The Budget Impact of Adding Pralsetinib to a U.S. Health Plan Formulary for the Treatment of Non-Small Cell Lung Cancer and Thyroid Cancer with Rearranged During Transfection Alterations

Steve Duff[1], Francesca Bargiacchi[2], Chelsea Norregaard[2], Melanie Brener[2], Erin Sullivan[2]

[1]Veritas Health Economics Consulting, Carlsbad, CA, USA; [2]Blueprint Medicines Corporation, Cambridge, MA, USA

Background

- Non-small cell lung cancer (NSCLC)-attributable deaths were estimated to exceed 135,000 in the U.S. in 2020[1]; on the other hand, thyroid cancer is associated with low mortality but a rapidly rising incidence[2] such that, in 2013, more than 630,000 patients were living with thyroid cancer in the U.S.[3]
- The *Re*arranged during *T*ransfection (RET) alteration is an actionable biomarker present in a broad number of solid tumors including NSCLC and thyroid cancer.
- Recent advancements in targeting RET-altered tumors led to the FDA approval of two potent, selective RET inhibitors in 2020—selpercatinib (May) and pralsetinib (September).
- Pralsetinib (GAVRETO™) is an oral, selective RET inhibitor that potently targets RET-altered kinases including gatekeeper mutations (V804) associated with resistance to multikinase inhibitors; it is an FDA-approved treatment for the indicated populations of focus in this analysis.[4]

Objective: To conduct a budget impact analysis, adding pralsetinib to a U.S. health plan formulary for treatment of patients with metastatic RET fusion-positive NSCLC, advanced or metastatic RET-mutant medullary thyroid cancer (MTC), or advanced or metastatic RET fusion-positive thyroid cancer.

Methods

Model Approach and Key Model Inputs

- The budget impact model (BIM) takes the perspective of a hypothetical health plan of 1-million members with demographic characteristics reflective of the overall U.S. population and a payer distribution (Commercial, Medicare, and Medicaid) consistent with the U.S. insured population.
- The analysis time horizon is 3 years and accounts for the shift in treatment options from current therapies (without pralsetinib) to a future mix that includes pralsetinib.
- A comprehensive set of medical resources, including RET testing, was used to estimate the costs of each treatment regimen in the model and, after adjusting for market share, the total costs and incremental budget impact (per-member per-month; PMPM) were calculated (**Figure 1**).

