BLU-808, a Potent and Selective Small Molecule Inhibitor of Wild-Type KIT for Mast Cell Disorders

Poster Number 189

Alexandra Grassian,[1] Andrew Haidle,[1] Sophia AuDuong,[1] James Baker,[1] Guangyan Du,[1] Megan Hatlen,[1] Melissa Hiki,[1] Karen Ho,[1] Wei Hu,[1] Kevin Keane,[1] Thiwanka Samarakoon Mudiyanselage,[1] Emanuele Perola,[1] Christine Pien,[1] Priyanka Sawant,[1] Doug Wilson,[1] Rob Meissner,[1] Jason Brubaker,[1] Scott Ribich,[1] Joseph Kim[1]

[1]Blueprint Medicines Corporation, Cambridge, MA, USA

Background

KIT is a transmembrane receptor tyrosine kinase, which plays important roles in mast cell differentiation, proliferation, survival, chemotaxis, and activation. The KIT ligand, more commonly termed Stem Cell Factor (SCF), binds to KIT and induces the formation of homodimers/oligomers, trans phosphorylation, and activation of downstream intracellular signaling cascades.[1] In murine models with inactivating mutations in either SCF or KIT, a dramatic decrease in the number of mast cells is observed.[2–7] Conversely, in patients with systemic mastocytosis, activating KIT mutations lead to a pathological overabundance of mast cells.[8–10] Other diseases, such as chronic urticaria, are characterized by inappropriate mast cell activation but are wild-type (WT) for KIT[11,12] and could benefit from a small molecule therapeutic agent targeting WT KIT.

Figure 1. Mast cells in allergic reactions and immune response; WT KIT is a key player in mast cells

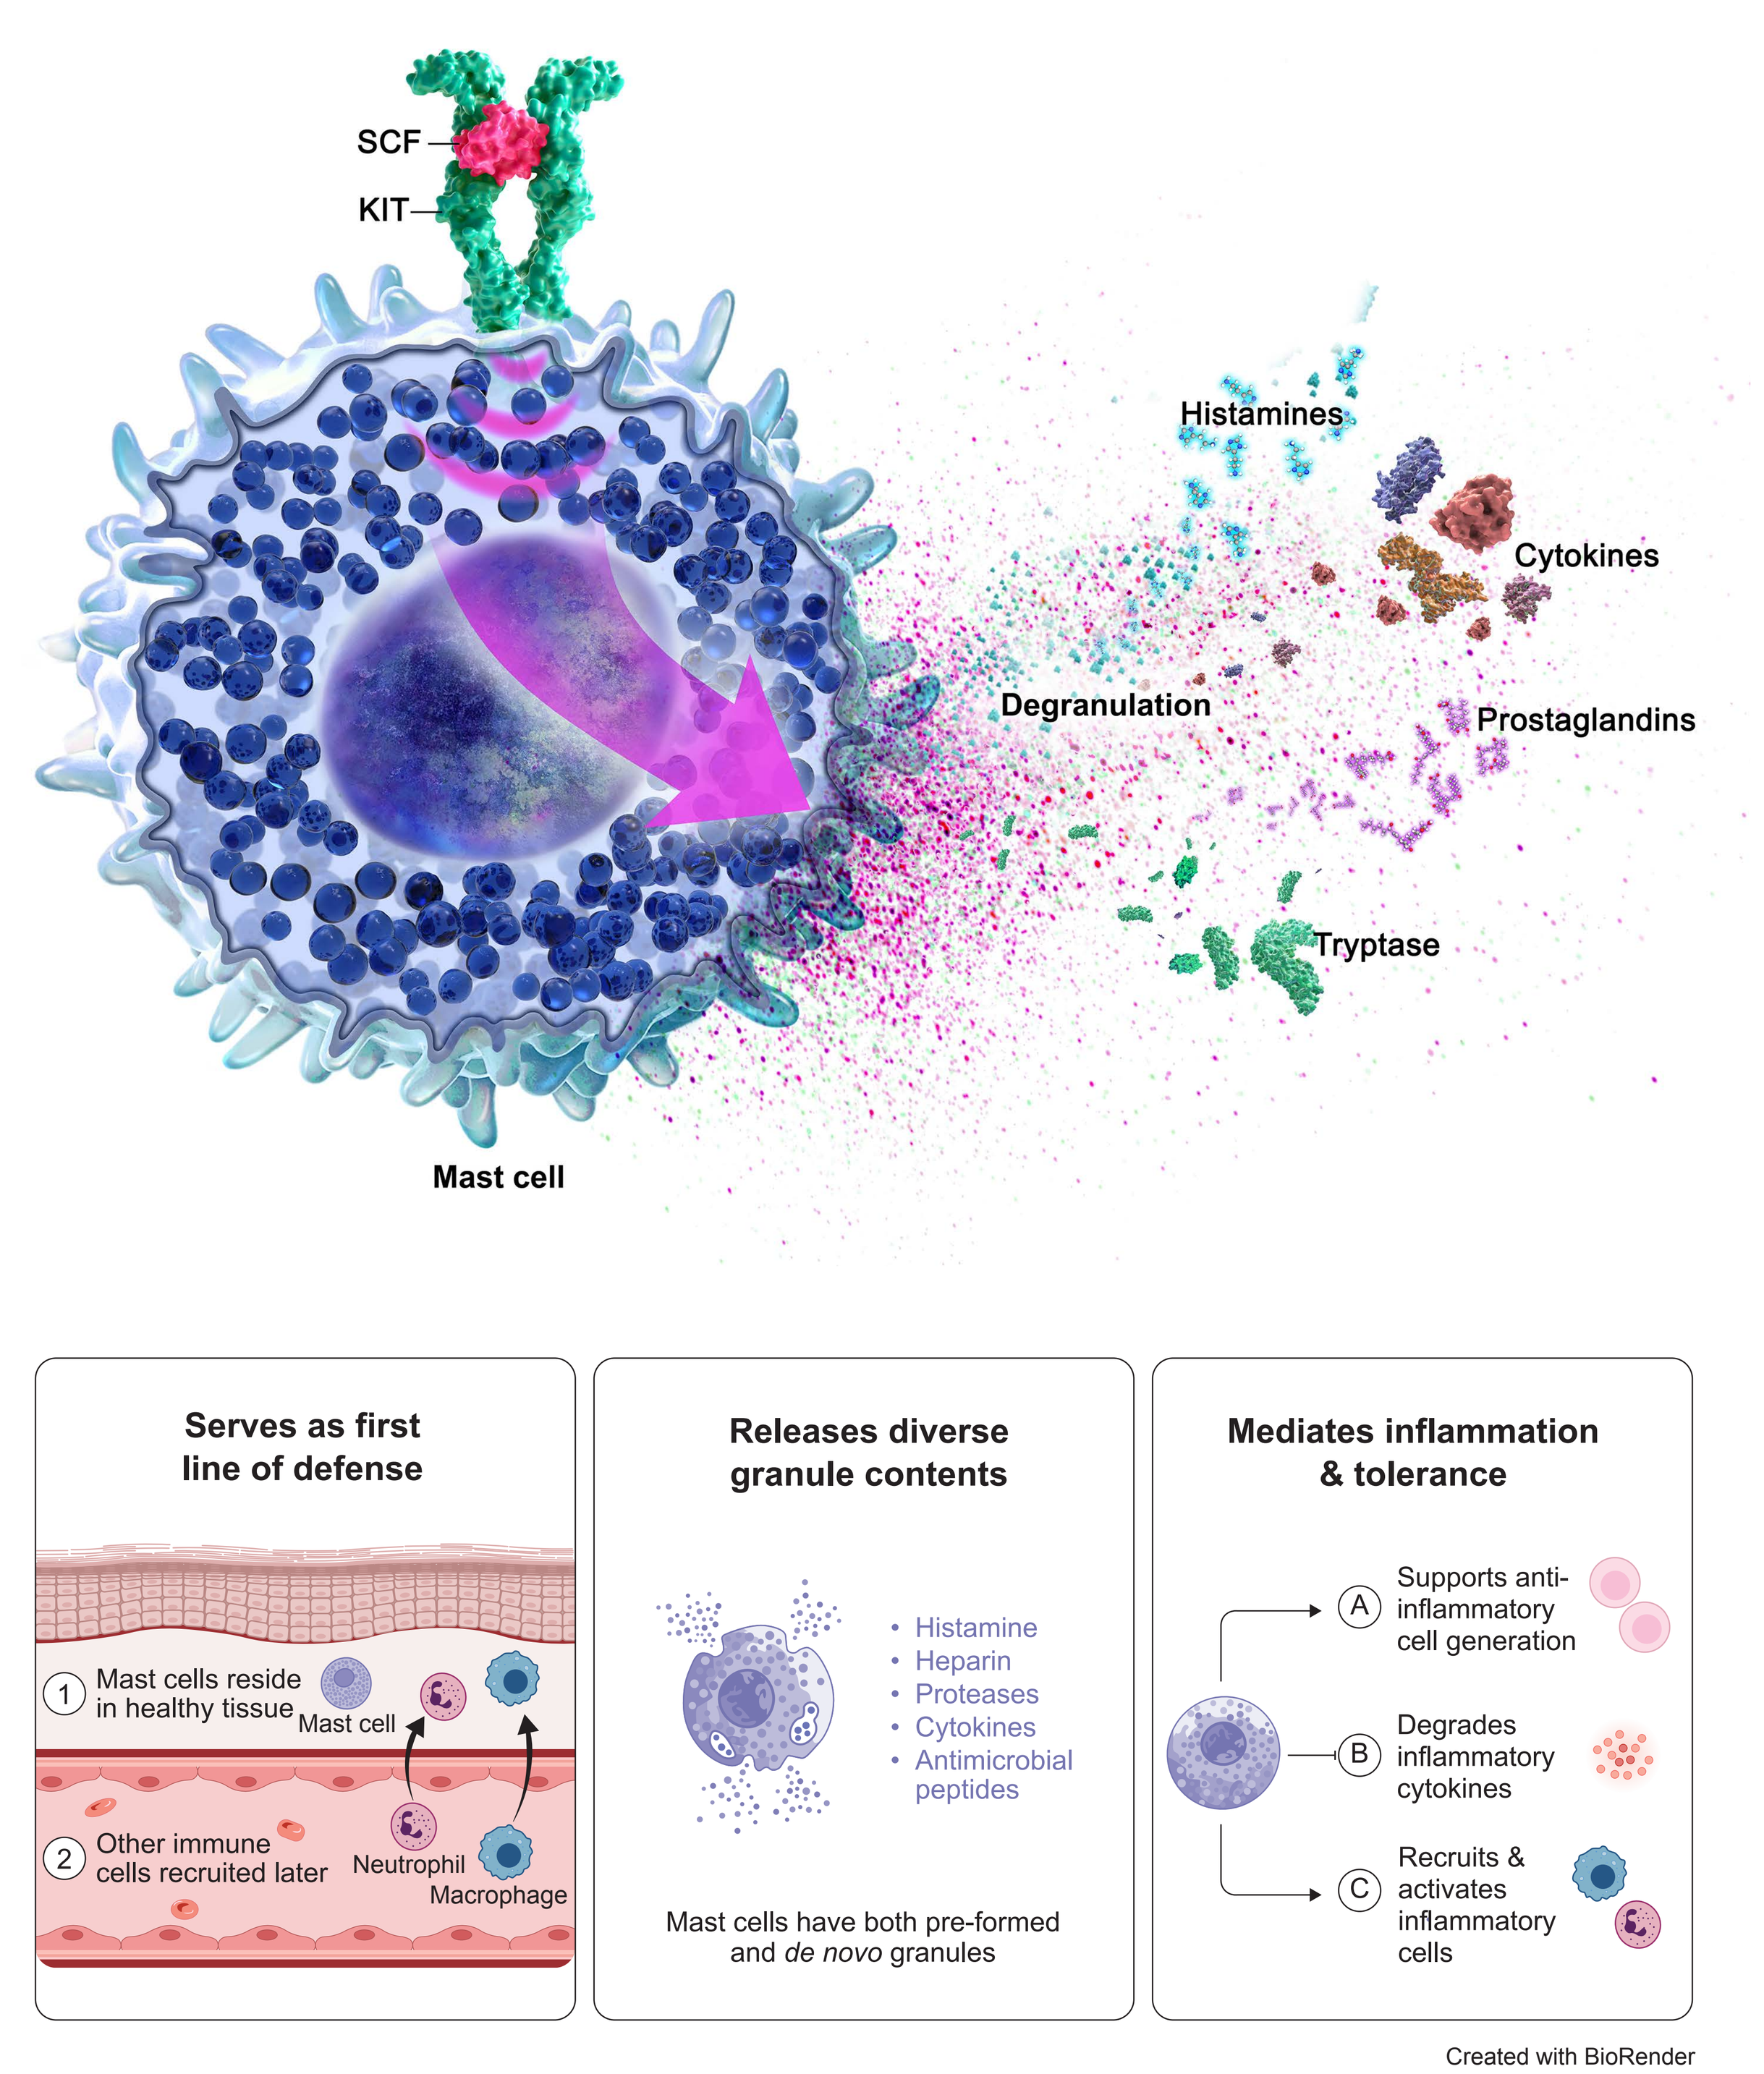
SCF
KIT
Histamines
Cytokines
Degranulation
Prostaglandins
Tryptase
Mast cell
Serves as first line of defense
Mast cells reside in healthy tissue
Mast cell
Other immune cells recruited later
Neutrophil
Macrophage
Releases diverse granule contents
Histamine
Heparin
Proteases
Cytokines
Antimicrobial peptides
Mast cells have both pre-formed and *de novo* granules
Mediates inflammation & tolerance
Supports anti-inflammatory cell generation
Degrades inflammatory cytokines
Recruits & activates inflammatory cells
Created with BioRender

SCF, stem cell factor; WT, wild-type.

