TPS9156

A phase 1/2 study of BLU-945 in patients with common activating *EGFR*-mutant non-small cell lung cancer (NSCLC) (SYMPHONY trial-in-progress)

Elaine Shum,[1] Yasir Elamin,[2] Zofia Piotrowska,[3] David R Spigel,[4] Karen L Reckamp,[5] Julia Rotow,[6] Daniel SW Tan,[7] Sun Min Lim,[8] Tae Min Kim,[9] Chia-Chi Lin,[10] Terufumi Kato,[11] Jagan Parepally,[12] Faris Albayya,[12] Melinda Louie-Gao,[12] Tessa Weinig,[12] Alena Zalutskaya,[12] Koichi Goto[13]

[1]Perlmutter Cancer Center, New York University Langone Health, New York, NY, USA; [2]MD Anderson Cancer Center, University of Texas, Houston, TX, USA; [3]Massachusetts General Hospital, Boston, MA, USA; [4]Sarah Cannon Research Institute/Tennessee Oncology, Nashville, TN, USA;
[5]Department of Medicine, Cedars-Sinai Medical Center, Los Angeles, CA, USA; [6]Dana-Farber Cancer Institute, Boston, MA, USA; [7]Division of Medical Oncology, National Cancer Center Singapore, Singapore, Singapore; [8]Department of Internal Medicine, Yonsei University, Seoul, Republic of Korea; [9]Department of Internal Medicine, Seoul National University Hospital, Seoul National University College of Medicine, Seoul, Republic of Korea; [10]Department of Oncology, National Taiwan University Hospital, Taipei, Taiwan; [11]Department of Thoracic Oncology, Kanagawa Cancer Center, Yokohama, Japan; [12]Blueprint Medicines Corporation, Cambridge, MA, USA; [13]National Cancer Center Hospital East, Kashiwa, Japan

Background

- The most frequent oncogenic drivers of non-small cell lung cancer (NSCLC) are epidermal growth factor receptor mutations (*EGFRm*), with the most common mutations being exon 19 deletion (ex19del) and L858R[1–3]
- Although EGFR-targeted therapies such as tyrosine kinase inhibitors (TKIs) have improved outcomes in patients with *EGFRm* NSCLC, on- and off-target resistance mutations to these drugs is inevitable[4]
- The most frequent mutations, *EGFR* T790M and *EGFR* C797S, can occur simultaneously within an individual patient and develop after treatment with first- and second-generation TKIs and third-generation TKIs, respectively[4,5]
- There are unmet medical needs for patients with *EGFRm* NSCLC and resistance mutations, highlighted by the current lack of approved therapies after progression on available therapies[5]
- BLU-945 is an investigational oral next-generation EGFR TKI designed to selectively target the L858R activating mutation, and the C797S and T790M on-target resistance EGFR mutations with nanomolar potency while sparing wildtype EGFR[6] (**Figure 1**)
- Preclinically, it has shown activity as monotherapy in treatment-naïve *EGFRm* xenograft and osimertinib-resistant, *EGFRm* patient-derived xenograft (PDX) models[7,8]
- Importantly, as BLU-945 has high selectivity for EGFR mutations while sparing wildtype, this could be advantageous for combination treatment with complementary therapies, such as osimertinib and the next-generation EGFR TKI BLU-701. Such combinations may provide the benefit of treating multiple mutations without increased toxicity. BLU-945 in combination with other agents has previously shown enhanced activity in *EGFRm* PDX models[7,8]

Figure 1: Combination of EGFR inhibitors provides broadest coverage of common EGFR resistance mutations

