Blood-Based Proteomics for Deeper Insights into Indolent Systemic Mastocytosis: The PIONEER Trial Experience

Yu-An Kuo,[1] Ehsan Irajizad,[1] Annie Hoai Nguyen,[1] Min Chen,[2] Guang Yang,[2] Benjamin Lampson,[2] Rachel L. Erlich,[2] Hussein A. Abbas[1]

[1]The University of Texas MD Anderson Cancer Center, Houston, TX, USA; [2]Blueprint Medicines Corporation, Cambridge, MA, USA

Poster Number PS1838

Introduction

- Systemic mastocytosis (SM), including its most common subtype indolent SM (ISM), is a clonal mast cell (MC) disease driven by the *KIT* D816V mutation in ~95% of patients[1–4]
- The prevalence of SM has been estimated at up to 1 in 5000 people[5–8]
- Patients with ISM often experience debilitating symptoms across multiple organ systems[9–12]
 - These symptoms are likely due to direct MC activation, mediator release, and inflammatory immune system alterations promoted by aberrant MCs[12]
- Avapritinib is a potent, oral inhibitor that selectively targets the KIT D816V mutation[13]
- Avapritinib was approved in adult patients with ISM in the USA and in adult patients with moderate-to-severe ISM in the EU, based on the findings from the randomized, double-blind, placebo-controlled PIONEER study (NCT03731260)[14,15]
- A better understanding of the inflammatory proteome may help facilitate the diagnosis and treatment of ISM. We therefore conducted high throughput profiling of the baseline inflammatory plasma proteome across the large and well-characterized cohort of patients enrolled in PIONEER

Methods

- The Olink® Explore 384 Inflammation protein panel (Uppsala, Sweden) measured 363 soluble proteins in baseline plasma samples from 168 patients with ISM enrolled in PIONEER and 39 age-matched healthy donors. Based on plasma sample quality and availability of baseline symptom information, 155 patients with ISM and all healthy donors were included in the final analysis. (**Figure 1**)

Figure 1. Study design

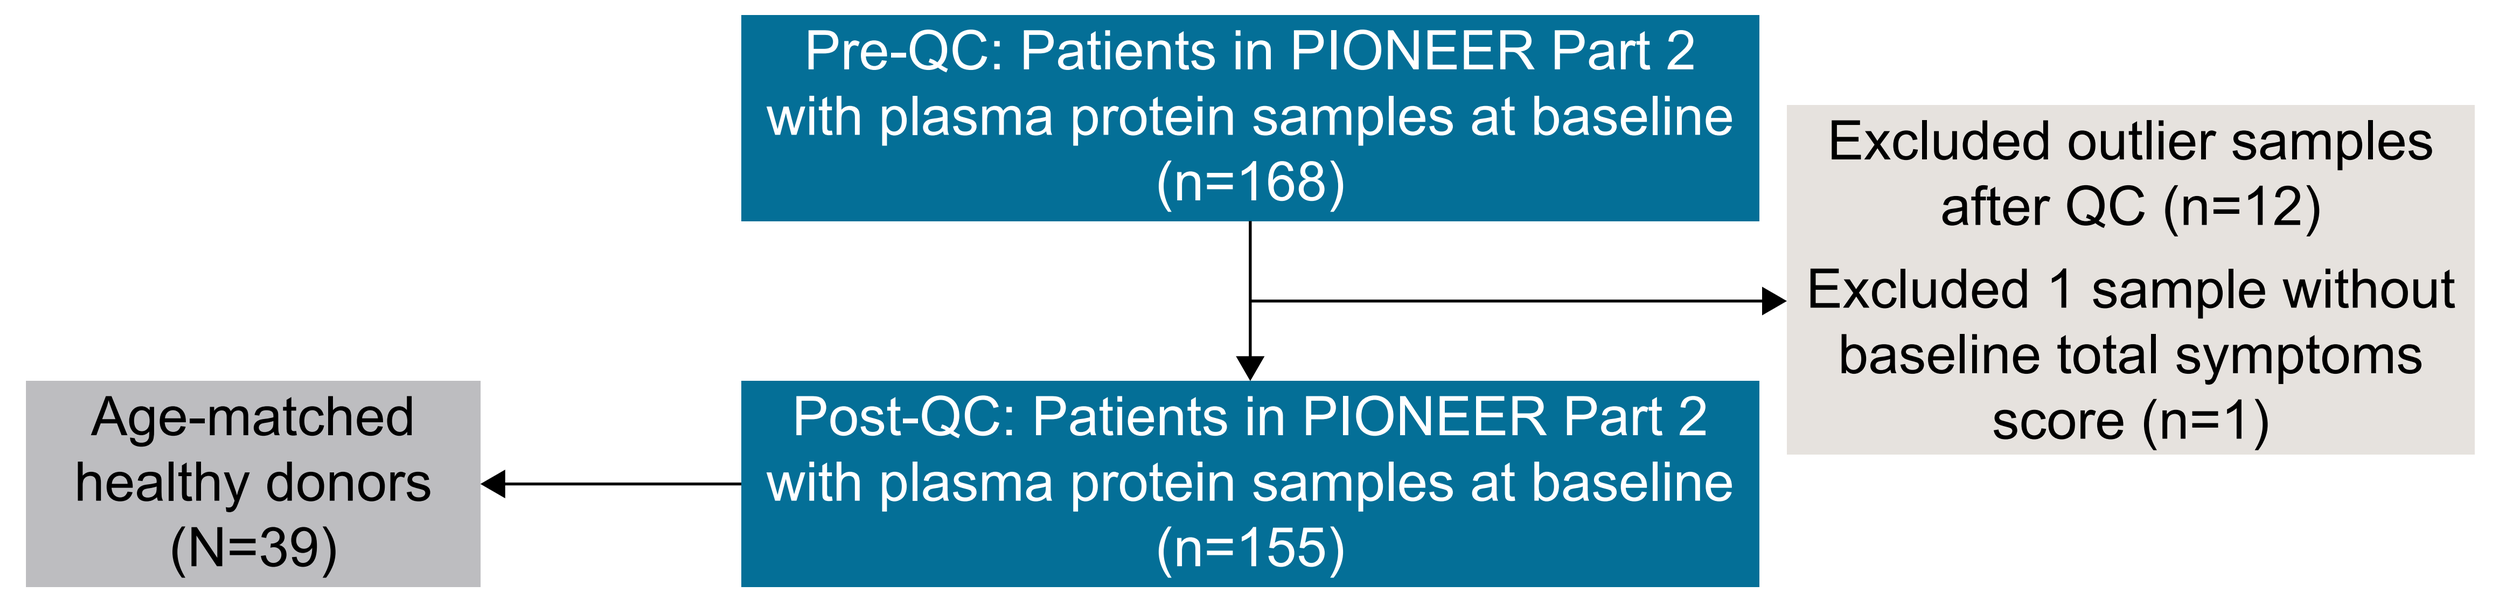

Pre-QC: Patients in PIONEER Part 2 with plasma protein samples at baseline (n=168)
Excluded outlier samples after QC (n=12)
Excluded 1 sample without baseline total symptoms score (n=1)
Age-matched healthy donors (N=39)
Post-QC: Patients in PIONEER Part 2 with plasma protein samples at baseline (n=155)

QC, quality control.

- A t-test was performed for each baseline plasma protein measurement and, unless otherwise specified, the resulting P-values were adjusted for multiple comparisons using the Benjamini-Hochberg method to control the false discovery rate (FDR)
 - An FDR of <0.05 was considered significant
- Gene Ontology (GO) pathway analysis was employed to ascertain the functional roles of differential soluble proteins
- The correlation of cytokine expression (as measured via Olink) with disease burden and symptoms (which included clinical metrics such as symptom scores, age, and serum tryptase levels) was assessed using R (version 4.4.1; R Foundation for Statistical Computing, Vienna, Austria)
- K-means clustering was performed in high dimensional space and visualized using Uniform Manifold Approximation and Projection (UMAP) to identify distinct populations of patients based on cytokine levels and expression
 - Identified population clusters were assessed to identify any association between disease burden and symptoms, and specific cluster

Results

- Patient demographics can be found in **Table 1**

Table 1. Baseline demographics

	Patients enrolled in PIONEER Part 2 who received either avapritinib 25 mg QD or placebo (n=155)
Age (years), median (range)	52 (22–79)
Female, n (%)	118 (76)
ISM symptom burden	
Baseline TSS, mean (SD)	51.7 (19.8)
MC burden	
Median (range) baseline serum tryptase, ng/mL	43.7 (3.6–501.6)
Median (range) baseline BMBMC, %	7 (1–70)
Median (range) baseline *KIT* D816V VAF in peripheral blood, %[a]	0.4 (undetectable–36.7)
Median (range) baseline alkaline phosphatase, U/L	78 (41–219)

[a]Limit of detection, 0.02%.
BMBMC, bone marrow biopsy mast cells; ISM, indolent systemic mastocytosis; QD, once daily; SD, standard deviation; TSS, total symptom score; VAF, variant allele frequency.