Methods

- Cell assays to measure KIT-dependent proliferation and SCF-mediated KIT phosphorylation were used to assess potency
- Selectivity against the structurally related kinases platelet-derived growth factor receptor alpha/beta (PDGFRA/B), FMS-like tyrosine kinase 3 (FLT3), and colony stimulating factor 1 receptor (CSF1R), as well as the broader kinome was assessed by Kinomescan
- Inhibition of degranulation was evaluated in human-derived mast cells and in an *in vivo* rodent histamine release model
- Inhibition of asthma-like symptom was assessed in a rodent ovalbumin-induced asthma model

Results

Table 1. BLU-808 is an investigational potent and selective inhibitor of WT KIT

	BLU-808
pKIT cellular IC_{50} (nM)	0.37
WT KIT-dependent proliferation IC_{50} (nM)	1.3
Human-derived CD34⁺ mast cells: inhibition of CD63 extracellular expression IC_{50} (nM)	2.7
Human-derived CD34⁺ mast cells: inhibition of histamine degranulation IC_{50} (nM)	8.6
PDGFRA/B/FLT3 selectivity[a]	>300x/>400x/>9600x
S(10) @ 3 µM	0.042
CSF1R Kd selectivity	>800x
Brain penetrance ($Kp_{u,u}$)	0.021

CSF1R, colony stimulating factor 1 receptor; FLT3, FMS-like tyrosine kinase 3; IC_{50}, half-maximal inhibitory concentration; PDGFRA/B, platelet-derived growth factor receptor alpha/beta; pKIT, phosphorylated KIT; S(10) @ 3 µM, selectivity score at a concentration of 3 µM; $Kp_{u,u}$, unbound brain to plasma partition coefficient.
[a]Determined in a cellular assay.

Figure 2. BLU-808 inhibits WT KIT *in vitro* and is selective over key off-targets PDGFRA/B and FLT3

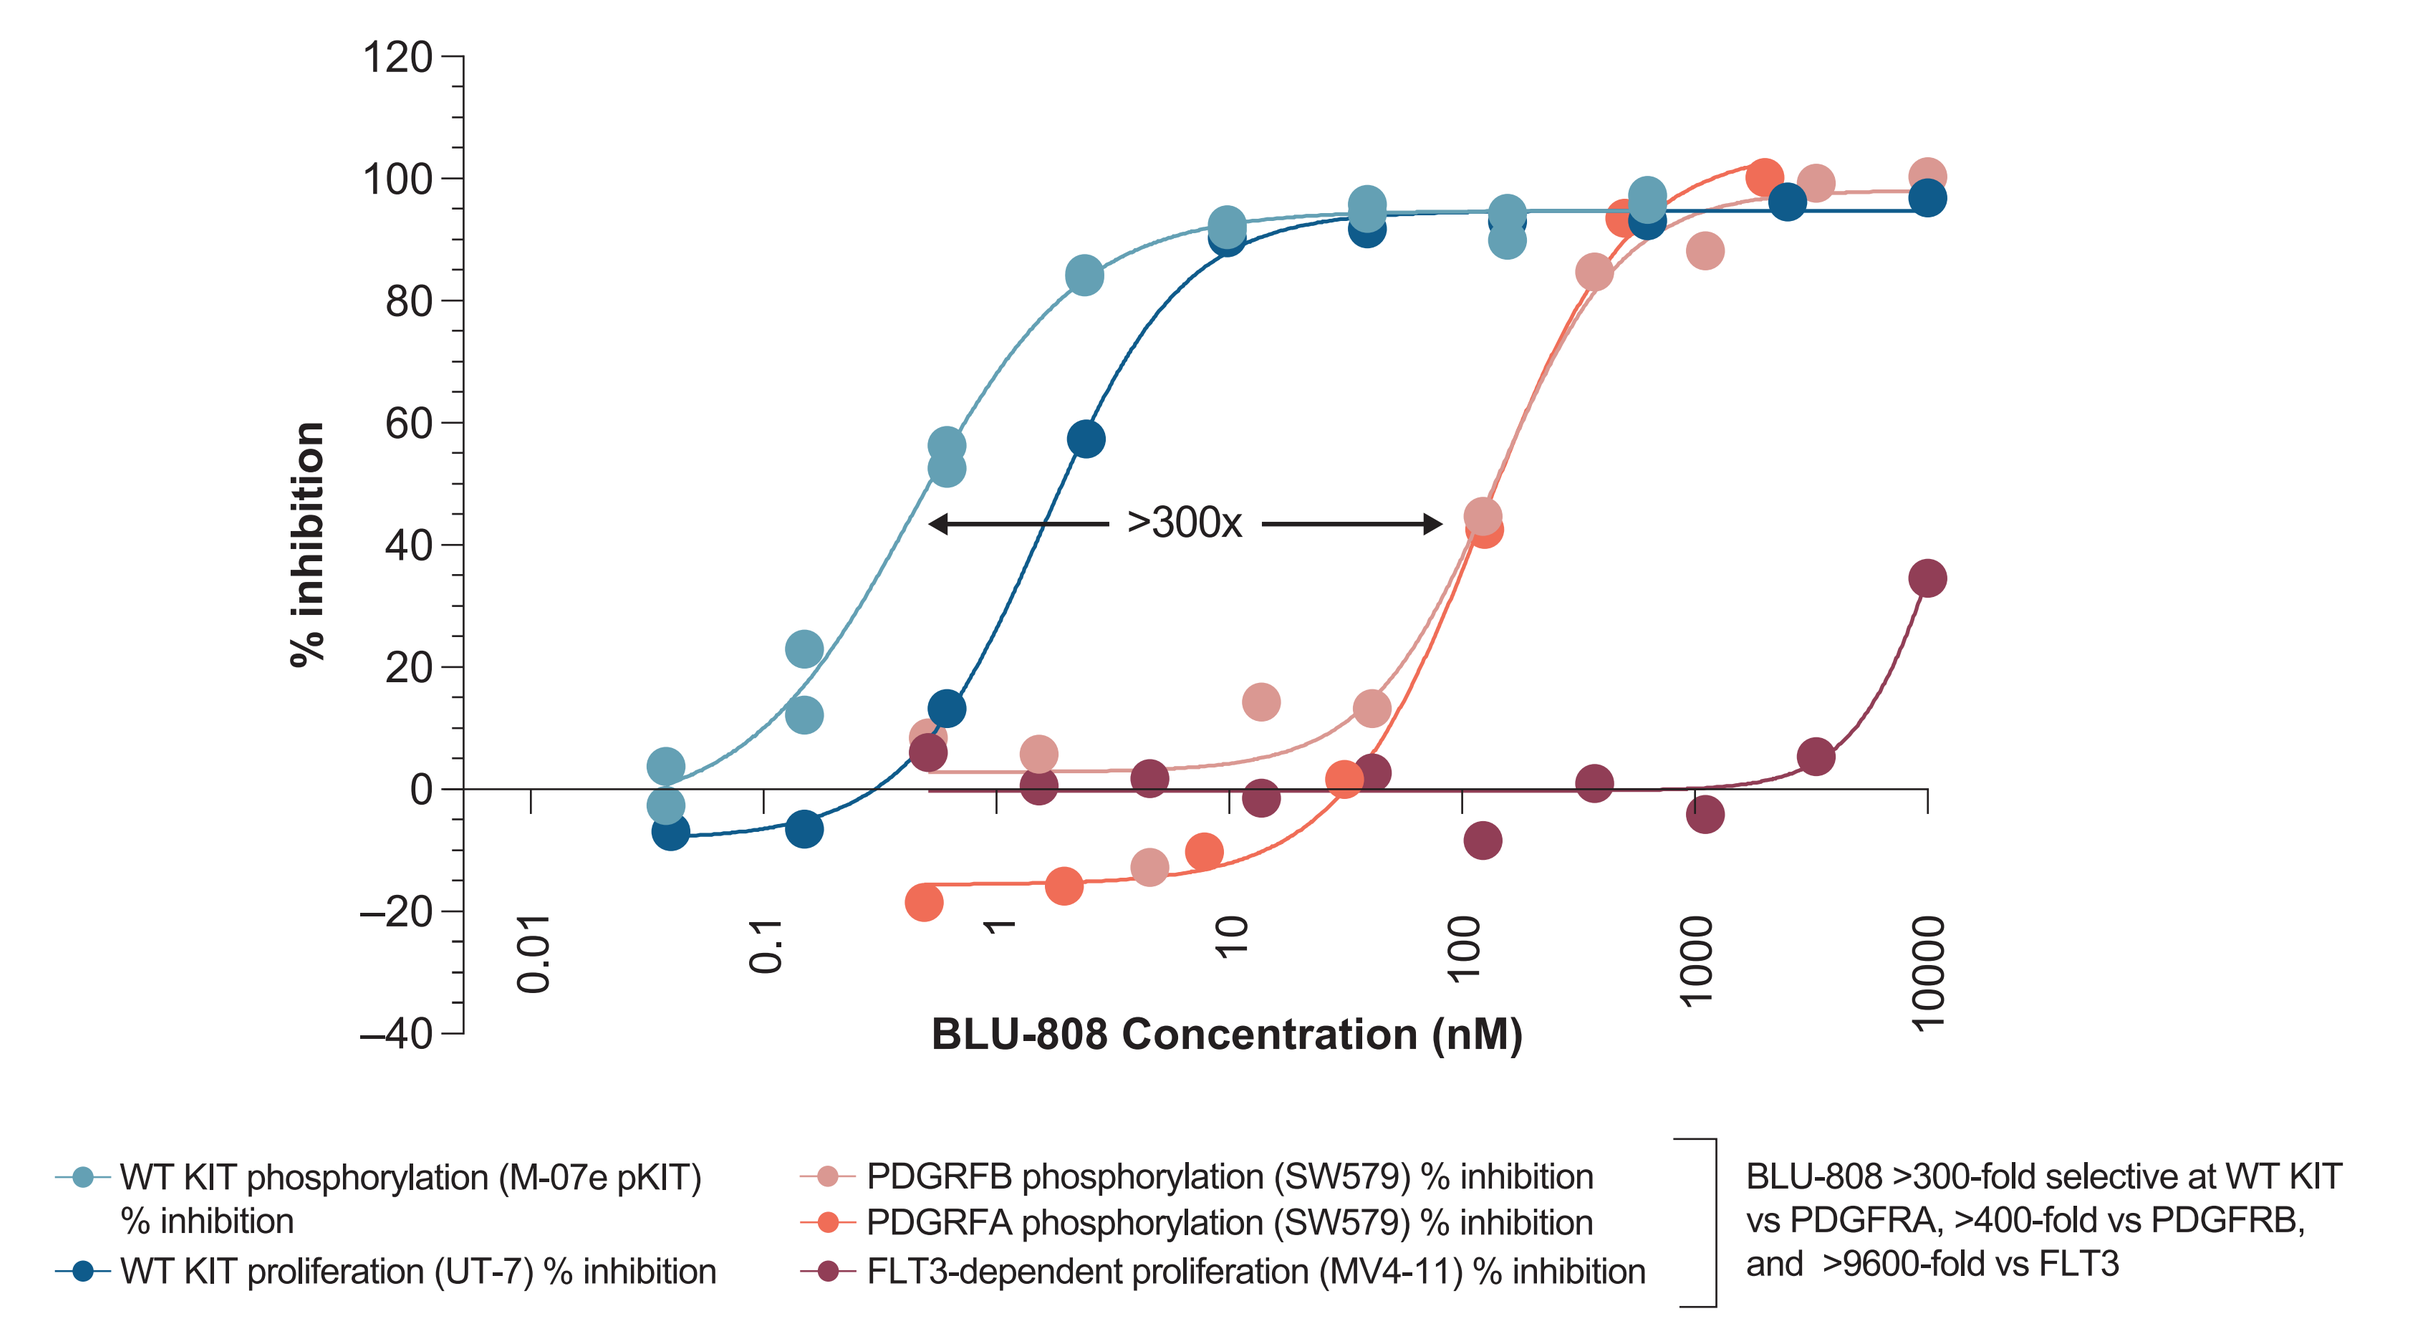
% inhibition
>300x
BLU-808 Concentration (nM)
WT KIT phosphorylation (M-07e pKIT) % inhibition
WT KIT proliferation (UT-7) % inhibition
PDGRFB phosphorylation (SW579) % inhibition
PDGRFA phosphorylation (SW579) % inhibition
FLT3-dependent proliferation (MV4-11) % inhibition
BLU-808 >300-fold selective at WT KIT vs PDGFRA, >400-fold vs PDGFRB, and >9600-fold vs FLT3

- BLU-808 shows potent inhibition of SCF-stimulated pKIT in M-07e cells and SCF-stimulated proliferation in UT-7 cells (**Figure 2**). BLU-808 is selective over key off-target proteins, PDGFRA and B phosphorylation in PDGFBB-stimulated SW579 cells and FLT3-dependent proliferation in MV4-11 cells

