagnosis?
Next-generation sequencing	Commercial use	~200x less sensitive than ddPCR[21]	Multiple exon 17 mutations	Isolated DNA from blood or bone marrow aspirate	Only detects *KIT* D816V in ~30% of patients[22]
ddPCR	Commercial use	**Current gold standard** LOD: 0.022%[18]	D816V only	Isolated DNA from blood or bone marrow aspirate	Positive in ~85% of patients[18]
RCA-based assay	Research use	At least ~30x more sensitive than ddPCR[a] (this poster)	D816V only	Isolated DNA from blood or bone marrow aspirate	Positive in >95% of patients (this poster)

[a]Lower LOD still to be validated.
ddPCR, droplet digital polymerase chain reaction; LOD, limit of detection; RCA, rolling circle amplification.

- However, even with the use of ddPCR, detection of the *KIT* D816V mutation in the peripheral blood of patients with ISM and low mast cell burden can be challenging, potentially leading to diagnostic delays and prolonged disease burden
- Diagnostic delays may be further exacerbated as another minor diagnostic criterion, serum tryptase level >20 ng/mL, may be absent in up to 30% of patients with ISM[13,14,17]
- Therefore, more sensitive assays are needed to help physicians identify patients with the *KIT* D816V mutation and underlying clonal mast cell disorders, including ISM
- superRCA® (Rarity Bioscience, Uppsala, Sweden), a rolling circle amplification technology, has been used to specifically detect *KIT* D816V mutations, with ~30-fold greater sensitivity for *KIT* D816V mutation detection *versus* ddPCR
- We used whole blood DNA samples from patients with ISM enrolled in the PIONEER study (NCT03731260) to investigate whether superRCA could detect *KIT* D816V in patients without detectable mutations by ddPCR
 - The randomized, double-blind PIONEER study (**Figure 2**) showed the efficacy of avapritinib, a potent, oral inhibitor that targets the *KIT* D816V mutation, compared with placebo and best supportive care in patients with ISM[18]
 - Avapritinib is approved in the USA and Europe for the treatment of adults with ISM, in the USA for adults with advanced SM (AdvSM), and in Europe for adults with AdvSM after ≥1 prior systemic therapy[19,20]