Figure 1. Model Schematic

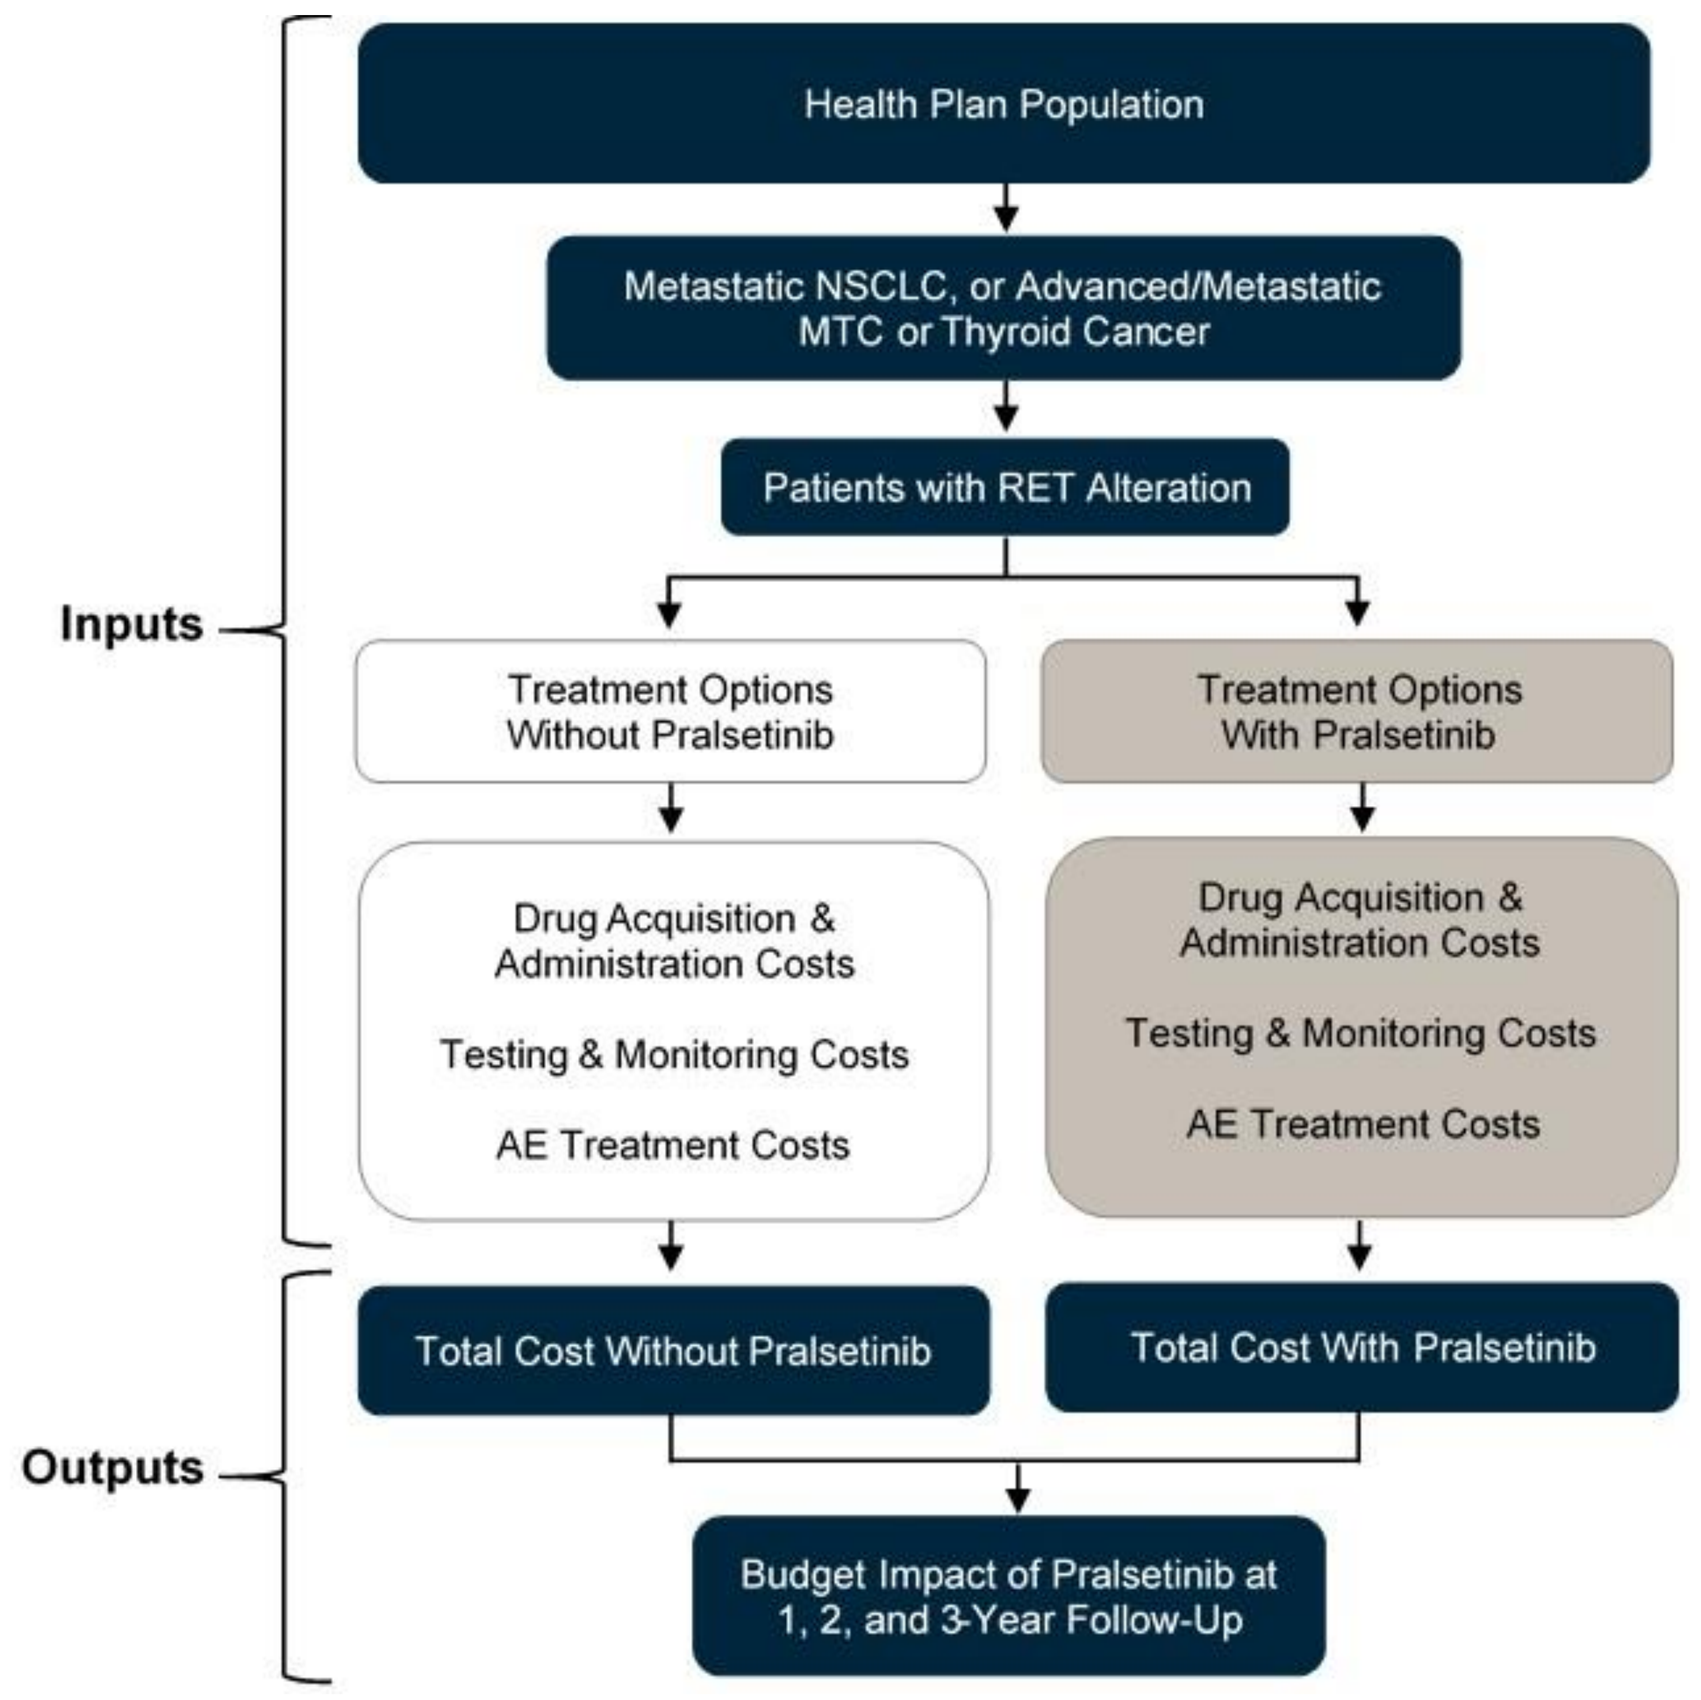
Inputs
Outputs
Health Plan Population
Metastatic NSCLC, or Advanced/Metastatic MTC or Thyroid Cancer
Patients with RET Alteration
Treatment Options Without Pralsetinib
Treatment Options With Pralsetinib
Drug Acquisition & Administration Costs
Testing & Monitoring Costs
AE Treatment Costs
Drug Acquisition & Administration Costs
Testing & Monitoring Costs
AE Treatment Costs
Total Cost Without Pralsetinib
Total Cost With Pralsetinib
Budget Impact of Pralsetinib at 1, 2, and 3-Year Follow-Up