Figure 3. Human mast cell assays correlate with inhibition of WT KIT

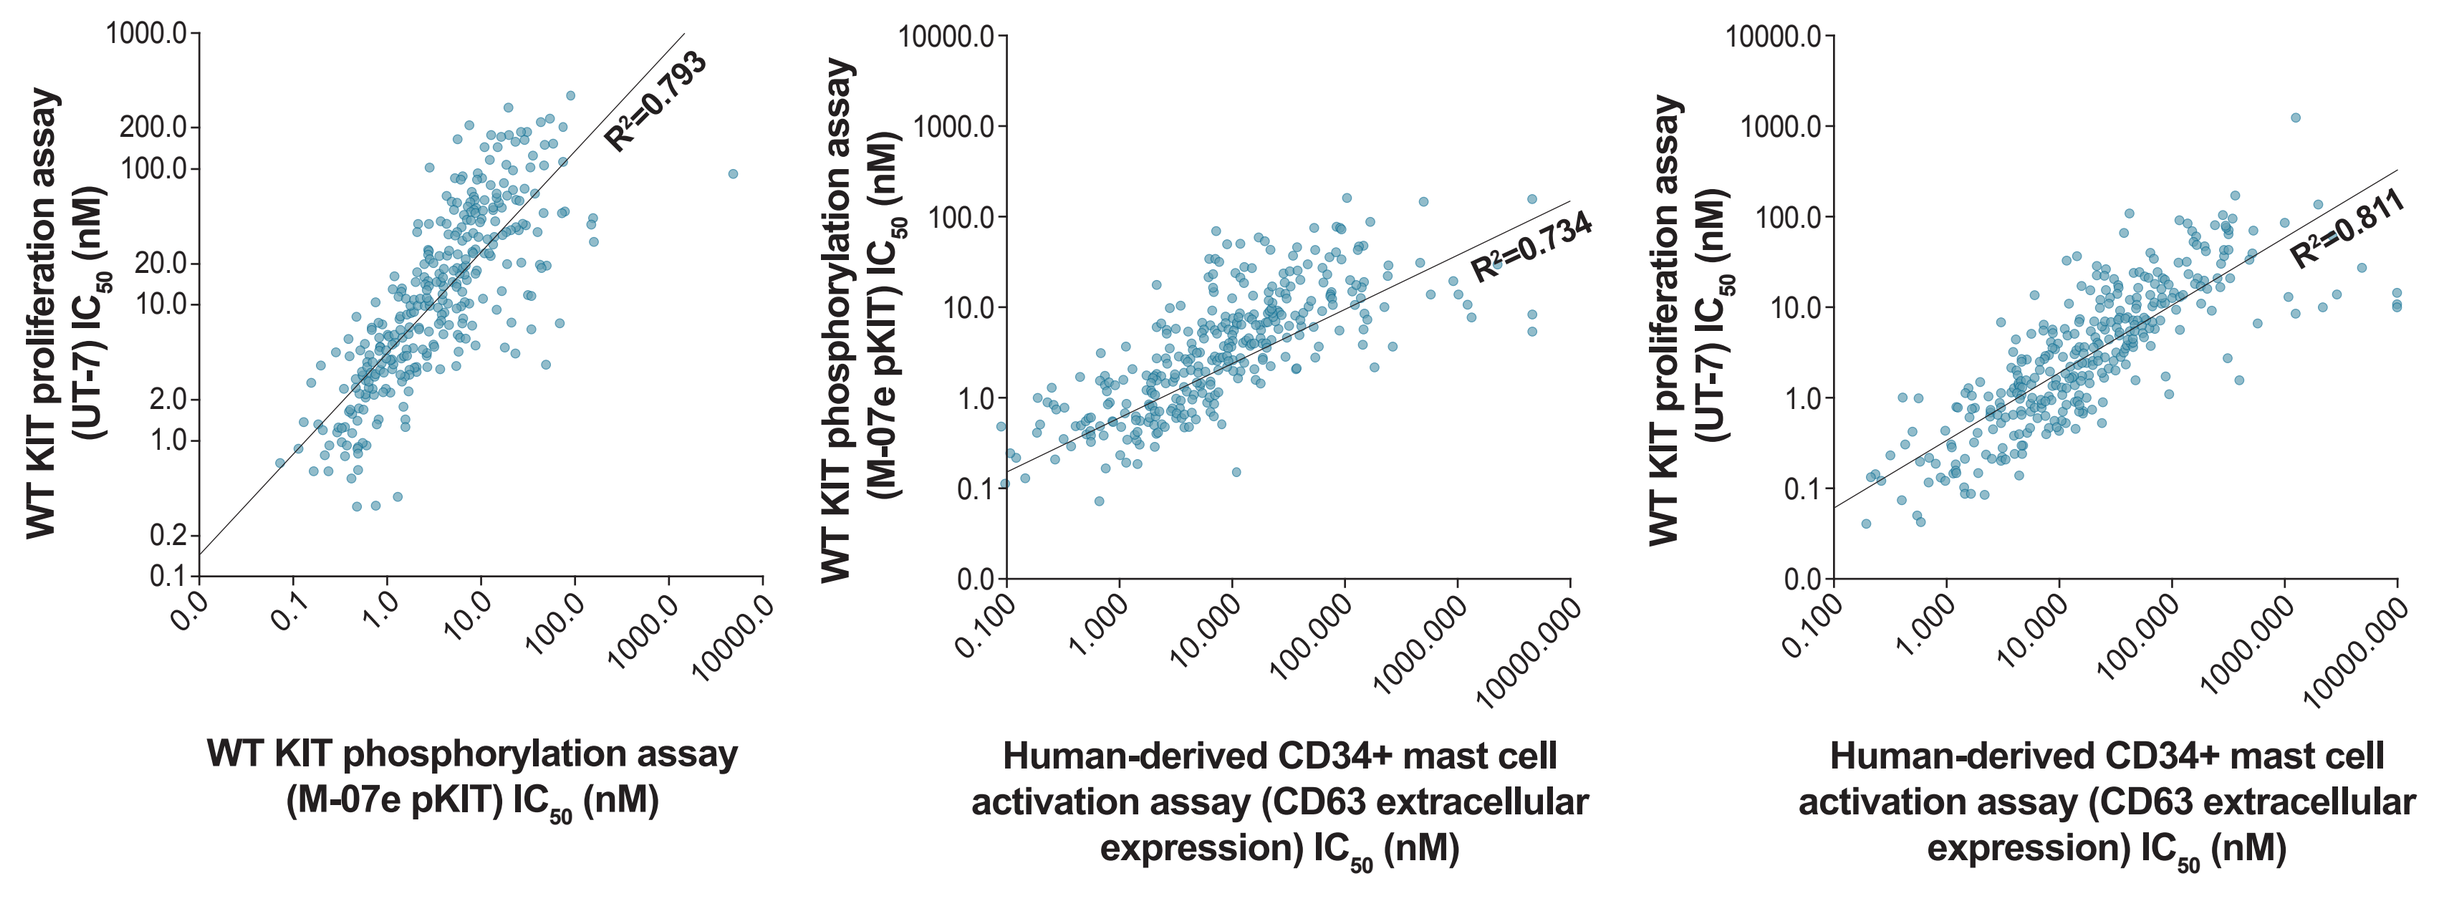
WT KIT proliferation assay (UT-7) IC_{50} (nM)
R^2=0.793
WT KIT phosphorylation assay (M-07e pKIT) IC_{50} (nM)
WT KIT phosphorylation assay (M-07e pKIT) IC_{50} (nM)
R^2=0.734
Human-derived CD34+ mast cell activation assay (CD63 extracellular expression) IC_{50} (nM)
WT KIT proliferation assay (UT-7) IC_{50} (nM)
R^2=0.811
Human-derived CD34+ mast cell activation assay (CD63 extracellular expression) IC_{50} (nM)

- Human-derived CD34+ mast cell activation (CD63 expression assay) correlated with cellular WT KIT assays (**Figure 3**)

Figure 4. BLU-808 inhibits activation of human-derived CD34⁺ mast cells

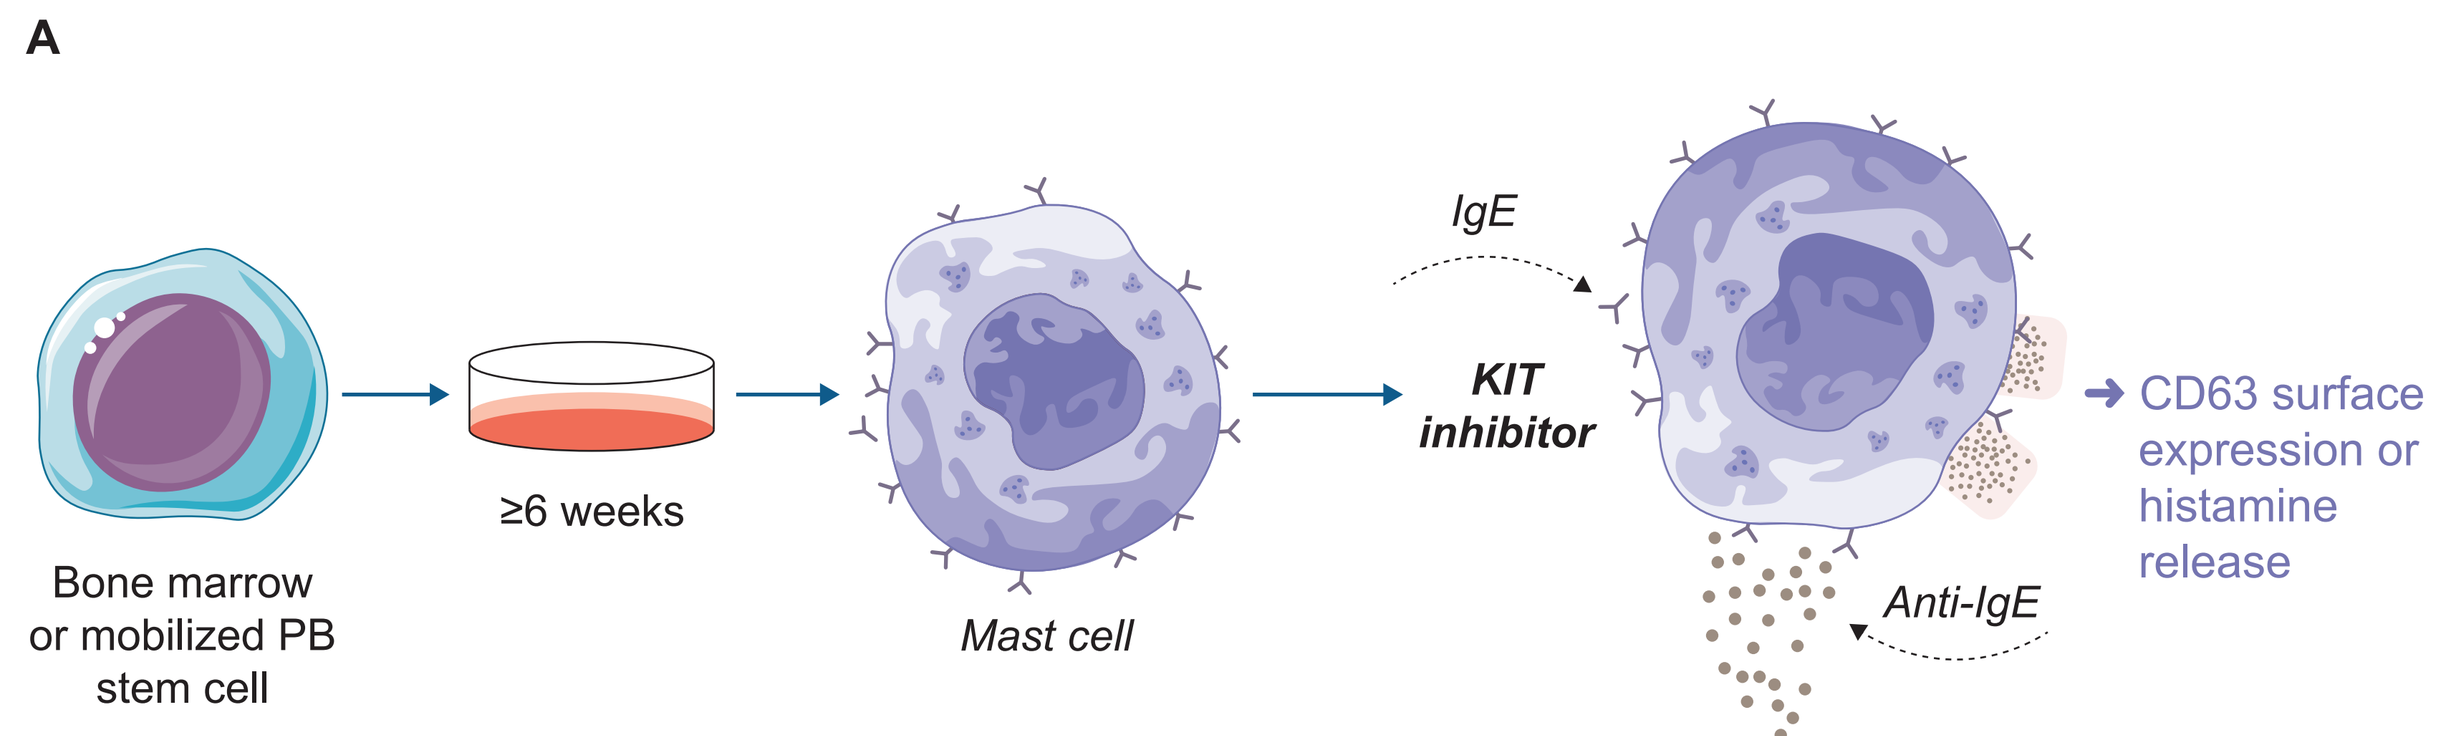
A
Bone marrow or mobilized PB stem cell
≥6 weeks
Mast cell
IgE
KIT inhibitor
Anti-IgE
CD63 surface expression or histamine release