Comparison of individual plasma proteins in patients with ISM versus healthy donors and Gene Ontology pathway analysis

- Compared with healthy individuals, 61 plasma proteins were significantly different in patients with ISM (**Figure 2**)
 - 51 plasma proteins were upregulated, including TPSAB1, MILR1, AGRP, CXCL 6, IL7, ITGA6, CCL13, CCL17, and CCL23
 - 10 plasma proteins were downregulated: MMP10, CKMT1A_CKMT1B, CCL21, LGALS4, MEPE, IL17A, MPIG6B, EGF, PLXNA4, and ARHGEF12
- GO analysis revealed that the upregulated plasma proteins were primarily associated with leukocyte chemotaxis and myeloid leukocyte migration (**Figure 3**)
 - It should be noted that the full 363-protein panel was enriched for the "leukocyte migration" and "chemotaxis" pathways

Figure 2. Plasma protein expression patients with ISM *versus* healthy donors

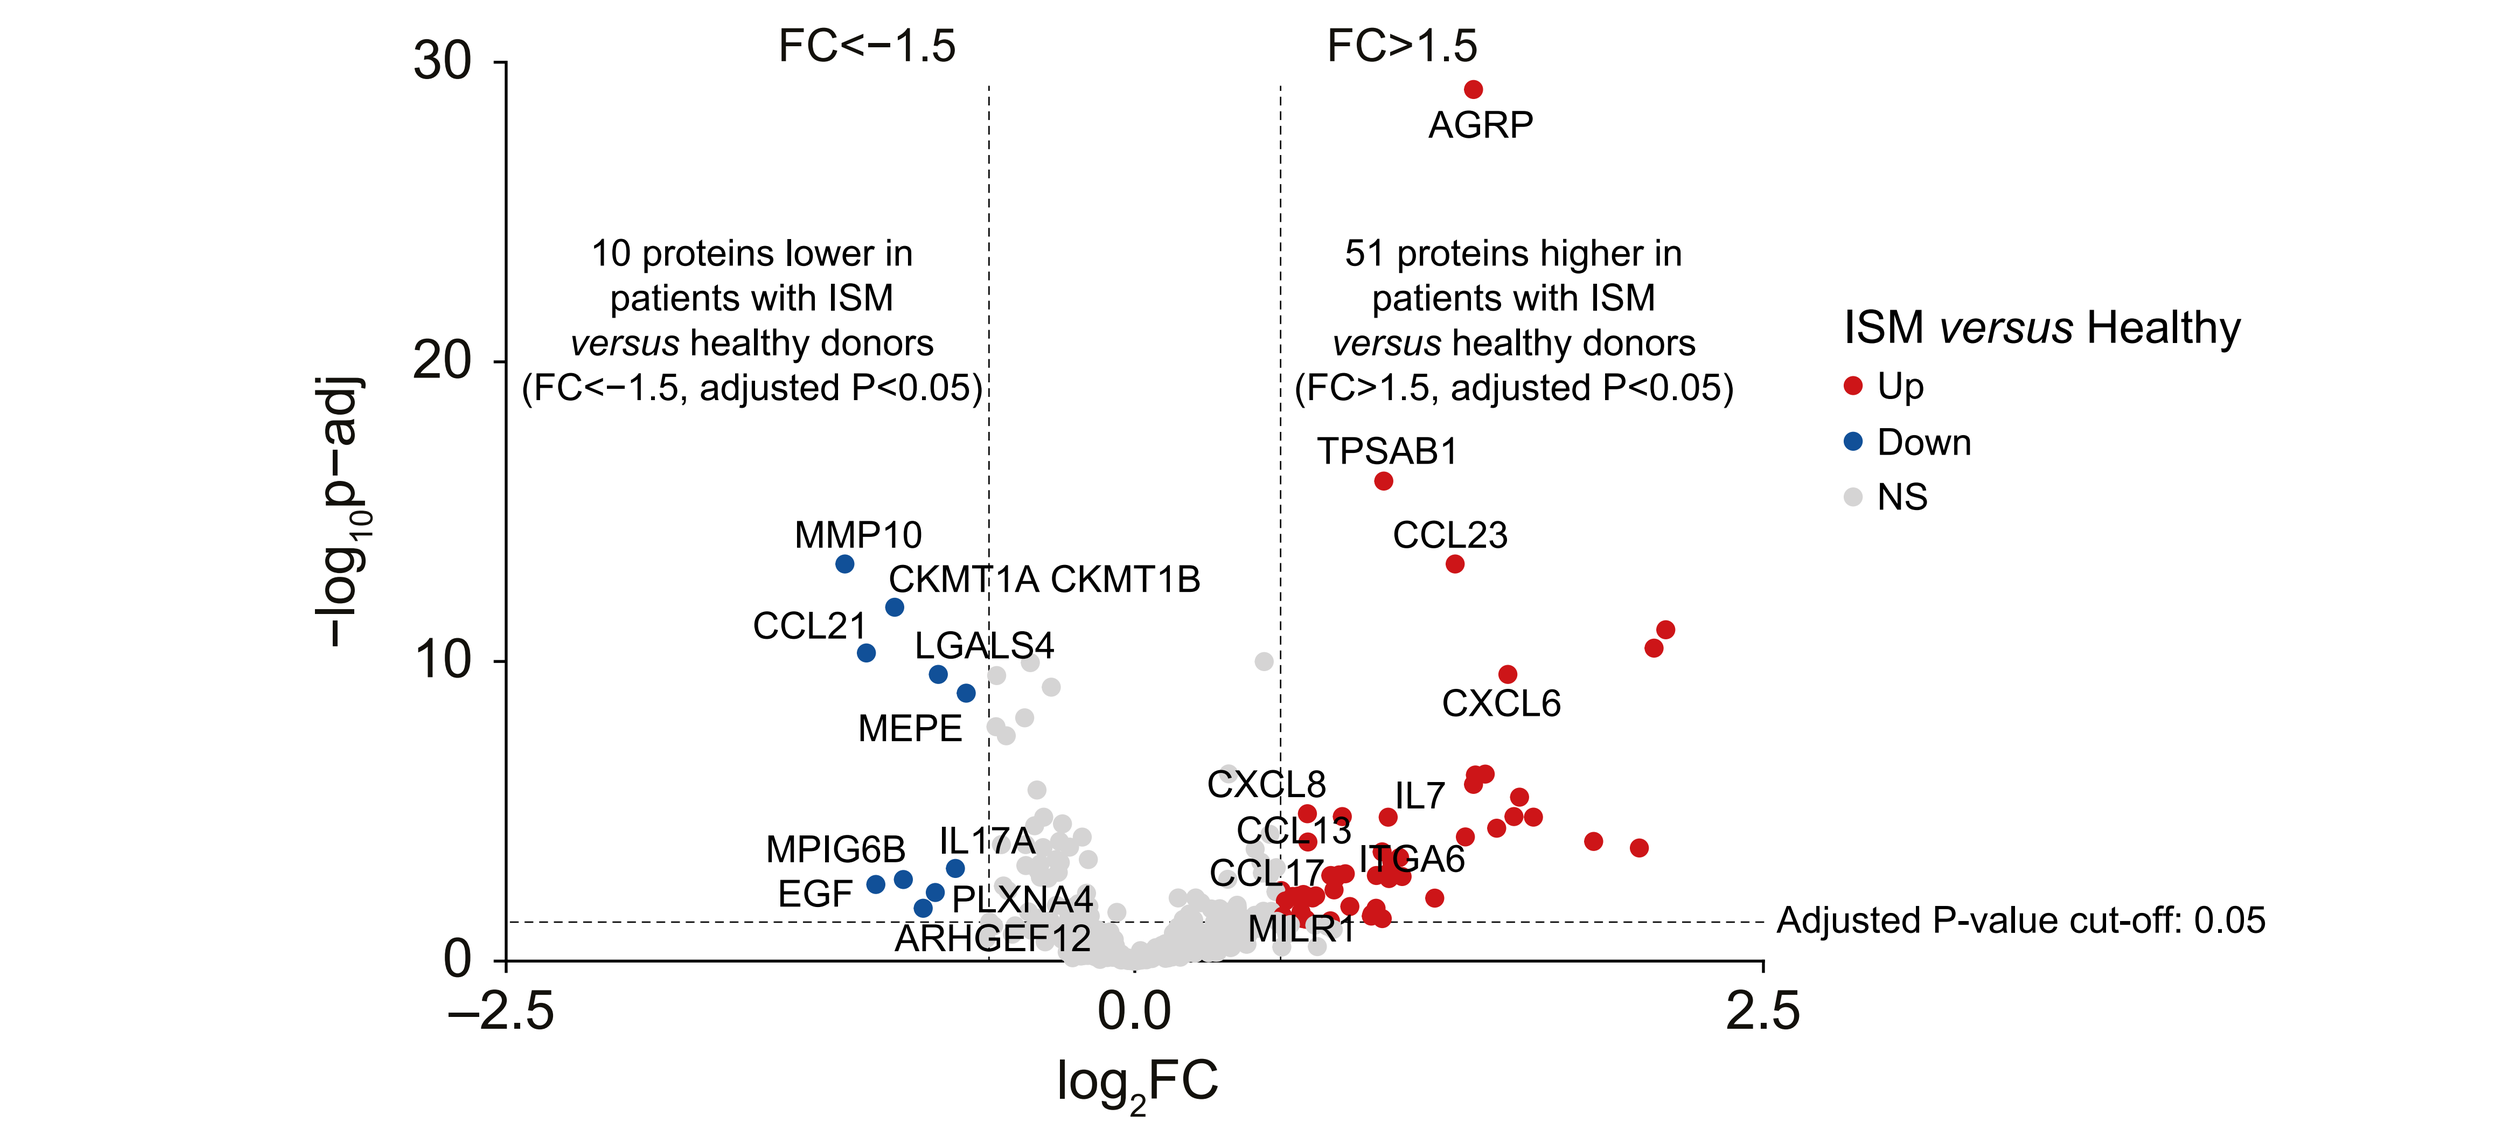

FC<−1.5
FC>1.5
10 proteins lower in patients with ISM *versus* healthy donors (FC<−1.5, adjusted P<0.05)
51 proteins higher in patients with ISM *versus* healthy donors (FC>1.5, adjusted P<0.05)
ISM *versus* Healthy
Up
Down
NS
AGRP
TPSAB1
CCL23
CXCL6
CXCL8
CCL13
CCL17
IL7
ITGA6
MILR1
MMP10
CKMT1A CKMT1B
CCL21
LGALS4
MEPE
MPIG6B
IL17A
EGF
PLXNA4
ARHGEF12
Adjusted P-value cut-off: 0.05
−log10p−adj
log2FC
−2.5
0.0
2.5
0
10
20
30

FC, fold change; NS, not significant; P-adj, adjusted P-value.