Epidemiology and Treatments

- The number of treated patients is calculated from the age and sex distribution of covered lives, the age- and sex-adjusted incidence of disease, and the proportion of patients diagnosed with advanced or metastatic disease. The epidemiologic and disease-related parameters for these calculations are presented in **Table 1**.
- Treatments included in the model were indication-specific and represent commonly used therapies.
- The two selective RET inhibitors (pralsetinib and selpercatinib) were the only treatments assumed to potentially gain market share over time with Year 3 shares at ≥86%, split equally between the two RET inhibitors in the market scenario that includes pralsetinib.

Costs and Analyses

- A comprehensive set of medical resources/costs (2020 U.S. dollars) were included in the analysis (**Table 2**) based on prescribing information, Medicare reimbursement, and Redbook wholesale acquisition costs.
- RET testing cost varied by indication ($1,220-$1,845) due to the different distribution of tests employed including next generation sequencing panel tests.
- Base case analyses calculated total and incremental costs for each indication and as a total of all three combined; sensitivity and scenario analyses were conducted.

Table 1. Epidemiology Parameters

NSCLC			
Lung Cancer Incidence by Age (per 100,000)	**Male**	**Female**	**Age-/Sex-Adjusted**
18 – 64 years	23.2	23.9	23.0
≥65 years	343.3	270.7	303.3
Age- and Sex-Adjusted	**113.3**	**100.2**	**106.4**
Other Cancer-Specific Parameters			
% NSCLC		85.0%	
% metastatic		46.4%	
RET-Related Parameters	**Year 1**	**Year 2**	**Year 3**
RET fusion rate	2%	2%	2%
RET fusion testing	30%	50%	70%
MTC			
Thyroid Cancer Incidence by Age (per 100,000)[a]	**Male**	**Female**	**Age-/Sex-Adjusted**
12 – 17 years	2.6	10.2	6.4
18 – 64 years	9.0	28.9	19.2
≥65 years	17.6	26.1	22.3
Age- and Sex-Adjusted	**10.6**	**26.5**	**18.9**
Other Cancer-Specific Parameters			
% MTC		4.0%	
% advanced/metastatic		25.6%	
% requiring systemic therapy		53.0%	
RET-Related Parameters	**Year 1**	**Year 2**	**Year 3**
RET mutation rate	75%	75%	75%
RET mutation testing	100%	100%	100%
Thyroid Cancer			
Thyroid Cancer Incidence by Age (per 100,000)	**Male**	**Female**	**Age-/Sex-Adjusted**
12 – 17 years	2.6	10.2	6.4
18 – 64 years	9.0	28.9	19.2
≥65 years	17.6	26.1	22.3
Age- and Sex-Adjusted	**10.6**	**26.5**	**18.9**
Other Cancer-Specific Parameters			
% PTC, FTC, ATC (not MTC)		96.0%	
% advanced/metastatic		14.0%	
% requiring systemic therapy		90.0%	
% RAI-refractory		30.0%	
RET-Related Parameters	**Year 1**	**Year 2**	**Year 3**
RET fusion rate	17%	17%	17%
RET fusion testing	50%	70%	70%

Sources: Drilon 2018; Elisei 2019; Herbst 2018 Hirsch 2017; Lee 2017; Pietrowska 2017; Prescott 2015; Randle 2017; US Census Bureau 2020, US NIH SEER 2020; Blueprint Medicines Data on File

Key assumptions: 1) 2019 U.S. population estimates were increased by a 1% growth factor to more accurately reflect the 2020 population size; 2) in order to better estimate % advanced and metastatic, assumptions about SEER data characterized as 'distant' or 'regional' were required

Table 2. Cost Parameters

NSCLC					
Parameter	**Pralsetinib**	**Selpercatinib**	**Cabozantinib**	**Pembrolizumab**	**Pemetrexed + Carboplatin**
Treatment duration (months)	17.5	17.5	11.2	5.2	8.9 / 2.8
Drug acquisition (per month)	$19,243	$20,600	$19,285	$13,892	$10,412 / $79
Drug administration (per mo.)	$0	$0	$0	$204	$204 / $99
Adverse events	$2,172	$1,042	$1,371	$754	$207
Monitoring	$170	$178	$172	$176	$178

MTC				
Parameter	**Pralsetinib**	**Selpercatinib**	**Cabozantinib**	**Vandetanib**
Treatment duration (months)	22.0	22.0	11.2	22.6
Drug acquisition (per month)	$19,243	$20,600	$19,285	$14,680
Drug administration (per mo.)	$0	$0	$0	$0
Adverse events	$1,881	$1,050	$1,382	$633
Monitoring	$170	$178	$172	$179

Thyroid Cancer						
Parameter	**Pralsetinib**	**Selpercatinib**	**Cabozantinib**	**Vandetanib**	**Lenvatinib**	**Sorafenib**
Treatment duration (months)	18.4	18.4	11.2	22.6	18.3	10.8
Drug acquisition (per month)	$19,243	$20,600	$19,285	$14,680	$19,021	$19,981
Drug administration (per mo.)	$0	$0	$0	$0	$0	$0
Adverse events	$1,885	$1,050	$1,388	$633	$2,006	$701
Monitoring	$170	$178	$172	$179	$185	$170

Sources: AHRQ HCUPNet 2020; CMS 2020; Redbook 2020; US BLS 2020; Prescribing information for all compactors (2020)

Key assumptions: 1) treatment duration of pralsetinib and selpercatinib assumed equivalent given lack of evidence on PFS and duration of response (see Limitations); 2) clinical expert opinion was used to estimate some medical resource use for adverse event and ongoing patient management; 3) the WAC for the starting dose of both pralsetinib and selpercatinib was used as a simplifying assumption noting that patients on either therapy may have the dose reduced which would decrease drug acquisition costs

Conclusions

- Given the rarity of the RET-altered cancers in this budget impact analysis, only ~6 patients in a 1-million member U.S. health plan would be expected to be eligible for treatment annually with pralsetinib by Year 3.
- Pralsetinib adoption is projected to result in a manageable budget impact to a U.S. health plan, with a modest overall cost reduction when compared to existing standard of care treatment options.
- Differences in drug acquisition cost are a key cost driver of the incremental budget impact; all other medical resource categories were cost-neutral or only nominally cost saving or additive in the budget impact analysis.