Decreased CD63 expression and histamine release in treated human-derived CD34+ mast cells stimulated with IgE and anti-IgE

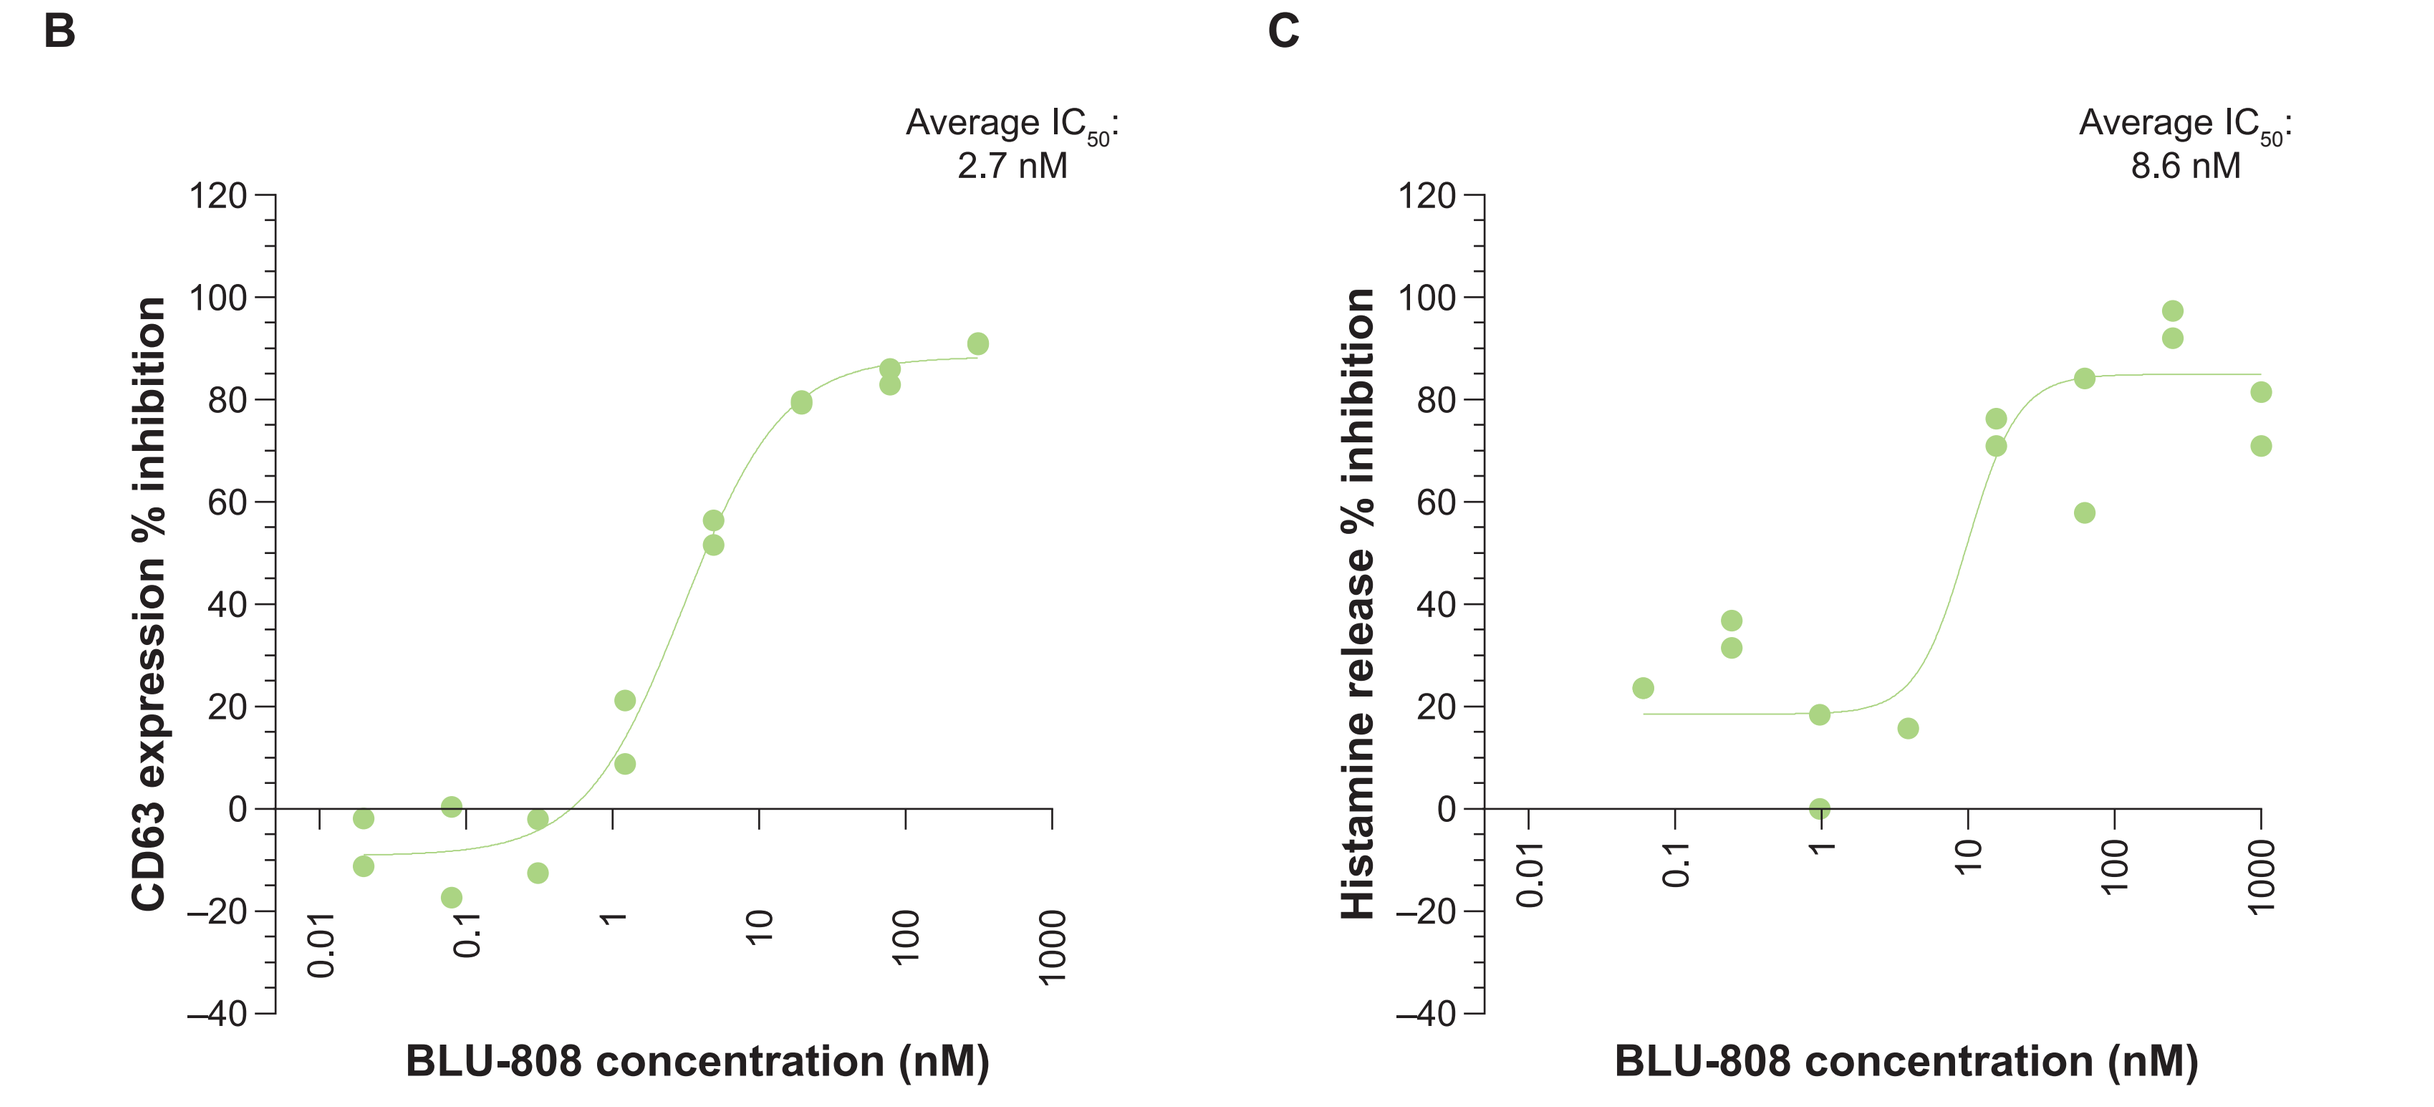
B
Average IC_{50}: 2.7 nM
CD63 expression % inhibition
BLU-808 concentration (nM)
C
Average IC_{50}: 8.6 nM
Histamine release % inhibition
BLU-808 concentration (nM)

IgE, immunoglobulin E; PB, peripheral blood.

- BLU-808 is potent in two human-derived CD34⁺ mast cell assays (**Figure 4A**), CD63 extracellular staining (**Figure 4B**) and histamine release (**Figure 4C**)