Figure 2. PIONEER study design

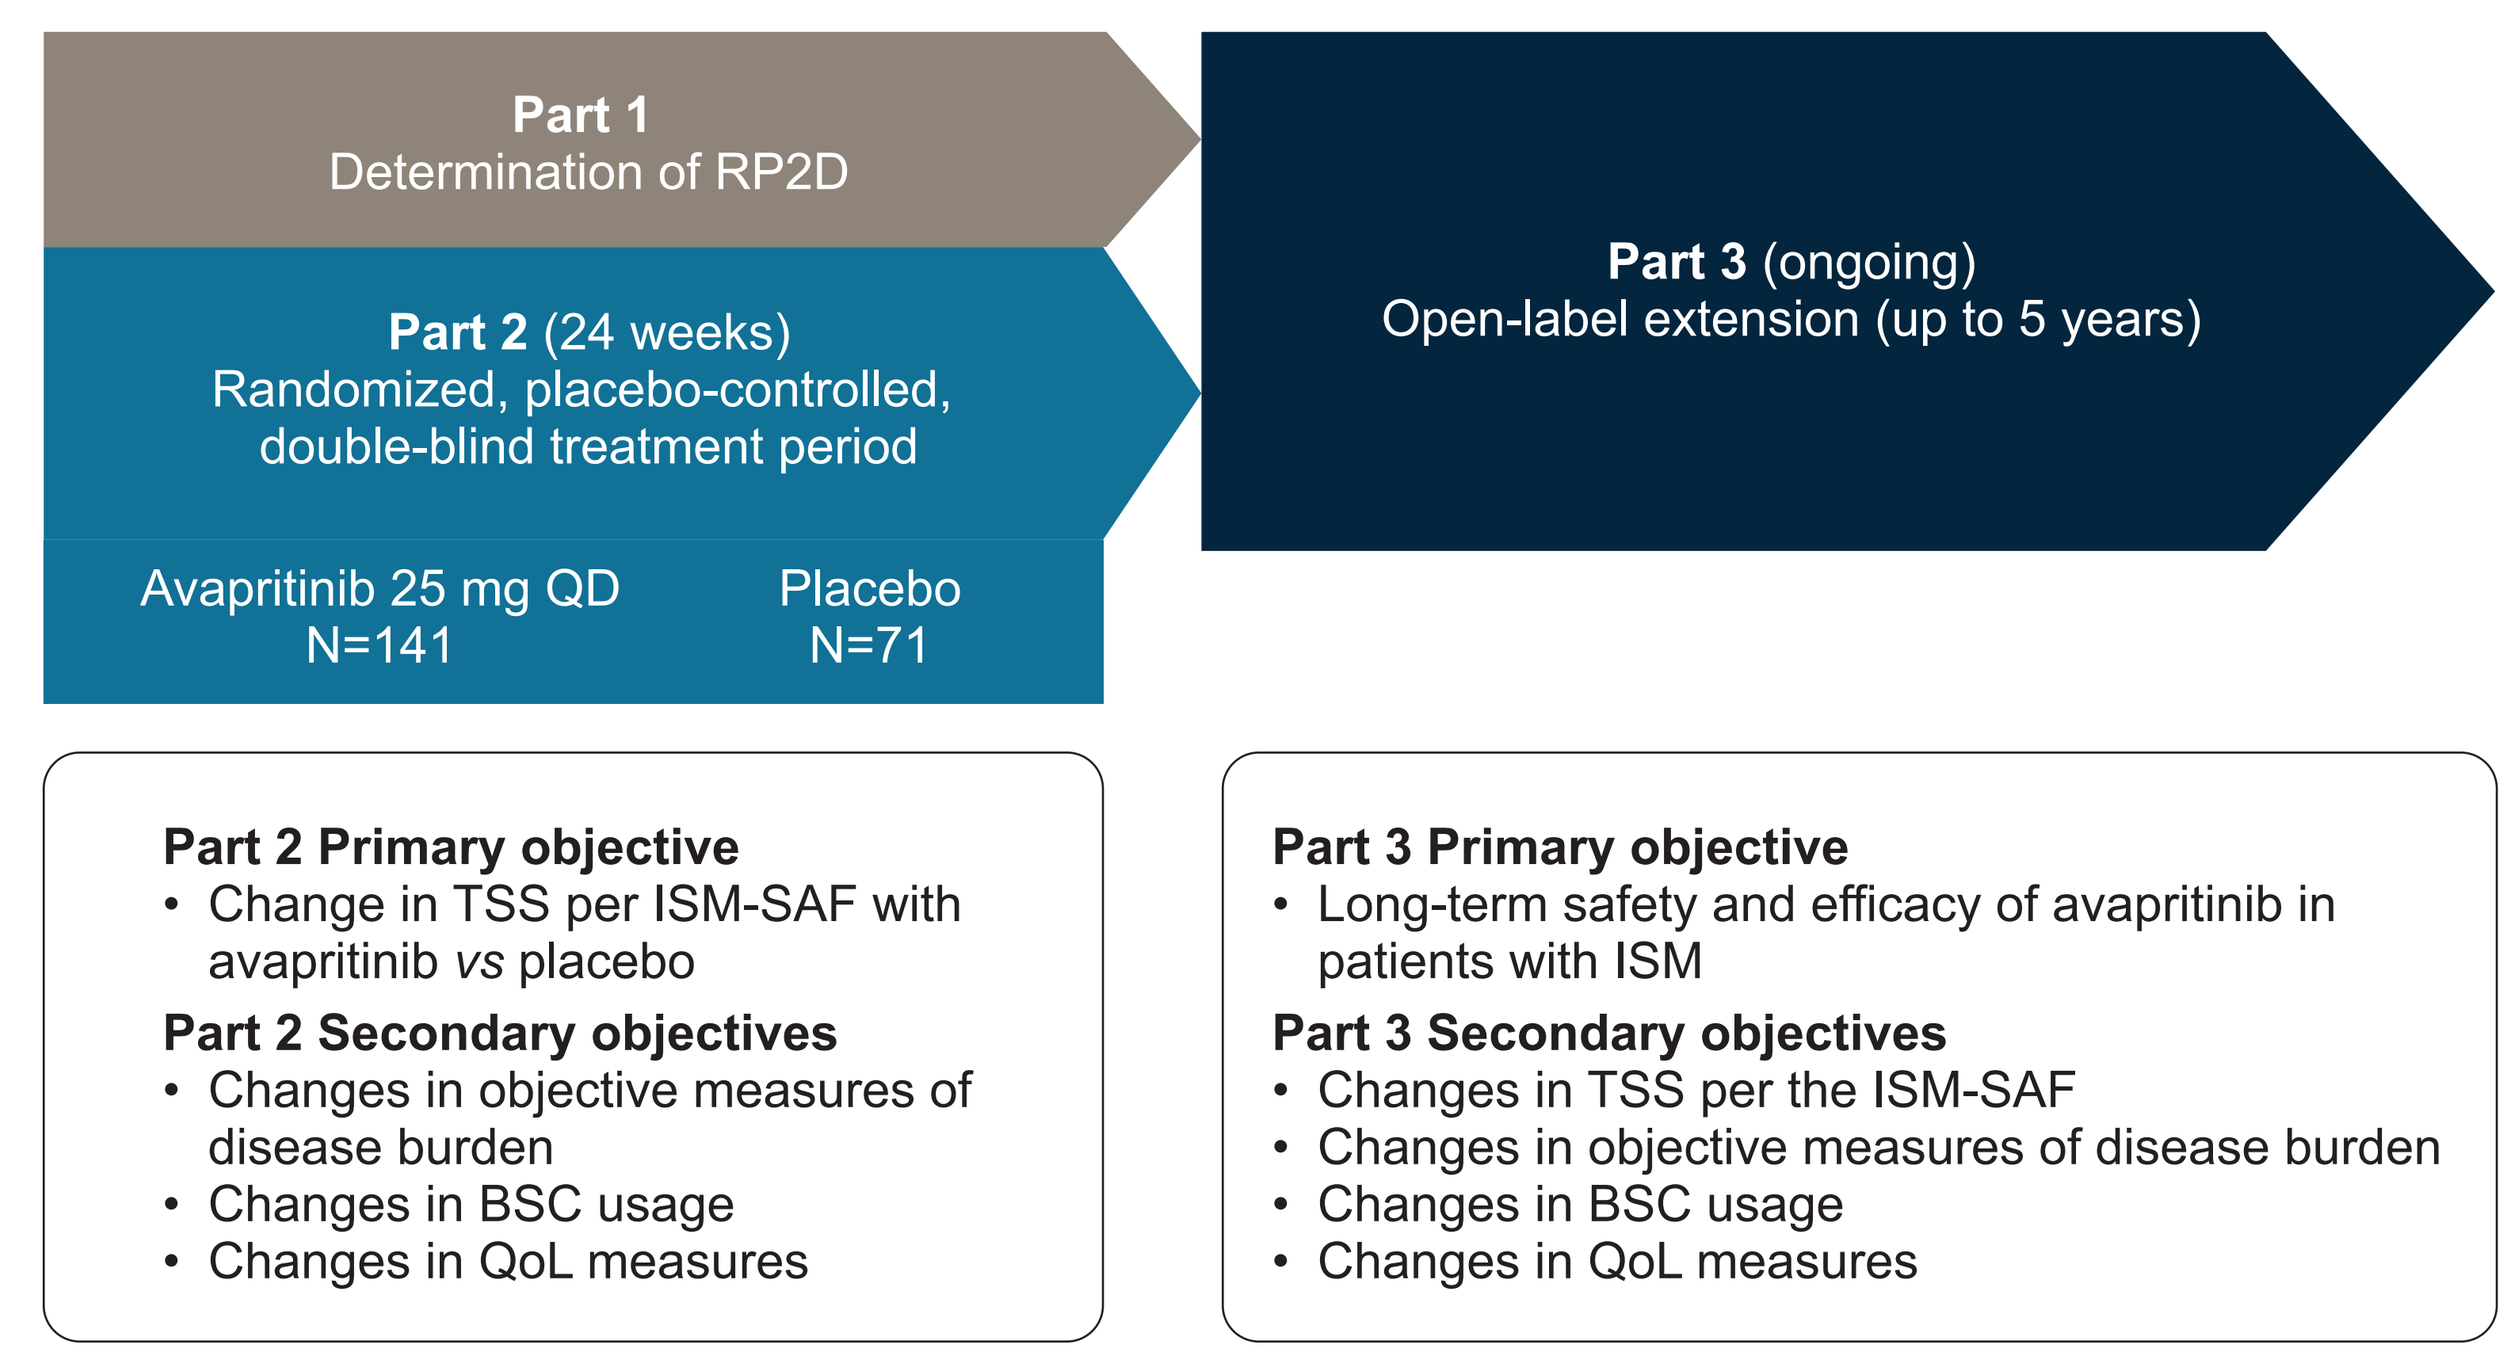

Part 1
Determination of RP2D
Part 2 (24 weeks)
Randomized, placebo-controlled, double-blind treatment period
Avapritinib 25 mg QD
N=141
Placebo
N=71
Part 3 (ongoing)
Open-label extension (up to 5 years)
Part 2 Primary objective
• Change in TSS per ISM-SAF with avapritinib vs placebo
Part 2 Secondary objectives
• Changes in objective measures of disease burden
• Changes in BSC usage
• Changes in QoL measures
Part 3 Primary objective
• Long-term safety and efficacy of avapritinib in patients with ISM
Part 3 Secondary objectives
• Changes in TSS per the ISM-SAF
• Changes in objective measures of disease burden
• Changes in BSC usage
• Changes in QoL measures

BSC, best supportive care; ISM-SAF, Indolent Systemic Mastocytosis Symptom Assessment Form; QD, once daily; QoL, quality of life; RP2D, recommended phase 2 dose; TSS, total symptom score.