Figure 3. Functional roles of upregulated cytokines

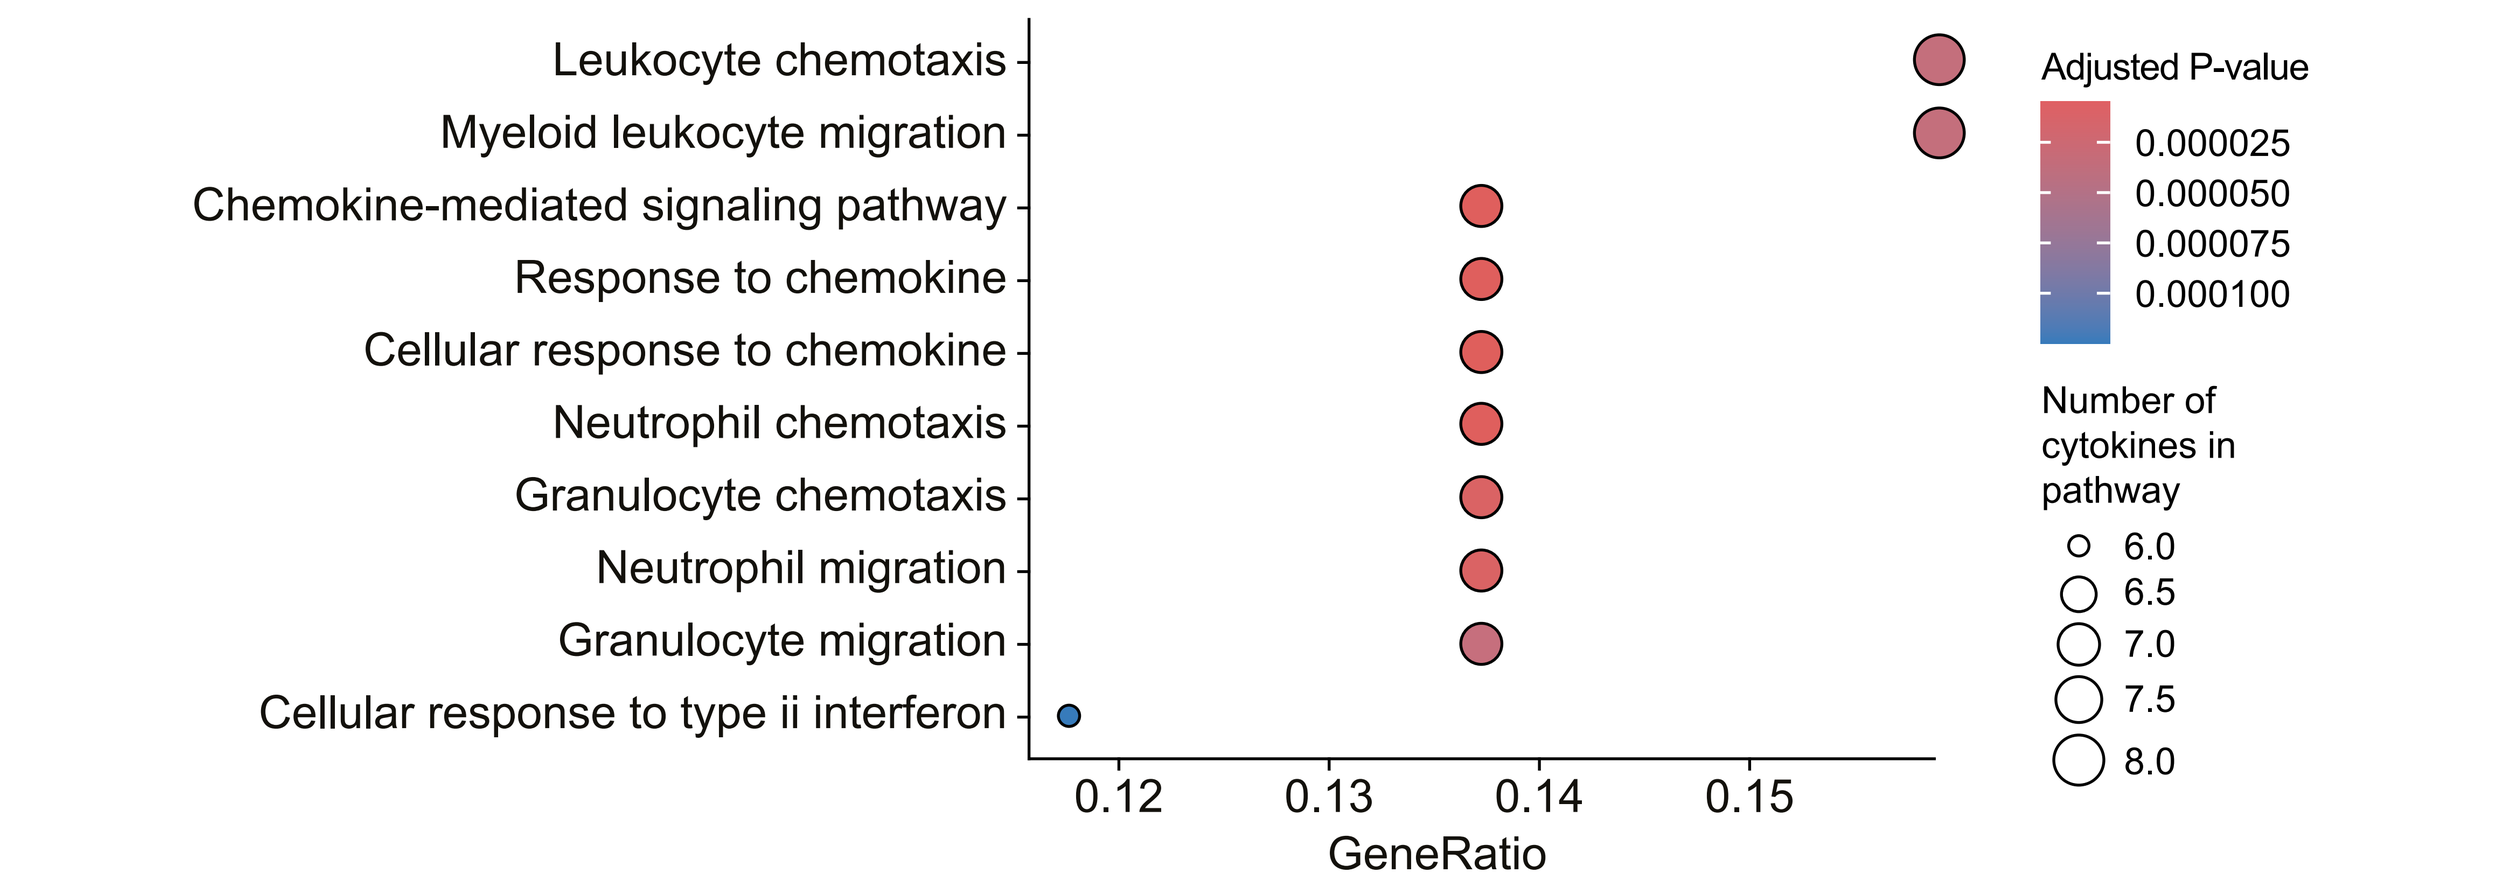

Leukocyte chemotaxis
Myeloid leukocyte migration
Chemokine-mediated signaling pathway
Response to chemokine
Cellular response to chemokine
Neutrophil chemotaxis
Granulocyte chemotaxis
Neutrophil migration
Granulocyte migration
Cellular response to type ii interferon
0.12
0.13
0.14
0.15
GeneRatio
Adjusted P-value
0.000025
0.000050
0.000075
0.000100
Number of cytokines in pathway
6.0
6.5
7.0
7.5
8.0

Correlation of cytokines with symptoms and disease burden measures

- Correlations between measured cytokines and symptom scores and disease burden measures were determined. P<0.05 are considered as significant (**Figure 4**)