Results

Base Case and Sensitivity Analysis Results

- By Year 3, 4.6 NSCLC patients, 0.7 MTC patients, and 0.7 thyroid cancer patients would initiate treatment annually in the hypothetical 1-million member U.S. health plan (**Table 3**).
- Expenditures were highest for NSCLC patients (~$1.5M in Year 3), but modest cost savings were achieved after adoption of pralsetinib across all three indications: -$6,495 to -$36,498 depending on indication (-$0.0005 to -$0.0030 PMPM).
- Drug acquisition cost was a key cost driver of cost savings; RET testing cost was highest for NSCLC patients given the greater number of patients tested in the plan (326 NSCLC patients vs. 6 thyroid cancer patients vs. 1 MTC patient)

Table 3. Base Case Budget Impact Results

Result	**NSCLC**			**MTC**			**Thyroid Cancer**		
	Year 1	**Year 2**	**Year 3**	**Year 1**	**Year 2**	**Year 3**	**Year 1**	**Year 2**	**Year 3**
Patient Count									
Number of new patients treated	2.0	3.3	4.6	0.7	0.7	0.7	0.5	0.7	0.7
With Pralsetinib Available									
Drug cost	$265,910	$606,671	$1,183,436	$138,197	$203,359	$239,832	$106,387	$190,793	$229,992
Administration cost	$1,486	$1,584	$676	$0	$0	$0	$0	$0	$0
Monitoring costs	$2,778	$5,768	$10,540	$1,324	$1,937	$2,163	$1,005	$1,793	$2,076
Adverse event costs	$1,937	$3,967	$6,785	$790	$860	$939	$620	$923	$1,002
RET testing cost	$119,169	$198,616	$278,062	$1,610	$1,610	$1,610	$4,629	$6,481	$6,481
Total	**$391,281**	**$816,606**	**$1,479,499**	**$141,921**	**$207,767**	**$244,544**	**$112,641**	**$199,990**	**$239,550**
Without Pralsetinib Available									
Drug cost	$269,727	$622,734	$1,221,911	$139,475	$207,302	$247,024	$107,356	$194,362	$236,707
Administration cost	$1,486	$1,584	$676	$0	$0	$0	$0	$0	$0
Monitoring costs	$2,802	$5,868	$10,779	$1,332	$1,961	$2,208	$1,011	$1,815	$2,117
Adverse event costs	$1,672	$2,973	$4,569	$725	$714	$694	$570	$766	$741
RET testing cost	$119,169	$198,616	$278,062	$1,610	$1,610	$1,610	$4,629	$6,481	$6,481
Total	**$394,856**	**$831,775**	**$1,515,997**	**$143,143**	**$211,586**	**$251,536**	**$113,566**	**$203,425**	**$246,045**
Incremental Results									
Drug cost	-$3,817	-$16,063	-$38,475	-$1,279	-$3,942	-$7,192	-$969	-$3,569	-$6,715
Administration cost	$0	$0	$0	$0	$0	$0	$0	$0	$0
Monitoring costs	-$24	-$100	-$239	-$8	-$24	-$45	-$6	-$22	-$42
Adverse event costs	$265	$994	$2,215	$65	$147	$245	$50	$157	$261
RET testing cost	$0	$0	$0	$0	$0	$0	$0	$0	$0
Total	**-$3,576**	**-$15,169**	**-$36,498**	**-$1,221**	**-$3,820**	**-$6,992**	**-$925**	**-$3,434**	**-$6,495**
Total (PMPM)	**-$0.0003**	**-$0.0013**	**-$0.0030**	**-$0.0001**	**-$0.0003**	**-$0.0006**	**-$0.0001**	**-$0.0003**	**-$0.0005**

SUMMARY RESULTS

- Approximately 6 new patients initiate treatment annually by Year 3
- Total cost of all 3 indications in Year 3:
 - Future Scenario ~$1.96M
 - Current Scenario ~$1.97M
- **Incremental cost of all 3 indications in Year 3:**
 - **Total: -$49,985**
 - **PMPM: -$0.0041**

- One-way sensitivity analyses were conducted by increasing and decreasing key values by 20%.
- Uncertainty in the RET testing assumption (no change in trajectory upon pralsetinib introduction), pralsetinib drug cost, and duration of treatment had the greatest impact on the potential incremental budget impact (**Figure 2**).
- Only these 3 parameters resulted in additive budget impact (PMPM range: $0.0028 to $0.0201).