Methods

- All patients enrolled in PIONEER met ISM diagnostic criteria, confirmed by central pathology review
- During screening, peripheral blood from all patients (N=246) was tested for the *KIT* D816V mutation by ddPCR; those without detectable mutations were retrospectively tested by superRCA
- Total symptom score (TSS) per the ISM-Symptom Assessment Form (ISM-SAF; ©2018 Blueprint Medicines Corporation) and serum tryptase levels were assessed at baseline and after 24 weeks of avapritinib therapy
 - ISM-SAF is a validated symptom assessment tool specifically developed for evaluation of ISM symptomology based on self-reported severity of 11 ISM symptoms. Scores range from 0 to 110[23,24]

superRCA assay

- superRCA is a sensitive method of mutation detection that relies on DNA amplification and flow cytometry to detect low abundance mutations
- The superRCA assay is reported to detect one mutant copy out of 100,000 wild-type DNA copies[25]
- A clear separation has been reported between the range of negative DNA controls that have been tested with superRCA *versus* positive patient samples and diluted positive cell lines

Results

- Baseline characteristics can be found in **Table 2**
- Baseline bone marrow mast cells were significantly lower in patients with ddPCR-undetectable, superRCA-detectable *KIT* D816V mutations compared with patients with ddPCR-detectable *KIT* mutations only

Table 2. Baseline characteristics

Characteristic	*KIT* mutation detectable by ddPCR (n=209)	*KIT* mutation not detectable by ddPCR but detectable by superRCA (n=29)	P-value
Age, years (range)	51 (18–79)	50 (27–64)	0.3269
Female, %	153 (73)	20 (69)	0.6586
Median baseline serum tryptase, ng/mL (range)	43.1 (4.2–590.4)	20.2 (3.6–200)	<0.0001
Median BM MC, % (range)	10.0 (1.0–60.0)	5.0 (1.0–40.0)	<0.0001
Mean TSS per the ISM-SAF (SD)	49.1 (19.6)	48.9 (18.9)	0.9514

BM MC, bone marrow mast cell; SD, standard deviation.