Figure 4. Correlations of cytokine expression with disease burden and symptoms

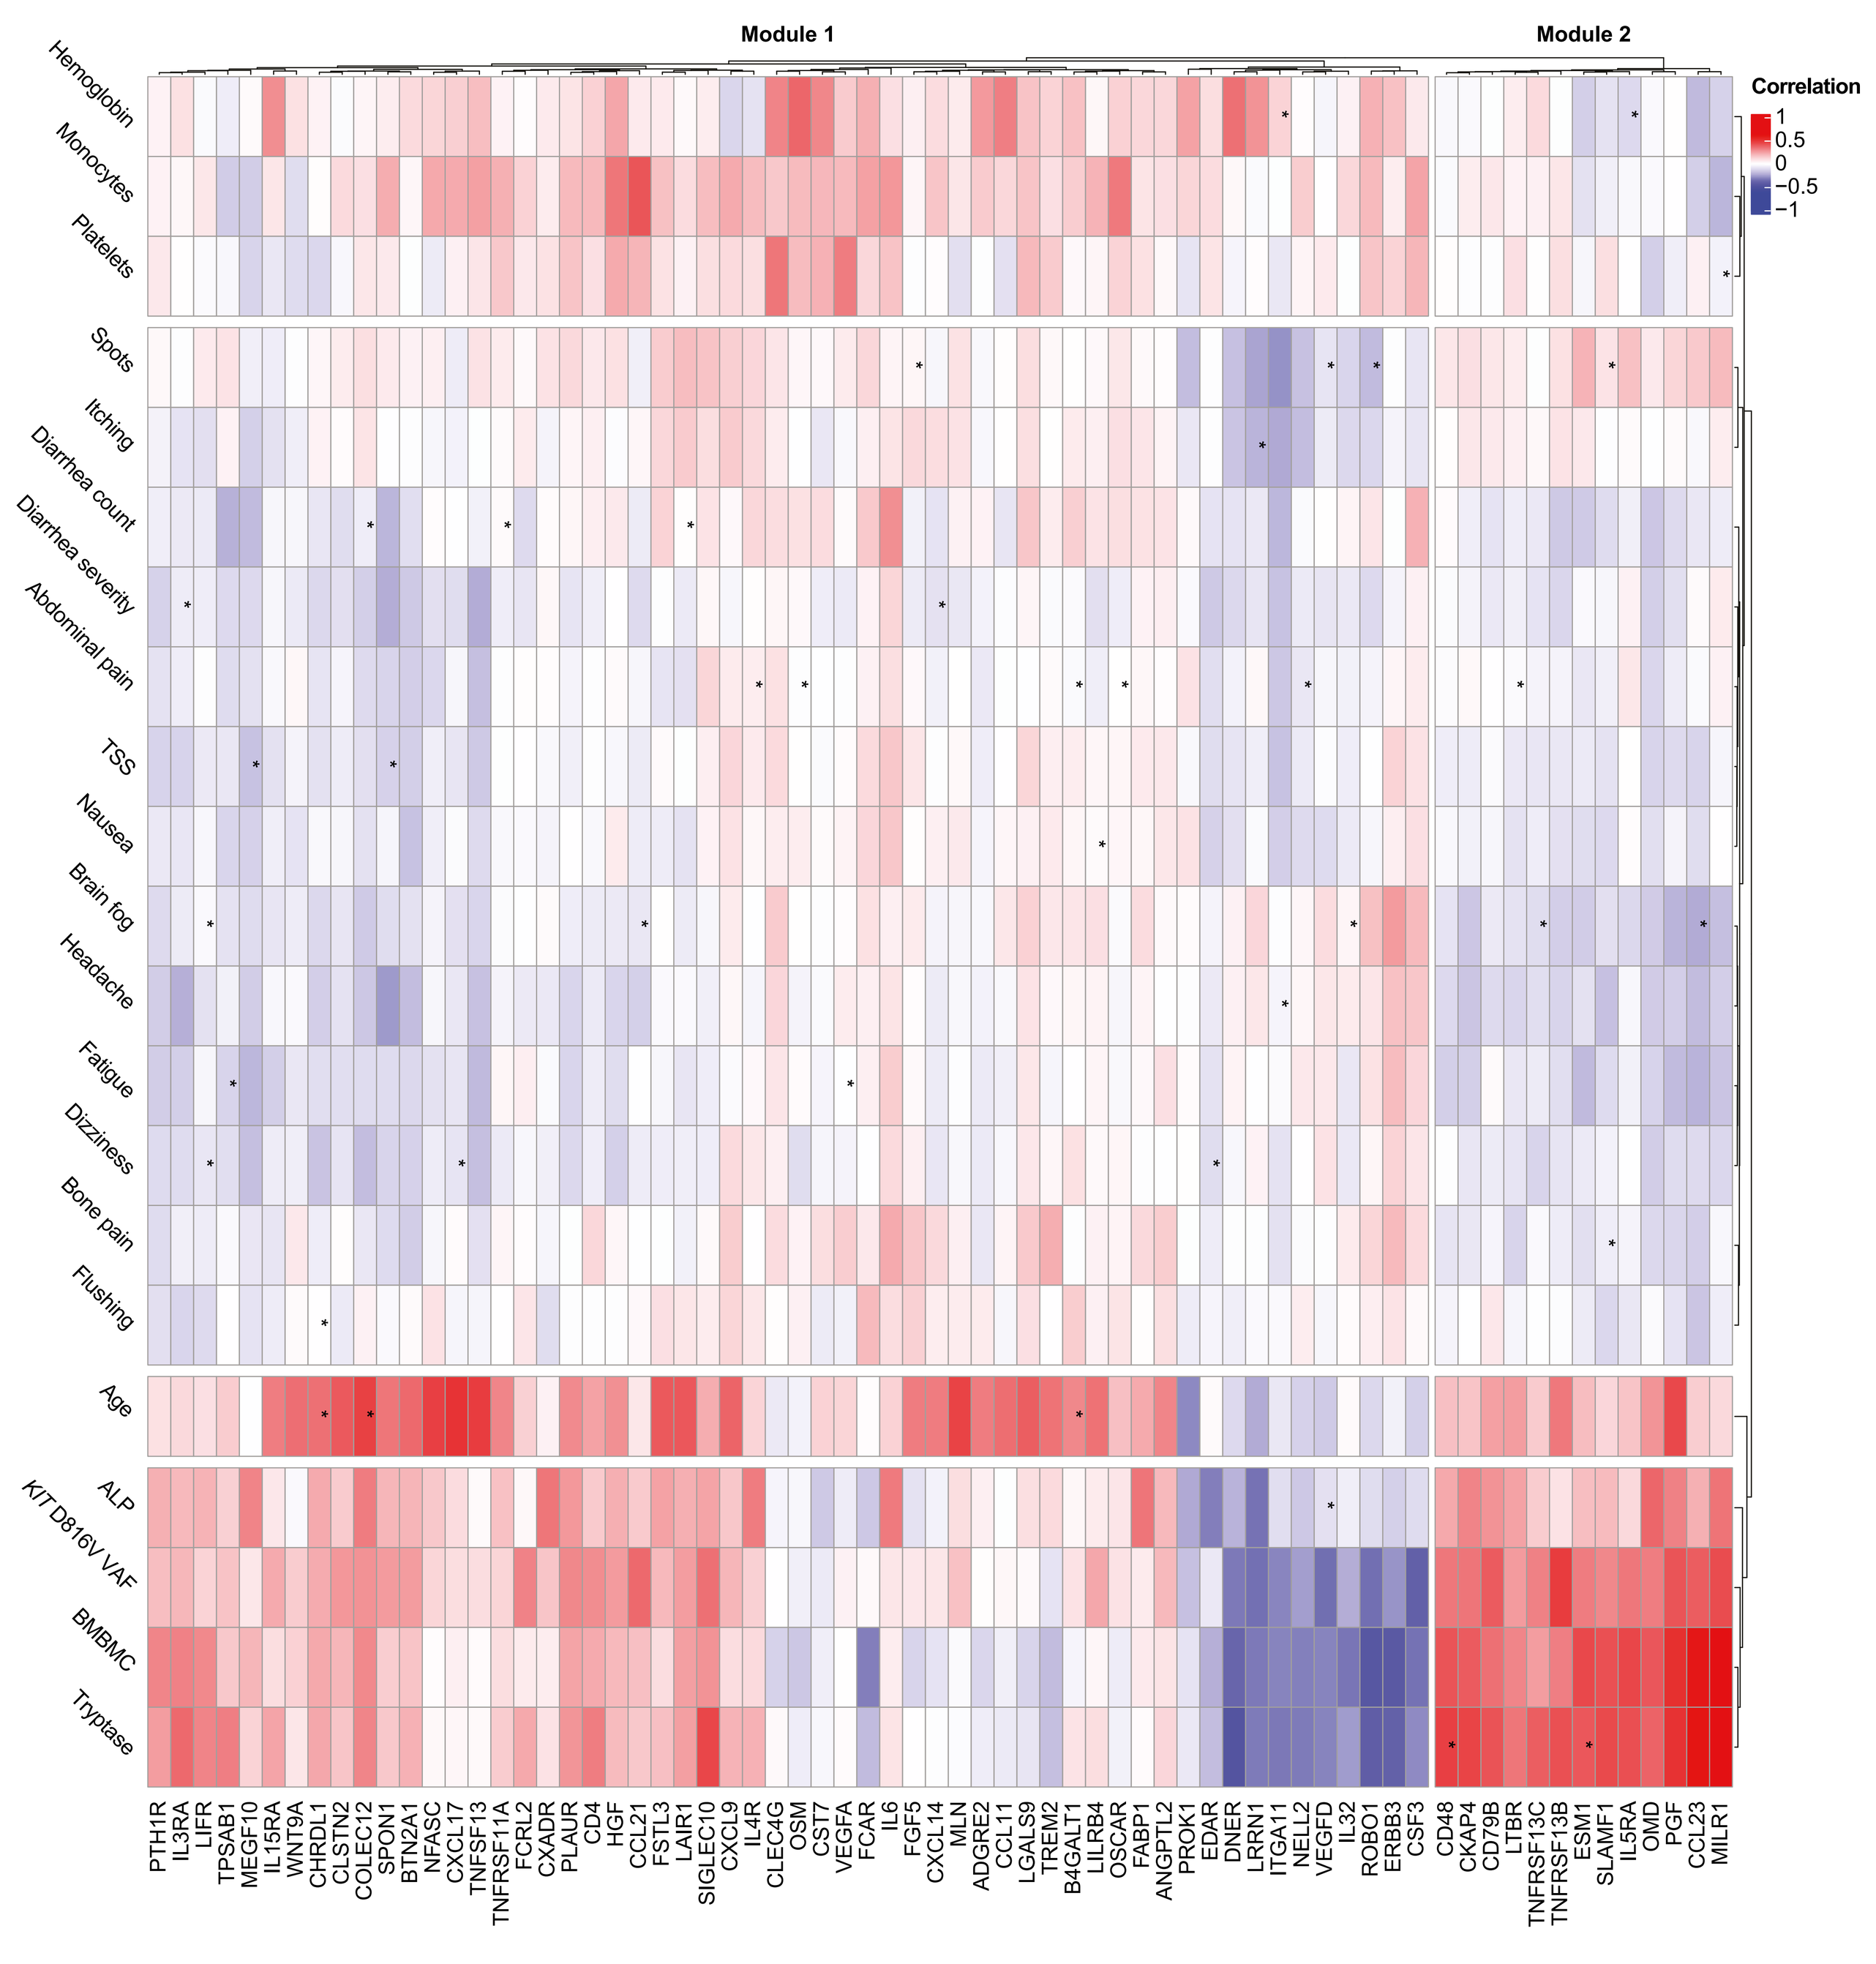

Module 1
Module 2
Correlation
1
0.5
0
−0.5
−1
Hemoglobin
Monocytes
Platelets
Spots
Itching
Diarrhea count
Diarrhea severity
Abdominal pain
TSS
Nausea
Brain fog
Headache
Fatigue
Dizziness
Bone pain
Flushing
Age
ALP
KIT D816V VAF
BMBMC
Tryptase
PTH1R IL3RA LIFR TPSAB1 MEGF10 IL15RA WNT9A CHRDL1 CLSTN2 COLEC12 SPON1 BTN2A1 NFASC CXCL17 TNFSF13 TNFRSF11A FCRL2 CXADR PLAUR CD4 HGF CCL21 FSTL3 LAIR1 SIGLEC10 CXCL9 IL4R CLEC4G OSM CST7 VEGFA FCAR IL6 FGF5 CXCL14 MLN ADGRE2 CCL11 LGALS9 TREM2 B4GALT1 LILRB4 OSCAR FABP1 ANGPTL2 PROK1 EDAR DNER LRRN1 ITGA11 NELL2 VEGFD IL32 ROBO1 ERBB3 CSF3 CD48 CKAP4 CD79B LTBR TNFRSF13C TNFRSF13B ESM1 SLAMF1 IL5RA OMD PGF CCL23 MILR1

*Adjusted P<0.05. ALP, alkaline phosphatase.