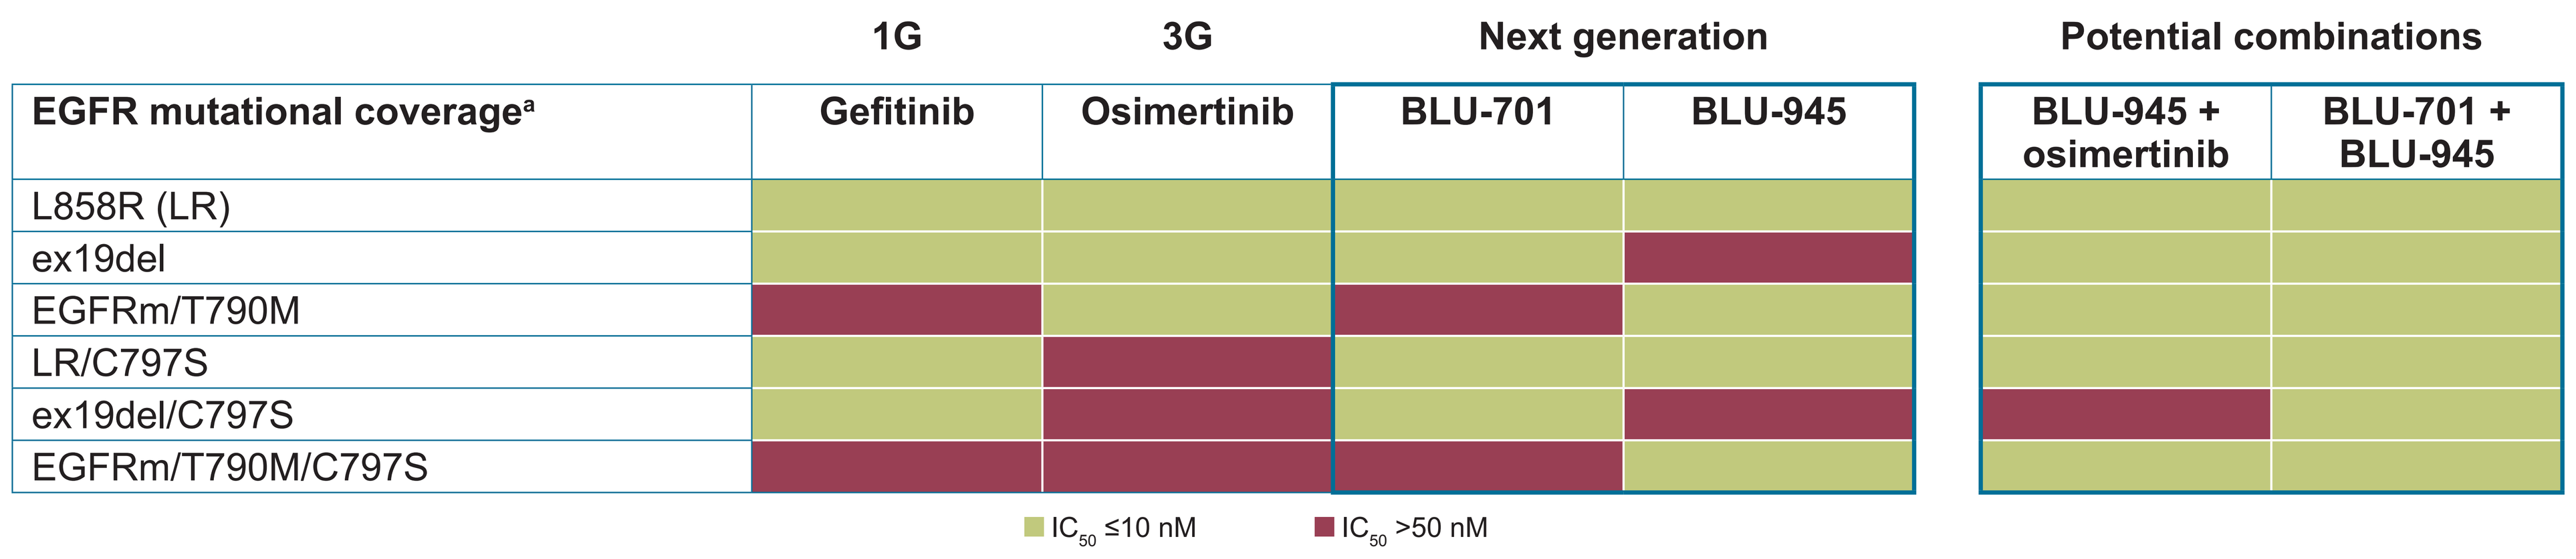
1G
3G
Next generation
Potential combinations
EGFR mutational coverage[a]
Gefitinib
Osimertinib
BLU-701
BLU-945
BLU-945 + osimertinib
BLU-701 + BLU-945
L858R (LR)
ex19del
EGFRm/T790M
LR/C797S
ex19del/C797S
EGFRm/T790M/C797S
IC_{50} ≤10 nM
IC_{50} >50 nM

[a]Based on biochemical IC_{50}. 1G, first-generation; 3G, third-generation; IC_{50}, half-maximal inhibitory concentration.
EGFRm: primary EGFR mutation, either L858R or ex19del.

Study objectives and design

- The SYMPHONY trial (BLU-945; NCT04862780) is an international, open-label, first-in-human, phase 1/2 study designed to evaluate safety, tolerability, and antitumor activity of BLU-945 as monotherapy and in combination with osimertinib in patients with *EGFRm* NSCLC (**Figure 2**)