Figure 3. *KIT* D816V mutations detected in patients enrolled in PIONEER

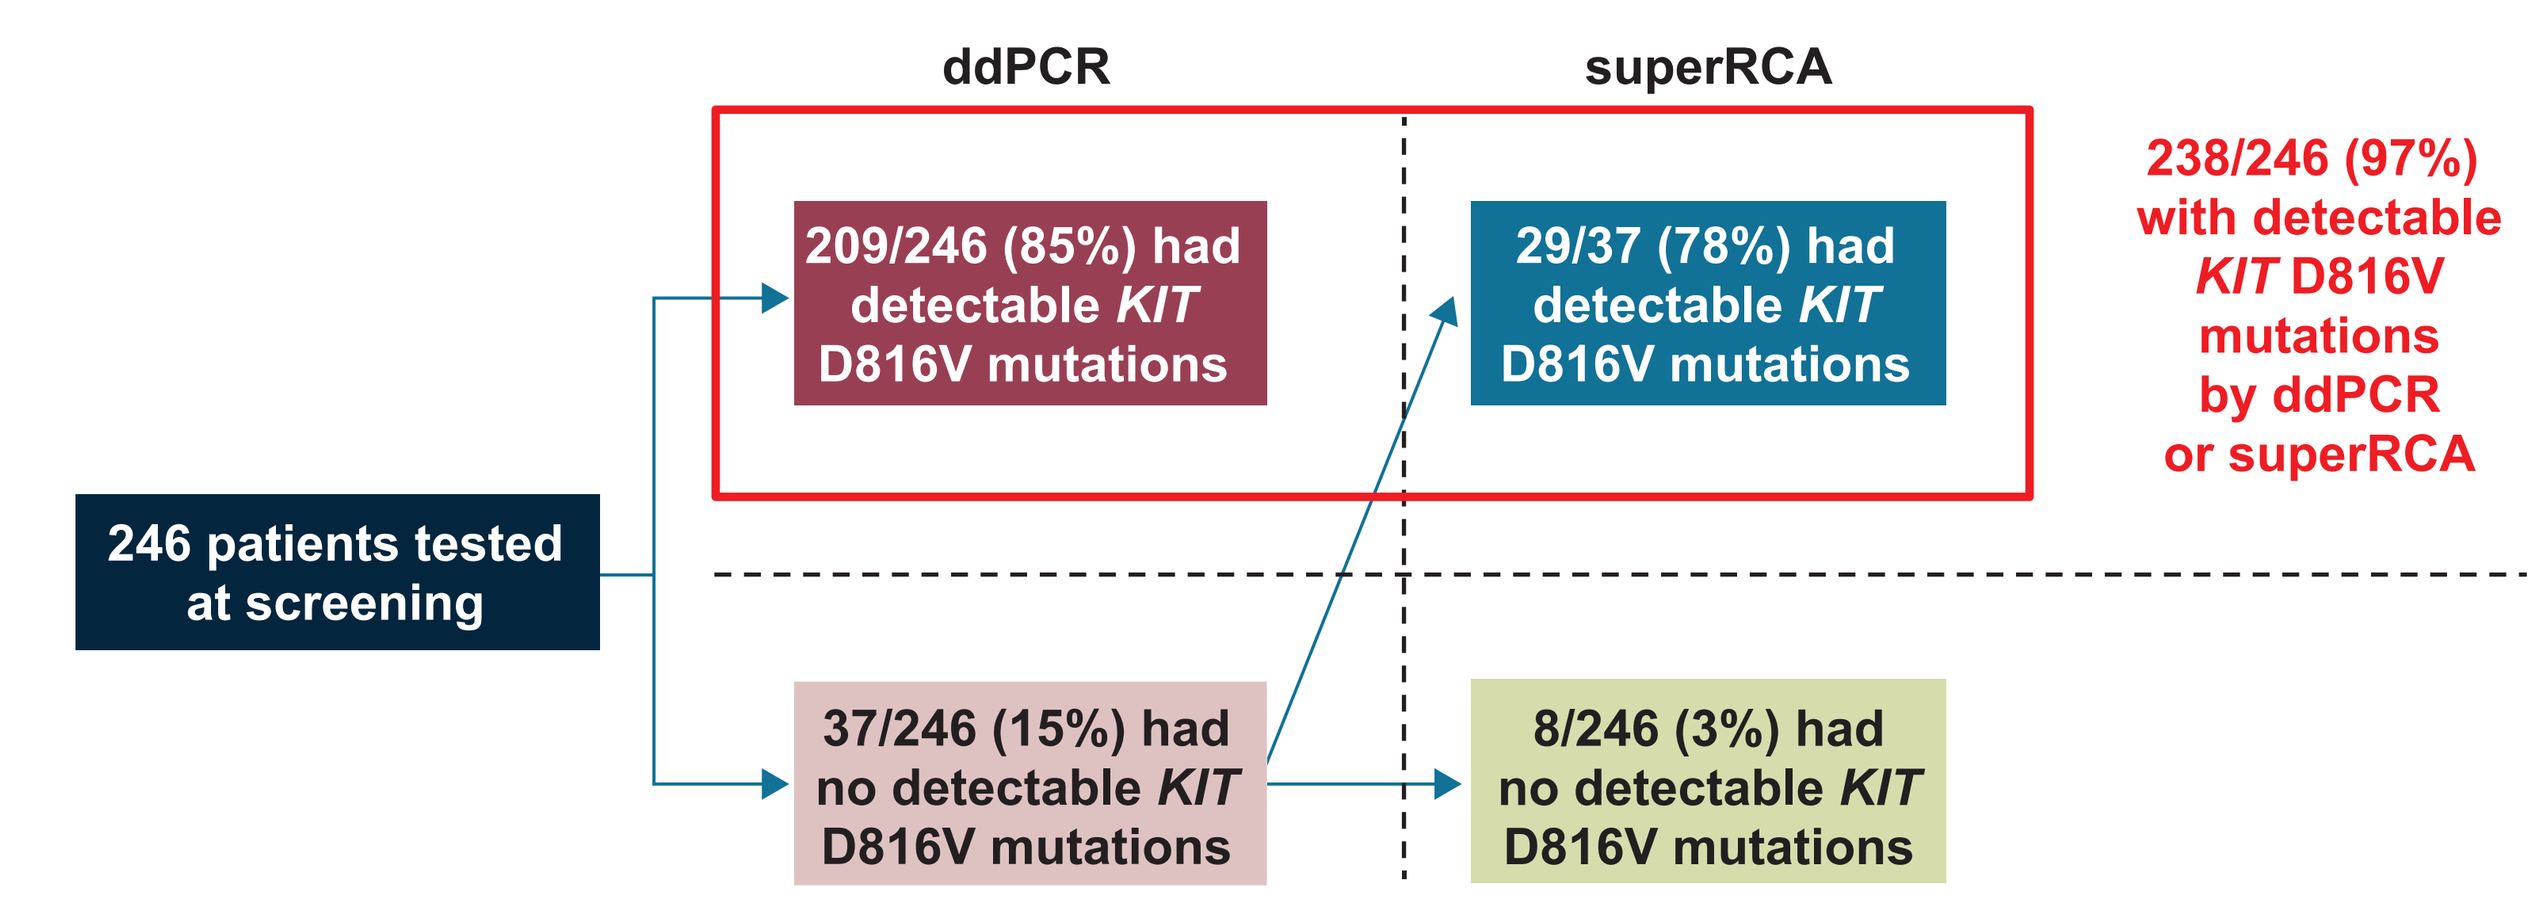

ddPCR
superRCA
246 patients tested at screening
209/246 (85%) had detectable KIT D816V mutations
29/37 (78%) had detectable KIT D816V mutations
238/246 (97%) with detectable KIT D816V mutations by ddPCR or superRCA
37/246 (15%) had no detectable KIT D816V mutations
8/246 (3%) had no detectable KIT D816V mutations

- In combination, superRCA and ddPCR detected *KIT* D816V mutations in the peripheral blood of 238/246 (97%) patients (**Figure 3**)
 - Using ddPCR, *KIT* D816V was detected in 209/246 (85%) peripheral blood samples
 - In patients with ISM who had undetectable *KIT* D816V by ddPCR, superRCA detected *KIT* D816V in 29/37 (78%) samples
- The range of detectable variant allele frequencies by superRCA was 0.00038–0.03389% with a median of 0.0047% (**Figure 4**)

Figure 4. *KIT* D816V VAF distribution by superRCA in ddPCR-undetectable patients