- Two distinct plasma protein modules were identified in unsupervised clustering, Module 1 and Module 2
 - Module 1, which included ITGA11, CSF3, and IL32, was consistently negatively correlated with disease burden
 - Module 2, including MILR1, CCL23, CD48, was positively correlated with disease burden
 - Several proteins from Module 2, including IL6, IL4R, CXADR, and FABP1, were positively correlated with alkaline phosphatase (ALP)
- ITGA11 encodes an integrin subunit and its expression is associated with gastric adenocarcinoma[16] and cutaneous squamous cell carcinoma.[17] Here, ITGA11 expression was negatively correlated with disease burden measures and symptoms, including the following:
 - All patient-reported gastrointestinal severity scores such as diarrhea and abdominal pain, and skin symptoms such as itching and spots
 - Levels of ALP, serum tryptase, bone marrow (BM) MC burden, *KIT* D816V variant allele frequency (VAF), and total symptom score (TSS)

Clustering of patients based on plasma proteomic profiles

- K-means clustering, performed on cytokine levels of patients with ISM and healthy donors in high dimensional space using UMAP, revealed two well-separated clusters (**Figure 5**)
 - Cluster 1 appeared more ISM-specific, with relatively few healthy donors
 - Cluster 2 included both patients with ISM and healthy donors, potentially suggesting a healthy donor-like ISM subpopulation
- Expression of specific cytokines in each patient cluster is shown in **Figure 6A**
- Overall, 171 cytokines had significantly higher expression in cluster 1 *versus* cluster 2, and 98 had significantly lower expression (**Figure 6B**)