Figure 2: Study design

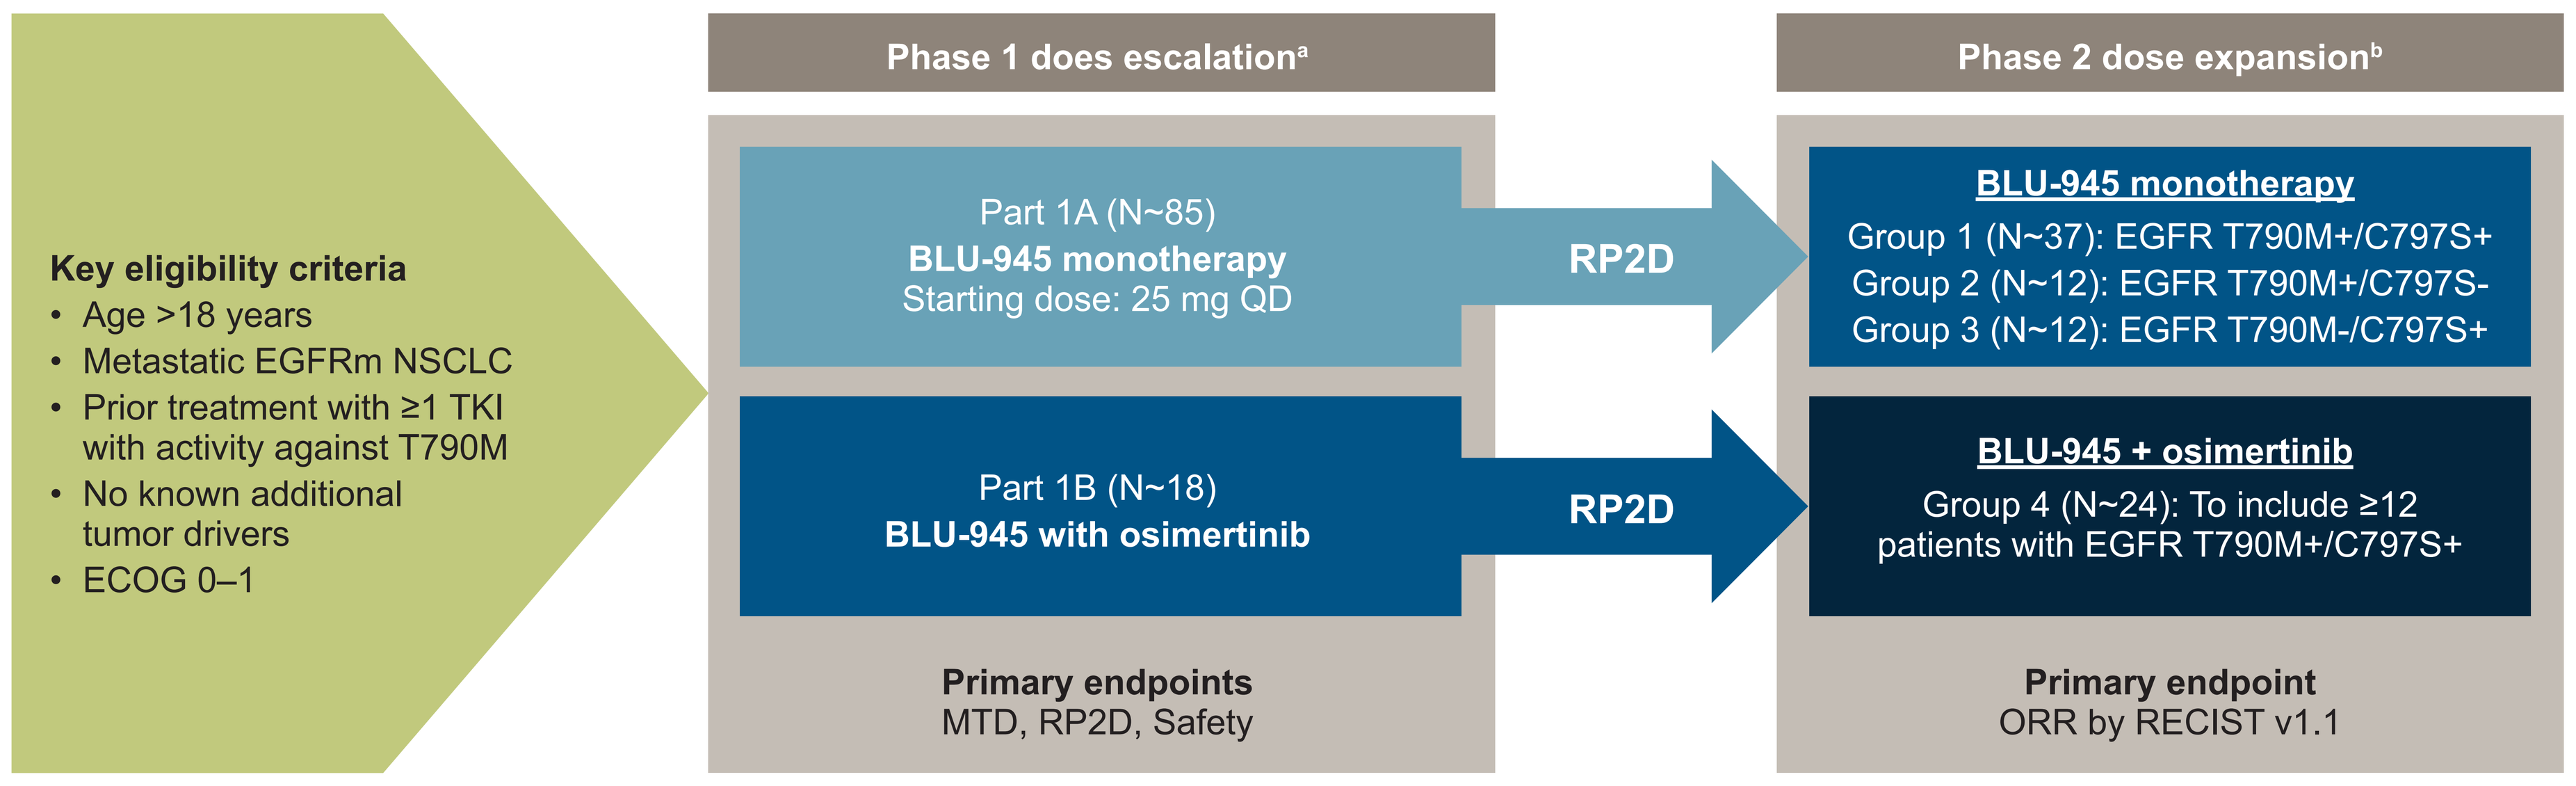
Key eligibility criteria
• Age >18 years
• Metastatic EGFRm NSCLC
• Prior treatment with ≥1 TKI with activity against T790M
• No known additional tumor drivers
• ECOG 0–1
Phase 1 does escalation[a]
Part 1A (N~85)
BLU-945 monotherapy
Starting dose: 25 mg QD
Part 1B (N~18)
BLU-945 with osimertinib
Primary endpoints
MTD, RP2D, Safety
RP2D
RP2D
Phase 2 dose expansion[b]
BLU-945 monotherapy
Group 1 (N~37): EGFR T790M+/C797S+
Group 2 (N~12): EGFR T790M+/C797S-
Group 3 (N~12): EGFR T790M-/C797S+
BLU-945 + osimertinib
Group 4 (N~24): To include ≥12 patients with EGFR T790M+/C797S+
Primary endpoint
ORR by RECIST v1.1