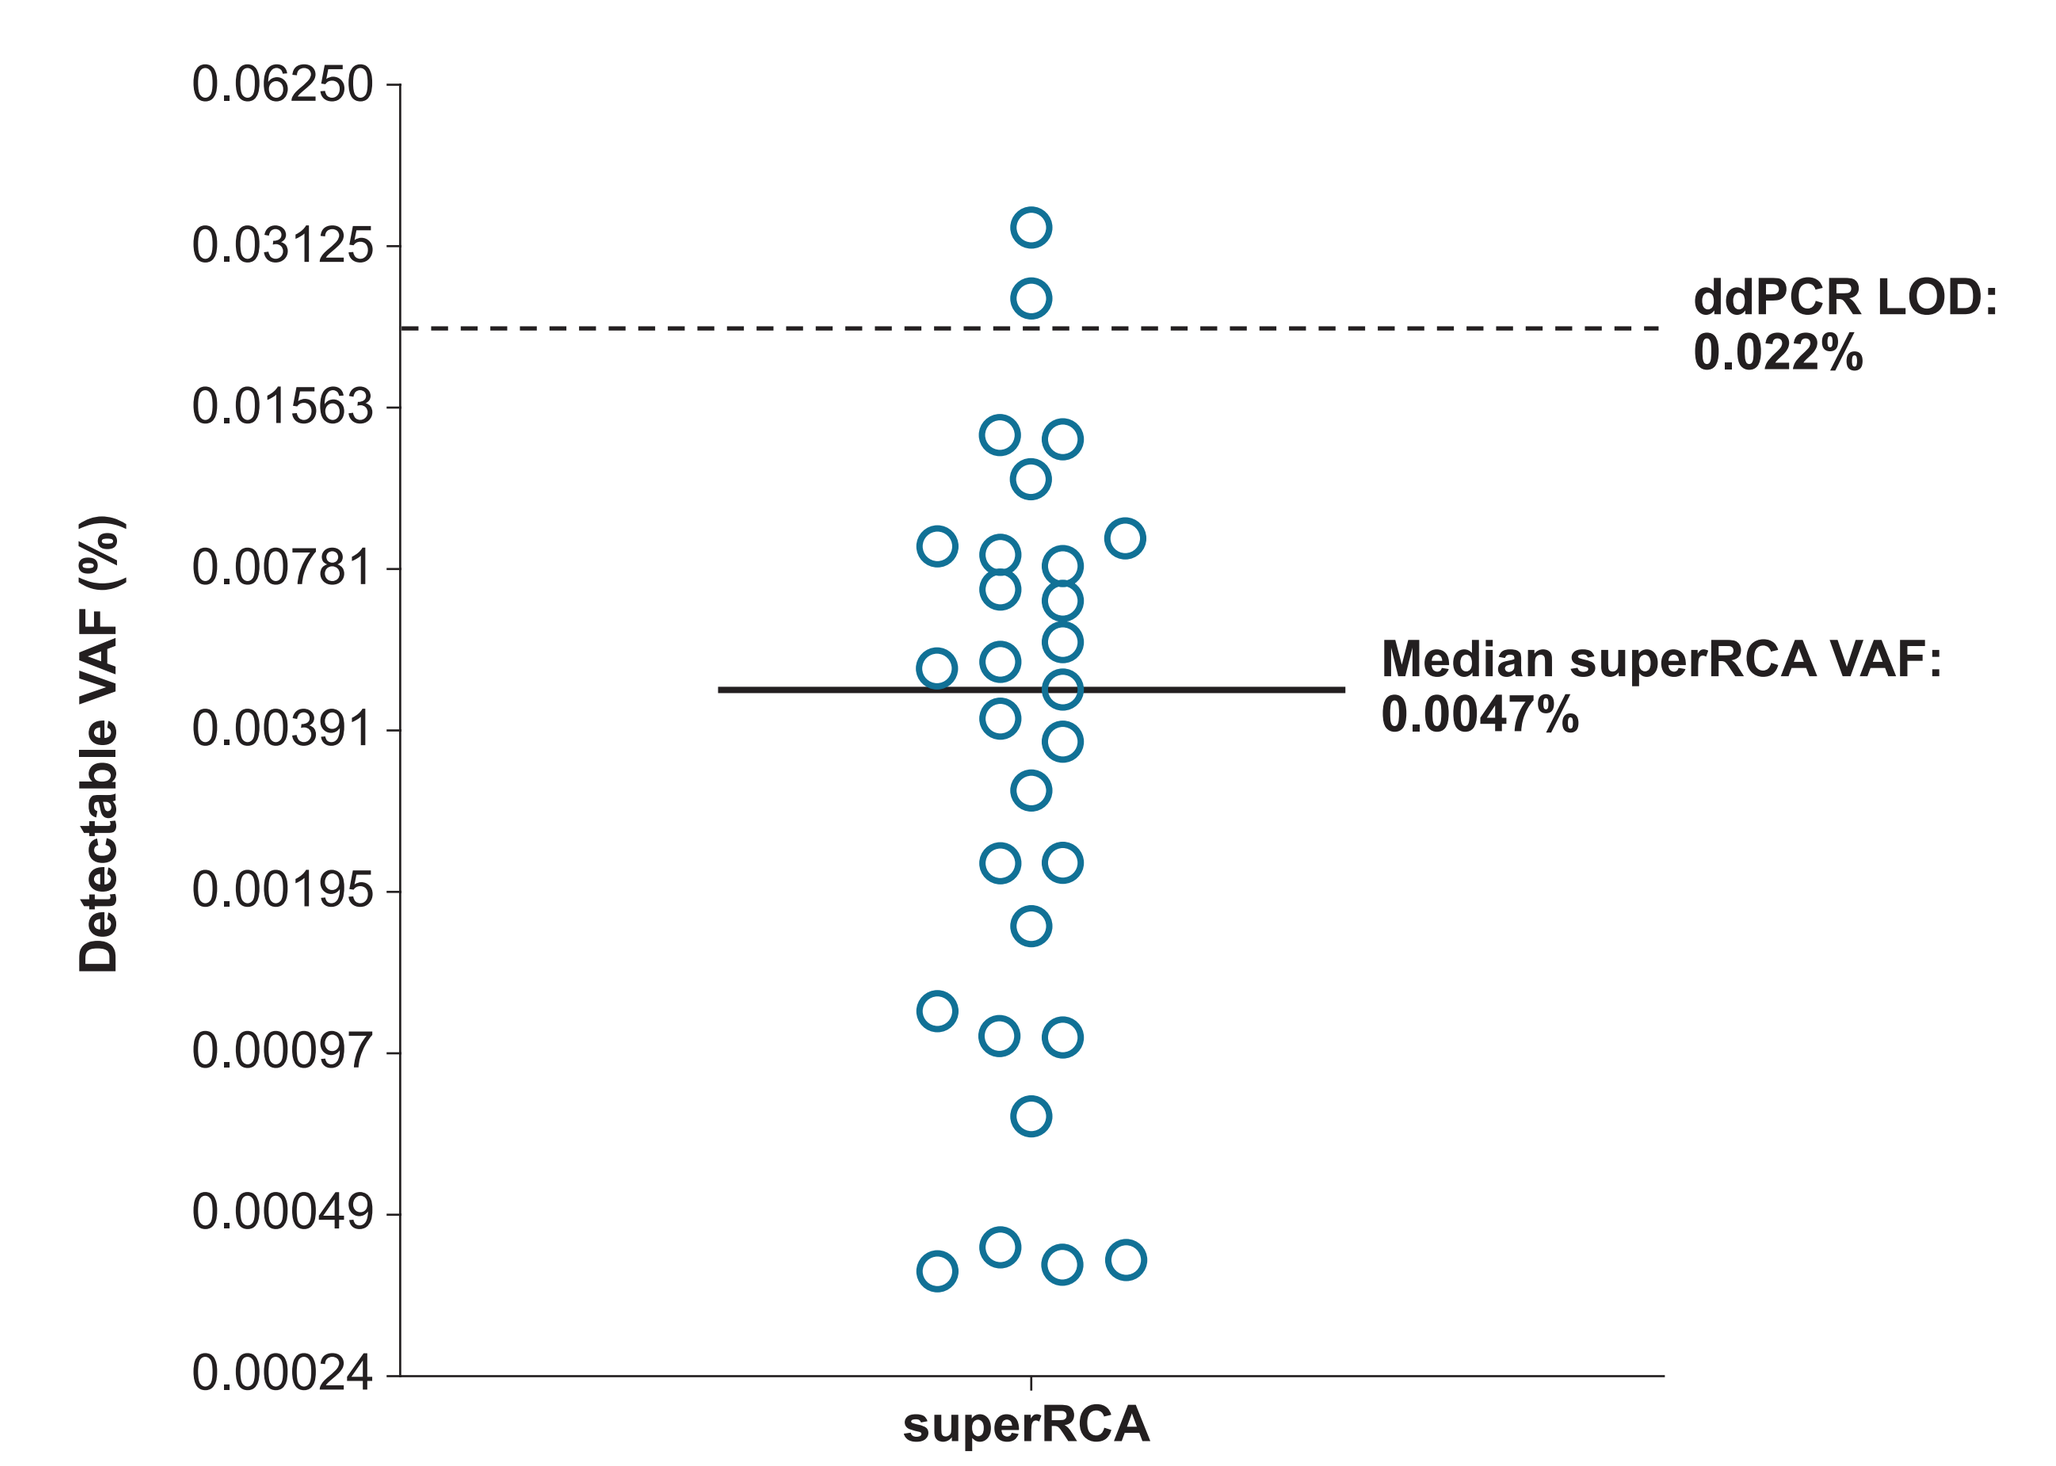

Detectable VAF (%)
0.06250
0.03125
0.01563
0.00781
0.00391
0.00195
0.00097
0.00049
0.00024
ddPCR LOD: 0.022%
Median superRCA VAF: 0.0047%
superRCA

VAF, variant allele frequency.

TSS and serum tryptase measures

- Baseline tryptase levels were significantly lower in patients with ddPCR-undetectable, superRCA-detectable *KIT* D816V mutations compared with patients with ddPCR-detectable mutations (**Table 2**)
- Baseline symptom burden, as measured by TSS per the ISM-SAF, was similar in patients with ddPCR-detected *versus* superRCA-detected *KIT* D816V (**Table 2**)
- After 24 weeks of therapy with avapritinib, patients with detectable *KIT* D816V mutations by superRCA, but undetectable *KIT* mutations by ddPCR, had improvements in ISM-SAF TSS and tryptase, with mean percentage changes from baseline of −20.7% and −55.6%, respectively

References

1. Mesa RA et al. *Cancer*. 2022;128:3691–3699; 2. Hermine O et al. *PLoS One*. 2008;3:e2266; 3. van Anrooij B et al. *Allergy*. 2016;71:1585–1593; 4. Hartmann K et al. *J Allergy Clin Immunol*. 2016;137:35–45; 5. Akin C et al. *J Allergy Clin Immunol*. 2022;149:1912–1918; 6. Brockow K. *Immunol Allergy Clin North Am*. 2014;34:283–295; 7. Cohen SS et al. *Br J Haematol*. 2014;166:521–528; 8. van Doormaal JJ et al. *J Allergy Clin Immunol*. 2013;131:1429–1431.e1421; 9. Bergstrom A et al. *Acta Oncol*. 2024;63:44–50; 10. Kristensen T et al. *Am J Hematol*. 2014;89:493–498; 11. Ungerstedt J et al. *Cancers*. 2022;14:3942; 12. Garcia-Montero AC et al. *Blood*. 2006; 108:2366–2372; 13. Verstovek S. ©International Agency for Research on Cancer. 2023. Systemic Mastocytosis. https://tumourclassification.iarc.who.iwent/chaptercontent/63/20. Accessed May 13, 2025; 14. Khoury JD, et al. *Leukemia*. 2022;36:1703–1719; 15. Valent P et al. 2021;5:e646; 16. Arber DA et al. *Blood*. 2022;140:1200–1228; 17. Sánchez-Muñoz L et al. *Mod Pathol*. 2011;24:1157–1168; 18. Gotlib J et al. *NEJM Evidence*. 2023;2:EVIDoa2200339; 19. Ayvakyt (avapritinib) Summary of Product Characteristics. Cambridge, MA; Blueprint Medicines Corporation; 2024; 20. Ayvakit (avapritinib) Prescribing Information. Cambridge, MA: Blueprint Medicines Corporation; 2024; 21. Illumina. TruSight™ Myeloid Sequencing Panel Data Sheet. https://www.illumina.com/content/dam/illumina/gcs/assembled-assets/marketing-literature/trusight-myeloid-data-sheet-m-gl-00320/trusight-myeloid-data-sheet-m-gl-00320.pdf Accessed April 29, 2025; 22. George T et al. Presented at the American Society of Hematology Annual Meeting and Exposition, 2020; Presentation #3004; 23. Shields AL et al. *Orphanet J Rare Dis*. 2023;18:69; 24. Taylor F et al. *Orphanet J Rare Dis*. 2021;16:414; 25. Chen L et al. *Nat Commun*. 2022;13:4033.