Figure 2. Sensitivity Analysis of Budget Impact Results (PMPM) for all Three Indications Combined

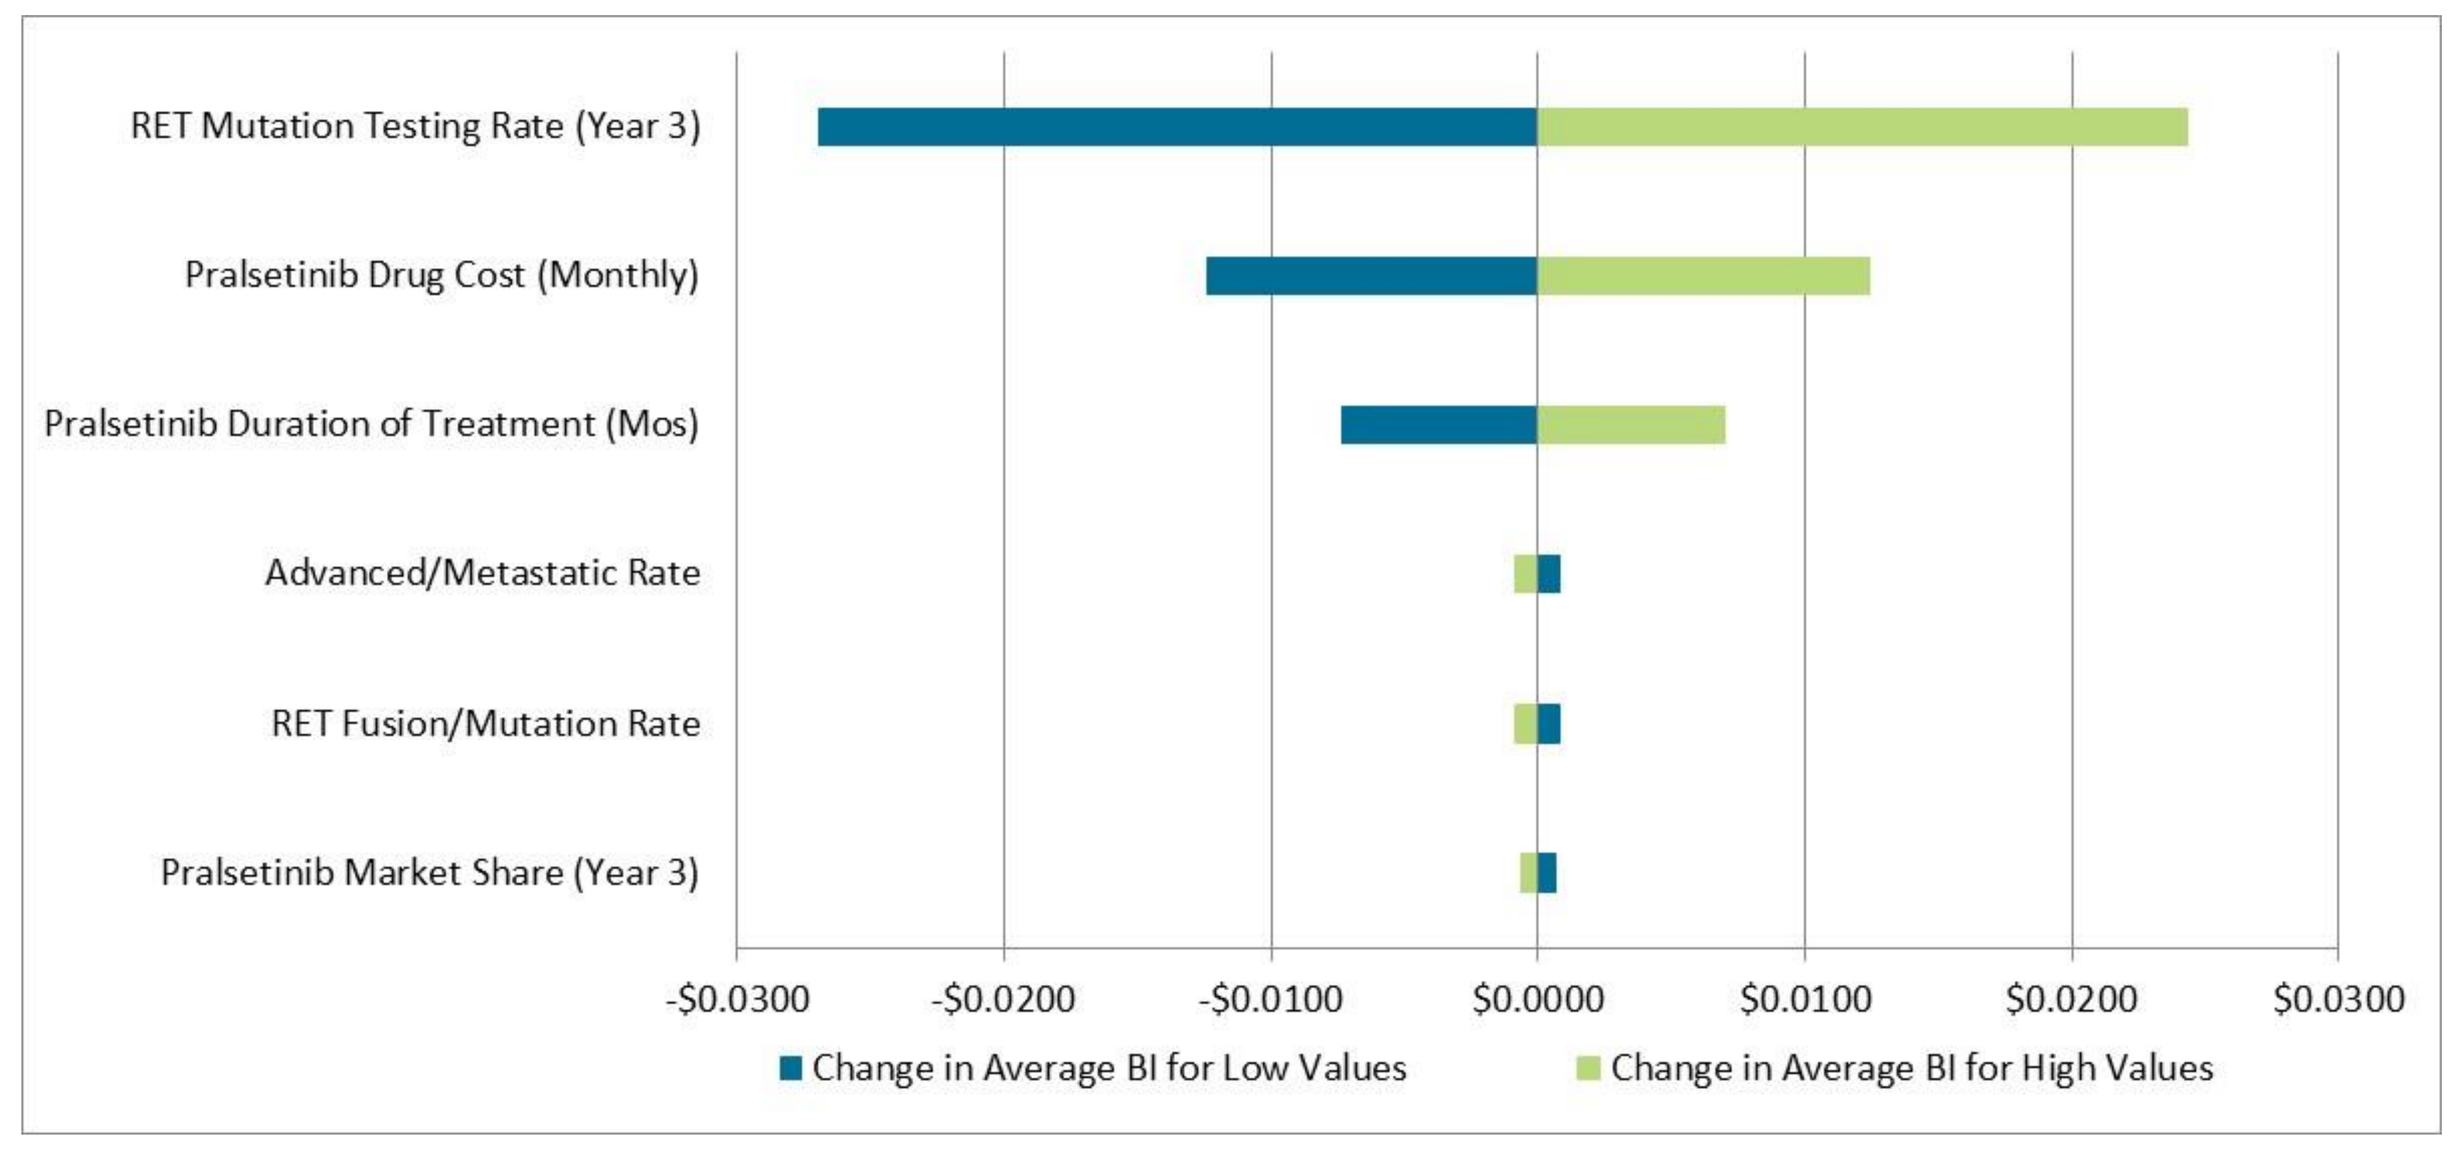
RET Mutation Testing Rate (Year 3)
Pralsetinib Drug Cost (Monthly)
Pralsetinib Duration of Treatment (Mos)
Advanced/Metastatic Rate
RET Fusion/Mutation Rate
Pralsetinib Market Share (Year 3)
-$0.0300
-$0.0200
-$0.0100
$0.0000
$0.0100
$0.0200
$0.0300
Change in Average BI for Low Values
Change in Average BI for High Values