[a]Based on Bayesian Optimal Interval escalation design (BOIN). [b]Initiation of phase 2 will be dependent on results of phase 1.
ECOG, Eastern Cooperative Oncology Group performance status; *EGFRm*, mutant epidermal growth factor receptor gene; MTD, maximum tolerated dose; NSCLC, non-small cell lung cancer; ORR, overall response rate; QD, once daily; RECIST 1.1, Response Evaluation Criteria in Solid Tumors version 1.1; RP2D, recommended phase 2 dose; TKI, tyrosine kinase inhibitor.

- Phase 1 dose escalation will be conducted using Bayesian optimal interval (BOIN) design with up to 12 patients evaluable for dose-limiting toxicities (DLTs) for any given dose level, and dose escalation will be considered complete when 12 patients are evaluable for DLT at one dose level. The DLT evaluation period is the first 28 days (cycle 1 of each cohort in the phase 1 dose escalation). Patients who experience a DLT or who receive at least 75% of the prescribed BLU-945 dose (i.e., ≥21 days) and complete the 28-day DLT evaluation period will be evaluable for DLT assessment
- Intra-patient dose escalation will be permitted for patients enrolled at previously tested dose levels that have been evaluated for safety
- BLU-945 and osimertinib combination treatment escalation will be initiated at 50% of the recommended phase 2 dose (RP2D) or 50% of the highest safe dose in the ongoing phase 1 BLU-945 monotherapy part of the study. Osimertinib will be given at the standard dose of 80 mg once daily (QD)
- In phase 1 part B, the dose level increase should be <100% of BLU-945 in the cohort(s) subsequent to cohort 1 and the next dose of BLU-945 will be selected such that the projected area under the plasma concentration time curve (AUC) of BLU-945 in combination with osimertinib will not be more than the AUC of BLU-945 as monotherapy at the maximum tolerated dose (MTD) or the highest dose deemed safe in part 1A in the combination dose-escalation
- The MTD will be determined based on the DLT rate and the dose for which the isotonic estimate of the toxicity rate is closest to the target toxicity rate (30%)
- The RP2D will not exceed the MTD and will be determined based on pharmacokinetics (PK), pharmacodynamics (PD), cumulative toxicity, and antitumor activity. If a MTD is not identified then PK, PD, and safety data, along with pertinent nonclinical data suggestive of a dose-effect relationship, will be used to define a RP2D
- In phase 2, patients will be enrolled based on specific *EGFRm* profiles at the RP2D determined in phase 1:
 - Groups 1–3 will be treated with BLU-945 monotherapy at the RP2D and schedule selected in phase 1 part A: *EGFR* T790M+/C797S+, *EGFR* T790M+/C797S-, *EGFR* T790M-/C797S+
 - Group 4 will be treated with BLU-945 combined with osimertinib at the RP2D and schedule selected in phase 1 part B: to include at least 12 patients with *EGFR* T790M+/C797S+
- Patients with disease progression may continue study treatment if ongoing clinical benefit is observed (as assessed by the investigator, and approved by the Sponsor)

Summary of key inclusion and exclusion criteria

Key inclusion criteria	Key exclusion criteria
• Age ≥18 years • Pathologically confirmed metastatic *EGFRm* NSCLC • Prior treatment with ≥1 EGFR-targeted TKI against T790M • Tumor mutation profile determined locally using tissue or plasma specimen via a sponsor-approved methodology (preferably NGS) • Pretreatment tumor sample (archival sample or pretreatment biopsy) submitted for central analysis • Part 1A: willing to undergo on-treatment biopsy at doses expected to result in efficacious exposure levels if safe and medically feasible • ECOG performance status 0–1	• Additional known tumor drivers[a] • NSCLC with mixed cell histology or with histologic transformation • Any immunotherapy or other antibody therapy within 28 days prior to first dose • Any other systemic anticancer therapy within 14 days or 5 half-lives (whichever is shortest) prior to first dose • Radiation within 14 days before first dose if including a vital organ, or 7 days if not including a vital organ • CNS metastases or spinal cord compression associated with progressive neurological symptoms. Patients with asymptomatic brain metastases who are on stable doses of corticosteroids are allowed

[a]Including, but not limited to *EGFR* exon 20 insertion, or pathologic abnormalities of *KRAS, BRAF, V600E, NTRK1/2/3, HER2, ALK, ROS1, MET,* or *RET*.
CNS, central nervous system; ctDNA, circulating tumor DNA; ECOG, Eastern Cooperative Oncology Group; *EGFRm*, epithelial growth factor receptor mutant; NGS, next-generation sequencing; NSCLC, non-small cell lung cancer; TKI, tyrosine kinase inhibitor.