Figure 5. UMAP K-means clustering of patients with ISM and healthy donors

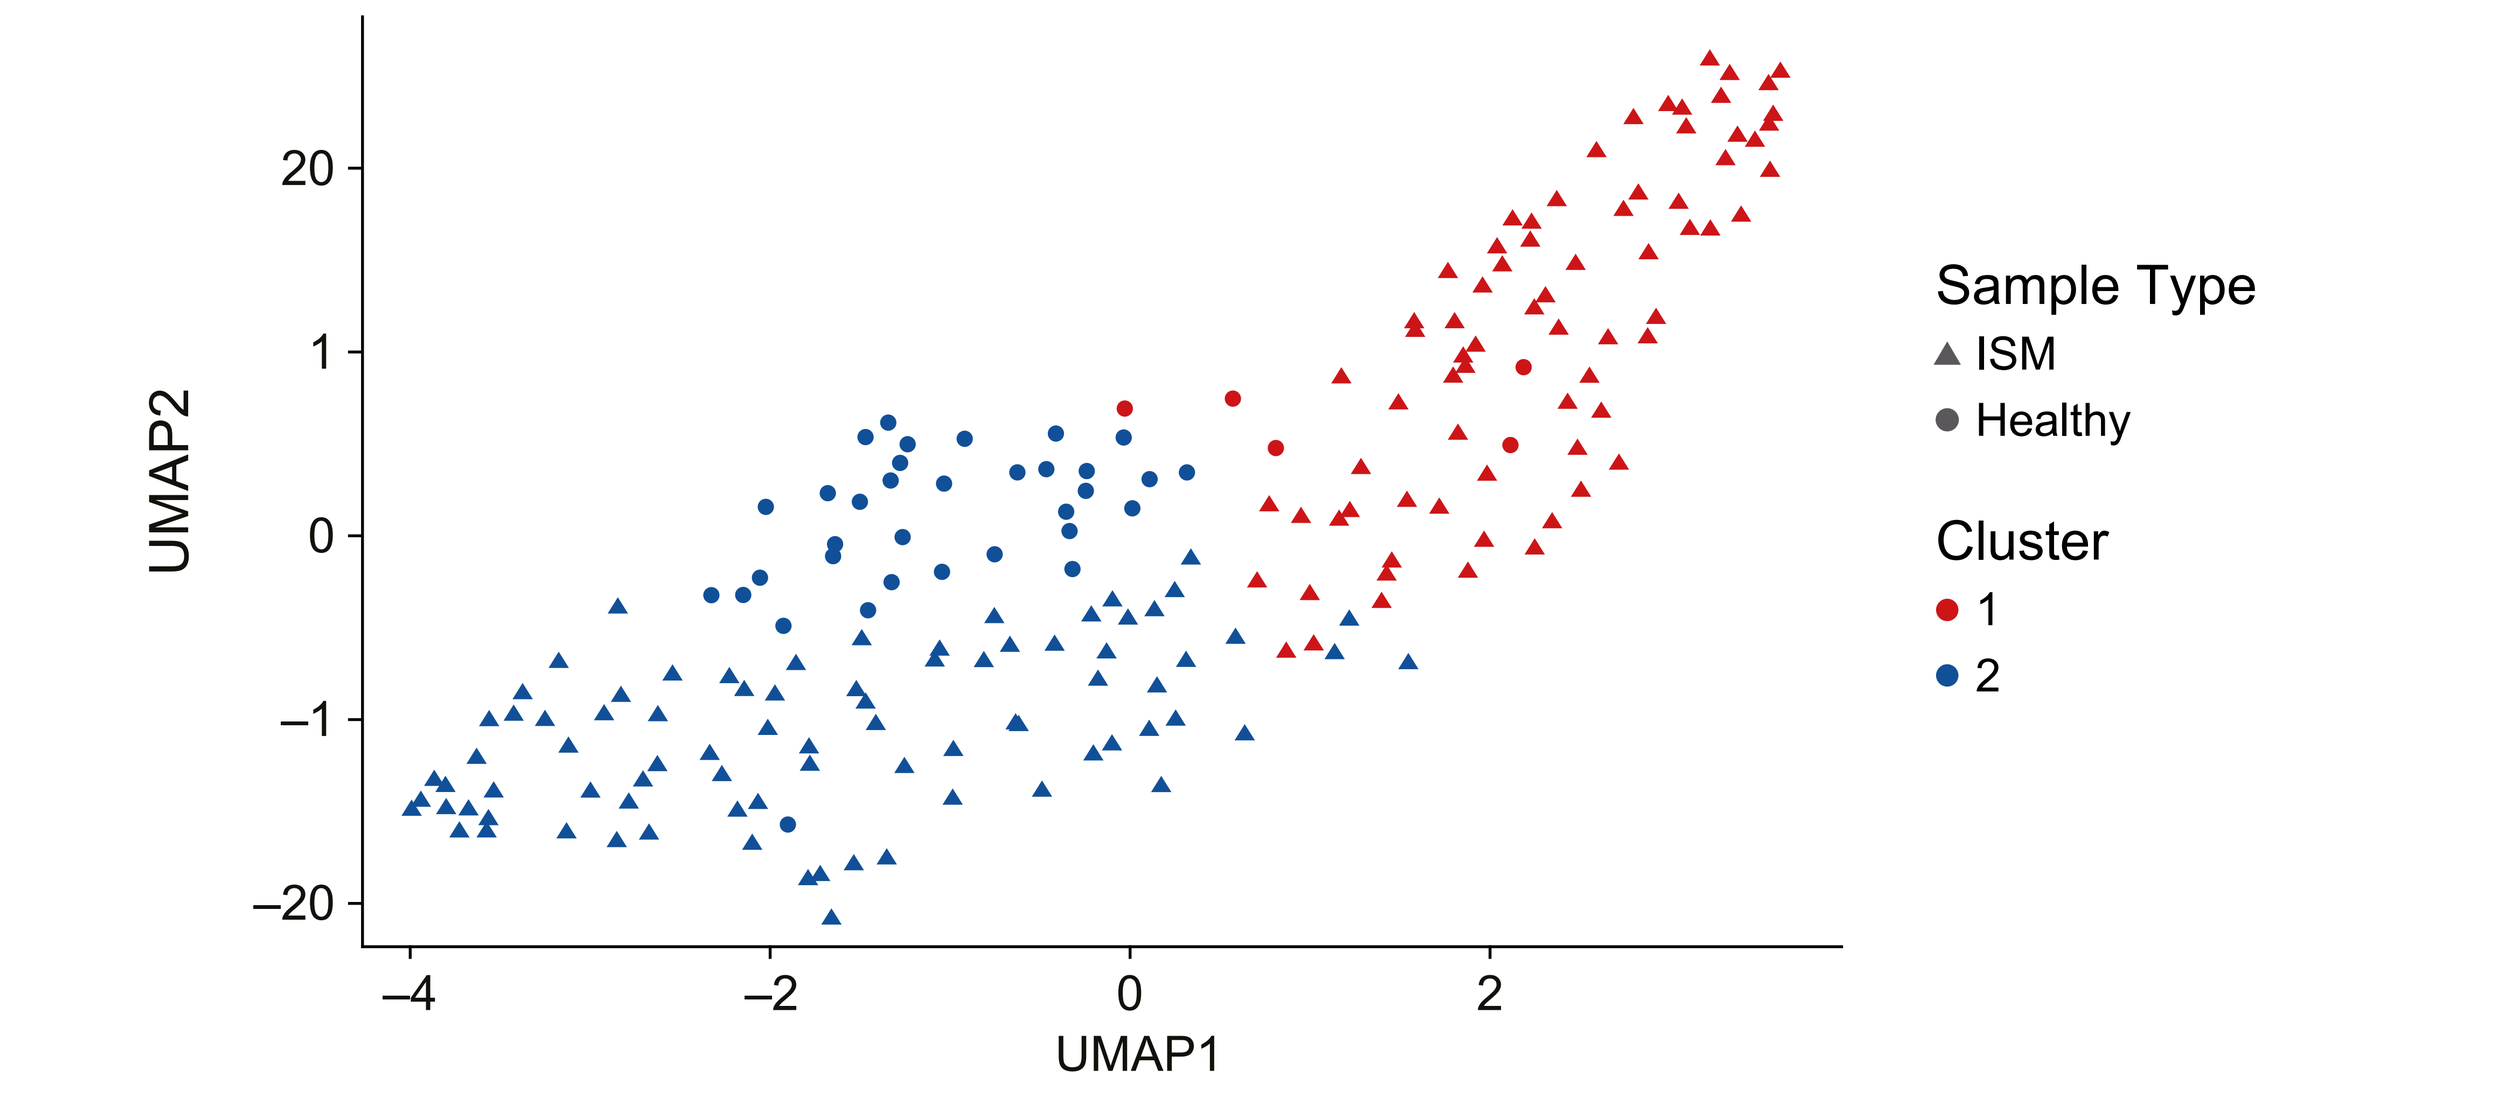

UMAP2
UMAP1
20
1
0
−1
−20
−4
−2
0
2
Sample Type
ISM
Healthy
Cluster
1
2

UMAP, uniform manifold approximation and projection.

Figure 6. Patterns of cytokine expression by patient cluster (A) and relative cytokine expression in each population (B)