Figure 5. BLU-808 inhibits degranulation of human-derived CD34⁺ mast cells

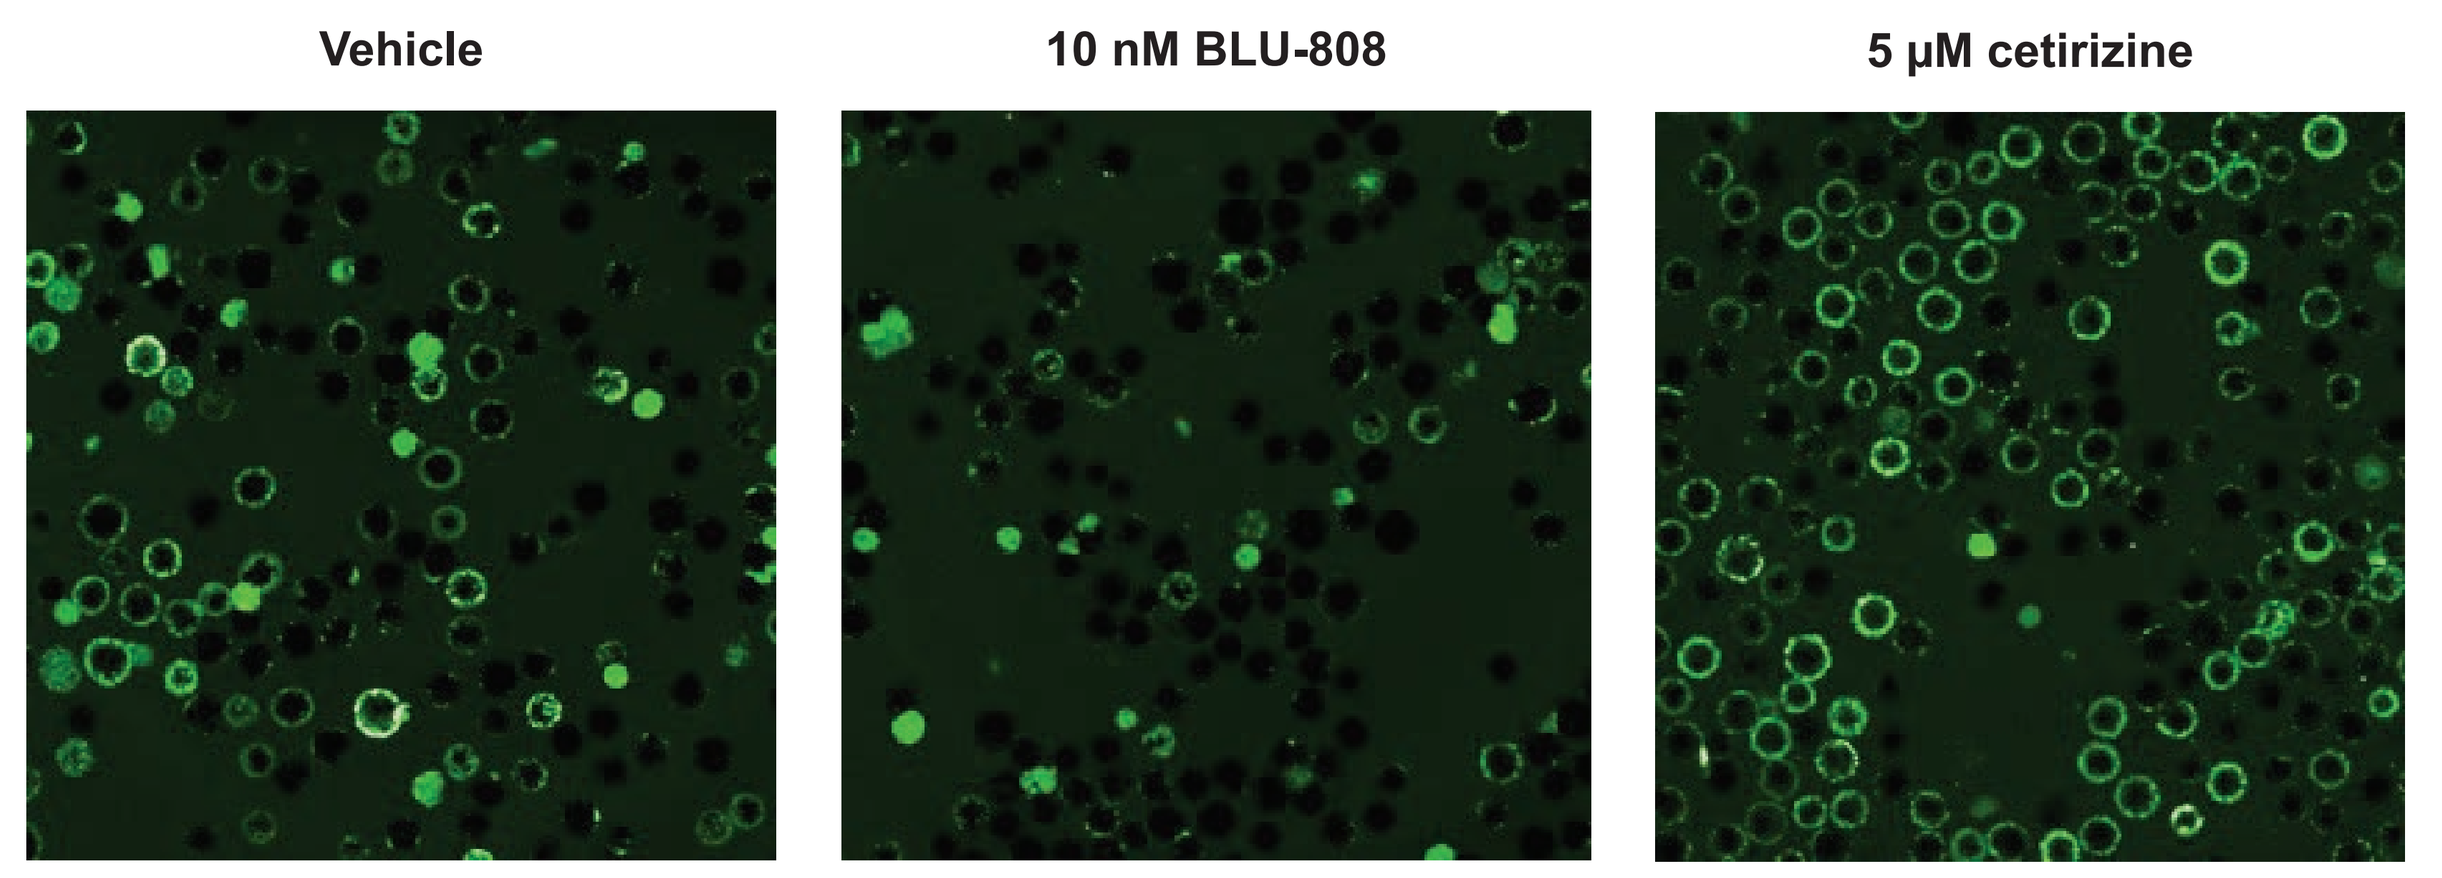
Vehicle
10 nM BLU-808
5 µM cetirizine

Time-lapse video of degranulation in mast cells treated with vehicle, 10 nM BLU-808, and 5 µM cetirizine can be accessed via this QR code:

- Cetirizine is an antihistamine that competes with histamine for binding to cells but does not affect degranulation in mast cells at lower concentrations[13,14]
- Mast cells were labeled with Avidin.488 to visualize degranulation. Following stimulation with immunoglobulin E (IgE) and anti-IgE, the green fluorescence indicates that degranulation occurred in mast cells treated with vehicle and 5 µM cetirizine. BLU-808 inhibited degranulation and reduced fluorescence intensity (**Figure 5**)

Figure 6. In an exposure-dependent manner, BLU-808 can decrease mast cell number after 7 days of dosing in rats

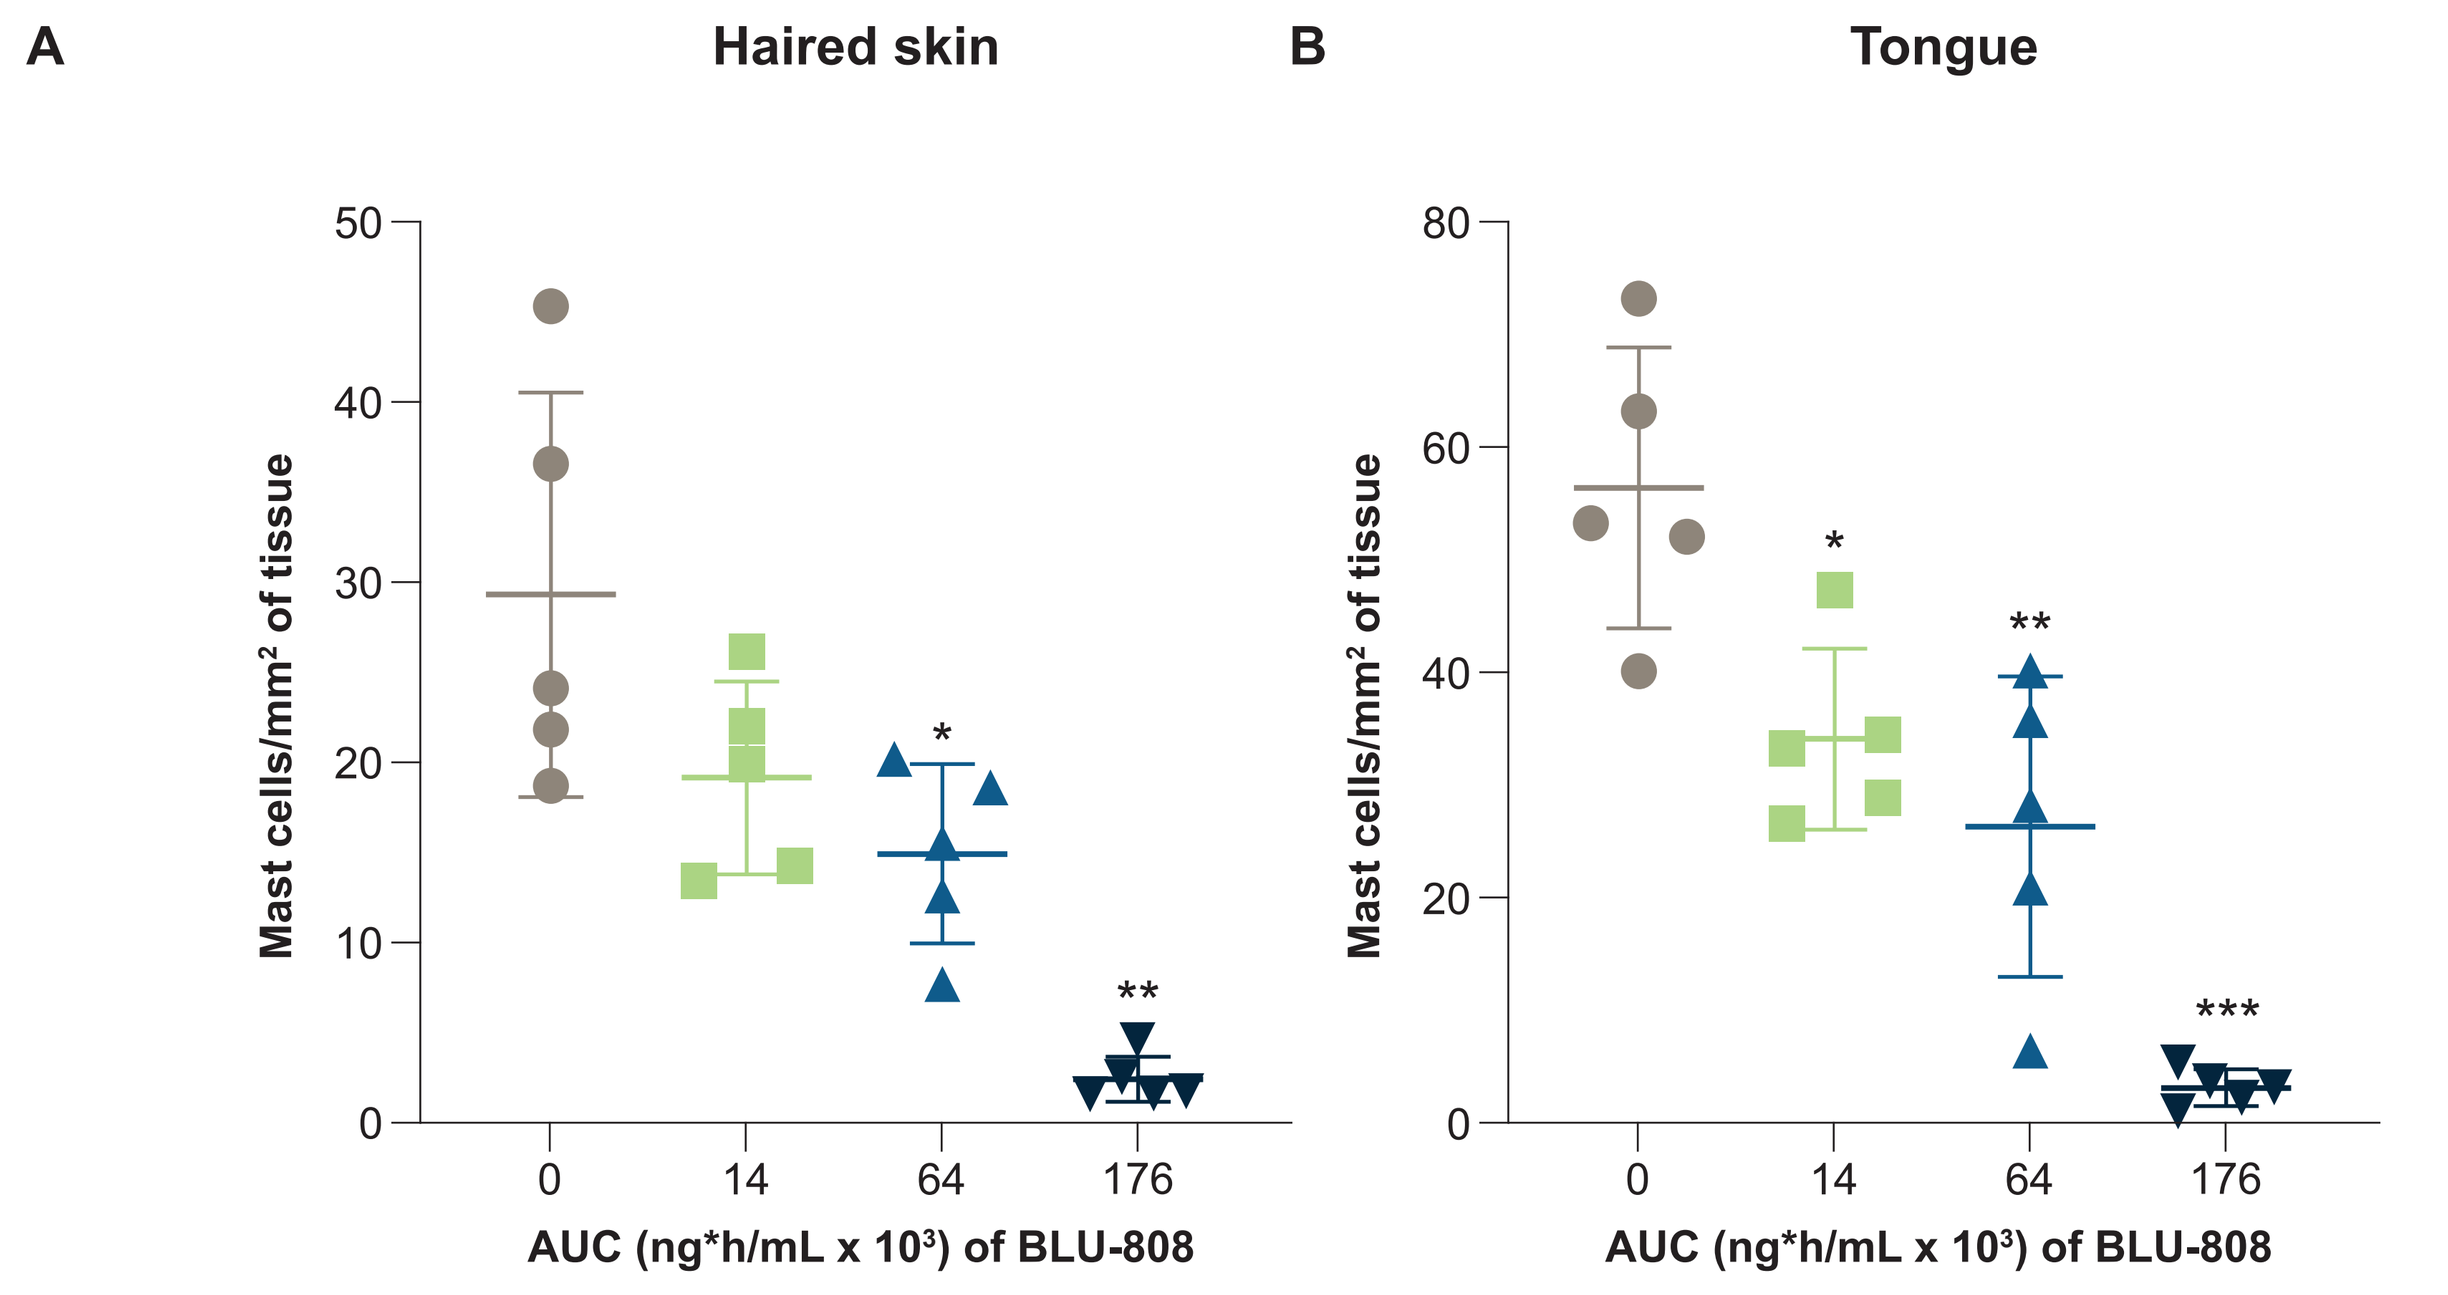
A
Haired skin
Mast cells/mm² of tissue
AUC (ng*h/mL x 10³) of BLU-808
B
Tongue
Mast cells/mm² of tissue
AUC (ng*h/mL x 10³) of BLU-808

Total mast cell % reduction from control:	--	35%	49%	92%	--	40%	54%	95%

AUC, area under the curve. Horizontal lines represent mean and standard deviation. *, *P*<0.05; **, *P*<0.005; ***, *P*<0.0001; all per 2-way analysis of variance with Tukey's correction for multiple comparisons.