Key study endpoints

Phase 1	Phase 2
• **Primary endpoints** – Maximum tolerated dose – Recommended phase 2 dose – Safety and tolerability • **Secondary endpoints** – Overall Response Rate (RECIST 1.1) – Duration of response – Pharmacokinetics – Modulation of *DUSP6* and *SPRY4* expression levels	• **Primary endpoint** – Overall Response Rate (RECIST 1.1) • **Secondary endpoints** – Duration of response – Disease control rate – Clinical benefit rate – Progression-free survival – Overall survival – Safety and tolerability – Cardiovascular parameters, including QTcF – Pharmacokinetics – CNS anticancer activity • Overall response rate (RECIST 1.1) • Duration of response • Progression rate

[a]ctDNA will be assessed in real-time using the FoundationOne Liquid CDx assay.
CNS, central nervous system; ctDNA, circulating tumor DNA; DUSP6, Dual specificity phosphatase 6; *EGFR*, epithelial growth factor receptor;
QTcF; QT interval corrected using Fridericia's formula; RECIST 1.1,
Response Evaluation Criteria in Solid Tumors version 1.1; SPRY4, sprouty RTK signaling antagonist 4.

Enrollment and status

- The phase 1 dose-escalation portion of the study is ongoing
- The study is planned for approximately 30 centers in North America, Europe, and Asia