Limitations

- Studies of RET-altered patients tend to be observational or of small sample size; therefore, we assumed that epidemiologic data from the cancers of interest applied to RET-altered populations.
- Pralsetinib and selpercatinib do not yet have published progression-free survival or response duration data available for all patient cohorts; thus, treatment duration estimates are uncertain.
- The BIM does not evaluate sequential lines of therapy and treatment algorithms. Market share estimates were considered to be cross-sectional and representative of the distribution of treatment comparators.

Background References: [1]Siegel RL. CA Cancer J Clin 2020;70(1):7-30; [2]Jung CK. J Clin Endocrinol Metab 2014;99(2):E276-E285; [3]American Thyroid Association 2020 https://www.thyroid.org/medullary-thyroid-cancer/; [4]Pralsetinib prescribing information 2020 https://www.accessdata.fda.gov/drugsatfda_docs/label/2020/214701s000lbl.pdf

Modeling References: Drilon A. Nat Rev Clin Oncol. 2018;15(3):151-167; Elisei R. Genes (Basel). 2019;10(9):698; Healthcare Cost and Utilization Project. Agency for Healthcare Research and Quality, Rockville, MD. https://hcupnet.ahrq.gov/ Accessed June 9, 2020; Herbst RS. Nature 2018;553(7689):446-454; Hirsch FR. Lancet 2017;389(10066):299-311; Lee MY. Cancer Res Treat. 2017;49(4):906-914; Pietrowska M. Biochimica et Biophysica Acta. 2017;1865(7):837-845; Prescott JD. Cancer 2015;121(13):2137- 2146; Randle RW. Surgery 2017;161(1):137-146; RED BOOK. IBM Micromedex. Accessed May 26, 2020; U.S. Census Bureau, Current Population Survey, Annual Social and Economic Supplement, 2019. Table 1. Population by Age and Sex: 2019; Internet release date April 2020; U.S. Centers for Medicare and Medicaid Services. 2020 Medicare Physician Fee Schedule and Clinical Diagnostic Fee Schedule, accessed in May/June 2020; U.S. NIH SEER 21 data for recent trends in SEER age-adjusted incidence rates 2000-2017, lung and bronchus cancer; U.S. NIH SEER 21 data for recent trends in SEER age-adjusted incidence rates 2000-2017, thyroid cancer; U.S. Prescribing Information for pralsetinib and all comparators

Support for this study was provided by Blueprint Medicines

Abbreviations: AE: adverse event; AHRQ: Agency for Healthcare Research and Quality; ATC: anaplastic thyroid cancer; BIM: budget impact model; BLS: Bureau of Labor Statistics: CMS: Centers for Medicare and Medicaid Services; FDA: Food and Drug Administration; FTC: follicular thyroid cancer; HCUPNet: Healthcare Cost and Utilization Project Network; M: million; Mo: month; MTC: medullary thyroid cancer; NIH: National Institutes of Health; NSCLC: non-small cell lung cancer; PFS: progression-free survival; PMPM: per-member per-month; PTC: papillary thyroid cancer; RAI: radioiodine-refractory; RET: rearranged during transfection; SEER: Surveillance, Epidemiology, and End Results Program; US: United States