Case study: Effectiveness of avapritinib in a patient with a *KIT* D816V mutation below the VAF detection threshold of ddPCR, but detectable by superRCA

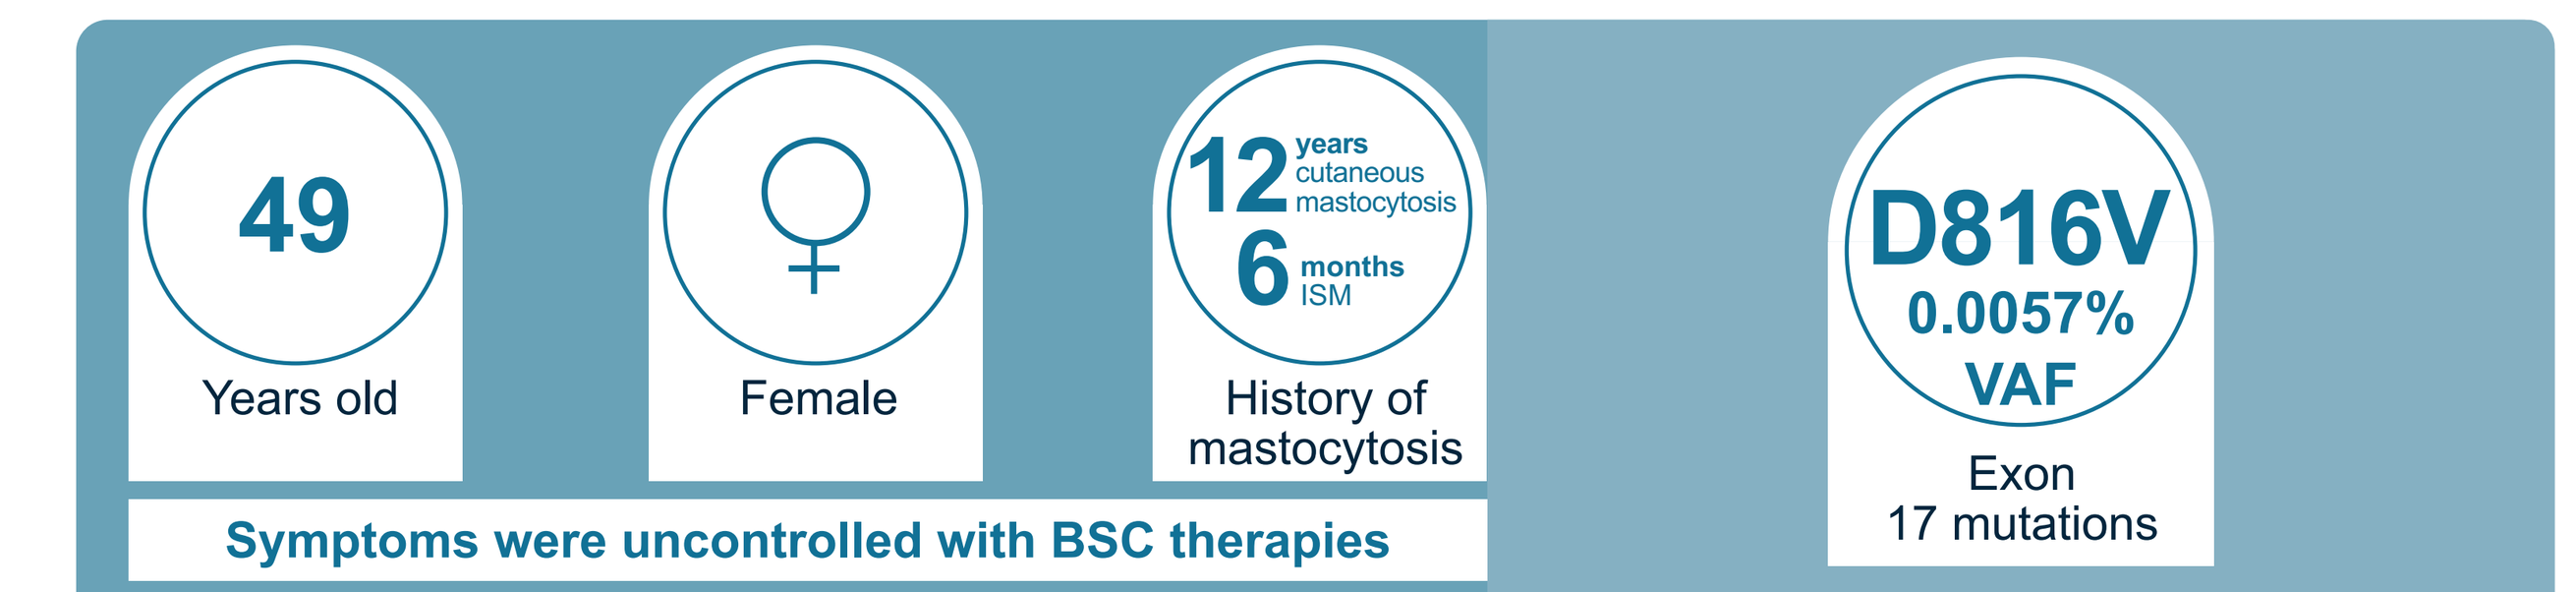

49
Years old
Female
12 years cutaneous mastocytosis
6 months ISM
History of mastocytosis
Symptoms were uncontrolled with BSC therapies
D816V
0.0057%
VAF
Exon 17 mutations

	% change from baseline to Week 24
ISM-SAF TSS	**−63%**
Skin domain score	−64%
Gastrointestinal domain score	−64%
Neuro domain score	−60%
Serum tryptase	**−65%**