Anticipated study locations

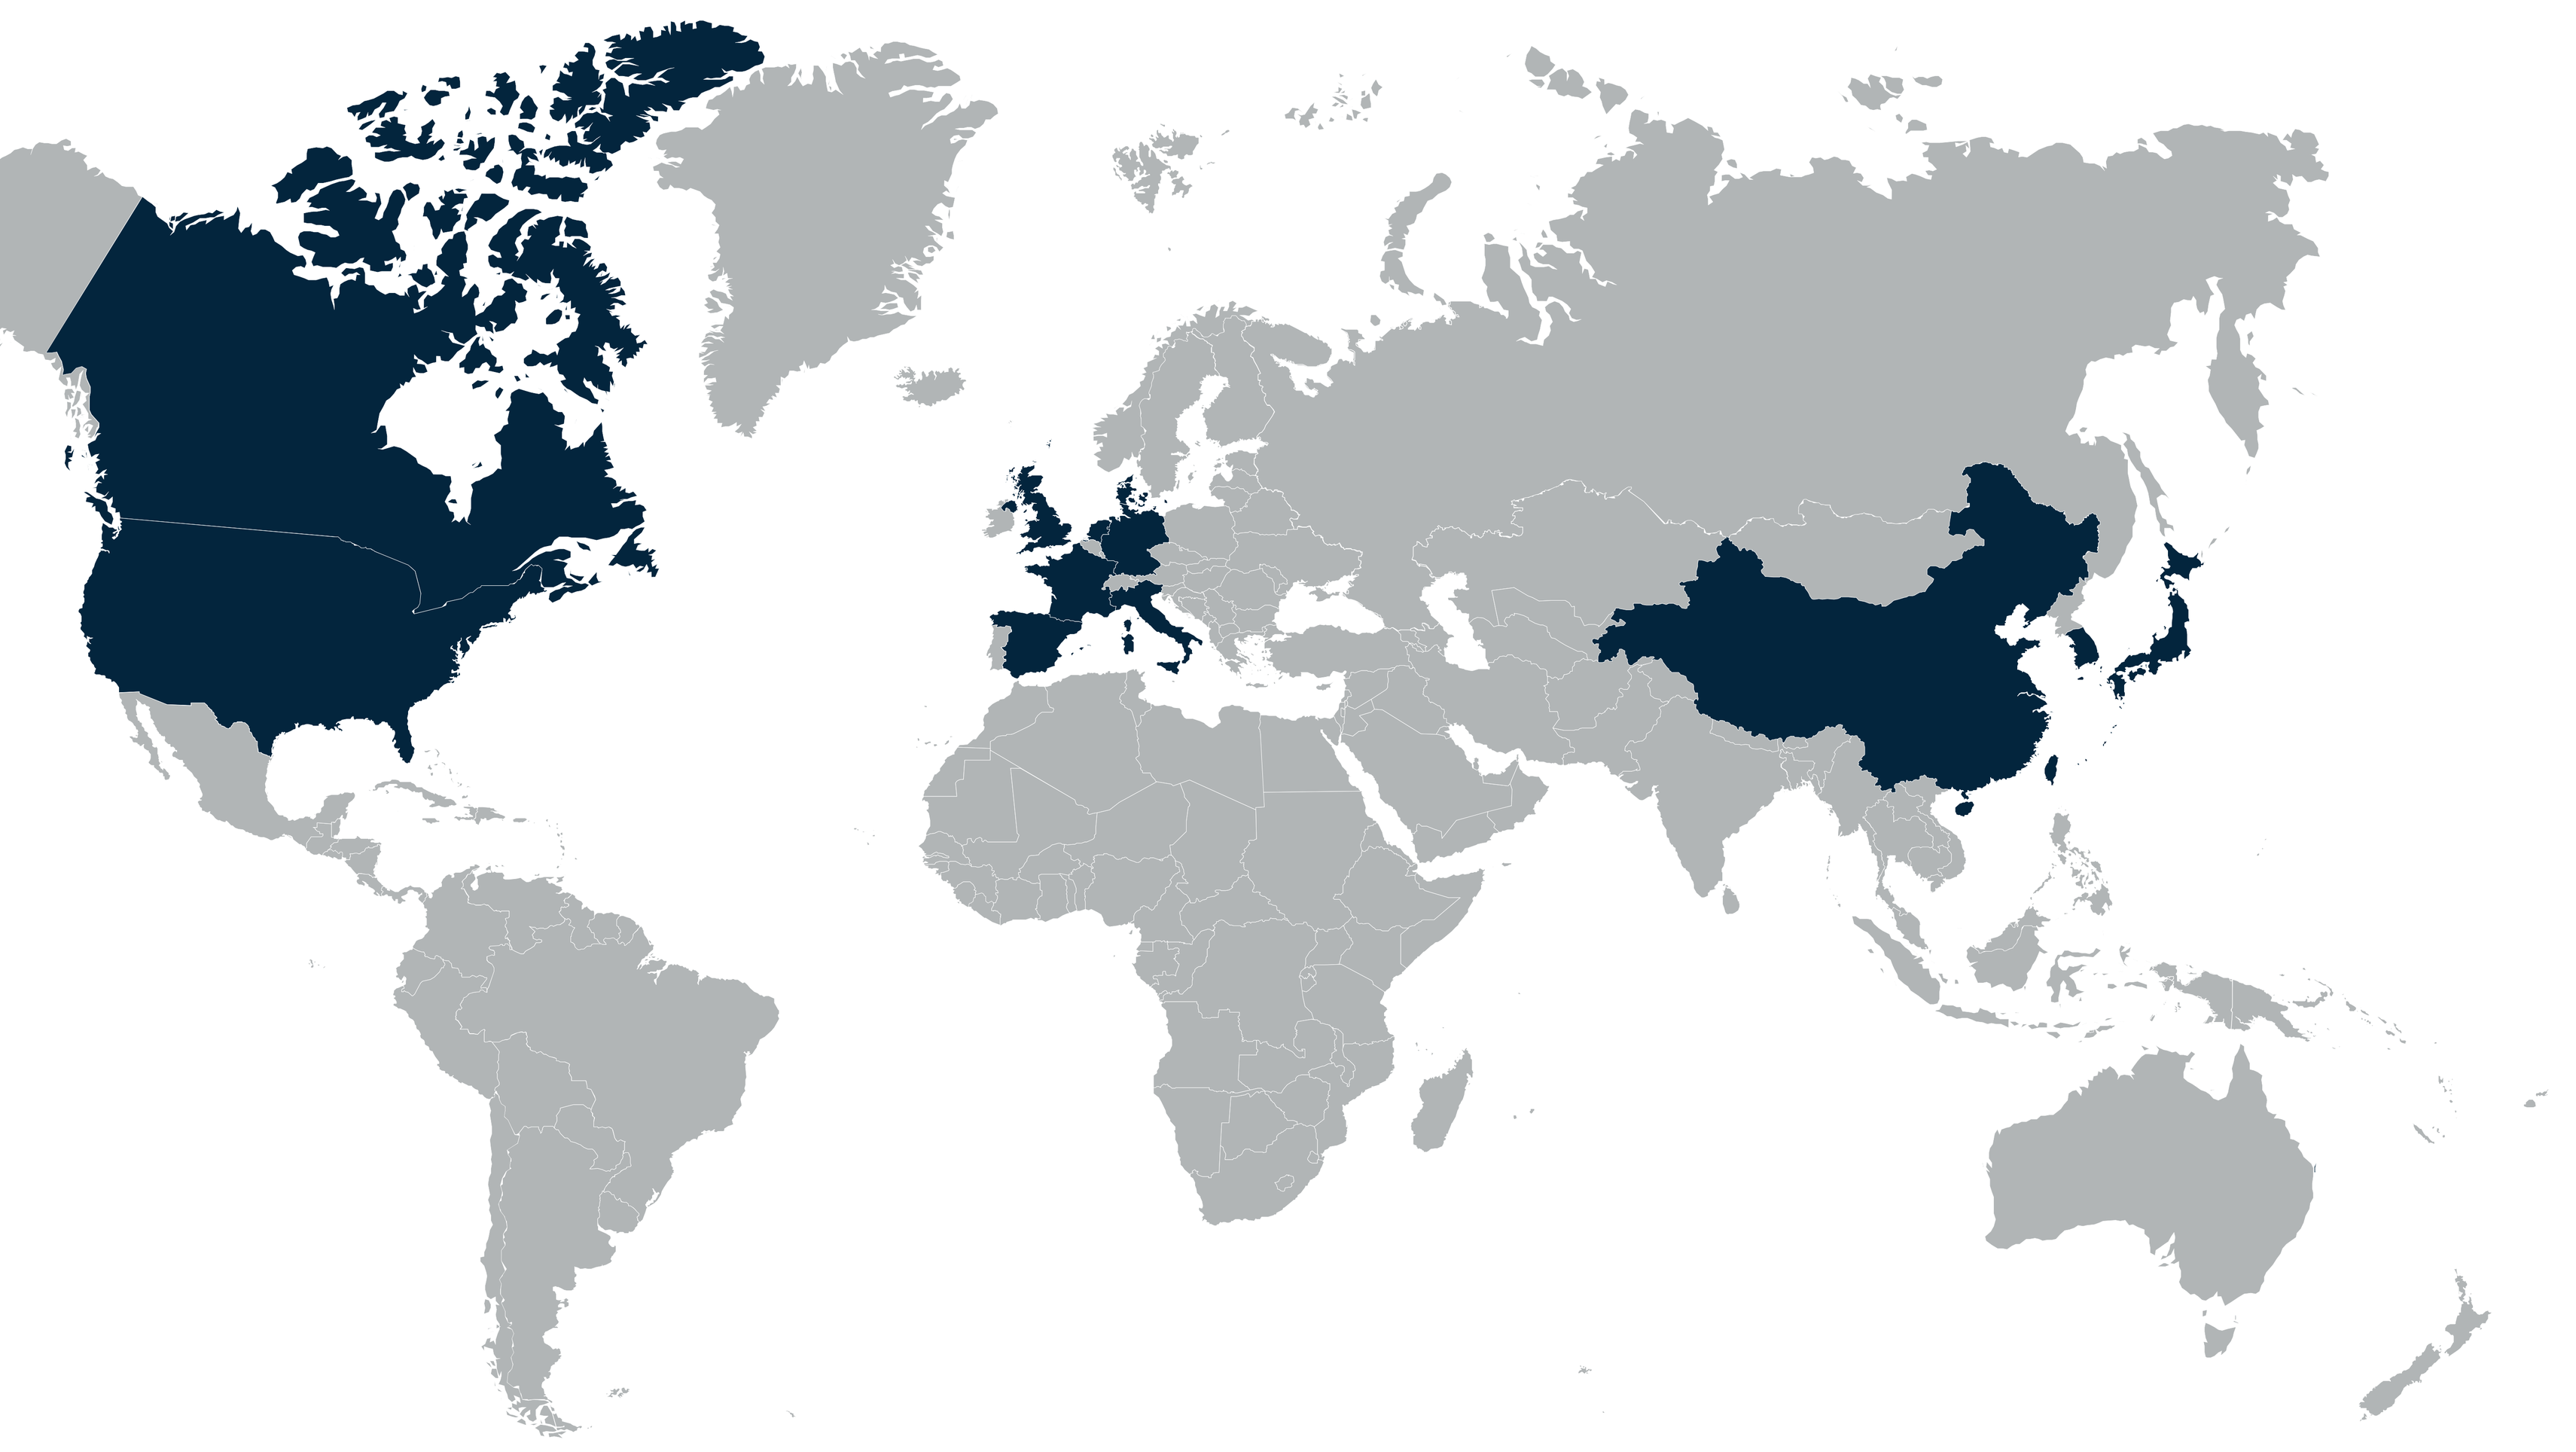

References

1. Zhang YL et al. *Oncotarget*. 2016;7:78985–78993.
2. Shi Y et al. *J Thorac Oncol*. 2014;9:154–162.
3. Leonetti A et al. *Br J Cancer*. 2019;121:725–737.
4. Niederst MJ et al. *Clin Cancer Res*. 2015;21:3924–3933.
5. Park S et al. *Cancer Res Treat*. 2020;52:1288–1290.
6. Schalm SS et al. *Ann Oncol*. 2020;31:S839.
7. Lim SM et al. *Cancer Res*. 2021;81:1467–1467.
8. Tavera L et al. Presented at AACR 2022. Poster #3328.

Acknowledgements

The authors thank the patients and their families for their participation in this study. Medical writing support was provided by Danielle Scheunemann, MS and Mhairi Foster, PhD, and editorial support was provided by Travis Taylor, BA, all of Paragon, Knutsford, UK, supported by Blueprint Medicines Corporation, Cambridge, MA, according to Good Publication Practice guidelines.