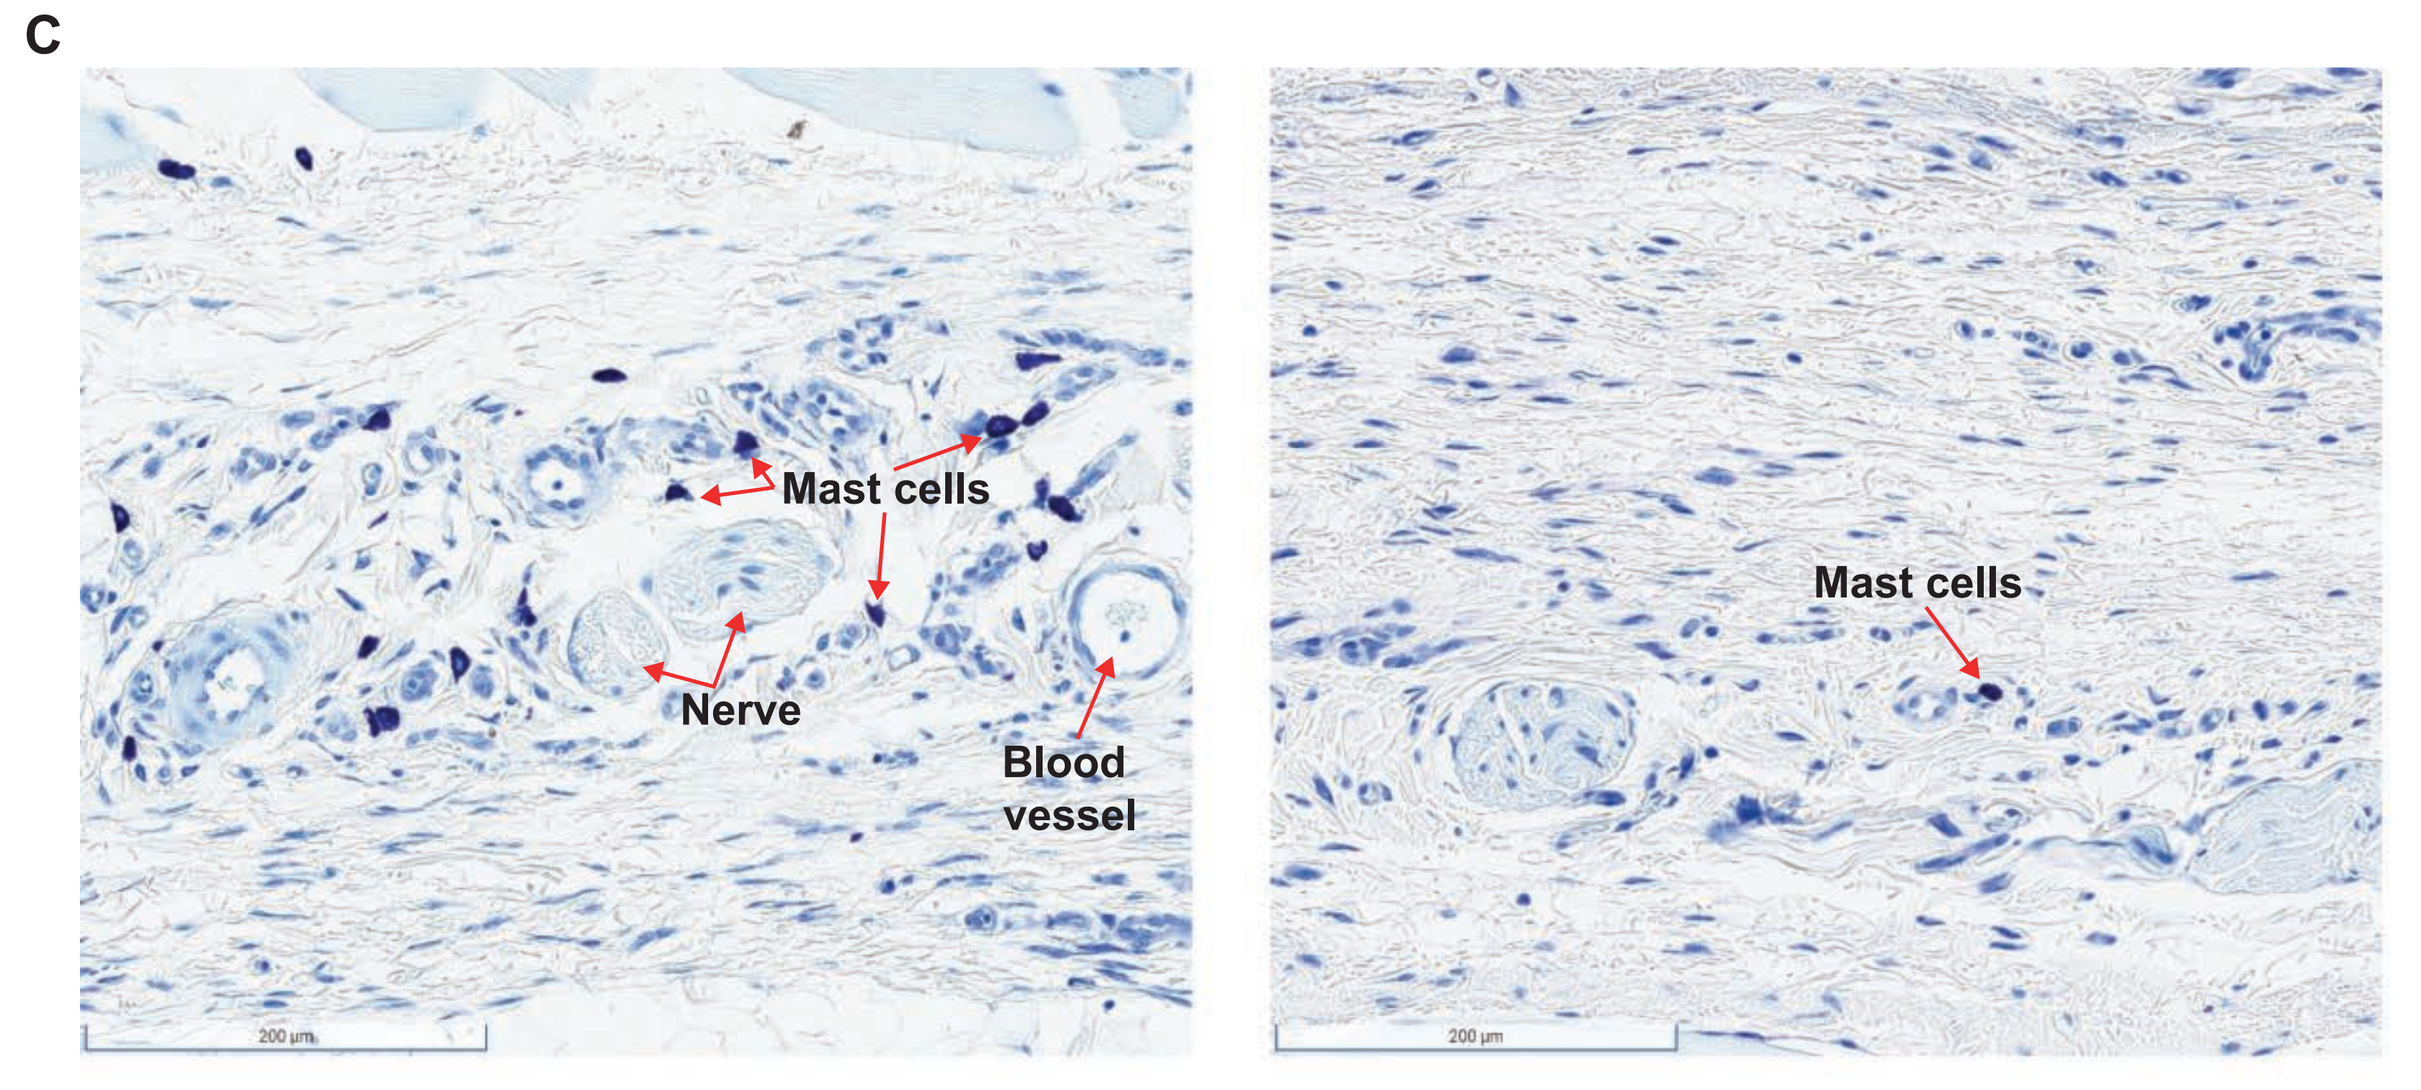
C
Mast cells
Nerve
Blood vessel
Mast cells