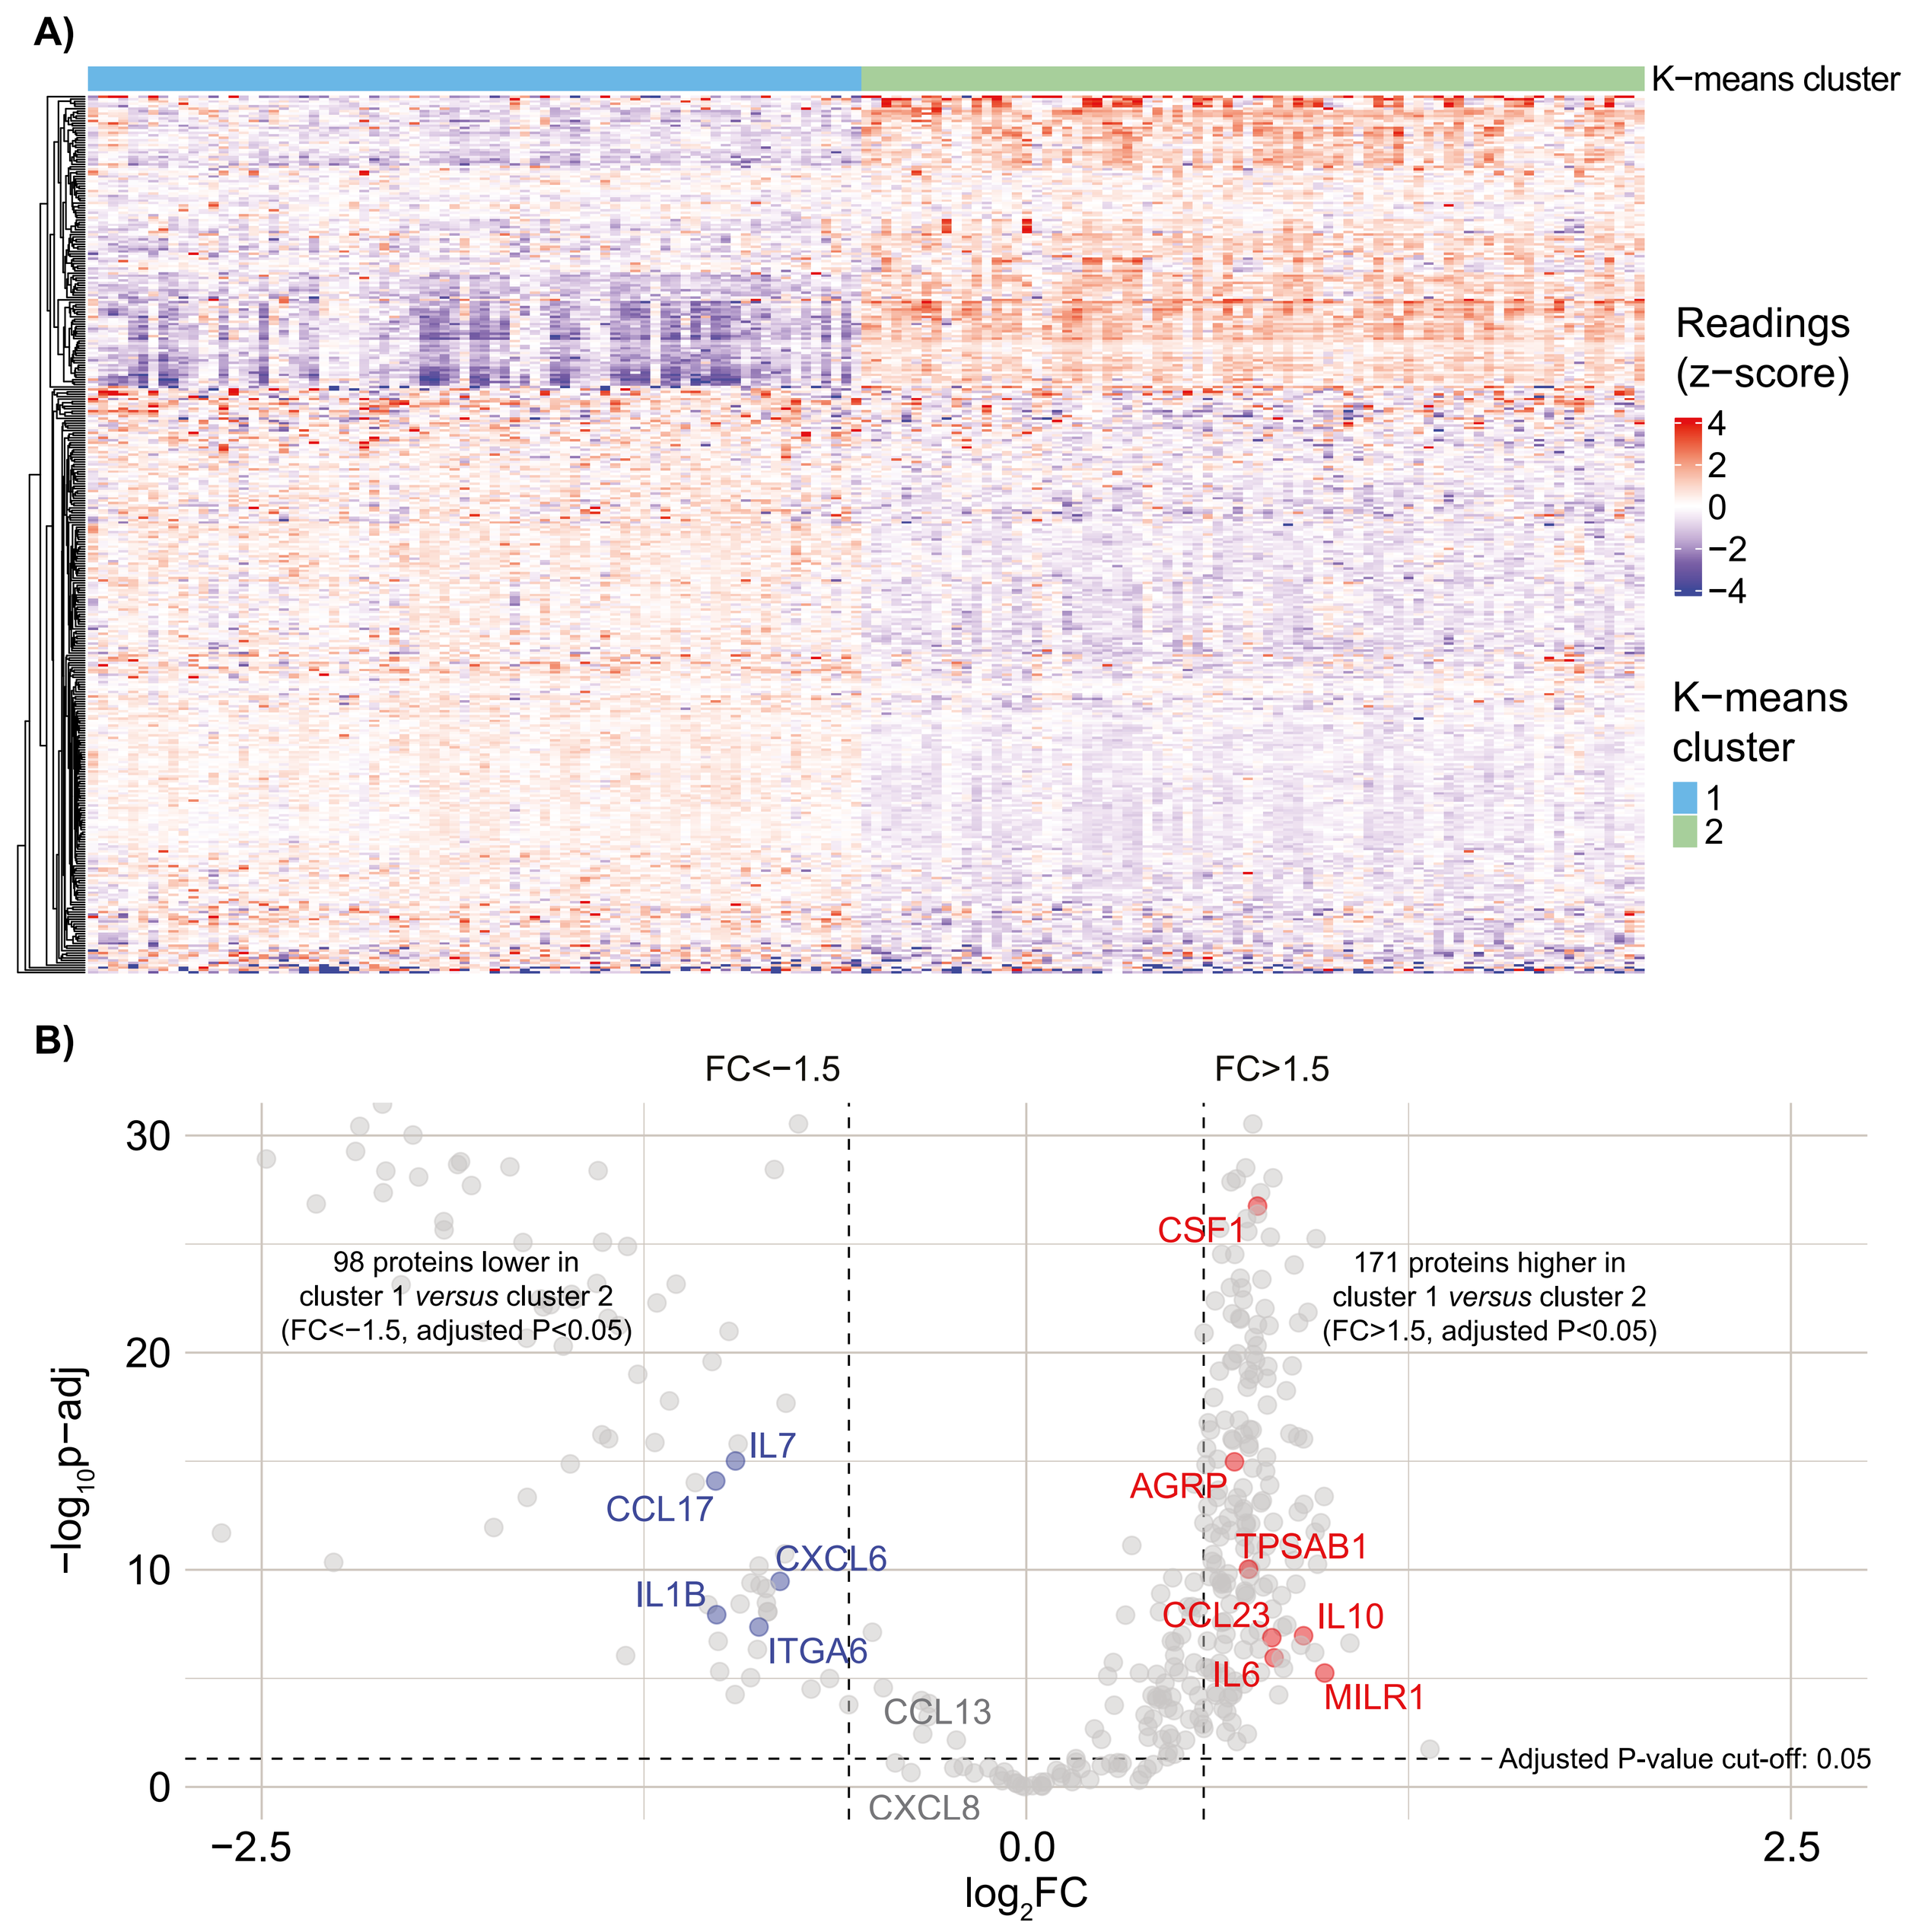

A)
K−means cluster
Readings (z−score)
4
2
0
−2
−4
K−means cluster
1
2
B)
FC<−1.5
FC>1.5
98 proteins lower in cluster 1 *versus* cluster 2 (FC<−1.5, adjusted P<0.05)
171 proteins higher in cluster 1 *versus* cluster 2 (FC>1.5, adjusted P<0.05)
IL7
CCL17
CXCL6
IL1B
ITGA6
CCL13
CXCL8
CSF1
AGRP
TPSAB1
CCL23
IL10
IL6
MILR1
Adjusted P-value cut-off: 0.05
−log10p−adj
log2FC
−2.5
0.0
2.5
0
10
20
30

Figure 7. Group analysis of disease burden measures in patients with ISM, by cluster