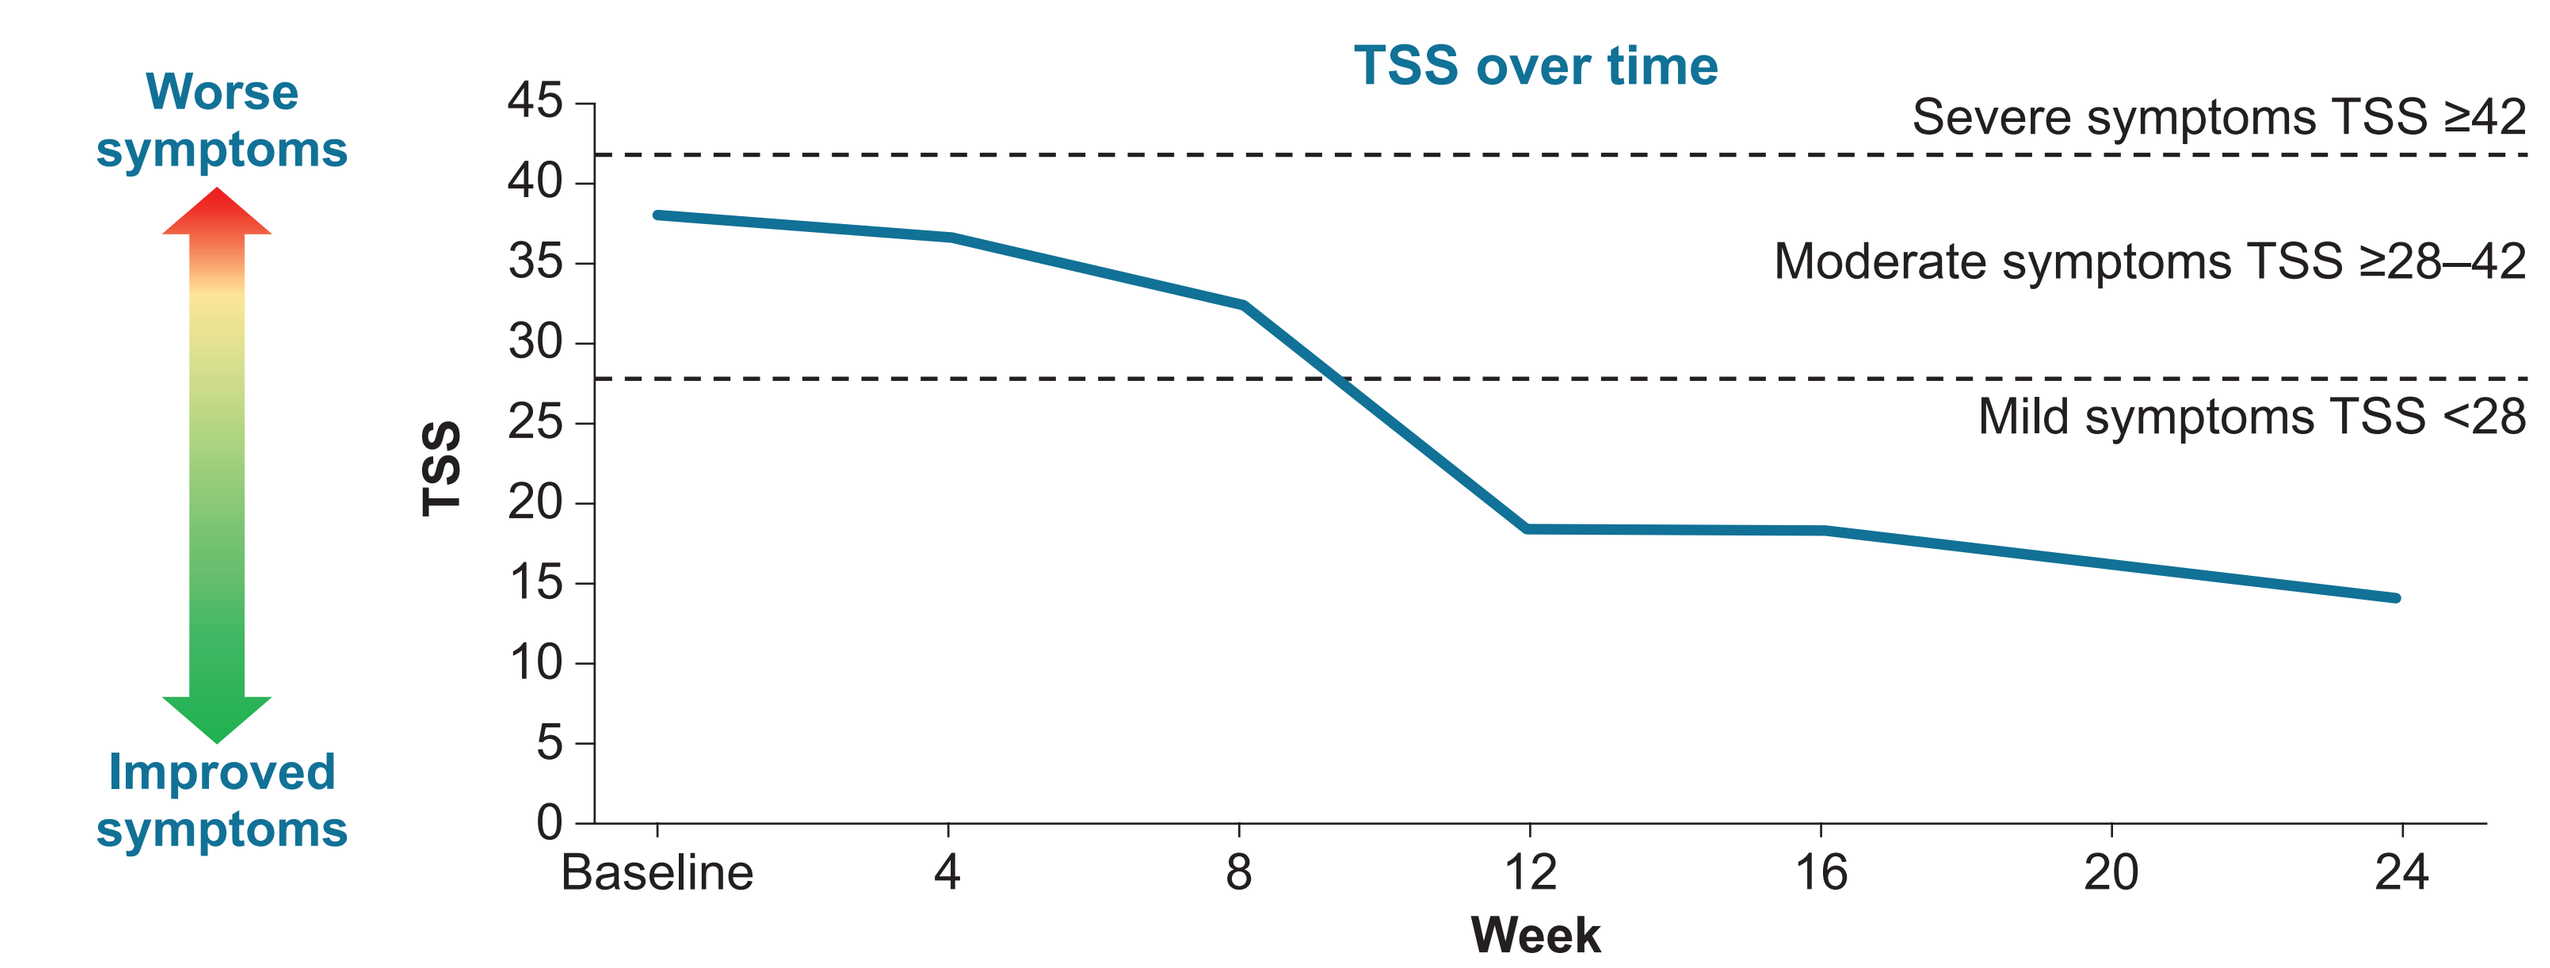

Worse symptoms
Improved symptoms
TSS over time
Severe symptoms TSS ≥42
Moderate symptoms TSS ≥28–42
Mild symptoms TSS <28
TSS
45
40
35
30
25
20
15
10
5
0
Baseline
4
8
12
16
20
24
Week