Control animal **BLU-808 treated**

Haired skin: Subcutis at high magnification, toluidine blue staining

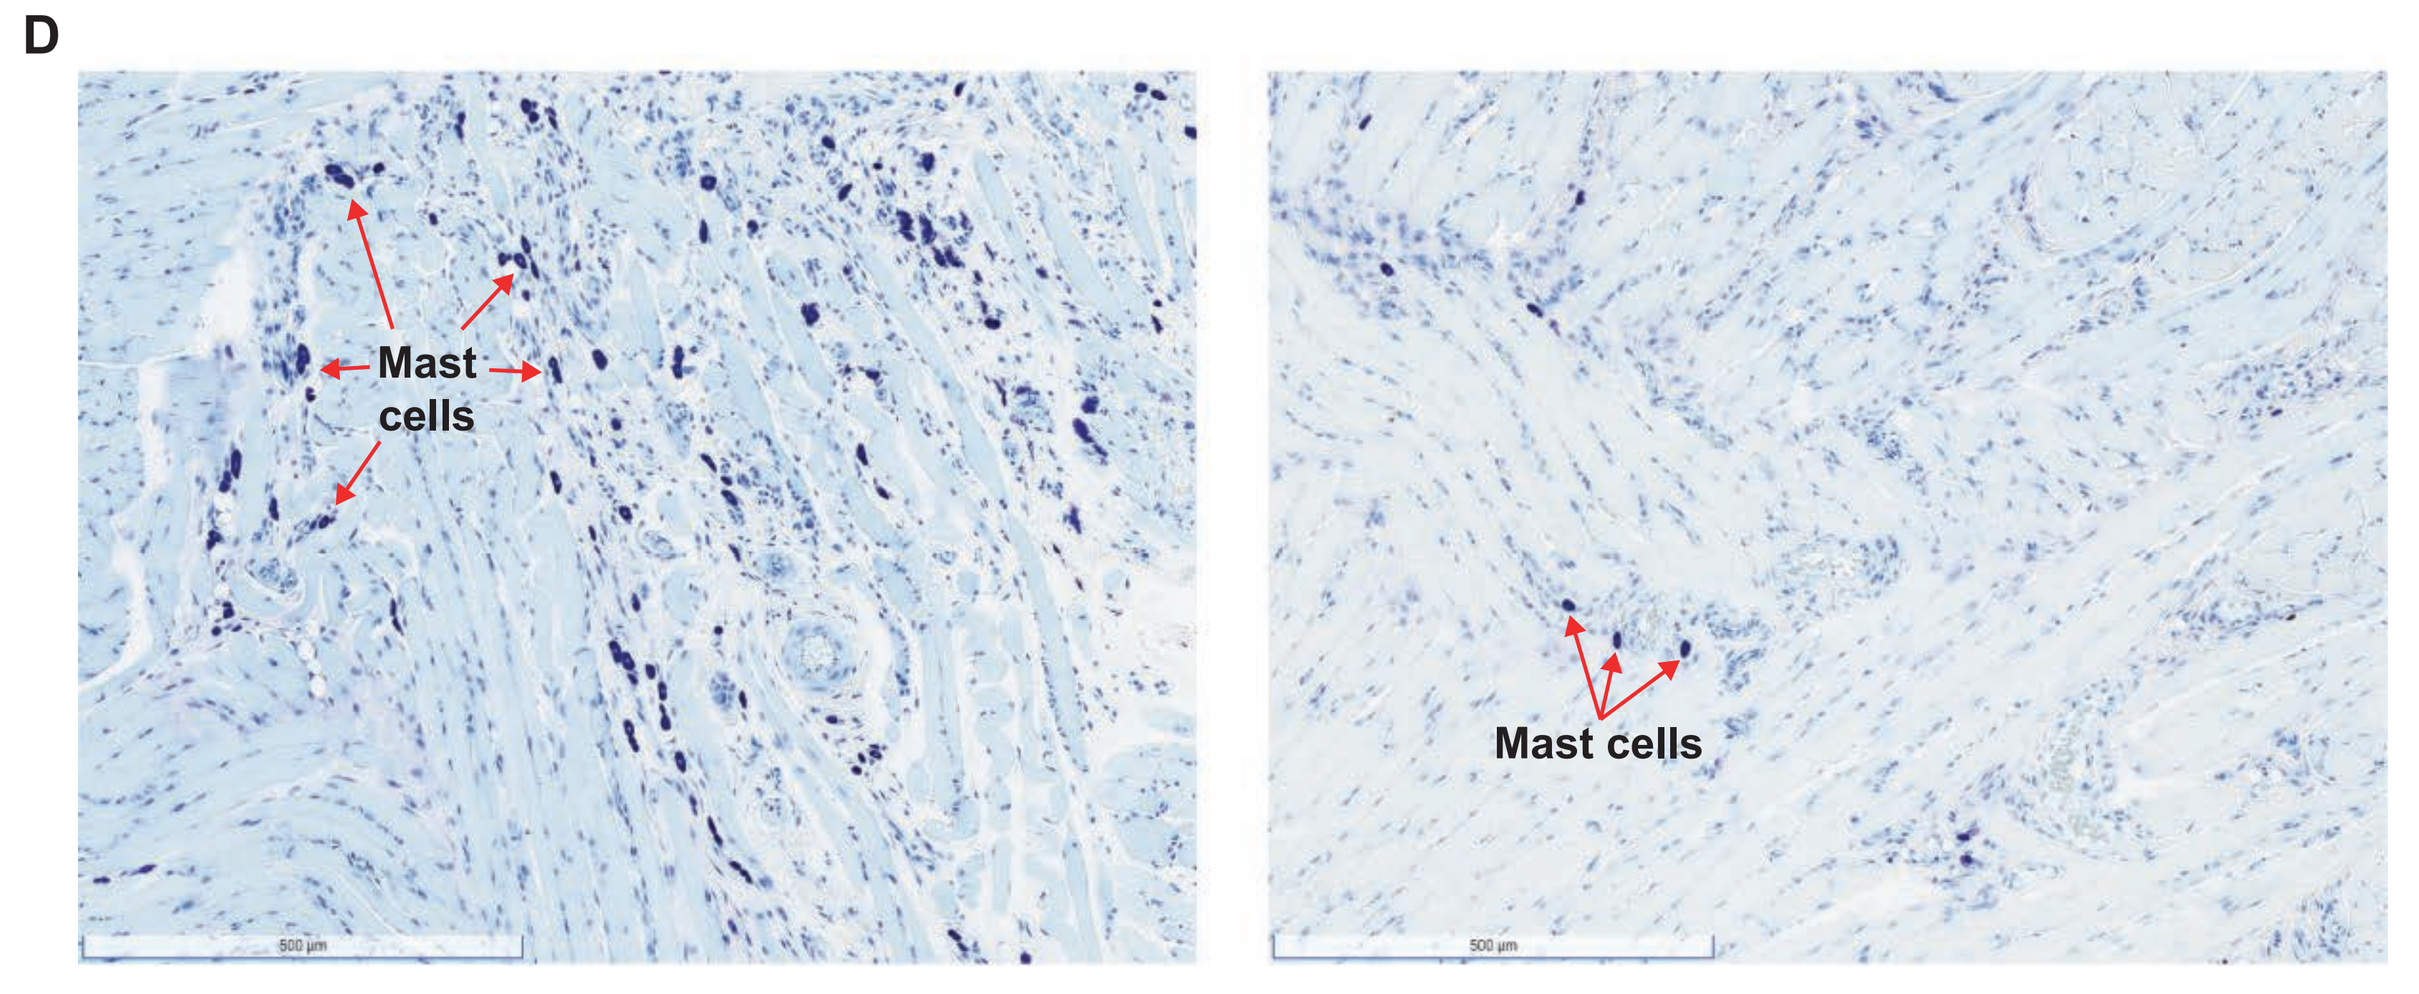
D
Mast cells
Mast cells

Control animal **BLU-808 treated**

Tongue: Muscular center at high magnification, toluidine blue staining

- BLU-808 was administered for 7 days at different specific doses in rats. Mast cells were quantified by toluidine blue staining and showed a dose-dependent reduction in the haired skin (**Figure 6A**) and tongue (**Figure 6B**). Representative images are shown in **Figures 6C & 6D**

Figure 7. A single dose of BLU-808 inhibits SCF-induced histamine release in mice

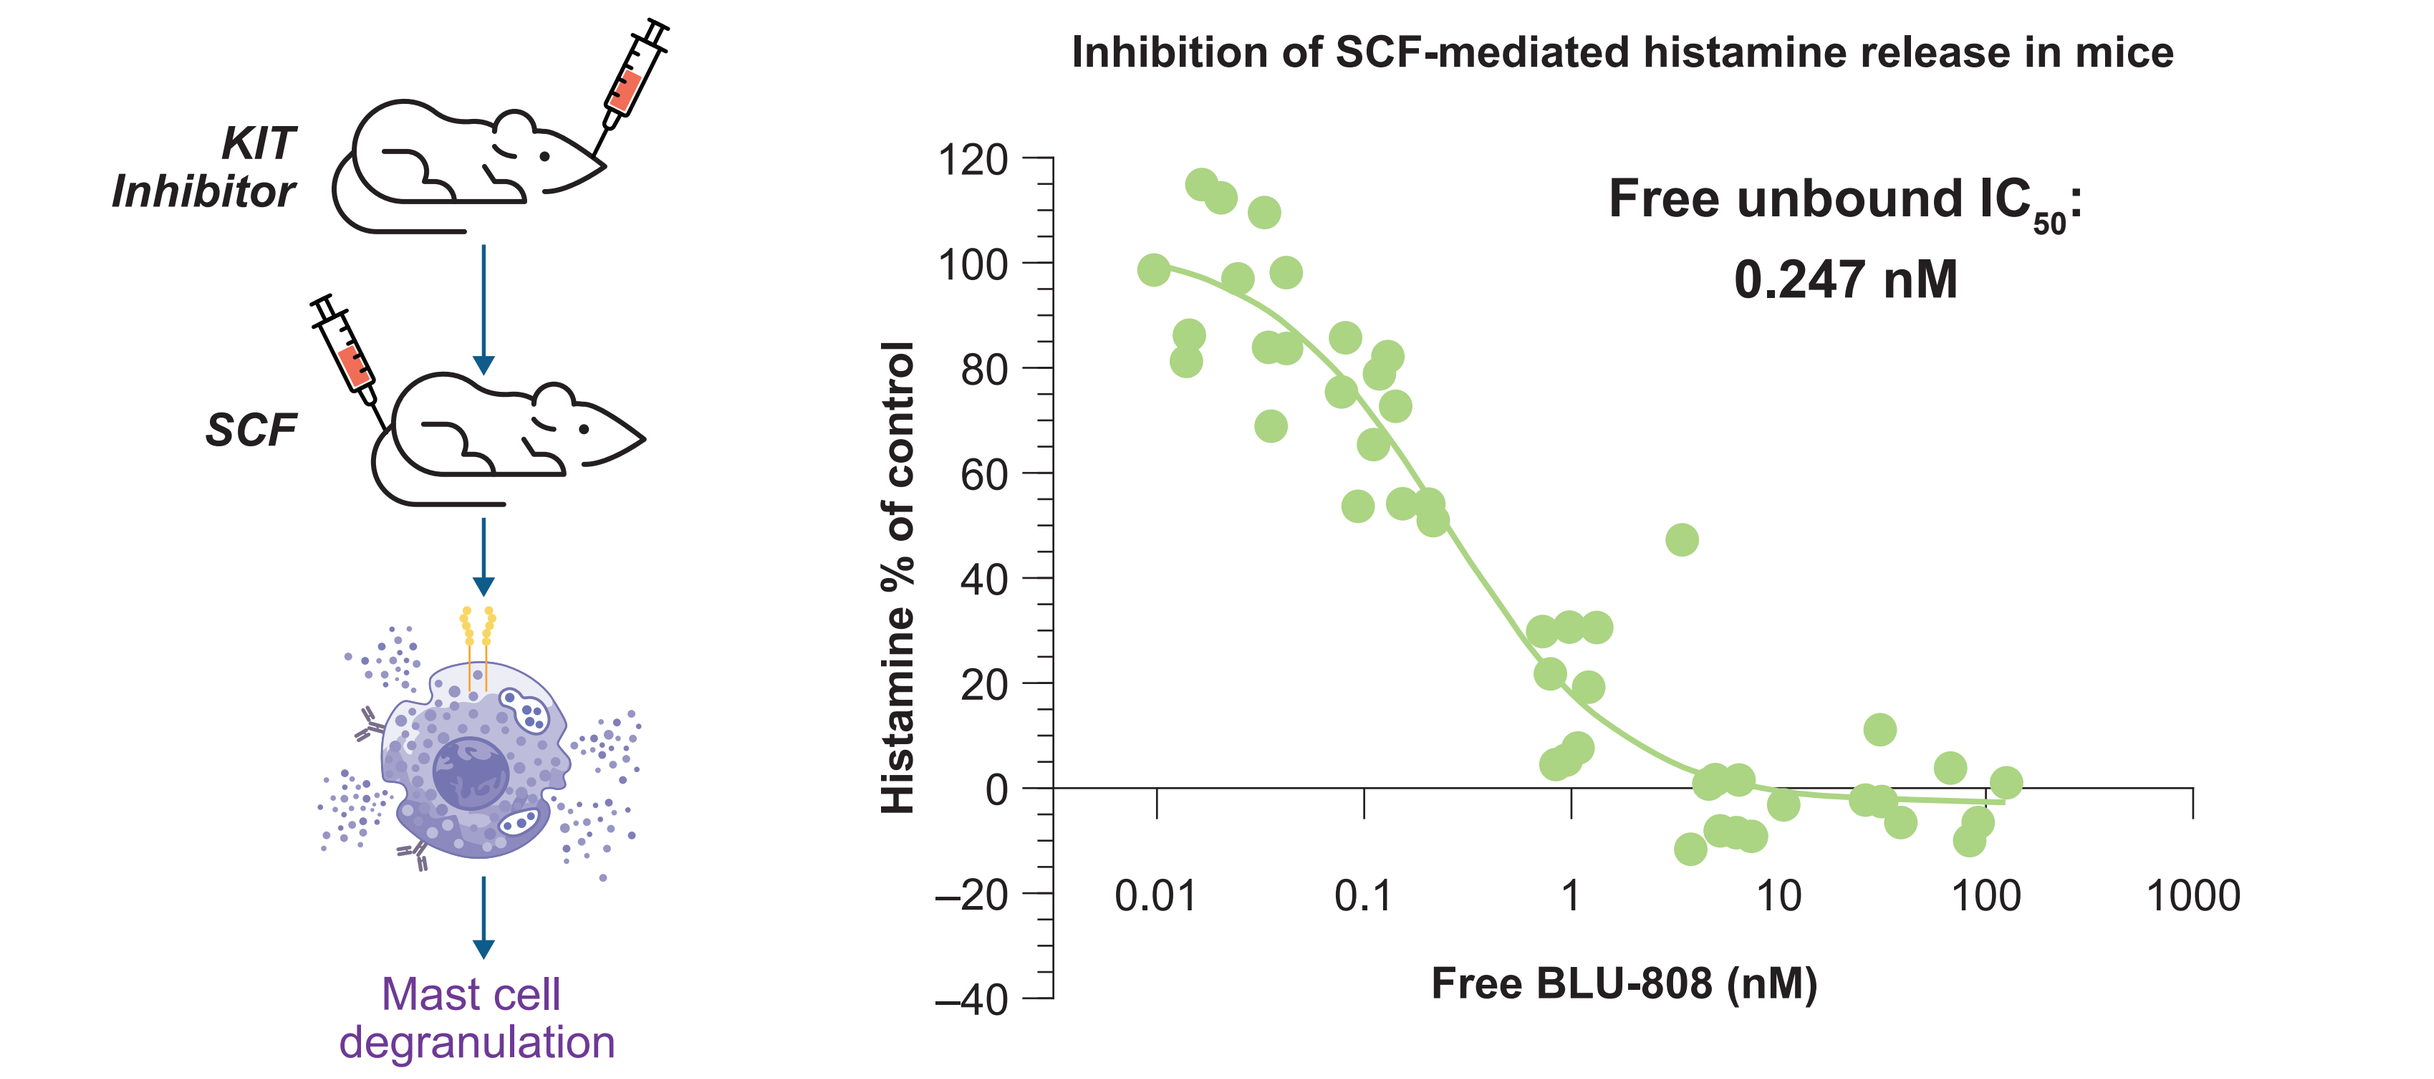
KIT Inhibitor
SCF
Mast cell degranulation
Inhibition of SCF-mediated histamine release in mice
Free unbound IC_{50}: 0.247 nM
Histamine % of control
Free BLU-808 (nM)

- Mast cell degranulation was modeled in mice by SCF induction. Mice were treated with BLU-808, and 3 hours later SCF was injected in the tail vein. Histamine release was measured in the plasma and BLU-808 was able to inhibit histamine release at an IC_{50} of 0.247 nM, corrected for free concentration of BLU-808 (**Figure 7**)

Figure 8. A single dose of BLU-808 inhibits asthma-like phenotypes in a rodent OVA-induced asthma model