Disclosures

E Shum had a consulting or advisory role for AstraZeneca, Genentech, Janssen, Boehringer-Ingelheim; and had research funding to institution from Delfi Diagnostics. Y Elamin has consulted or had an advisory role for AstraZeneca, Takeda, Sanofi, Turning Point Therapeutics, Takeda, Lilly. Research Funding: AstraZeneca, Spectrum Pharmaceuticals, Takeda, Turning Point Therapeutics, Blueprint Medicines Corporation, Lilly, Forward Pharmaceuticals, Precision Oncology, Xcovery. Z Piotrowksa has had a consulting or advisory role for C4 Therapeutics, Blueprint Medicines Corporation, Jazz Pharmaceuticals, Janssen, Takeda, Cullinan, Daiichi Sankyo. Research funding: Novartis, Takeda, Spectrum, AstraZeneca, Cullinan, Daiichi Sankyo, AbbVie, GSK/Tesaro, Janssen, Blueprint Medicines Corporation. D Spigel has had a leadership role for ASCO Guideline Committee - Adjuvant Lung Cancer, and a consulting or advisory role (payments to institution) for Genetech/Roche, Novartis, Celgene, Bristol-Myers Squib, AstraZeneca, Pfizer, Boehringer Ingelheim, AbbVie, Foundation Medicine, GlaxoSmithKline, Lilly, Merck, Moderna Therapeutics, Nektar, Takeda, Amgen, TRM Oncology, Precision Oncology, Evelo Biosciences, Illumina, PharmaMar; and has also received research Funding (payments to institution) from Genetech/Roche, Novartis, Celgene, Bristol-Myers Squib, AstraZeneca, Pfizer, Boehringer Ingelheim, AbbVie, Foundation Medicine, GlaxoSmithKline, Lilly, University of Texas-Southwestern, G1 Therapeutics, Neon Therapeutics, Takeda, Nektar, Celldex, Clovis Oncology, Daiichi Sankyo, EMD Serono, Acerta Pharma, OncoGenex, Astellas Pharma, GRAIL, Transgene, Aeglea Biotherapeutics, Tesaro, Ipsen, ARMO Biosciences (Lilly), Amgen, Millennium; travel and accommodations expenses from AstraZeneca, Boehringer Ingelheim, Celgene, Lilly, EMD Serono, Bristol-Myers Squib, Genetech, Genzyme, Intuitive Surgical, Merck, Pfizer, Purdue Pharma, Spectrum Pharmaceuticals, and Sysmex. K L Reckamp has received consulting honoraria to self from Amgen, Calithera, AstraZeneca, Blueprint Medicines Corporation, Boehringer Ingelheim, Daiichi Sankyo, EMD Soreno, Genentech, GlaxoSmithKline, Janssen, Lilly, Merck KGA, Mirati, Takeda, Tesaro and non-financial support from Seagen Inc.; institution research support from Calithera, Blueprint Medicines Corporation, Daiichi Sankyo, Genentech, Elevation Oncology and Janssen outside the submitted work. J Rotow had a Consulting role for AstraZeneca, AbbVie, Gritstone, Lilly, Takeda and Sanofi-Genzyme; received Honoraria from Jazz, Johnson and Johnson, Merck, Pfizer, Lilly and Regeneron. D SW Tan has received personal fees from Novartis, Bayer, Boehringer Ingelheim, AstraZeneca, Eli Lilly, LOXO Oncology, Merck, Pfizer, Roche, Takeda, and Merrimack, and grants from Novartis, Bayer, AstraZeneca, Pfizer, and GlaxoSmithKline. SM Lim has received personal fees from Yuhan, AstraZeneca, Boehringer Ingelheim, Lilly, Takeda, and J INTS BIO and research support to institution from AstraZeneca, Boehringer Ingelheim, GSK, Roche, Hengrui, BridgeBio Pharma, Inc., Oscotec, and Janssen. TM Kim has received honoraria or had an advisory role with AstraZeneca, Boryung, F. Hoffmann-La Roche Ltd/Genentech, Inc, Janssen, Novartis, Sanofi, Takeda, Yuhan; and has received research funding outside this work from AstraZeneca-Korea Health Industry Development Institute. C-C Lin has received personal fees from AbbVie, Bayer, BeiGene, Blueprint Medicines Corporation, Boehringer-Ingelheim, Bristol Myers Squibb, Daiichi Sankyo, Eli Lilly, Merck KGaA, Novartis, PharmaEngine, and Roche. T Kato has received honoraria or had an advisory or consulting role with AstraZeneca, Boehringer Ingelheim, Bristol-Myers Squibb, Chugai Pharma, Daiichi Sankyo, Lilly, Merck Serono, Merck Sharp & Dohme, Nippon Kayaku, Novartis, Ono Pharmaceutical, Pfizer, Taiho Pharmaceutical, Takeda; and has received research funding from AbbVie, Amgen, AstraZeneca, Bristol-Myers Squibb, Chugai Pharma, Lilly, Merck Serono, Merck Sharp & Dohme, Novartis, Ono Pharmaceutical, Pfizer, Regeneron, and Taiho Pharmaceutical. J Parepally, F Albayya, M Louie-Gao, T Weinig, and A Zalutskaya are current employees and shareholders of Blueprint Medicines Corporation. K Goto has received grants or contracts to institution from Amgen Astellas BioPharma, Amgen, Boehringer Ingelheim Japan, Bristol-Myers Squibb, Chugai Pharmaceuticals, Daiichi Sankyo, Eisai, Eli Lilly Japan, Ignyta, Janssen Pharmaceutical, Kissei Pharmaceutical, Kyowa Kirin, Loxo Oncology, Medical and Biological Laboratories, Merck Biopharma, Merus, MSD, NEC, Ono Pharmaceutical, Pfizer Japan, Sumitomo Dainippon Pharma, Spectrum Pharmaceuticals, Sysmex, Haihe Biopharma, Taiho Pharmaceuticals, Takeda, Turning Point Therapeutics, and Life Technologies Japan; has had advisory board or data safety monitoring board participation for Takeda, Amgen, Bayer US, Eli Lilly Japan, Medpace Japan, and Janssen Pharmaceuticals, and honoraria from Amgen Astellas BioPharma, Amgen, Amoy Diagnostics, AstraZeneca, Boehringer Ingelheim Japan, Bristol-Myers Squibb, Bayer Yakuhin, Chugai Pharmaceuticals, Daiichi Sankyo, Eisai, Eli Lilly Japan, Guardant Health, Janssen Pharmaceutical, Kyowa Kirin, Life Technologies Japan, MSD, Novartis, Ono Pharmaceutical, Otsuka Pharmaceutical, Pfizer Japan, Taiho Pharmaceutical, and Takeda.

Poster available for download at:

Presented at **ASCO Annual Meeting, June 3–7, 2022**. Please contact medinfo@blueprintmedicines.com for permission to reprint and/or distribute.