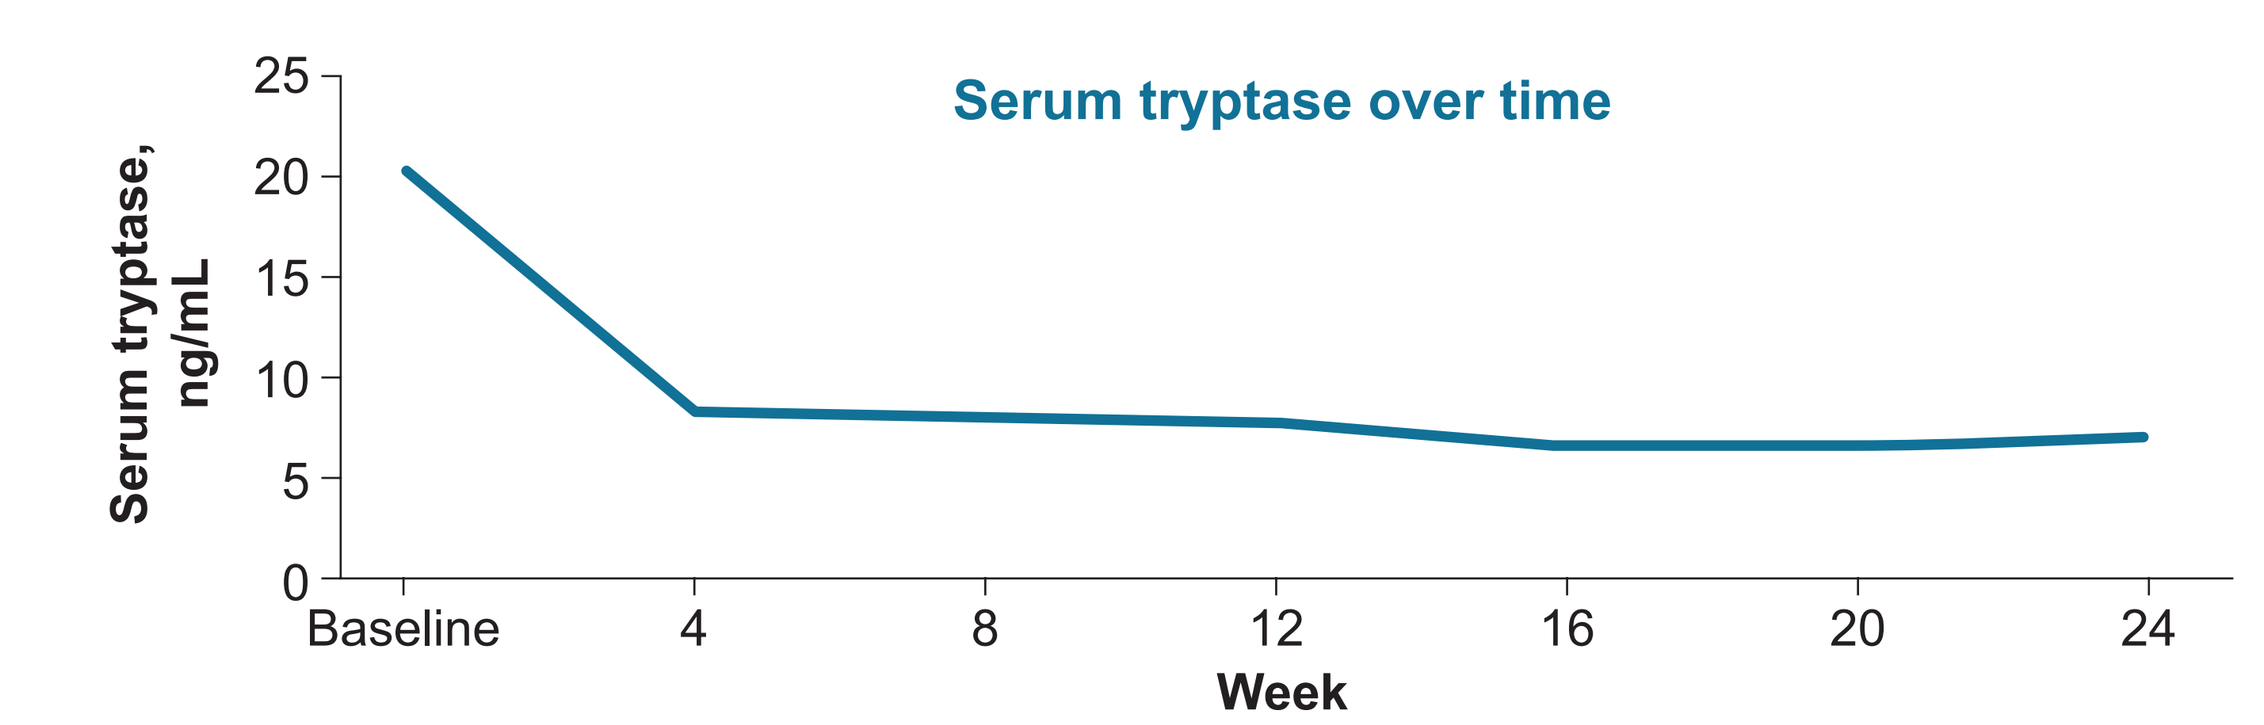

Serum tryptase over time
Serum tryptase, ng/mL
25
20
15
10
5
0
Baseline
4
8
12
16
20
24
Week

Conclusions

- More sensitive assays are needed to aid clinicians in identifying *KIT* D816V mutations, an important minor diagnostic criterion for SM
- *KIT* D816V mutations were detected in 97% of patients when combining ddPCR and superRCA-based assay findings
 - In patients with ISM who had undetectable *KIT* D816V by ddPCR, superRCA detected *KIT* D816V in 78%
- The superRCA assay increased sensitivity limits for mutant-*KIT* D816V in peripheral blood and would facilitate the diagnosis of ISM in patients without detectable *KIT* D816V by currently commercially available tests
- A high symptom burden can still be experienced by patients without detectable *KIT* D816V by ddPCR
- Avapritinib can effectively reduce symptoms and serum tryptase levels in patients who do not have detectable *KIT* D816V by ddPCR
- Further development of superRCA may improve the detection of clonal mast cell disorders
- Until such research-based tests are widely available, bone marrow biopsy should still be performed and remains the standard-of-care for evaluating SM

Acknowledgements

We thank the patients and their families for making the PIONEER study possible. We also thank the investigators and clinical trial teams who participated in the study. Medical writing support was provided by Matthew Nicolas MSc, and Sarah Christopher, PhD of Paragon (a division of Prime, Knutsford, UK). Funded by Blueprint Medicines Corporation. The sponsor reviewed and provided feedback on the presentation. However, the authors had full editorial control and provided final approval of all content.

Conflicts of interest/disclosures

Dr George has received consulting fees and is a study steering committee member for Blueprint Medicines Corporation, BMS/Celgene, Cogent Biosciences, and Incyte.
For all author disclosures, please contact medinfo@blueprintmedicines.com.

Presented at EAC 2025 Congress, May 29–June 01 2025, Palm Beach, FL, USA. Please contact medinfo@blueprintmedicines.com for permission to reprint or distribute.