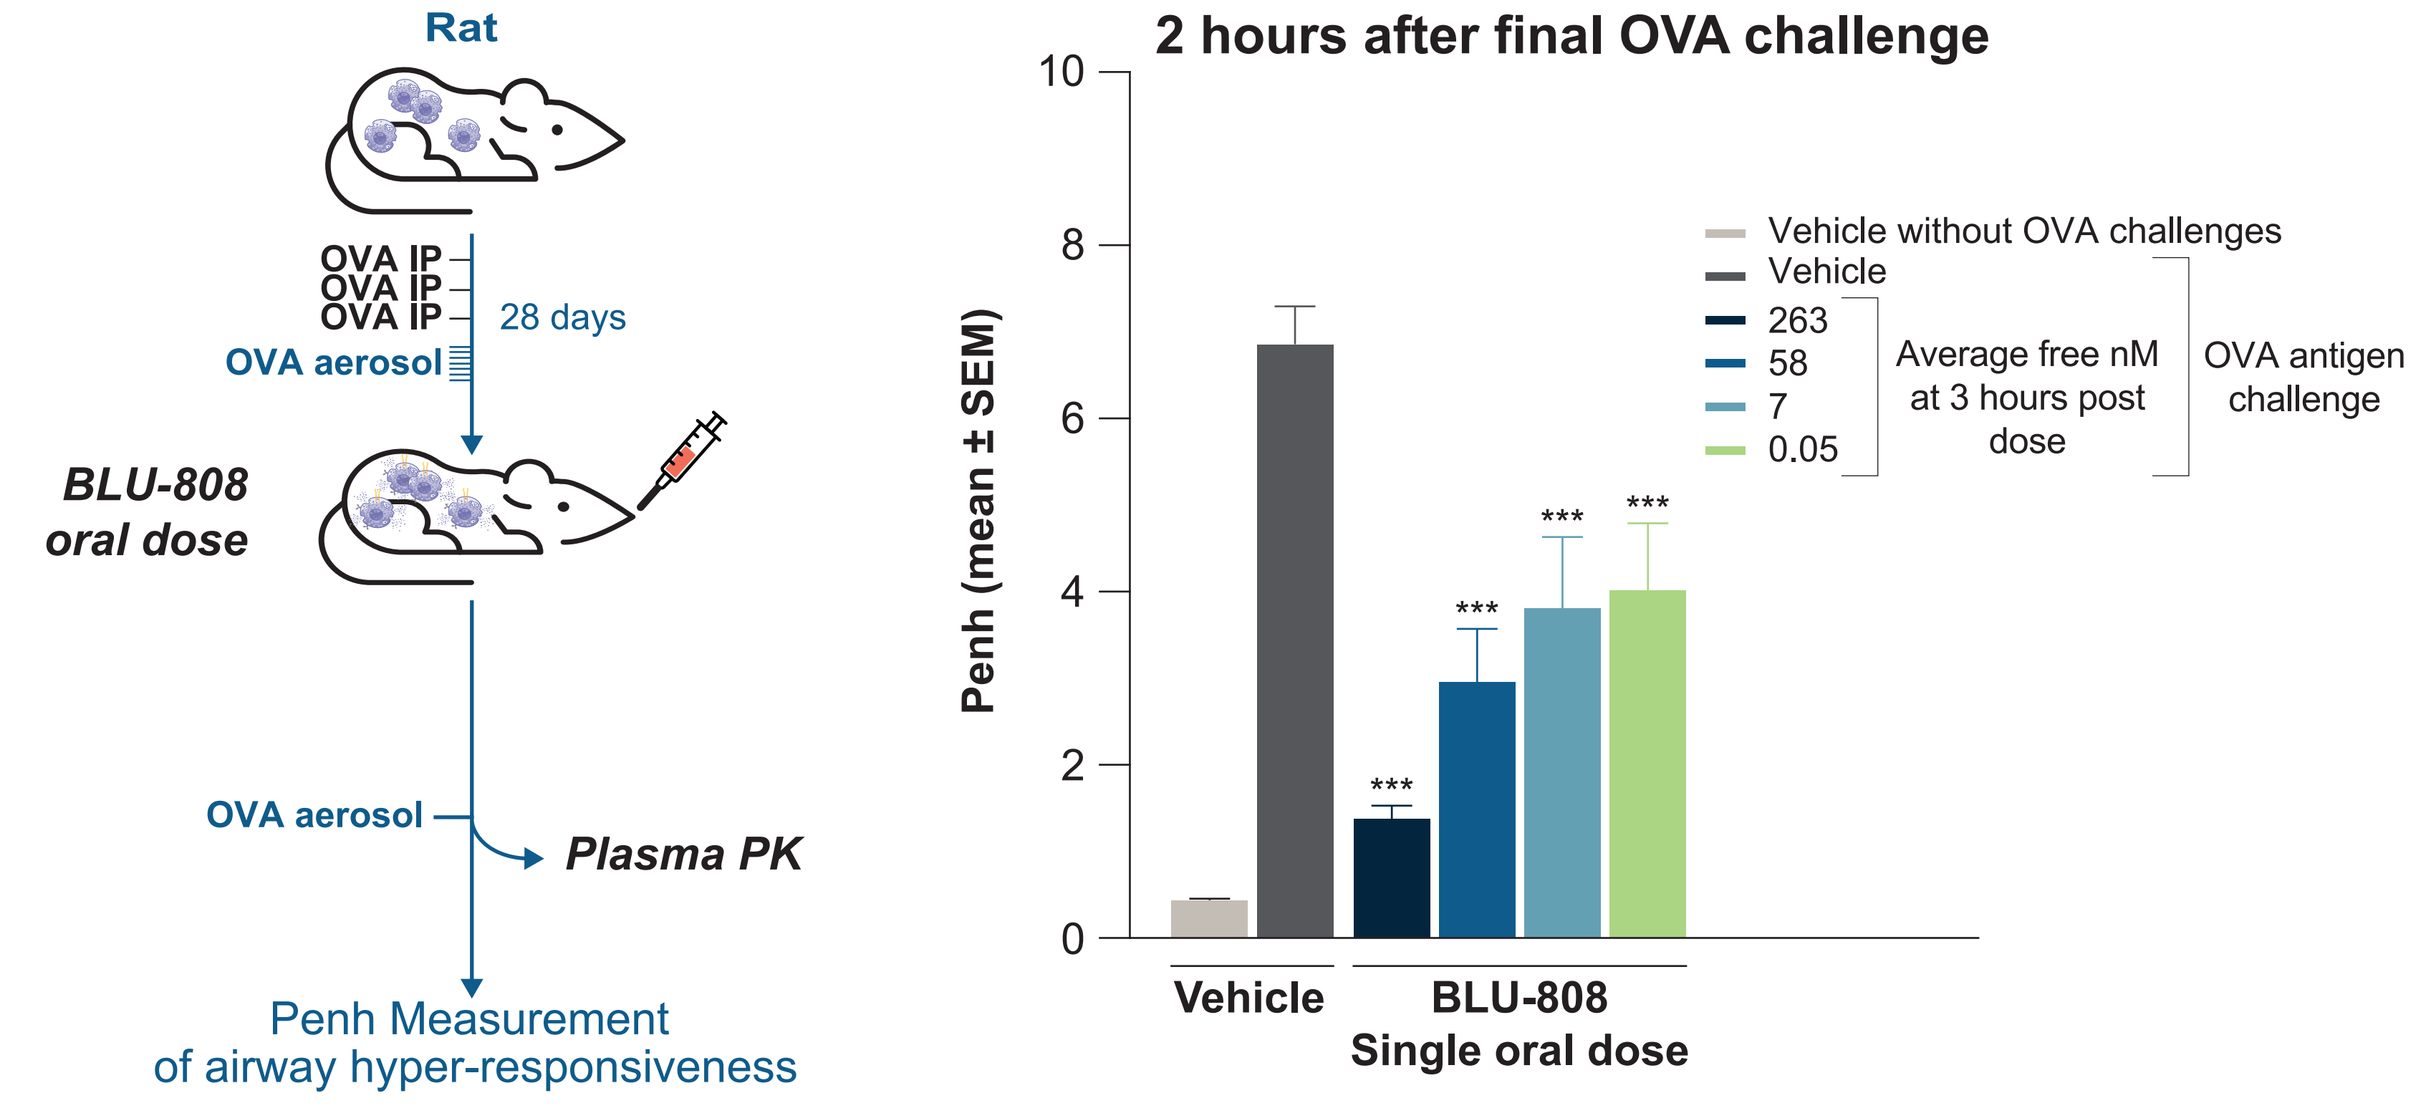
Rat
OVA IP
OVA IP
OVA IP
28 days
OVA aerosol
BLU-808 oral dose
OVA aerosol
Plasma PK
Penh Measurement of airway hyper-responsiveness
2 hours after final OVA challenge
Penh (mean ± SEM)
Vehicle without OVA challenges
Vehicle
263
58
7
0.05
Average free nM at 3 hours post dose
OVA antigen challenge
Vehicle
BLU-808 Single oral dose

IP, intraperitoneal; OVA, ovalbumin; PK, pharmacokinetics; SEM, standard error of the mean ***, *P*<0.0001 per 2-way analysis of variance with Dunnett's correction for multiple comparisons.

- KIT inhibition by BLU-808 decreased OVA-induced airway hyper-responsiveness in mice *in vivo* (**Figure 8**)

Conclusions

- BLU-808 is a potent and selective investigational WT KIT inhibitor that is CNS sparing and orally bioavailable
- BLU-808 demonstrates potent inhibition of human-derived CD34+ mast cell activation and *in vivo* inhibition of SCF-induced histamine release. BLU-808 can also decrease mast cell number *in vivo*
- BLU-808 offers a potential best in class mast cell modulator that provides dosing flexibility to either fully deplete mast cells, or fully or partially inhibit their activity
- In an allergic model of asthma, a single dose of BLU-808 was able to improve lung function
- BLU-808 may provide benefit to patients with WT KIT mast cell diseases including chronic urticaria, asthma, and mast cell activation syndrome
- BLU-808 Investigational New Drug application will be filed mid-2024

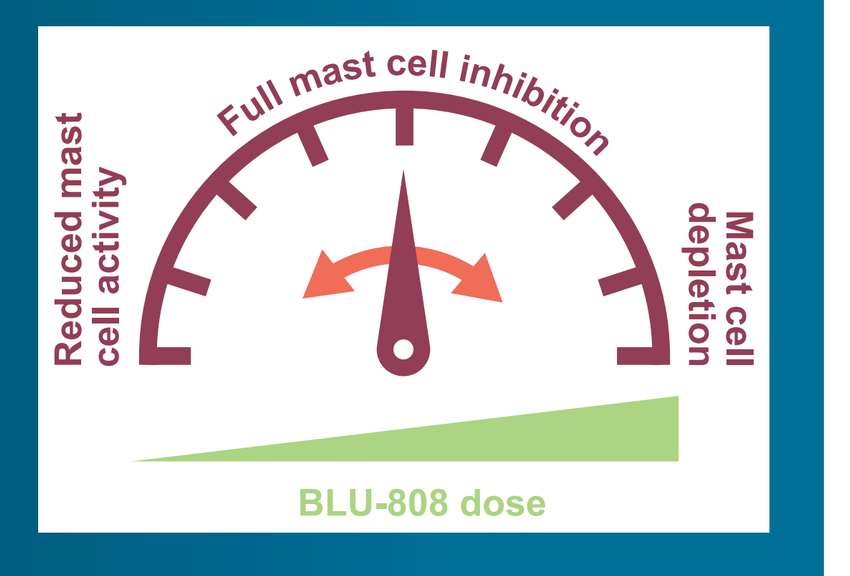
Full mast cell inhibition
Reduced mast cell activity
Mast cell depletion
BLU-808 dose

References

1. Lennartsson J et al. *Physiol Rev.* 2012;92:1619–1649; 2. Chervenick PA et al. *J Cell Physiol.* 1969;73:25–30; 3. Kitamura Y et al. *Blood.* 1979;53:492–497; 4. Kitamura Y et al. *Blood.* 1978;52:447–452; 5. Nakayama H et al. *Development.* 1988;102:107–116; 6. Nocka K et al. *EMBO J.* 1990;9:3287–3294; 7. Tsai M et al. *Chem Immunol Allergy.* 2005;87:179–197; 8. Arber DA et al. *Blood.* 2022;140:1200–1228; 9. Cohen SS et al. *Br J Haematol.* 2014;166:521–528; 10. Kristensen T et al. *J Mol Diagn.* 2011;13:180–188; 11. Church MK et al. *Immunol Rev.* 2018;282:232–247; 12. Elieh-Ali-Komi D et al. *Allergol Int.* 2023;72:359–368; 13. Church DS et al. *WAO Journal.* 2011;4:S22–S27; 14. Fujimura R et al. *Drug Discov Ther.* 2022;16:245–250.

Acknowledgements

Medical writing support was provided by Hannah Boyd, PhD, and editorial support was provided by Sarah Christopher, PhD, all of Paragon (a division of Prime, Knutsford, UK) supported by Blueprint Medicines Corporation, Cambridge, MA, according to Good Publication Practice guidelines. The authors would like to acknowledge the contributions of employees of Blueprint Medicines Corporation who contributed to the work on this project.

Disclosures

This research was funded by Blueprint Medicines Corporation. Blueprint Medicines Corporation reviewed and provided feedback on the poster. Alexandra Grassian is an employee of and equity holder in Blueprint Medicines Corporation. The authors had full editorial control of the poster and provided their final approval of all content. Full disclosures for all authors are available upon request at medinfo@blueprintmedicines.com

Poster available for download at:

Presented at the American Academy of Allergy Asthma & Immunology (AAAAI) 2024, February 23–26, in Washington, DC, USA. Please contact medinfo@blueprintmedicines.com for permission to reprint and/or distribute.