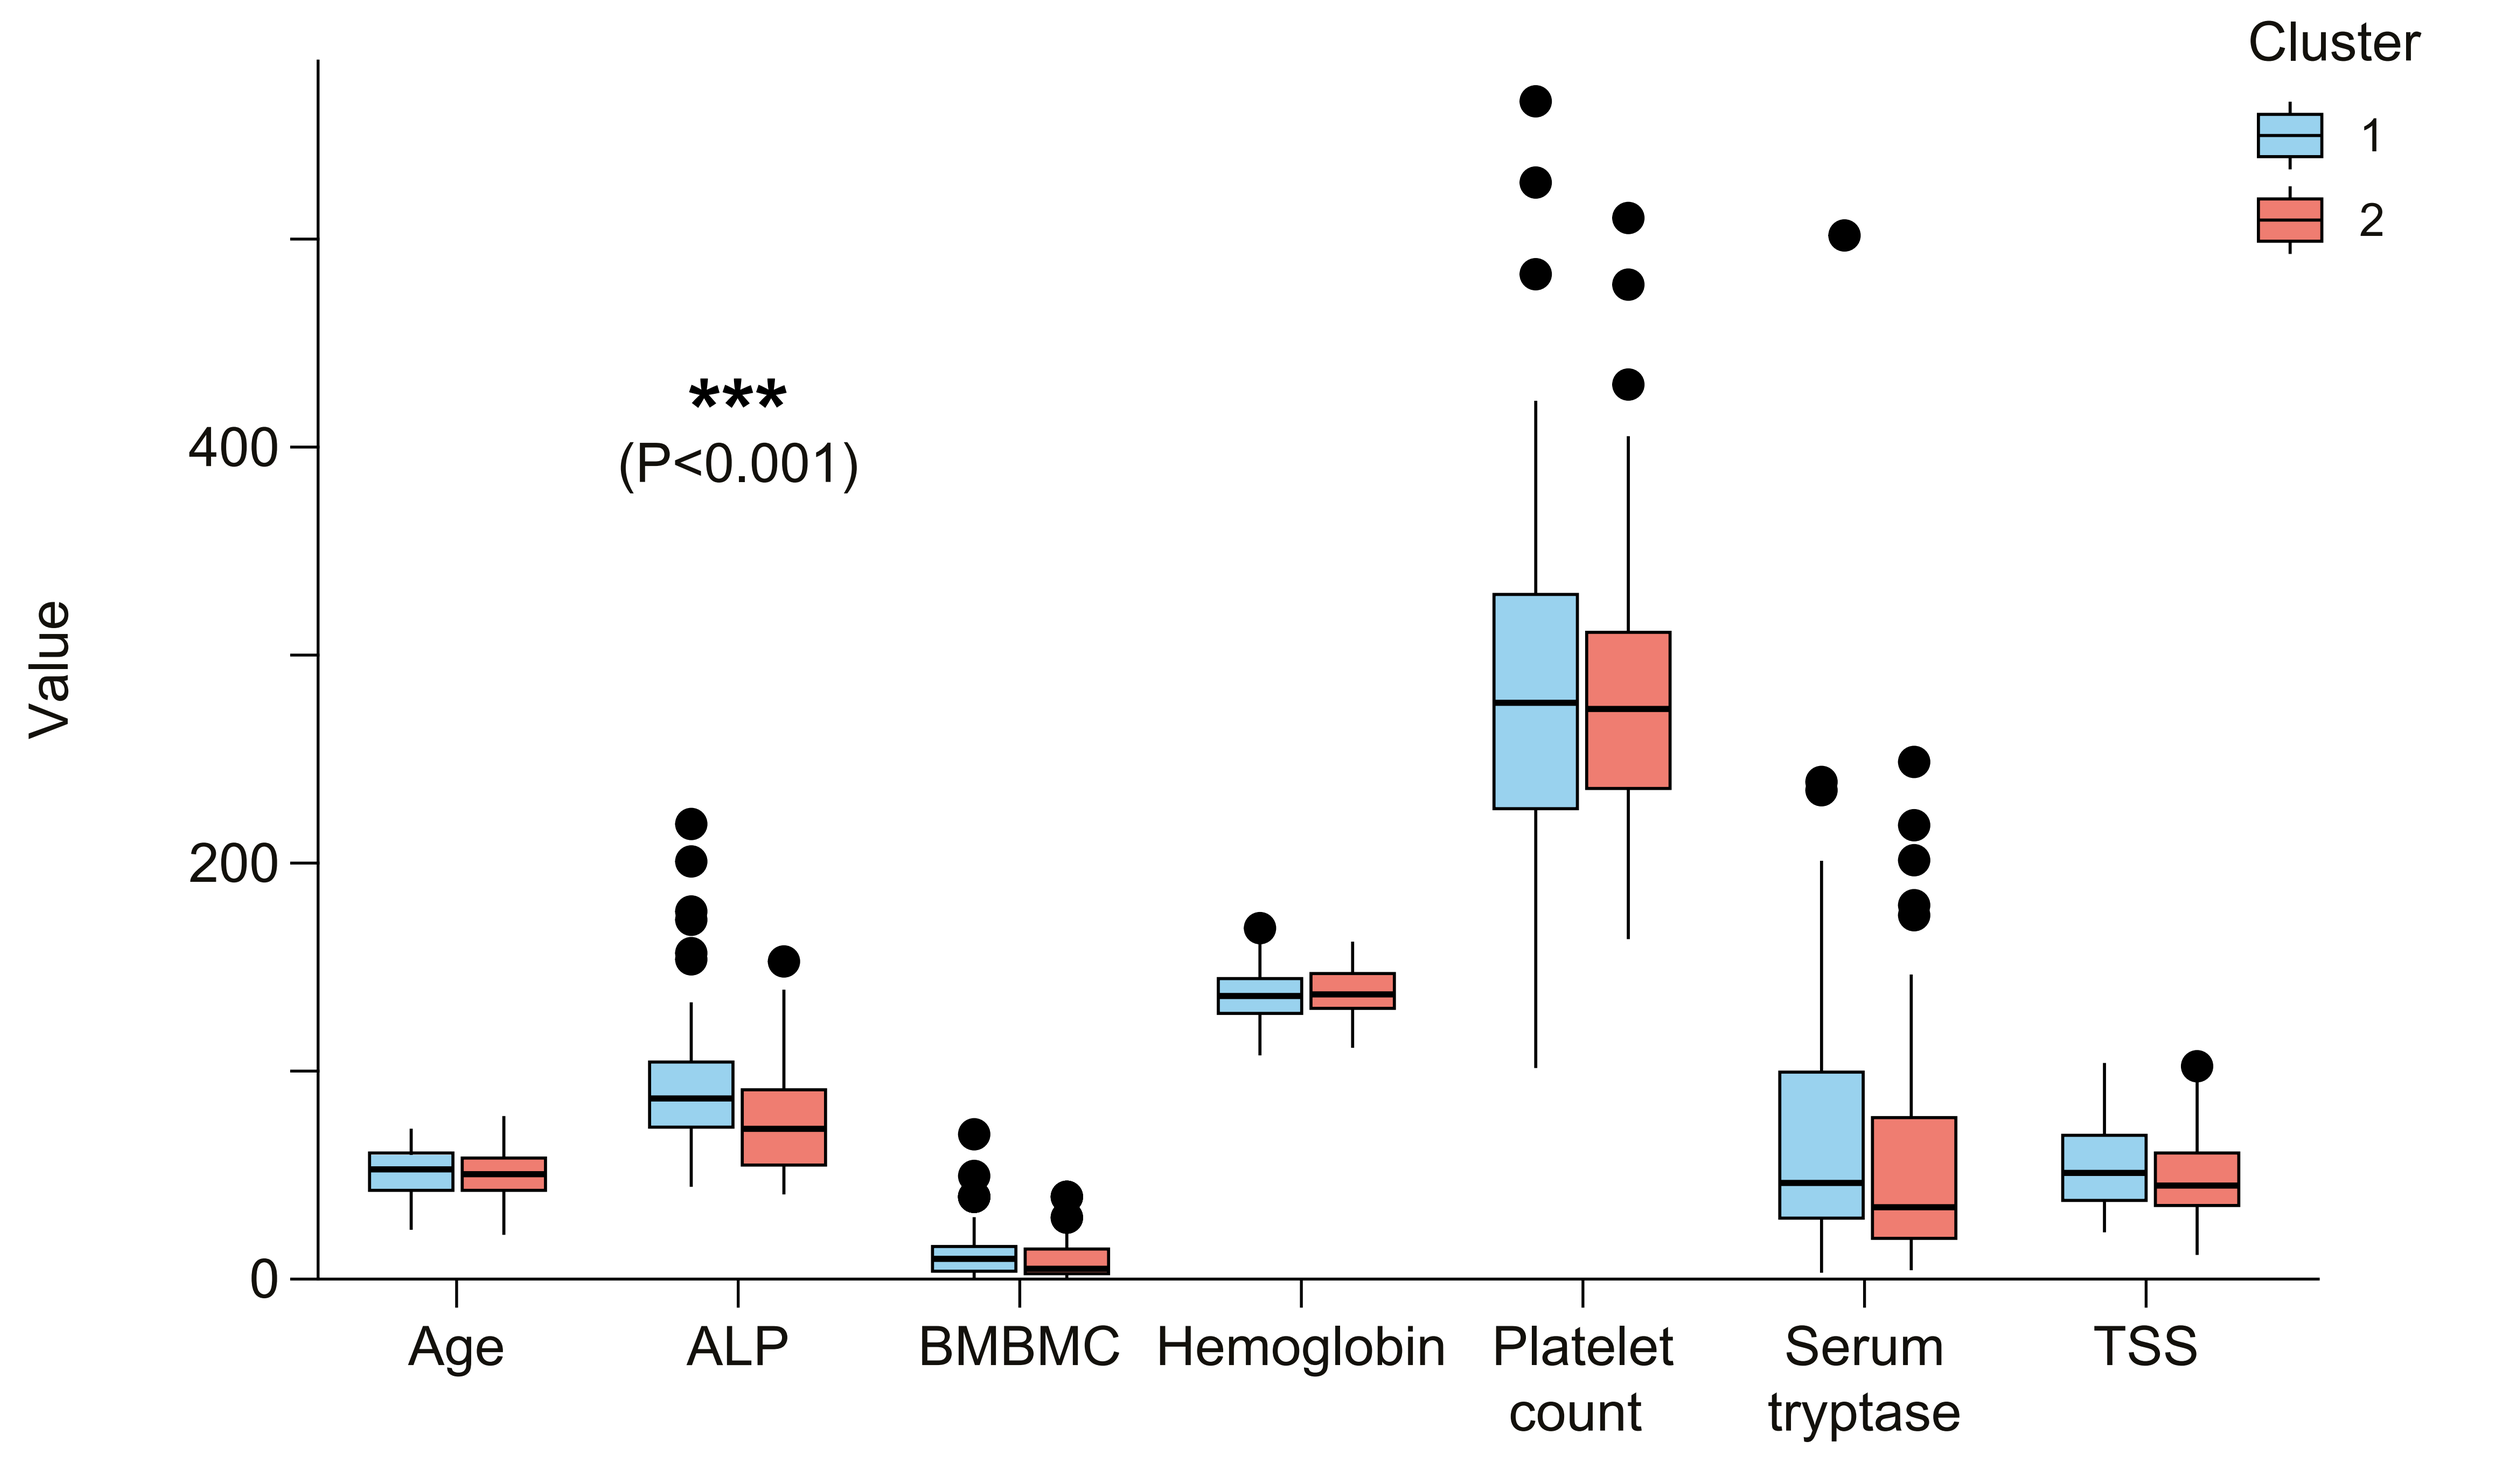

Cluster
1
2

(P<0.001)
Value
0
200
400
Age
ALP
BMBMC
Hemoglobin
Platelet count
Serum tryptase
TSS

- Baseline symptom scores and disease burden measures did not show any clear trends between clusters at a patient level
- When conducting comparisons between groups, ALP levels were statistically significantly higher in cluster 1 *versus* cluster 2 (raw unadjusted P< 0.001), while other markers of disease burden were similar between the clusters (**Figure 7**)

Conclusions

- Plasma proteomic analysis provides a new tool to understand and further characterize ISM
- Patients with ISM have many alterations in the plasma proteome compared with healthy individuals, highlighting the immune dysregulation seen with this disease
- Using this well-characterized cohort of patients, we identified two distinct plasma protein modules distinguished by negative (Module 1) *versus* positive (Module 2) correlations with disease burden
 - Module 1 included ITGA11, CSF3, and IL32, while Module 2 included MILR1, CCL23, and CD48
- Plasma protein ITGA11 was negatively correlated with disease burden and symptoms in patients with ISM
- K-means clustering of patient cytokine profiles and levels identified two distinct patient clusters within the overall population of patients with ISM
- Other than a difference in ALP expression, there were no differences in symptoms or disease burden measures between the two clusters
- Analysis of the inflammatory proteome present in patients with ISM provides a unique lens with which to view the disease and may eventually identify new therapeutic targets for its treatment

Acknowledgements

The authors thank the patients, their families, all other investigators and all investigational site members involved in this study. This study was funded by Blueprint Medicines Corporation. Medical writing support was provided by Matthew Nicolas, MSc and Michelle Seddon, Dip Psychol, both of Paragon (a division of Prime, Knutsford, UK), funded by Blueprint Medicines Corporation. Responsibility for all opinions, conclusions and data interpretation lies with the authors.

Scan the QR code to view our **Interactive Poster**

Conflicts of interest / Disclosures

Dr Kuo declares no conflict of interest. For all author disclosures, please contact medinfo@blueprintmedicines.com.

References

1. Kristensen T et al. *Am J Hematol*. 2014;89:493–498; 2. Ungerstedt J et al. *Cancers*. 2022;14:3942. 3. Garcia-Montero AC et al. *Blood*. 2006;108:2366–2372. 4. Pardanani A. *Am J Hematol*. 2023;98:1097–1116. 5. Bergstrom A et al. *Acta Oncol*. 2024;63:44–50; 6. Brockow K. *Immunol Allergy Clin North Am*. 2014;34:283–295; 7. Cohen SS et al. *Br J Haematol*. 2014;166:521–528; 8. van Doormaal JJ et al. *J Allergy Clin Immunol*. 2013;131:1429–1431.e1421; 9. Mesa RA et al. *Cancer*. 2022;128:3691–3699; 10. Hermine O et al. *PLoS One*. 2008;3:e2266; 11. van Anrooij B et al. *Allergy*. 2016;71:1585–1593; 12. Akin C et al. *J Allergy Clin Immunol*. 2022;149:1912–1918; 13. Evans EK et al. *Sci Transl Med*. 2017;9:eaao1690; 14.Blueprint Medicines Corporation. AYVAKYT® (avapritinib). Summary of Product Characteristics. 2024. Available at: https://www.ema.europa.eu/en/documents/product-information/ayvakyt-epar-product-information_en.pdf. Accessed June 4, 2024; 15. Blueprint Medicines Corporation. AYVAKIT® (avapritinib). Prescribing Information. 2024. Available at: https://www.blueprintmedicines.com/wp-content/uploads/uspi/AYVAKIT.pdf. Accessed May 2025; 16. Yang X et al. *Int J Gen Med*. 2024;17:471–483; 17. Martínez-Nieto GA et al. *Front Oncol*. 2022;12:981009.

Presented at the European Haematology Association (EHA) 2025; June 12–15, 2025, Milan, Italy. Please contact medinfo@blueprintmedicines.com for permission to reprint and/or distribute.